E-131

In Vitro Activity of ACHN-490 against Recent Respiratory Tract Pathogens

R. BADAL[1], S. LOB[1], M. HACKEL[1], S. BOUCHILLON[1], D. HOBAN[1], E. ARMSTRONG[2]

[1]International Health Management Associates, Inc., Schaumburg, IL USA
[2]Achaogen Inc., South San Francisco, CA USA

IHMA, Inc.
2122 Palmer Dr.
Schaumburg, IL 60173
Tel: 847.303.5003
Fax: 847.303.5601

Revised Abstract

Introduction: Plazomicin (ACHN-490), a broad-spectrum aminoglycoside with differentiated activity against multi-drug resistant *Enterobacteriaceae*, including carbapenemase-producing strains, is being developed as a first-line treatment for resistant bacterial pathogens. This study evaluated the *in vitro* efficacy of plazomicin and several comparators versus respiratory tract infection (RTI) pathogens from the US and 5 other countries with relatively high antimicrobial resistance rates. **Methods:** 1,026 recent (2007-2009) RTI isolates (US 572, Argentina 216, Brazil 34, Greece 101, India 46, Turkey 57) of 7 species (*Acinetobacter baumannii, Enterobacter aerogenes, E. cloacae, Escherichia coli, Klebsiella pneumoniae, Pseudomonas aeruginosa, Staphylococcus aureus*) from hospitalized patients were randomly selected to approximate current resistance rates in each region. Activity was determined for plazomicin (Plz) , amikacin (Ak), aztreonam (Azt), cefepime (Cpe), ciprofloxacin (Cp), daptomycin (Dap), gentamicin (Gm), imipenem (Imp), piperacillin-tazobactam (PT), tigecycline (Tig), and tobramycin (Tob) using CLSI broth microdilution. **Results:** $MIC_{50/90}$ values (mcg/ml) for each species are shown in the following table.

Species	n	Plz	Ak	Azt	Cpe	Cp	Dap	Gm	Imp	PT	Tig	Tob
		$MIC_{50/90}$										
A. baumannii	201	4/64	32/>128	>16/>16	>16/>16	>2/>2	na	32/>32	>8/>8	>64/>64	2/4	4/>32
E. aerogenes	37	0.5/1	1/2	≤1/>16	≤0.5/1	≤0.12/>2	na	≤0.25/0.5	1/1	4/>64	1/2	0.5/1
E. cloacae	64	≤0.25/0.5	1/4	≤1/>16	≤0.5/>16	≤0.12/>2	na	≤0.25/8	0.5/1	4/>64	1/2	0.5/16
E. coli	214	1/2	4/8	≤1/16	≤0.5/>16	0.25/>2	na	1/>32	≤0.25/0.5	4/>64	0.5/1	1/16
K. pneumoniae	210	≤0.25/0.5	1/16	≤1/>16	≤0.5/>16	≤0.12/>2	na	≤0.25/>32	≤0.25/1	8/>64	2/4	0.5/32
P. aeruginosa	200	8/16	4/64	8/>16	4/>16	0.25/>2	na	2/>32	2/>8	16/>64	>4/>4	0.5/>32
S. aureus, MRSA	50	1/2	16/128	na	>16/>16	>2/>2	1/1	0.5/>32	>8/>8	>64/>64	0.25/1	>32/>32
S. aureus, MSSA	50	1/2	4/8	na	4/4	0.25/0.5	1/1	0.5/1	≤0.25/≤0.25	2/2	0.25/0.5	0.5/16
		na=not applicable										

Conclusions: Plazomicin $MIC_{50/90}$ values were ± one log_2 dilution of each other for all organisms except *A. baumannii*, and were comparable to Imp and Tig. Plazomicin MIC_{90} values for all species tested were among the lowest values of all study drugs, particularly against *Enterobacteriaceae* (including ESBL-producers) and MRSA, of which 99% were inhibited by ≤2 mcg/ml of plazomicin. Plazomicin is a promising investigational antimicrobial compound with excellent *in vitro* activity against many RTI pathogens, including those resistant to many presently available drugs.

Introduction

As antimicrobial resistance—often manifested as multi-drug resistance (MDR)—continues to increase in many bacterial species worldwide, the need for new drugs with clinical utility against MDR bacterial pathogens is critical. Extended-spectrum beta-lactamase (ESBL) producing *Enterobacteriaceae*, especially *Escherichia coli* and *Klebsiella pneumoniae*, have become prevalent around the globe. MDR *Acinetobacter baumannii* and *Pseudomonas aeruginosa* have become especially worrisome, because for some strains, there are very few, and in some cases no effective drugs remaining. A much older problem is that of methicillin-resistant *Staphylococcus aureus* (MRSA), though recent approvals of several new antimicrobials with activity against gram-positive pathogens have reduced this concern.

Plazomicin (ACHN-490), a broad-spectrum aminoglycoside that retains activity in the presence of nearly all aminoglycoside-modifying enzymes, is being developed as a first-line treatment for resistant bacterial pathogens. The compound has demonstrated *in vitro* activity against a variety of pathogens, including MRSA, and ESBL- and carbapenemase-producing gram-negative bacteria (except those also harboring 16S rRNA methylases) [1, 2, 3, 4]. This study evaluates the *in vitro* activity of plazomicin and several comparators against pathogens isolated from patients with respiratory tract infection (RTI) from the United States and 5 other countries known to have relatively high antimicrobial resistance rates.

Materials & Methods

- 1,026 isolates collected during 2007, 2008, and 2009 from lower respiratory tract infections in hospitalized patients in 6 countries (United States 572, Argentina 216, Greece 101, Turkey 57, India 46, and Brazil 34) were evaluated. Species included *E. coli* (214, including 19 ESBL-positive), *K. pneumoniae* (210, including 54 ESBL-positive), *A. baumannii* (201), *P. aeruginosa* (200), *S. aureus* (50 MRSA, 50 MSSA), *Enterobacter cloacae* (64), and *E. aerogenes* (37). Other than the MRSA and MSSA, isolates were selected without regard for their antimicrobial susceptibility.
- Susceptibility of isolates to plazomicin, amikacin, aztreonam, cefepime, ciprofloxacin, daptomycin (*S. aureus* only), gentamicin, imipenem, piperacillin-tazobactam, tigecycline, and tobramycin was determined by broth microdilution following Clinical and Laboratory Standards Institute (CLSI) guidelines [5].
- Quality control was conducted following CLSI guidelines [6].
- $MIC_{50/90}$ values were determined for each species, by drug and by country.

References

1. Landman, D., Babu, E., *et al.* 2010. Activity of a novel aminoglycoside, ACHN-490, against clinical isolates of *Escherichia coli* and *Klebsiella pneumoniae* from New York City. J. Antimicrob. Chemother. 65:2123-2127.
2. Endimiani, A., Hujer, K., *et al.* 2009. ACHN-490, a Neoglycoside with Potent In Vitro Activity against Multidrug-Resistant *Klebsiella pneumoniae* Isolates. Antimicrob. Agents Chemother. 53:4504-4507.
3. Tenover, F., Tickler, I., Armstrong, E., *et al*. 2011. Activity of ACHN-490 against methicillin-resistant Staphylococcus aureus (MRSA) isolates from patients in US hospitals. Int. J. Antimicrob. Agents. (Epub ahead of print).
4. Livermore, D.M., Mushtaq, M.W., *et al*. 2010. Activity of aminoglycosides, including ACHN-490, against carbapenem-resistant Enterobacteriaceae isolates. J. Antimicrob. Chemother. 66:48-53.
5. Clinical Laboratory Standards Institute. 2009. Methods for Dilution Antimicrobial Susceptibility Tests for Bacteria That Grow Aerobically; Approved Standards -- Eighth Edition. CLSI document M07-A8 Wayne, PA.
6. Clinical and Laboratory Standards Institute. 2011. Performance Standards for Antimicrobial Susceptibility Testing; Twenty-First Informational Supplement. CLSI Document M100-S21. Wayne, PA.

Acknowledgments

This material is based upon work supported by the National Institute of Allergy and Infectious Diseases. Any opinions, findings and conclusions or recommendations expressed in this material are those of the authors and do not necessarily reflect the views of the National Institute of Allergy and Infectious Diseases.

This study was supported in part by Achaogen, Inc.

Results

Table 1. Comparison of $MIC_{50/90}$ of all study drugs, by species. Gray shading denotes no CLSI breakpoints defined; no shading denotes value in susceptible category; yellow shading denotes value in intermediate category; red shading denotes value in resistant category.

Species (n)	Plz		Ak		Azt		Cpe		Cp		Dap		Gm		Imp		PT		Tig		Tob	
	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}
A. baumannii (201)	4	64	32	256	>16	>16	>16	>16	>2	>2	na	na	32	>32	>8	>8	>64	>64	2	4	4	>32
E. aerogenes (37)	0.5	1	1	2	≤1	>16	≤0.5	1	≤0.12	>2	na	na	≤0.25	0.5	1	1	4	>64	1	2	0.5	1
E. cloacae (64)	0.25	0.5	1	4	≤1	>16	≤0.5	>16	≤0.12	>2	na	na	≤0.25	8	0.5	1	4	>64	1	2	0.5	16
E. coli (214)	1	2	4	8	≤1	16	≤0.5	>16	0.25	>2	na	na	1	>32	≤0.25	0.5	4	>64	0.5	1	1	16
K. pneumoniae (210)	0.25	0.5	1	16	≤1	>16	≤0.5	>16	≤0.12	>2	na	na	≤0.25	>32	≤0.25	1	8	>64	2	4	0.5	32
P. aeruginosa (200)	8	16	4	64	8	>16	4	>16	0.25	>2	na	na	2	>32	2	>8	16	>64	>4	>4	0.5	>32
S. aureus, MRSA (50)	1	2	16	128	na	na	>16	>16	>2	>2	1	1	0.5	>32	>8	>8	>64	>64	0.25	1	>32	>32
S. aureus, MSSA (50)	1	2	4	8	na	na	4	4	0.25	0.5	1	1	0.5	1	≤0.25	≤0.25	2	2	0.25	0.5	0.5	16

Figure 1.

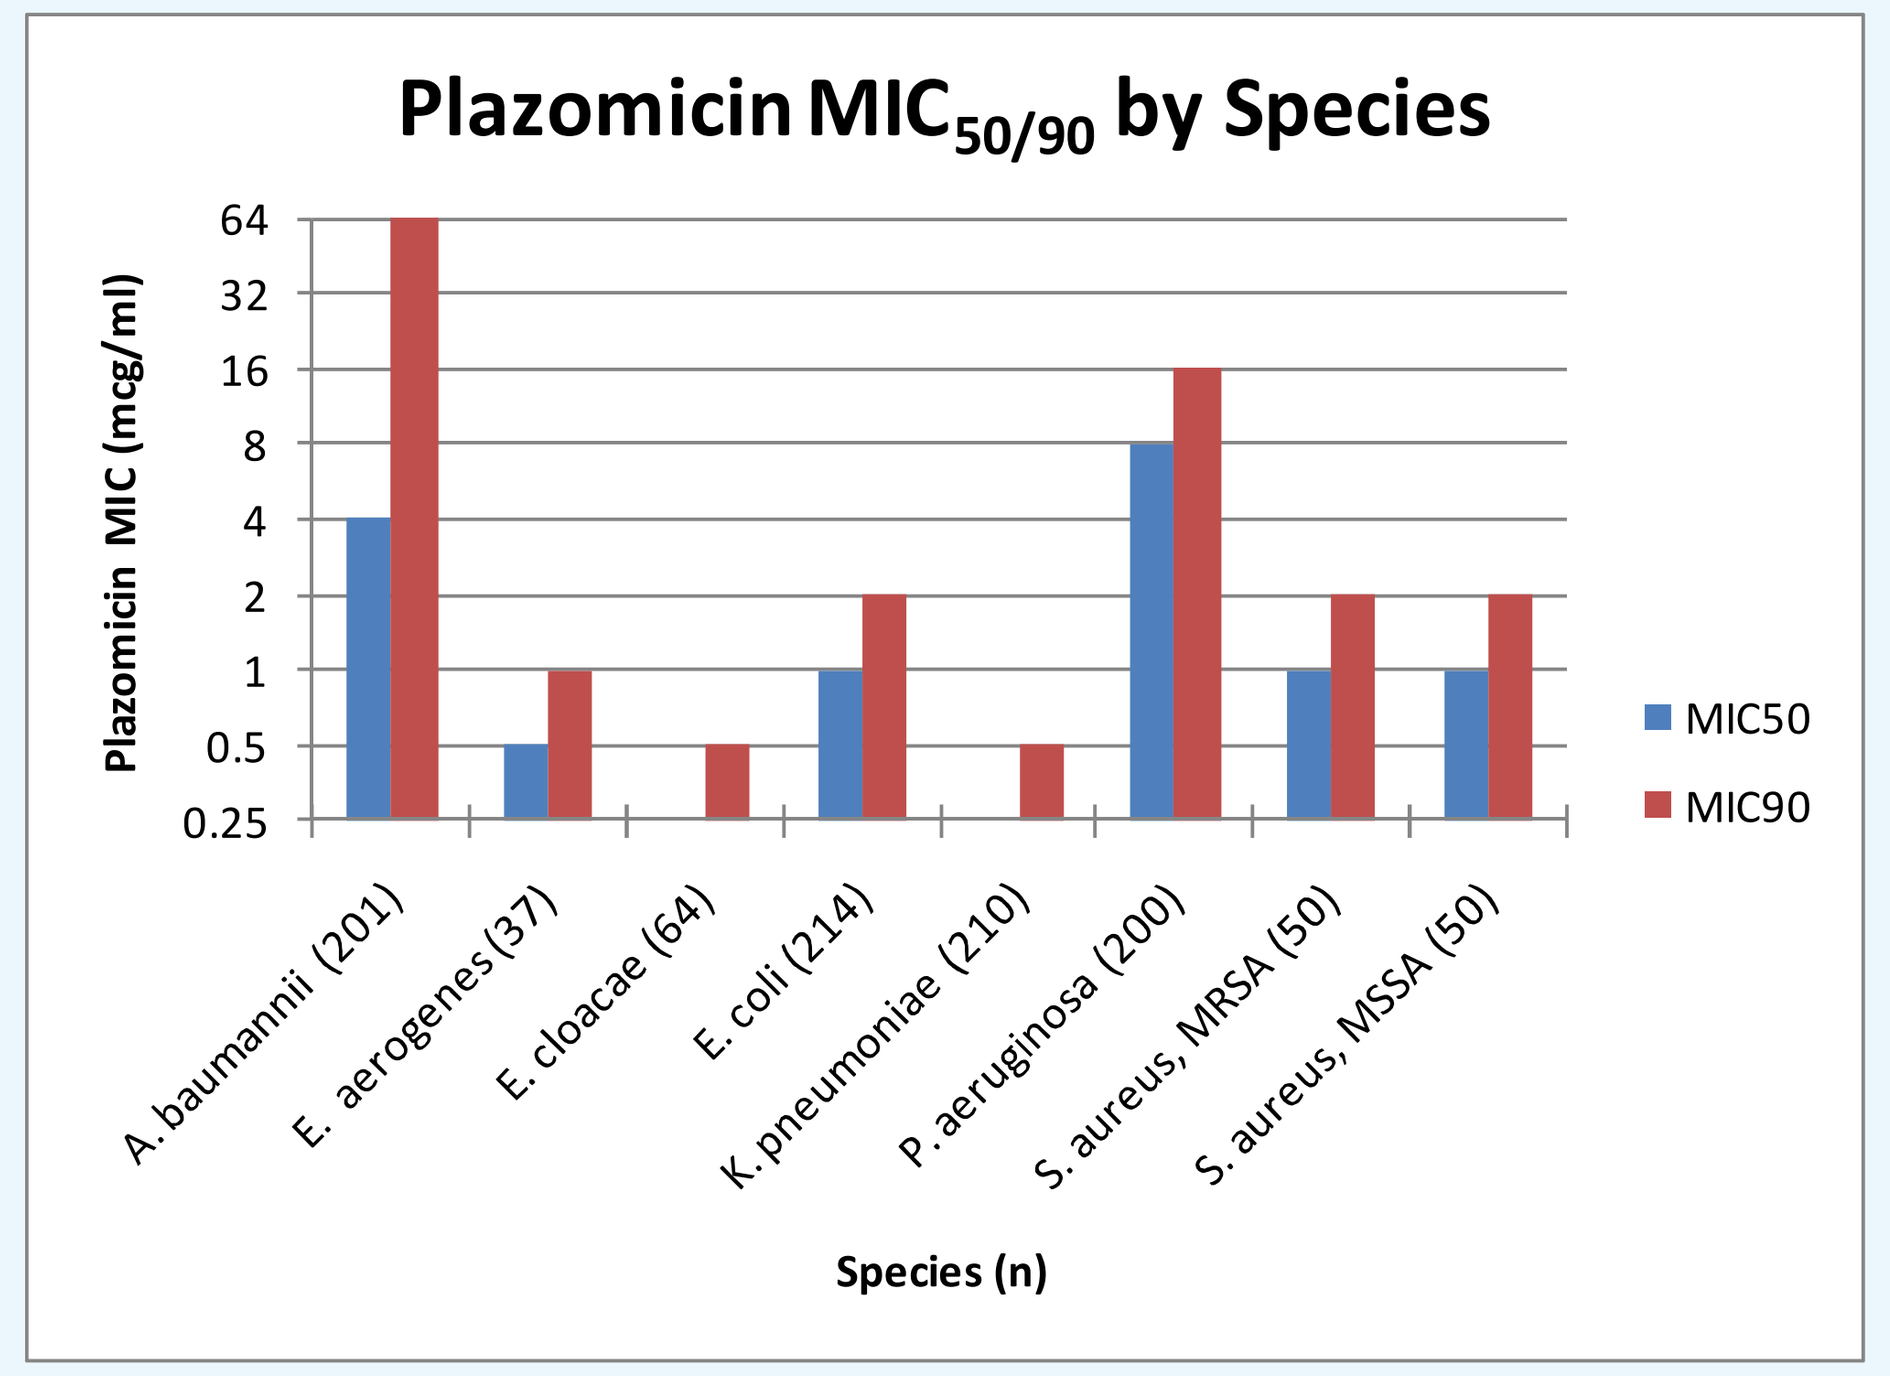
Plazomicin MIC50/90 by Species
Plazomicin MIC (mcg/ml)
64
32
16
8
4
2
1
0.5
0.25
A. baumannii (201)
E. aerogenes (37)
E. cloacae (64)
E. coli (214)
K. pneumoniae (210)
P. aeruginosa (200)
S. aureus, MRSA (50)
S. aureus, MSSA (50)
Species (n)
MIC50
MIC90

Figure 2.

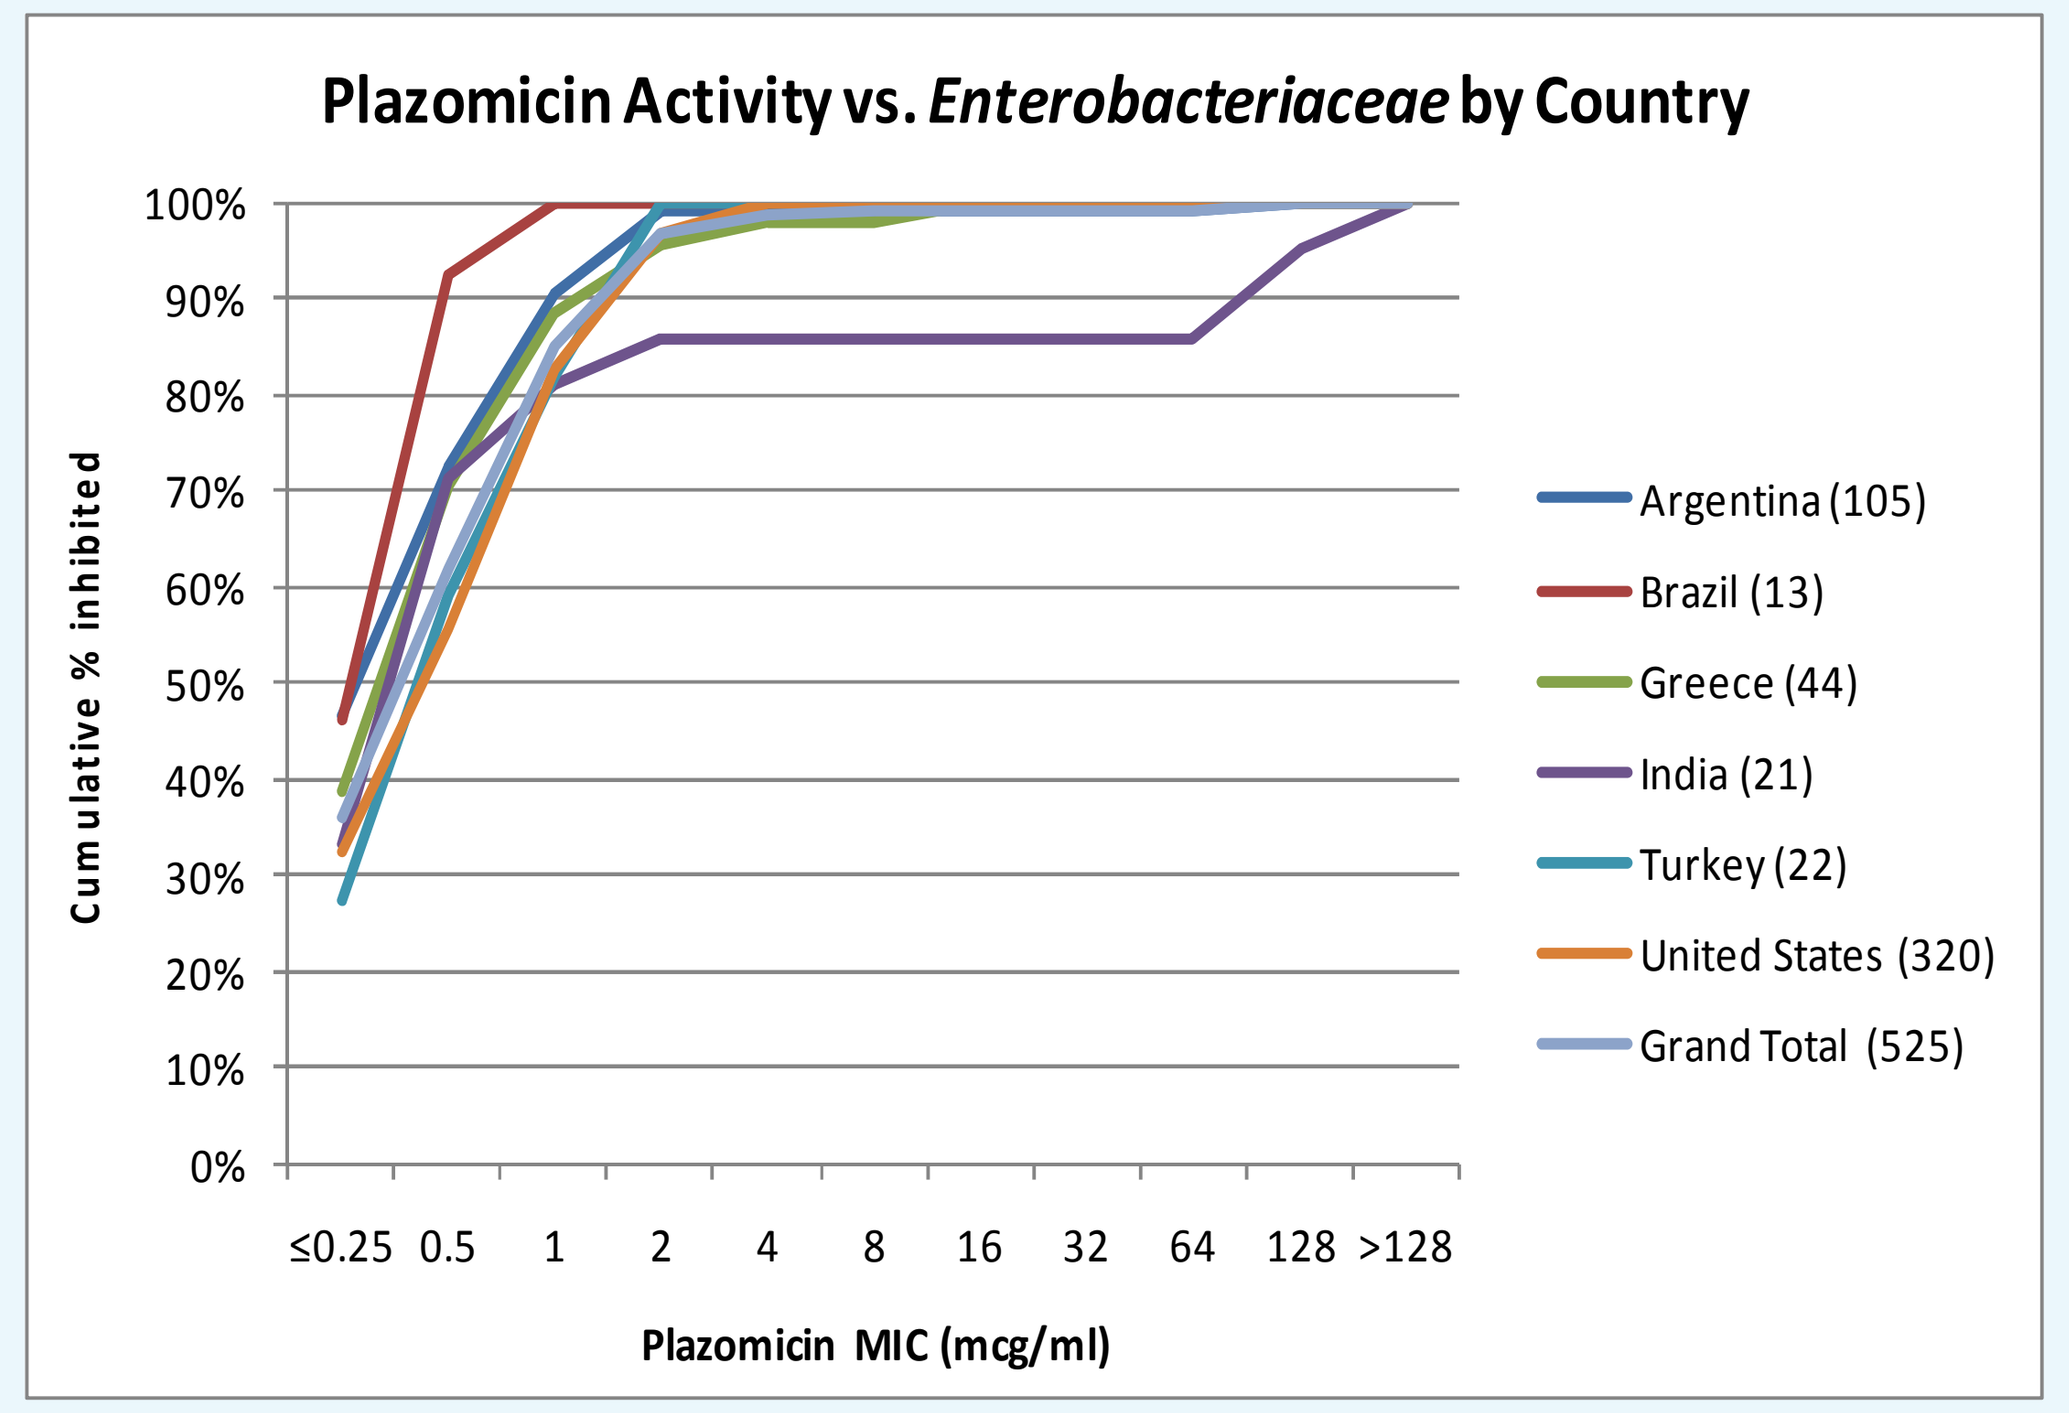
Plazomicin Activity vs. Enterobacteriaceae by Country
Cumulative % inhibited
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
≤0.25 0.5 1 2 4 8 16 32 64 128 >128
Plazomicin MIC (mcg/ml)
Argentina (105)
Brazil (13)
Greece (44)
India (21)
Turkey (22)
United States (320)
Grand Total (525)

Figure 3.

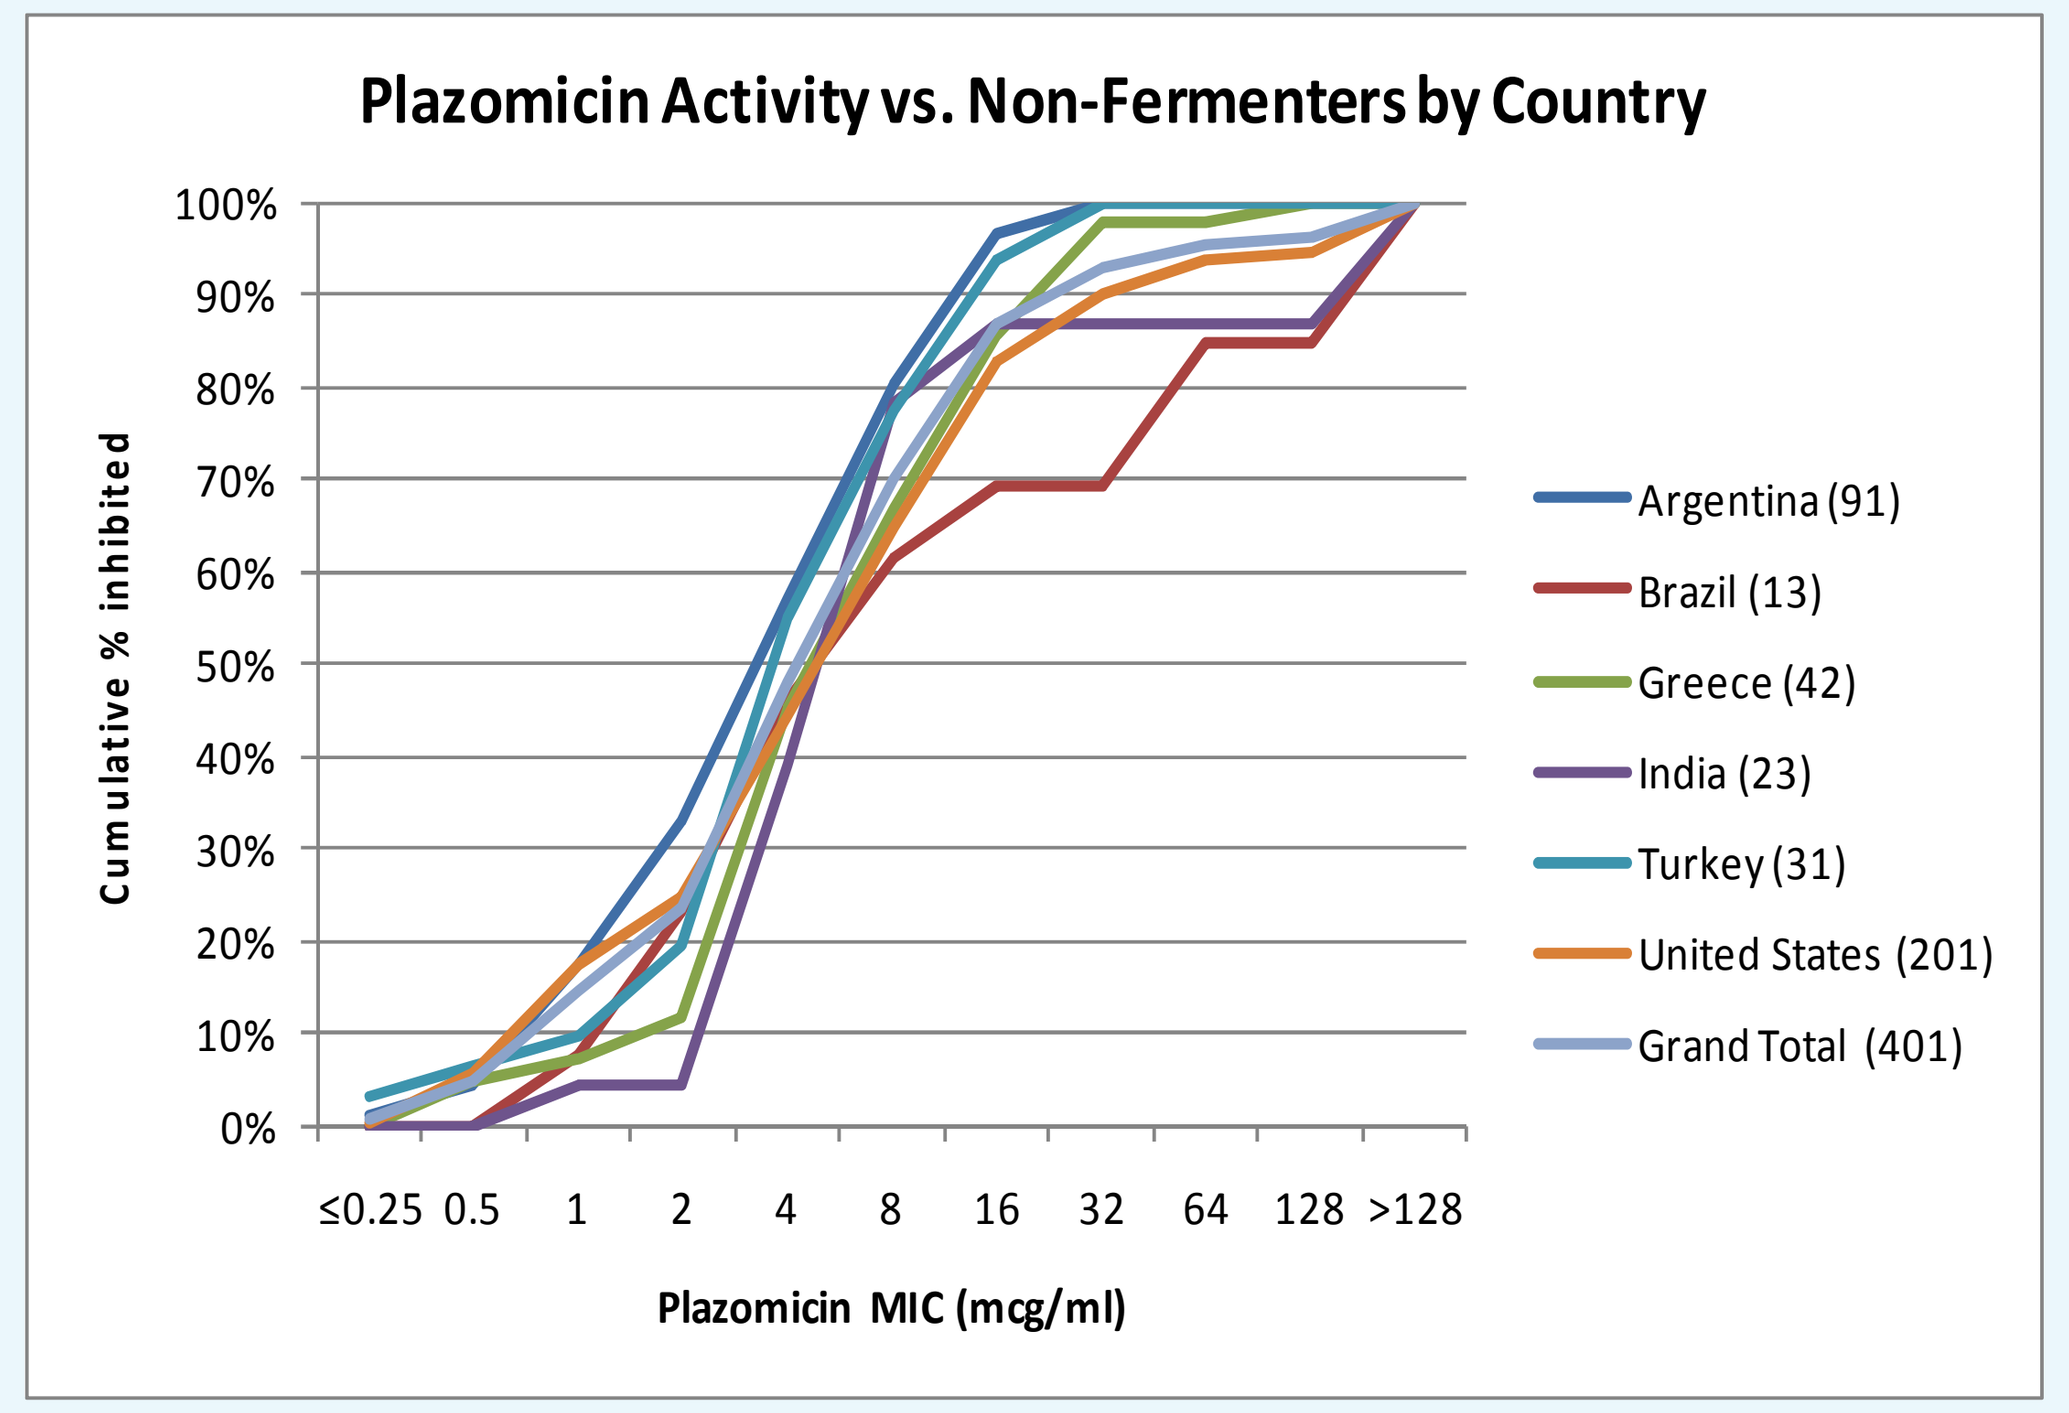
Plazomicin Activity vs. Non-Fermenters by Country
Cumulative % inhibited
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
≤0.25 0.5 1 2 4 8 16 32 64 128 >128
Plazomicin MIC (mcg/ml)
Argentina (91)
Brazil (13)
Greece (42)
India (23)
Turkey (31)
United States (201)
Grand Total (401)

Figure 4.

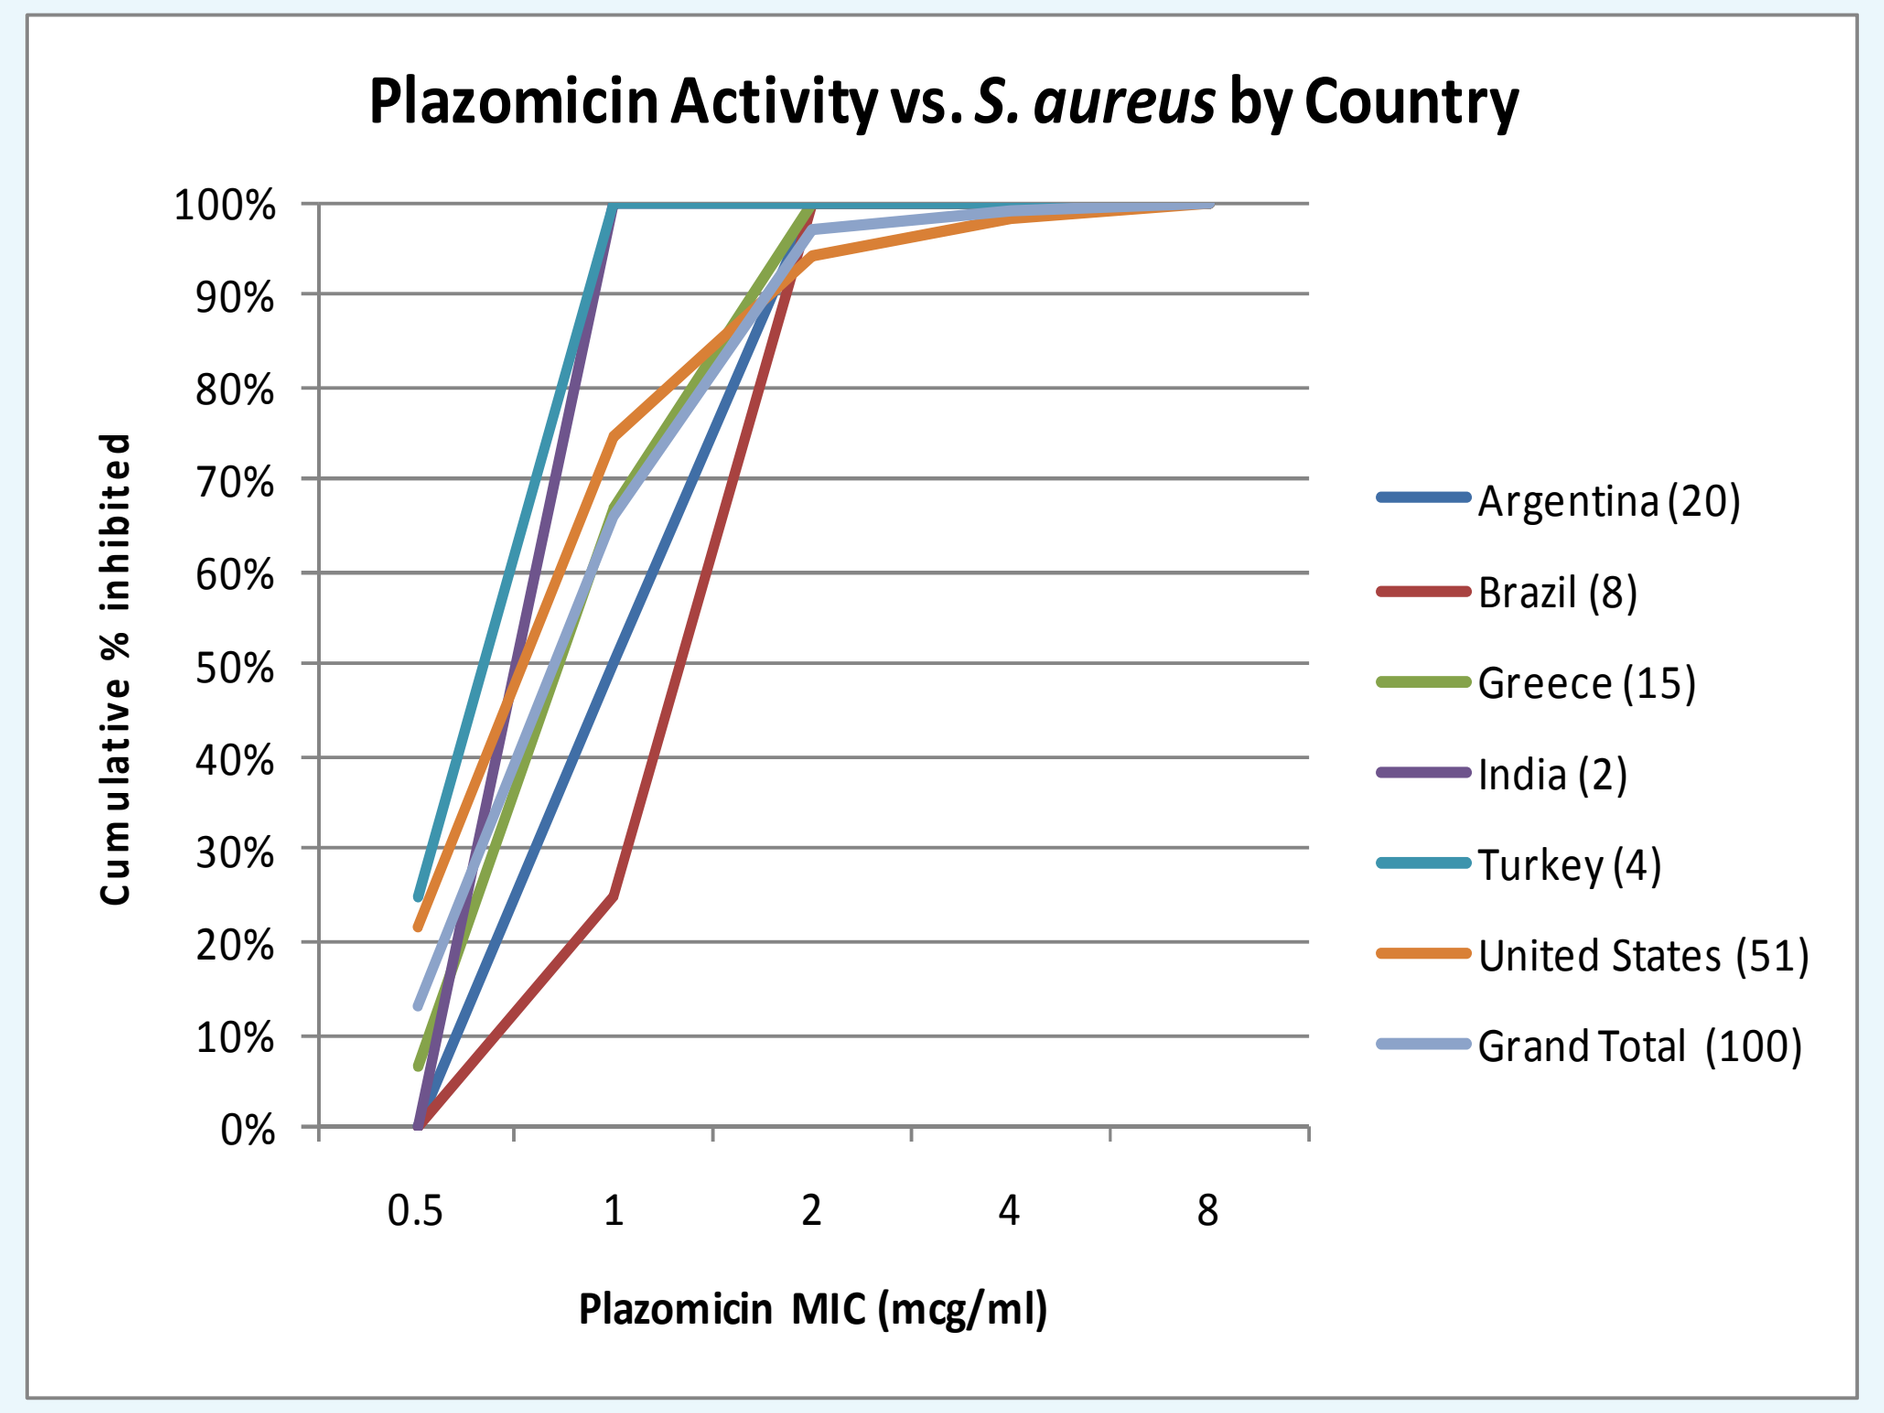
Plazomicin Activity vs. S. aureus by Country
Cumulative % inhibited
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
0.5 1 2 4 8
Plazomicin MIC (mcg/ml)
Argentina (20)
Brazil (8)
Greece (15)
India (2)
Turkey (4)
United States (51)
Grand Total (100)

Conclusions

- With the exception of *A. baumannii*, plazomicin $MIC_{50/90}$ values were within one log_2 dilution of each other for all organism groups, making its activity similar to that of tigecycline and imipenem in that respect.
- The MIC_{50}/MIC_{90} of plazomicin (no breakpoint yet determined) against *P. aeruginosa* was 8/16 mcg/ml; the MIC_{90} of each comparator drug was in the resistant range.
- With >90% of *Enterobacteriaceae* (including 73 ESBL-producers) and *S. aureus* (half of which were MRSA) inhibited by ≤2 mcg/ml, plazomicin demonstrated *in vitro* broad-spectrum activity against respiratory tract isolates that warrants consideration for development for the treatment of serious respiratory tract infections.