#E-821

Susceptibility Rates of Anaerobic Bacteria in Six European Nations in 2007-2008

R. Badal[1], S. Bouchillon[1], M. Renteria[1], M. Hackel[1], J. Johnson[1], D. Hoban[1], B. Johnson[1], S. Hawser[2], M. Dowzicky[3]

[1]International Health Management Assoc., Inc., Schaumburg, IL, USA
[2]IHMA Europe Sàrl, Epalinges, Switzerland
[3]Wyeth Pharmaceuticals, Collegeville, PA, USA

IHMA, Inc.
2122 Palmer Dr.
Schaumburg, IL
60173
Tel: 847.303.5003
www.ihmainc.com

Revised Abstract

Background: The Tigecycline European Surveillance Trial (TEST) monitors susceptibility of European aerobic and anaerobic bacteria to tigecycline (Tig), cefoxitin (Cfx), clindamycin (Cd), meropenem (Mer), piperacillin-tazobactam (PT), metronidazole (Met), and penicillin (P). This report summarizes susceptibility of gram-positive and –negative anaerobes in 6 countries to these drugs.

Methods: Anaerobic isolates collected by 29 cumulative labs in 6 countries in 2007-2008 were sent to a central lab in the US for agar dilution susceptibility testing, following CLSI guidelines. Results were interpreted by EUCAST guidelines; CLSI was used if no EUCAST breakpoints exist for a given drug.

Results: % Susceptible of each organism group in each country is summarized in the table below; lowest value for each drug/organism combination is highlighted.

Country	Organism	N	Cfx	Cd	Mer	Met	P-T	Tig
Belgium	Bacteroides spp.	45	77.78	73.33	95.56	100	91.11	95.56
Czech Republic	Bacteroides spp.	53	94.34	75.47	92.45	100	100	100
France	Bacteroides spp.	185	82.16	72.43	97.84	100	94.05	97.3
Germany	Bacteroides spp.	318	82.08	78.62	96.86	99.06	91.19	96.54
Hungary	Bacteroides spp.	171	87.72	82.46	97.66	99.42	91.23	98.83
United Kingdom	Bacteroides spp.	10	100	90	100	100	100	100

Country	Organism	N	Cfx	Cd	Mer	Met	P-T	Tig
Belgium	Prevotella spp.	26	100	65.38	100	96.15	100	100
Czech Republic	Prevotella spp.	38	100	84.21	100	100	100	100
France	Prevotella spp.	72	100	75	98.61	97.22	100	100
Germany	Prevotella spp.	166	98.19	86.75	100	97.59	98.19	99.4
Hungary	Prevotella spp.	28	92.86	60.71	100	100	96.43	96.43
United Kingdom	Prevotella spp.	3	100	100	100	100	100	100

Country	Organism	N	Cd	Mer	Met	P	P-T	Tig
Belgium	Clostridium spp.	32	84.38	100	100	50	90.63	100
Czech Republic	Clostridium spp.	40	60	97.5	100	77.5	90	100
France	Clostridium spp.	172	75	100	100	36.63	92.44	100
Germany	Clostridium spp.	226	63.27	98.67	99.56	31.86	96.02	98.67
Hungary	Clostridium spp.	53	64.15	100	100	71.7	92.45	100
United Kingdom	Clostridium spp.	3	66.67	100	100	66.67	100	100

Country	Organism	N	Cd	Mer	Met	P	P-T	Tig
Belgium	Peptostreptococcus spp.	16	100	100	100	75	100	100
Czech Republic	Peptostreptococcus spp.	16	93.75	100	93.75	100	100	100
France	Peptostreptococcus spp.	52	98.08	100	92.31	86.54	100	100
Germany	Peptostreptococcus spp.	86	98.84	98.84	97.67	82.56	98.84	100
Hungary	Peptostreptococcus spp.	18	100	100	100	94.44	100	100

Conclusions:
- Tig, P-T, Mer, and Met all inhibited >90% of isolates in all countries.
- Cfx, Cd, and P showed wide ranges in susceptibilities between countries and organism groups.
- No one country stood out as having lower or higher overall susceptibility levels.

Introduction

Susceptibility testing of anaerobic bacteria is rarely performed routinely by clinical microbiology laboratories, and antimicrobial therapy of infections presumed to be due to anaerobic pathogens is usually empiric. Data from current surveillance studies can be used to understand susceptibility levels of pathogens to commonly-prescribed drugs, but due to differences in patterns of drug usage and susceptibility in geographically diverse locations, it is important to collect such data from as broad a base as practical to provide more meaningful information to clinicians and microbiologists.

The Tigecycline European Surveillance Trial (TEST) began monitoring the susceptibility of anaerobes to tigecycline and six other drugs in Europe in 2007. This report summarizes the antibiograms of the gram-negative and –positive isolates collected and tested from 2007 through 2008, and presents the results by country to determine if there are regional differences in anaerobic susceptibility to these particular antimicrobial agents within Europe.

Materials & Methods

- All isolates were derived from blood, wounds, fluids, gastrointestinal, and complicated intra-abdominal infections. Isolates were identified to genus and species by the local laboratory. Only one isolate per patient was accepted.
- 1,879 clinical isolates were collected in 2007 to 2008 from 29 cumulative laboratories in six European countries (Belgium, Czech Republic, France, Germany, United Kingdom, and Hungary).
- All isolates were sent to a central laboratory in the USA (Laboratories International for Microbiology Studies, a subsidiary of International Health Management Associates, Inc.) for confirmation of identification and susceptibility testing.
- Minimum inhibitory concentrations (MICs) were determined by agar dilution as specified by the Clinical and Laboratory Standards Institute (CLSI, formerly NCCLS) [1]. The following antimicrobial agents were tested with their dilution ranges (expressed in mcg/ml): tigecycline (0.06-32); clindamycin (0.25-8); metronidazole (0.12-16); piperacillin/tazobactam (0.06/4-64/4); meropenem (0.06-8); penicillin (0.25-32, gram positive only); and cefoxitin (2-32, gram negative only).
- MIC interpretive criteria followed published breakpoints established by the European Committee on Antimicrobial Susceptibility Testing (EUCAST) where applicable [2]. Since no EUCAST guidelines were available for a cefoxitin, CLSI breakpoints [1] were used. FDA anaerobe breakpoints were used for tigecycline [3].
- Quality control testing was done following CLSI guidelines [1].

References

1. Clinical and Laboratory Standards Institute, Methods for Antimicrobial Susceptibility Tests of Anaerobic Bacteria; Approved Standard—Sixth Edition 2004: Clinical Laboratory Standards Institute (CLSI), 940 West Valley Road, Suite 1400, Wayne, Pennsylvania 19087-1898 USA.
2. European Committee on Antimicrobial Susceptibility Testing (EUCAST) website, http://www.eucast.org, 2008-06-27(version 1.2).
3. Tygacil®, 2005. Food and Drug Administration product information. Wyeth Pharmaceuticals, Inc., Philadelphia, PA, USA.

Acknowledgements

We gratefully acknowledge the contributions of the investigators in Europe who provided the anaerobic isolates for the TEST study: A. Rodloff (Germany), M. Marschall (Germany), B. Grabein (Germany), E. Nagy (Hungary), C-J. Soussy (France), R. Mutters (Germany), H. Seifert (Germany), H. Marchandin (France), C. Poyart (France), H. Bezirtzoglou (Greece), L. Dubreuil (France), J. Wagner (Germany), J. Szabo (Hungary), D. Pierard (Belgium), E. Chmelarova (Czech Republic), L. Szikra (Hungary), and Mandy Wootton (UK). This study was sponsored by Wyeth Pharmaceuticals.

Results

Figures 1-5 show susceptibility percentages of *Bacteroides* spp., *Clostridium* spp., *Peptoniphilus* spp., *Peptostreptococcus* spp., and *Prevotella* spp. to each of the six study drugs in each of the six countries, as well as combined Europe.
Drug abbreviations used are Cfx=cefoxitin, Met=metronidazole, Mer=meropenem, PT=piperacillin-tazobactam, Tig=tigecycline, Cd=clindamycin, and Pen=penicillin.

Figure 1.

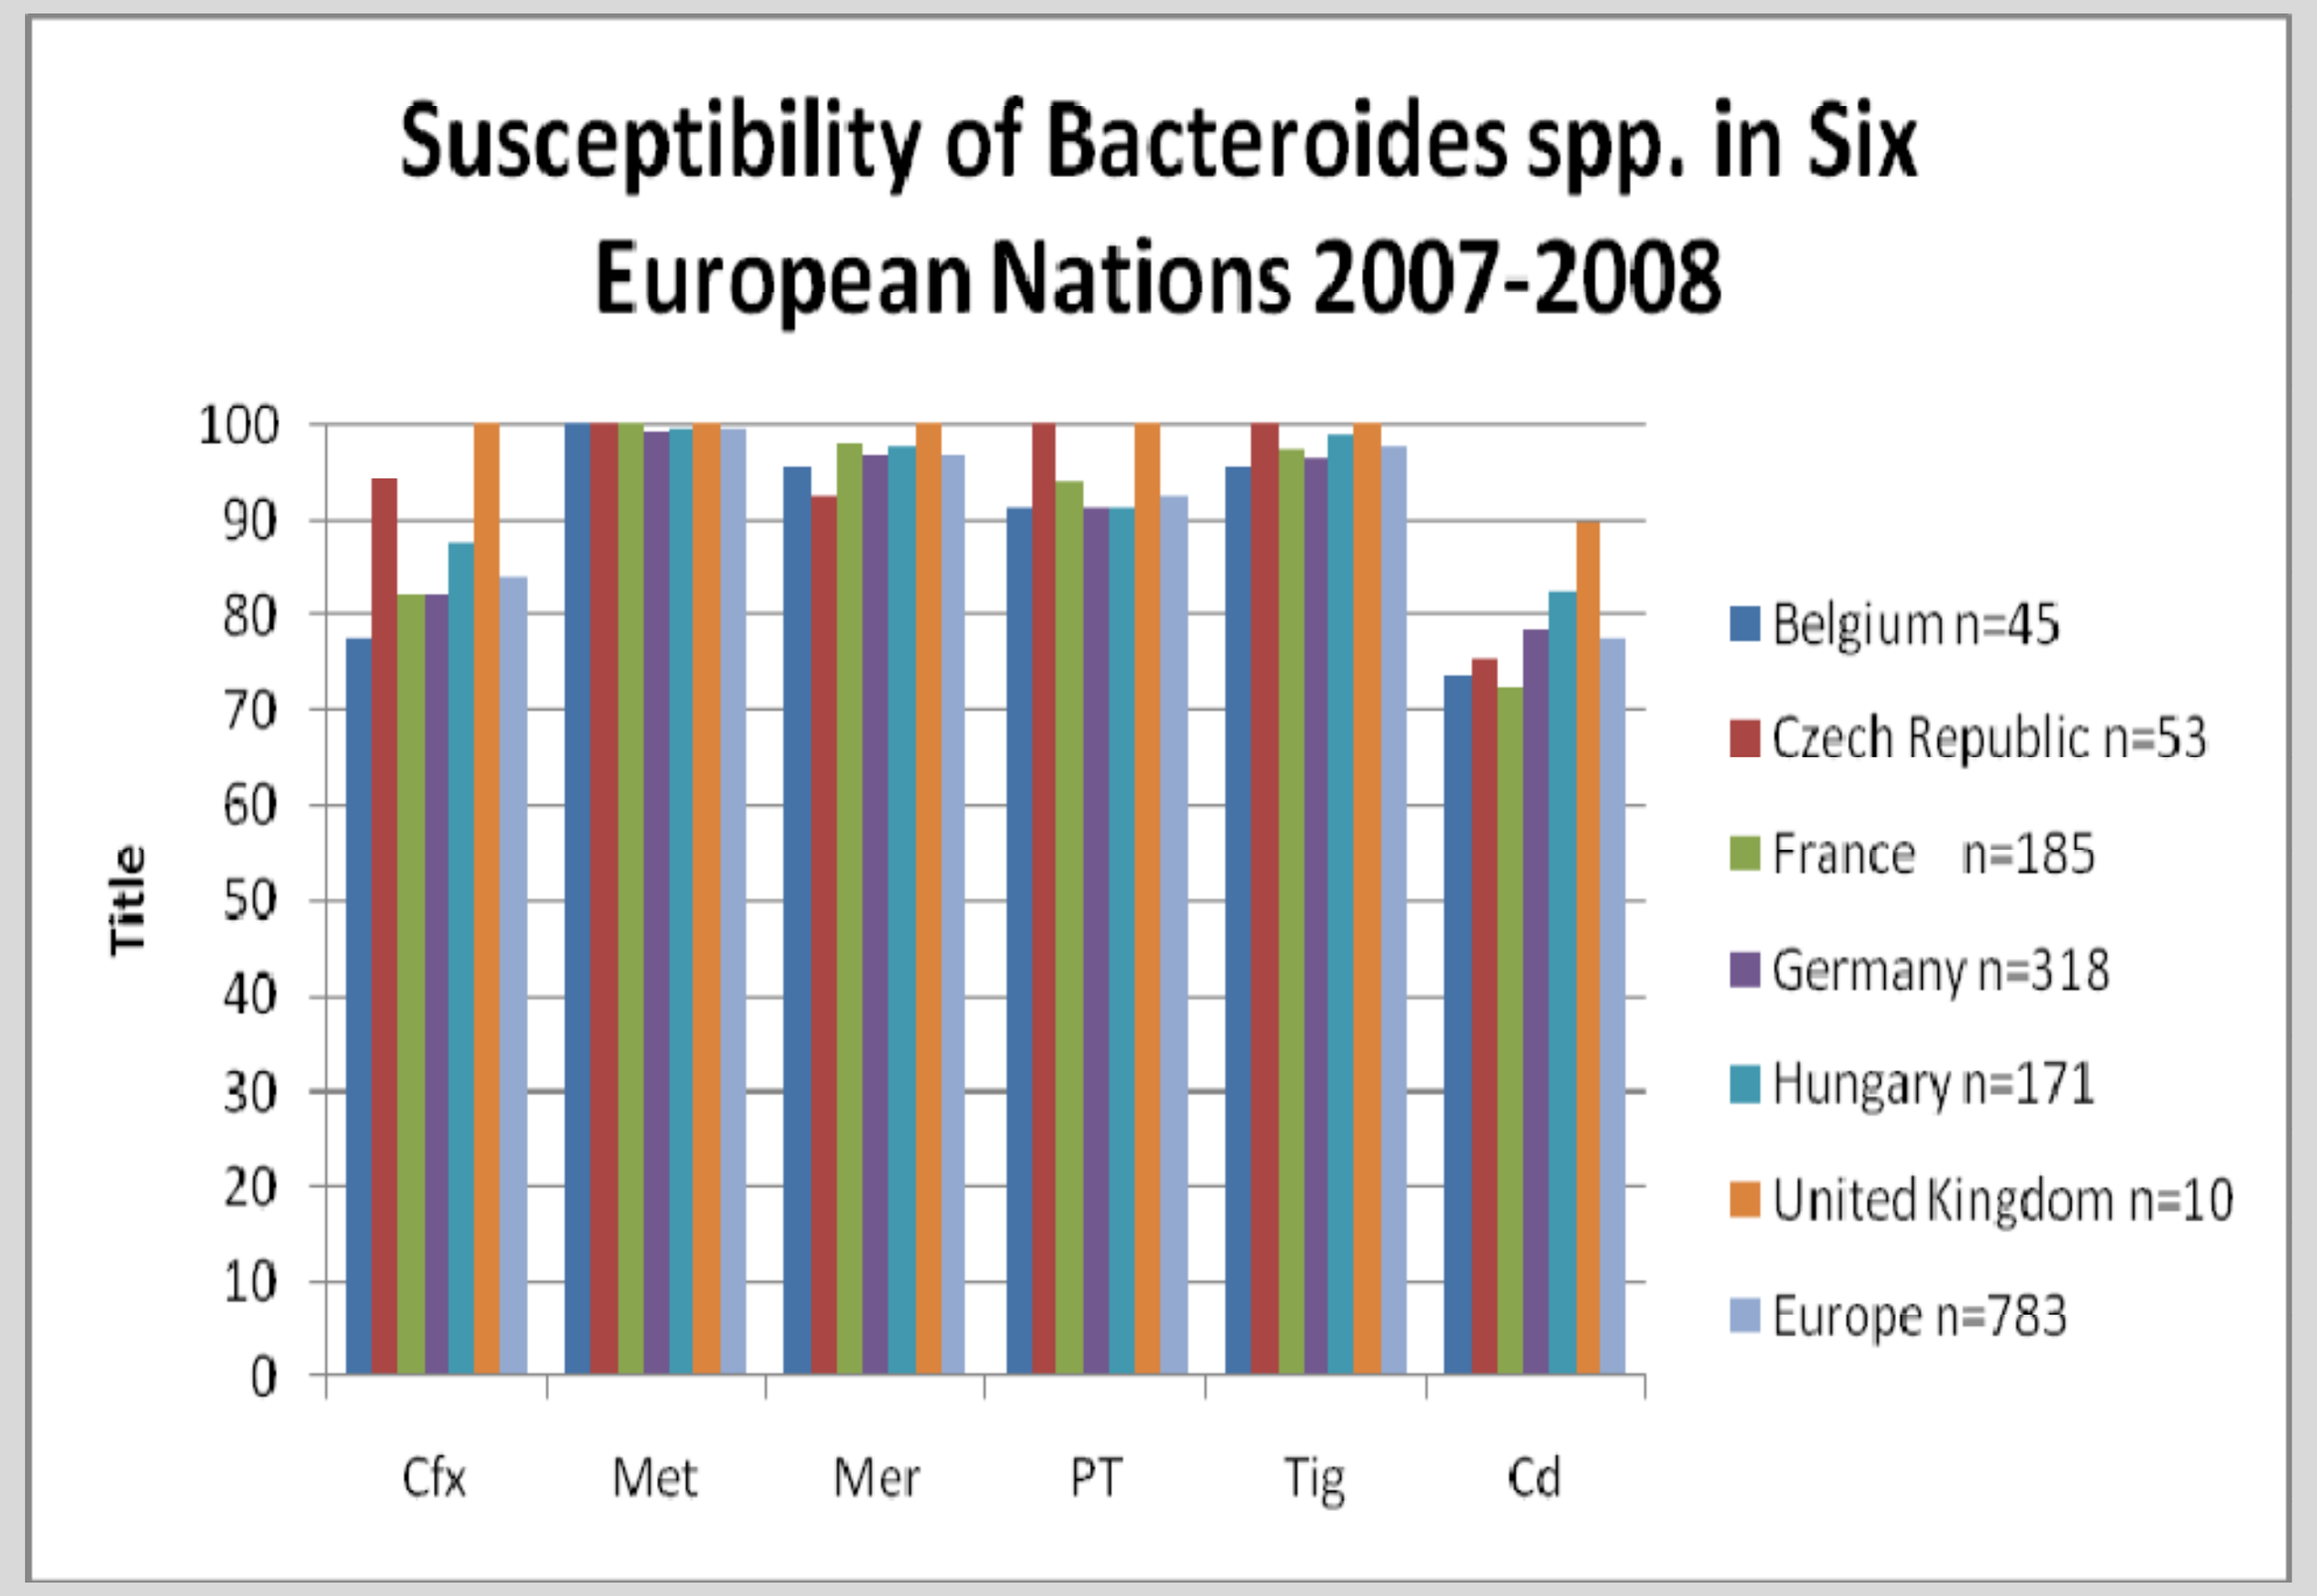
Susceptibility of Bacteroides spp. in Six European Nations 2007-2008
Title
Cfx
Met
Mer
PT
Tig
Cd
Belgium n=45
Czech Republic n=53
France n=185
Germany n=318
Hungary n=171
United Kingdom n=10
Europe n=783

Figure 2.

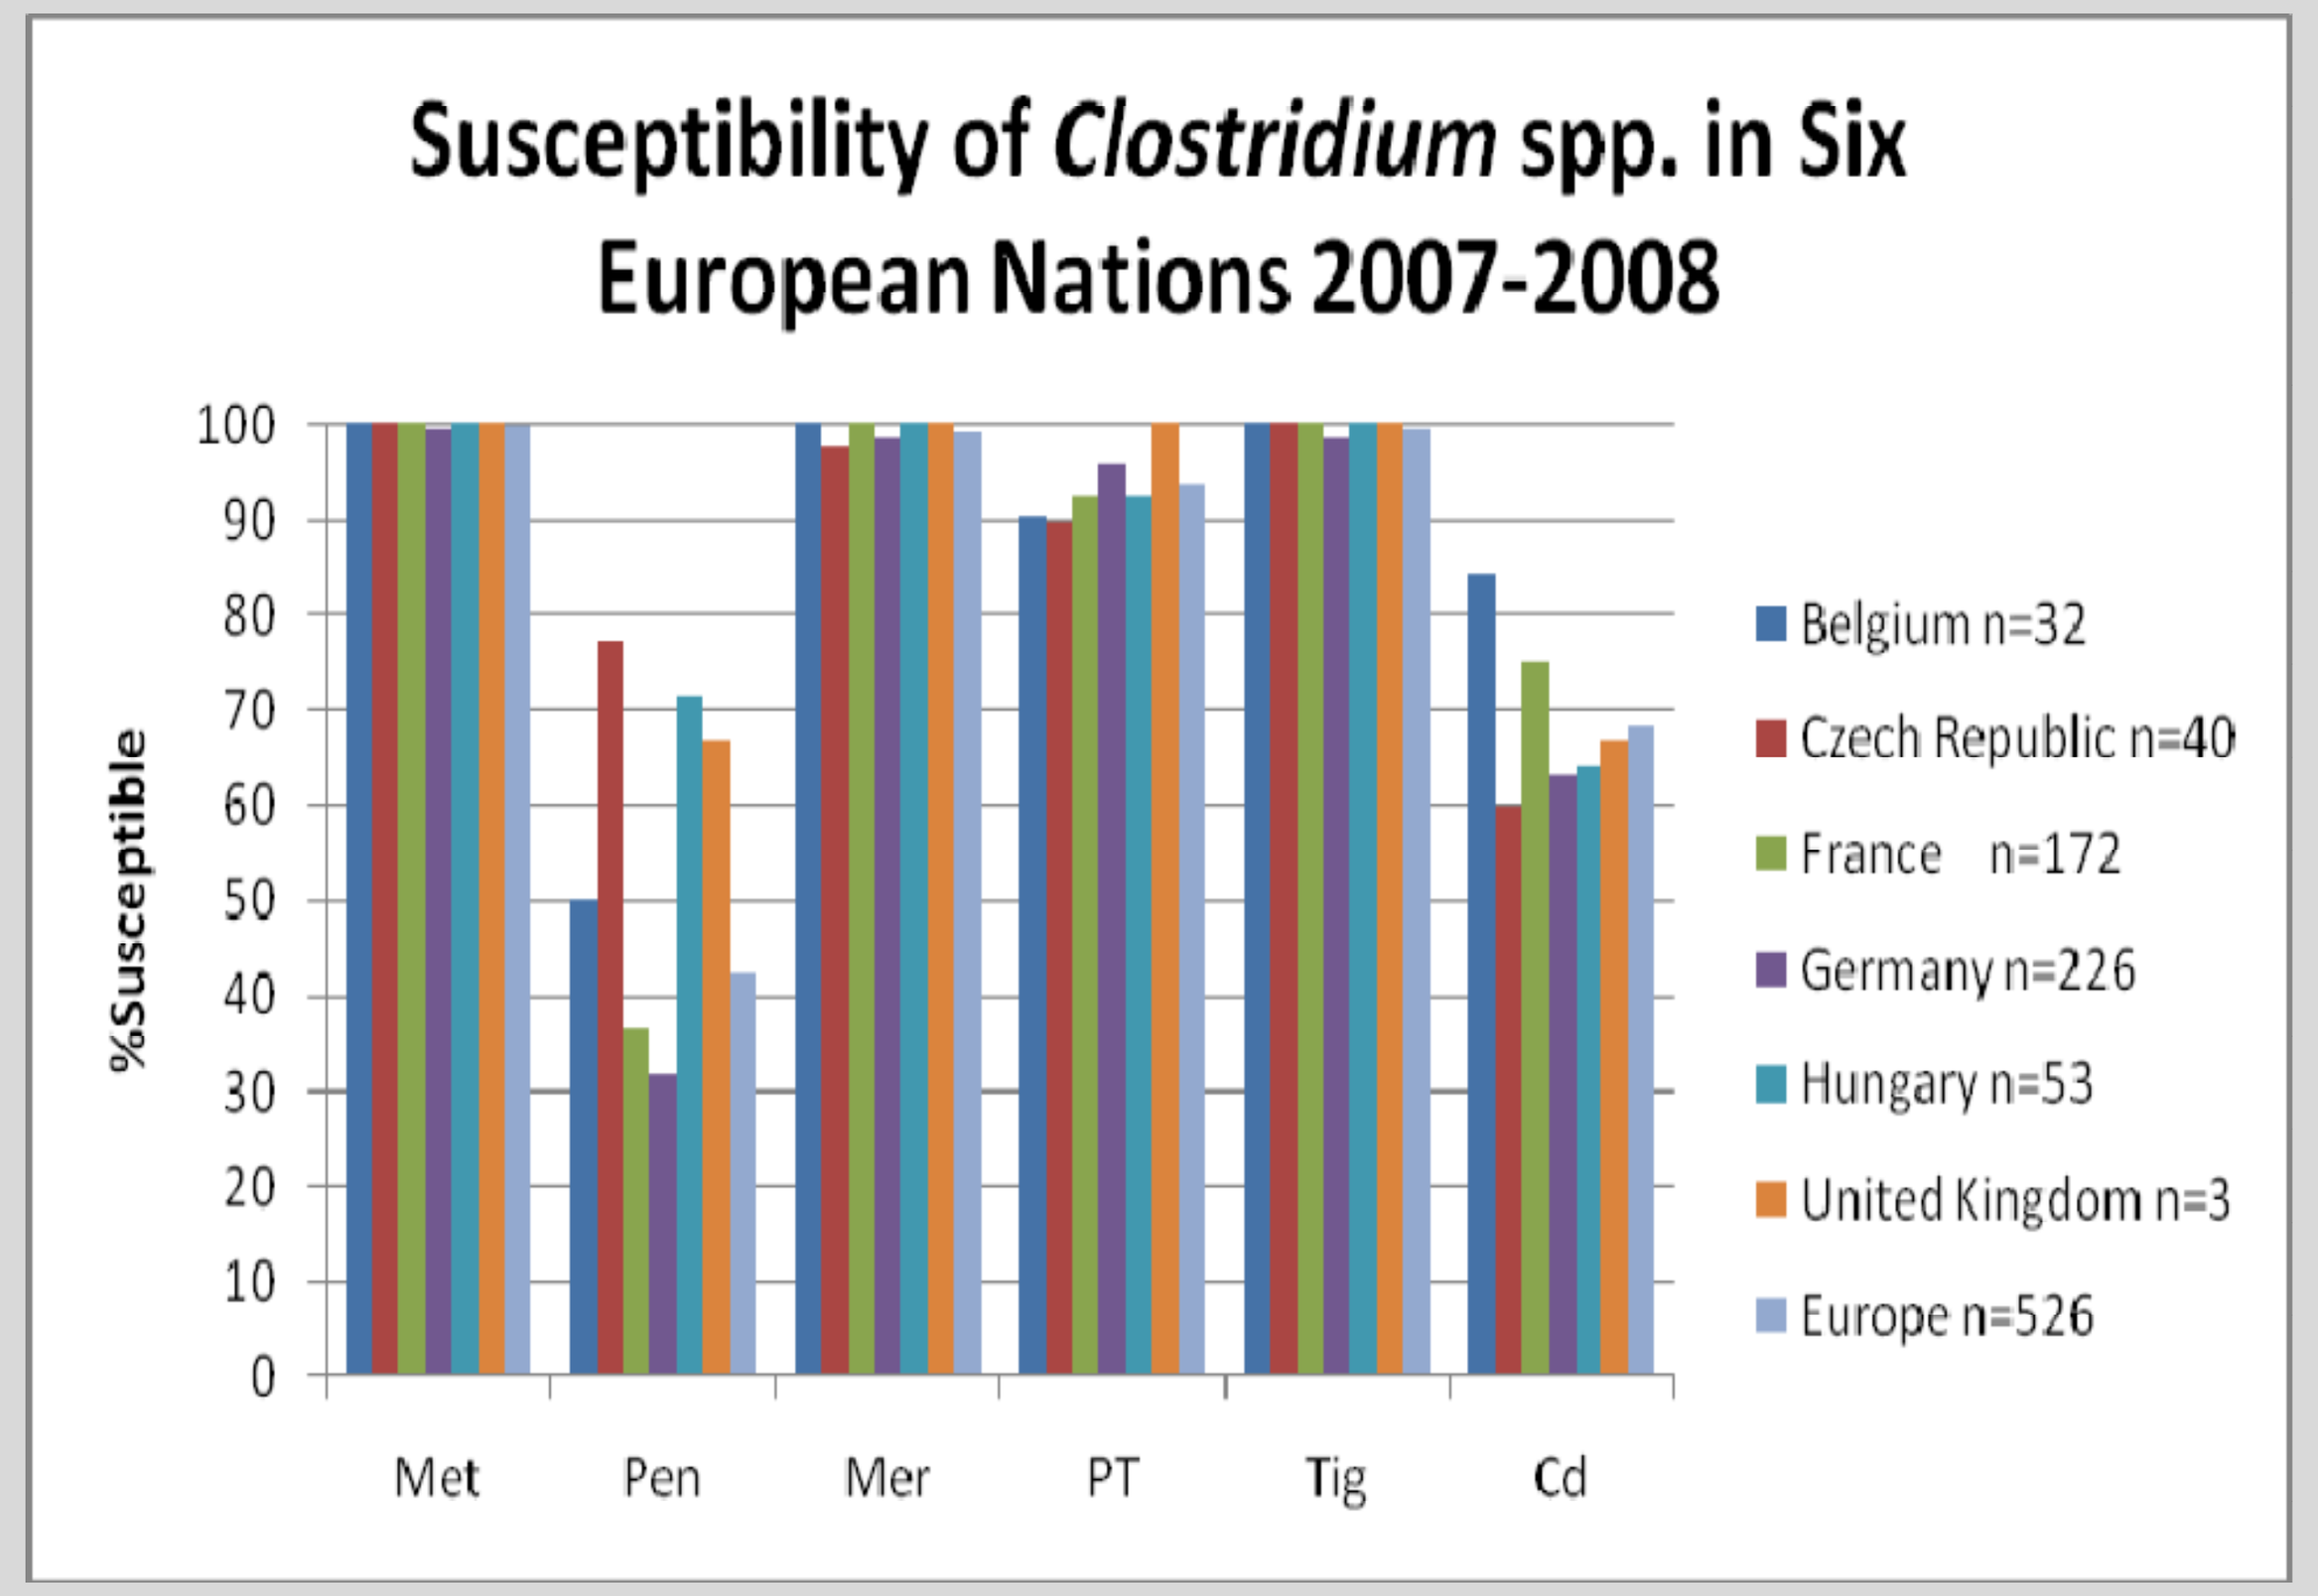
Susceptibility of *Clostridium* spp. in Six European Nations 2007-2008
%Susceptible
Met
Pen
Mer
PT
Tig
Cd
Belgium n=32
Czech Republic n=40
France n=172
Germany n=226
Hungary n=53
United Kingdom n=3
Europe n=526

Figure 3.

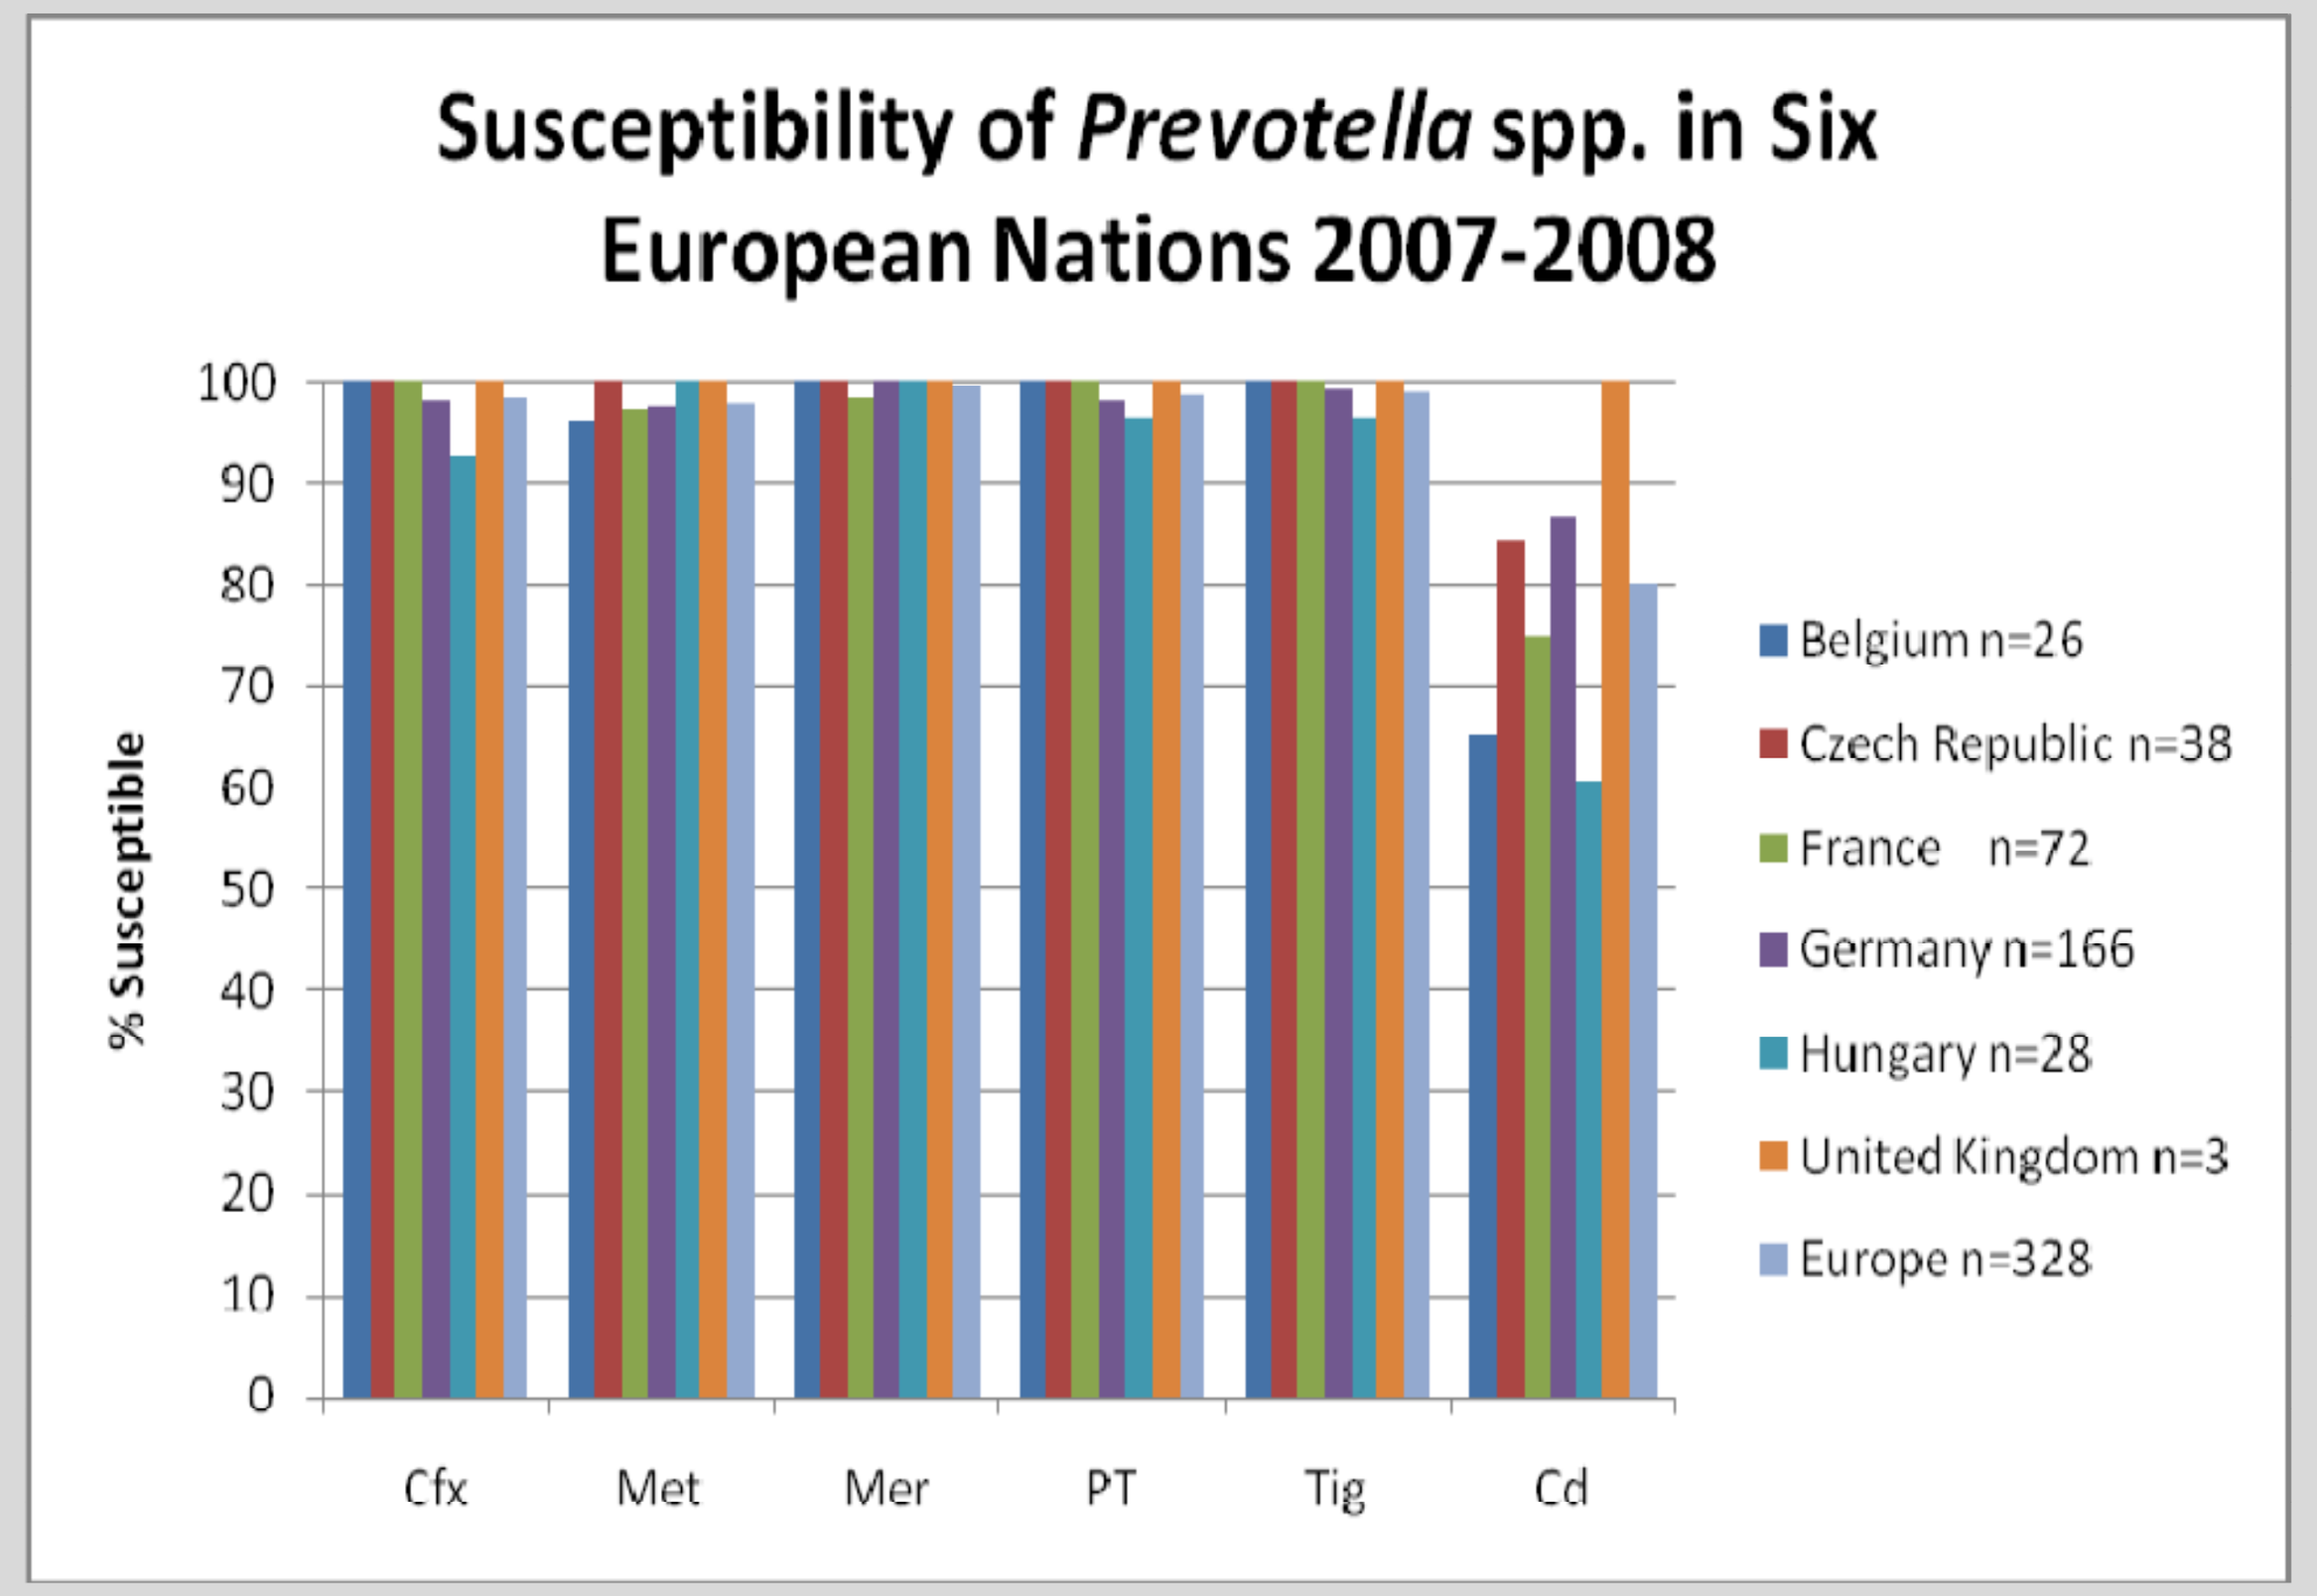
Susceptibility of *Prevotella* spp. in Six European Nations 2007-2008
% Susceptible
Cfx
Met
Mer
PT
Tig
Cd
Belgium n=26
Czech Republic n=38
France n=72
Germany n=166
Hungary n=28
United Kingdom n=3
Europe n=328

Figure 4.

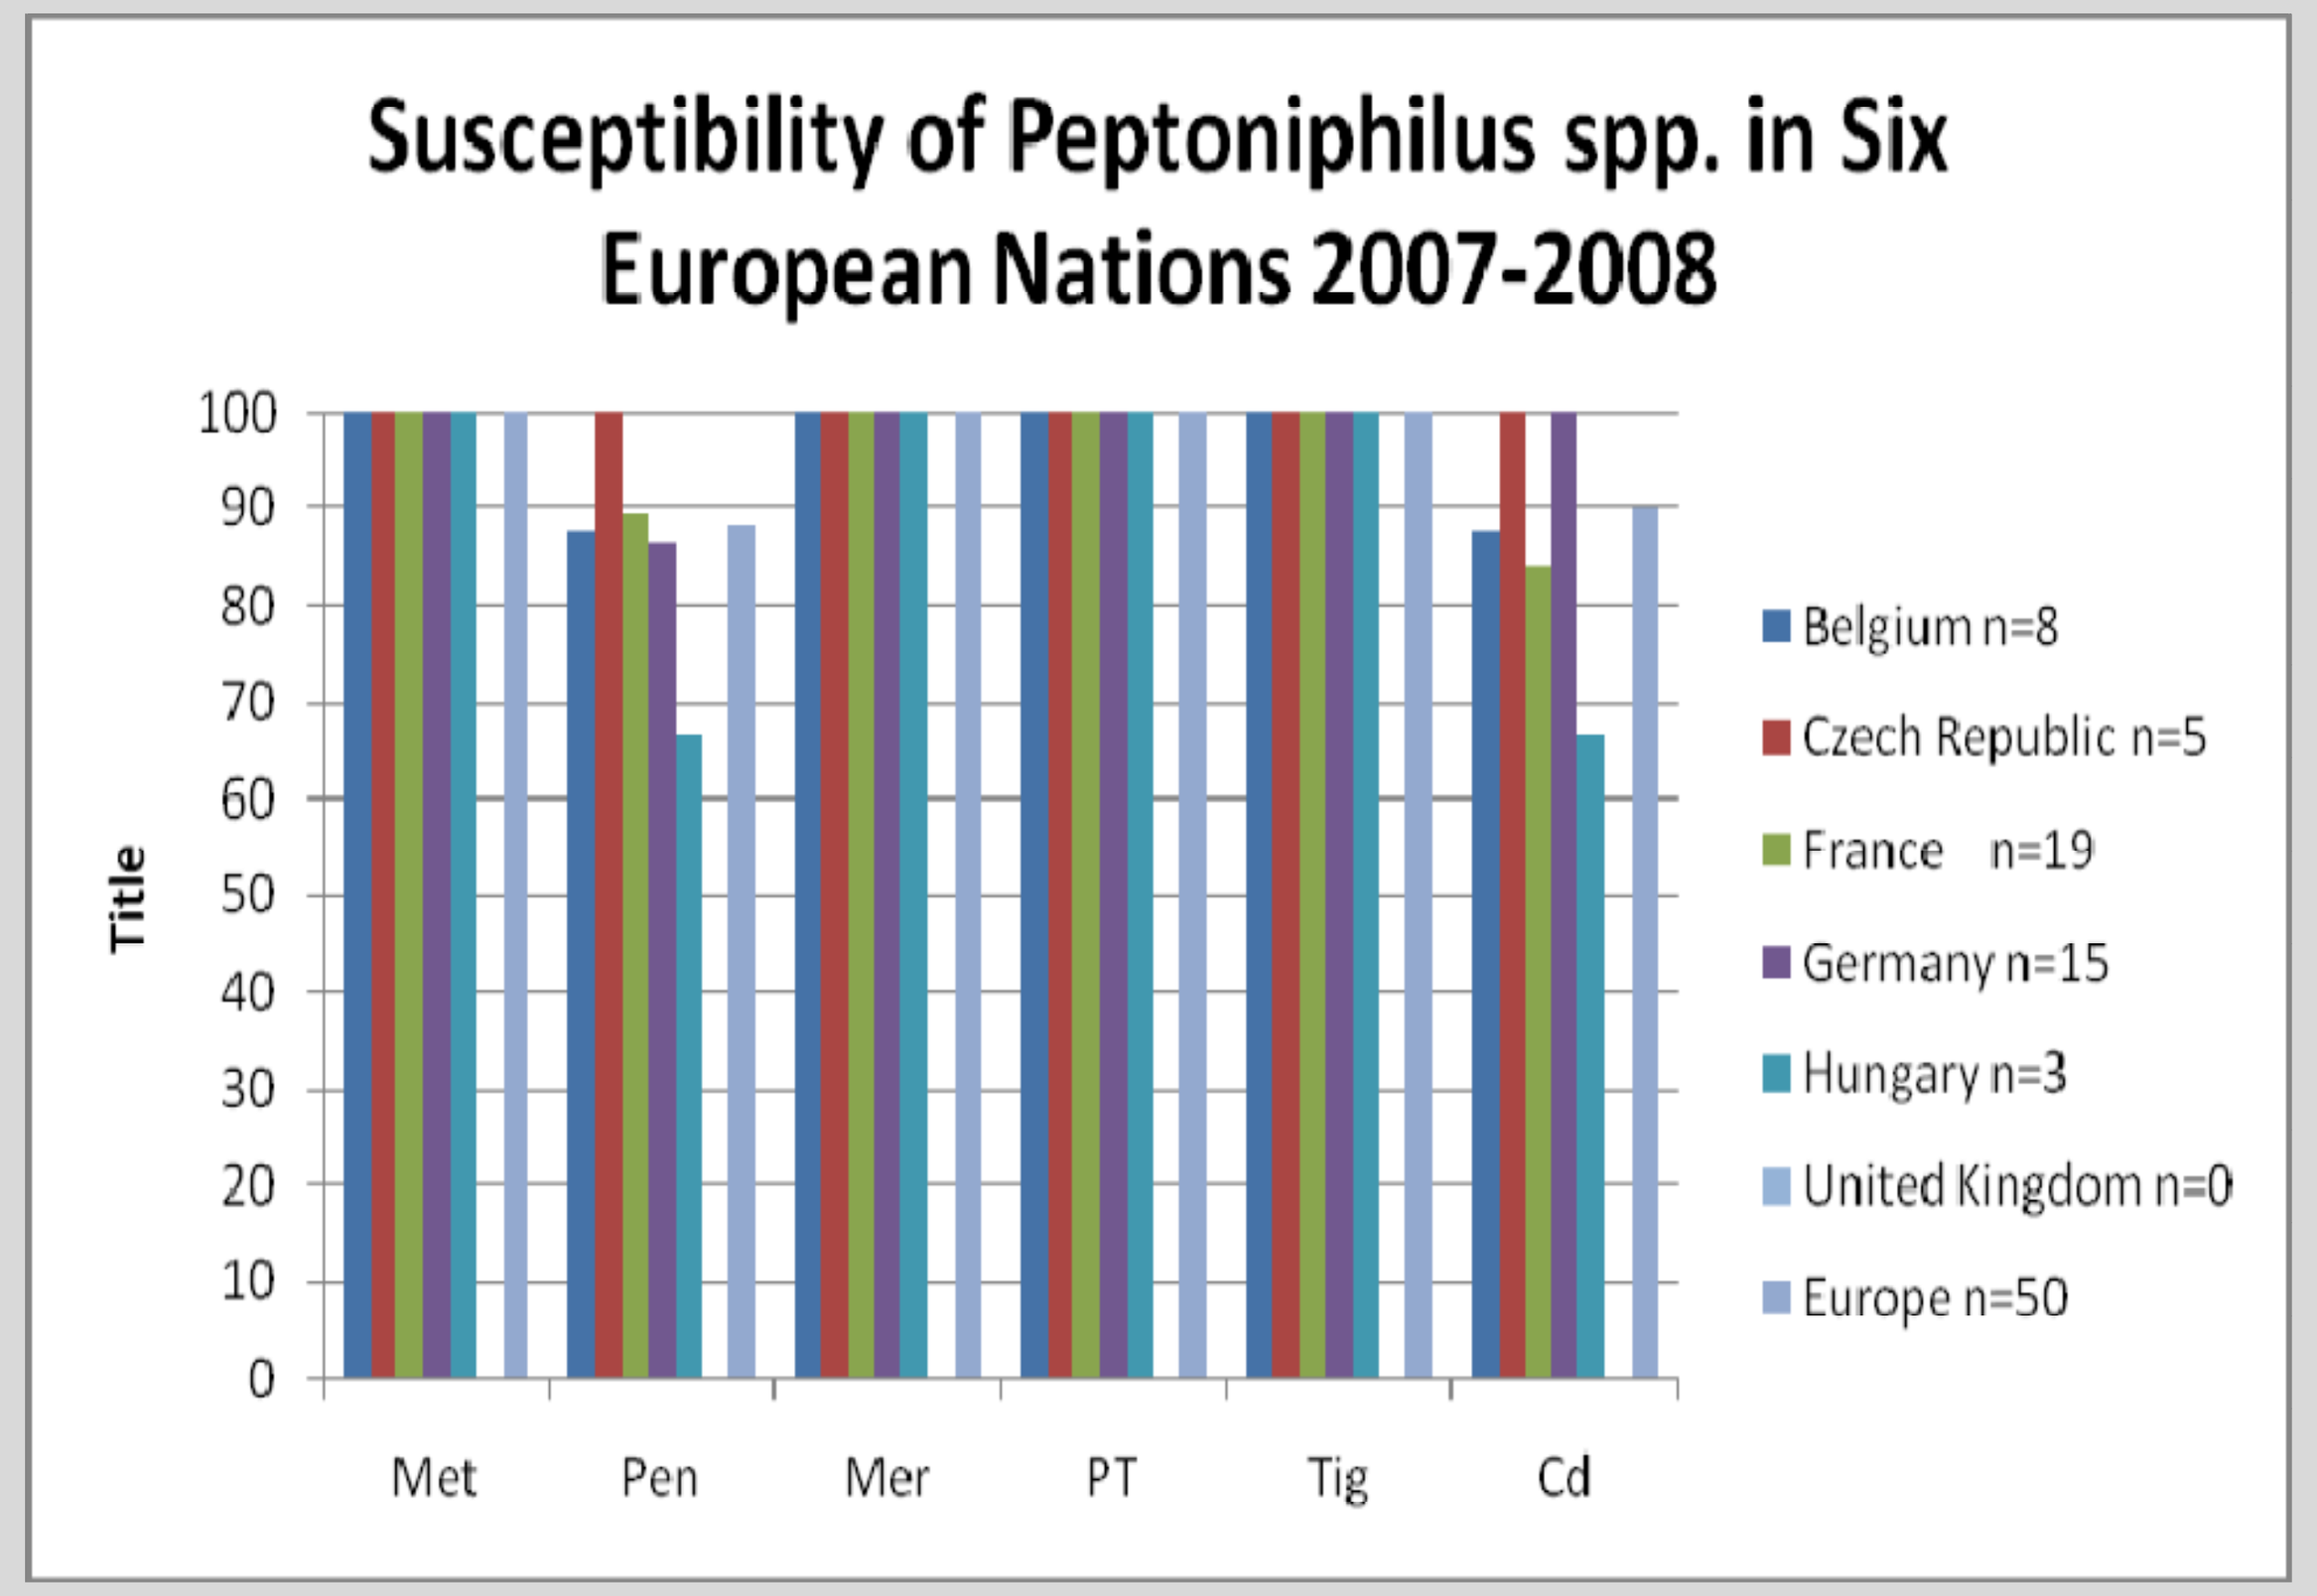
Susceptibility of Peptoniphilus spp. in Six European Nations 2007-2008
Title
Met
Pen
Mer
PT
Tig
Cd
Belgium n=8
Czech Republic n=5
France n=19
Germany n=15
Hungary n=3
United Kingdom n=0
Europe n=50

Figure 5.

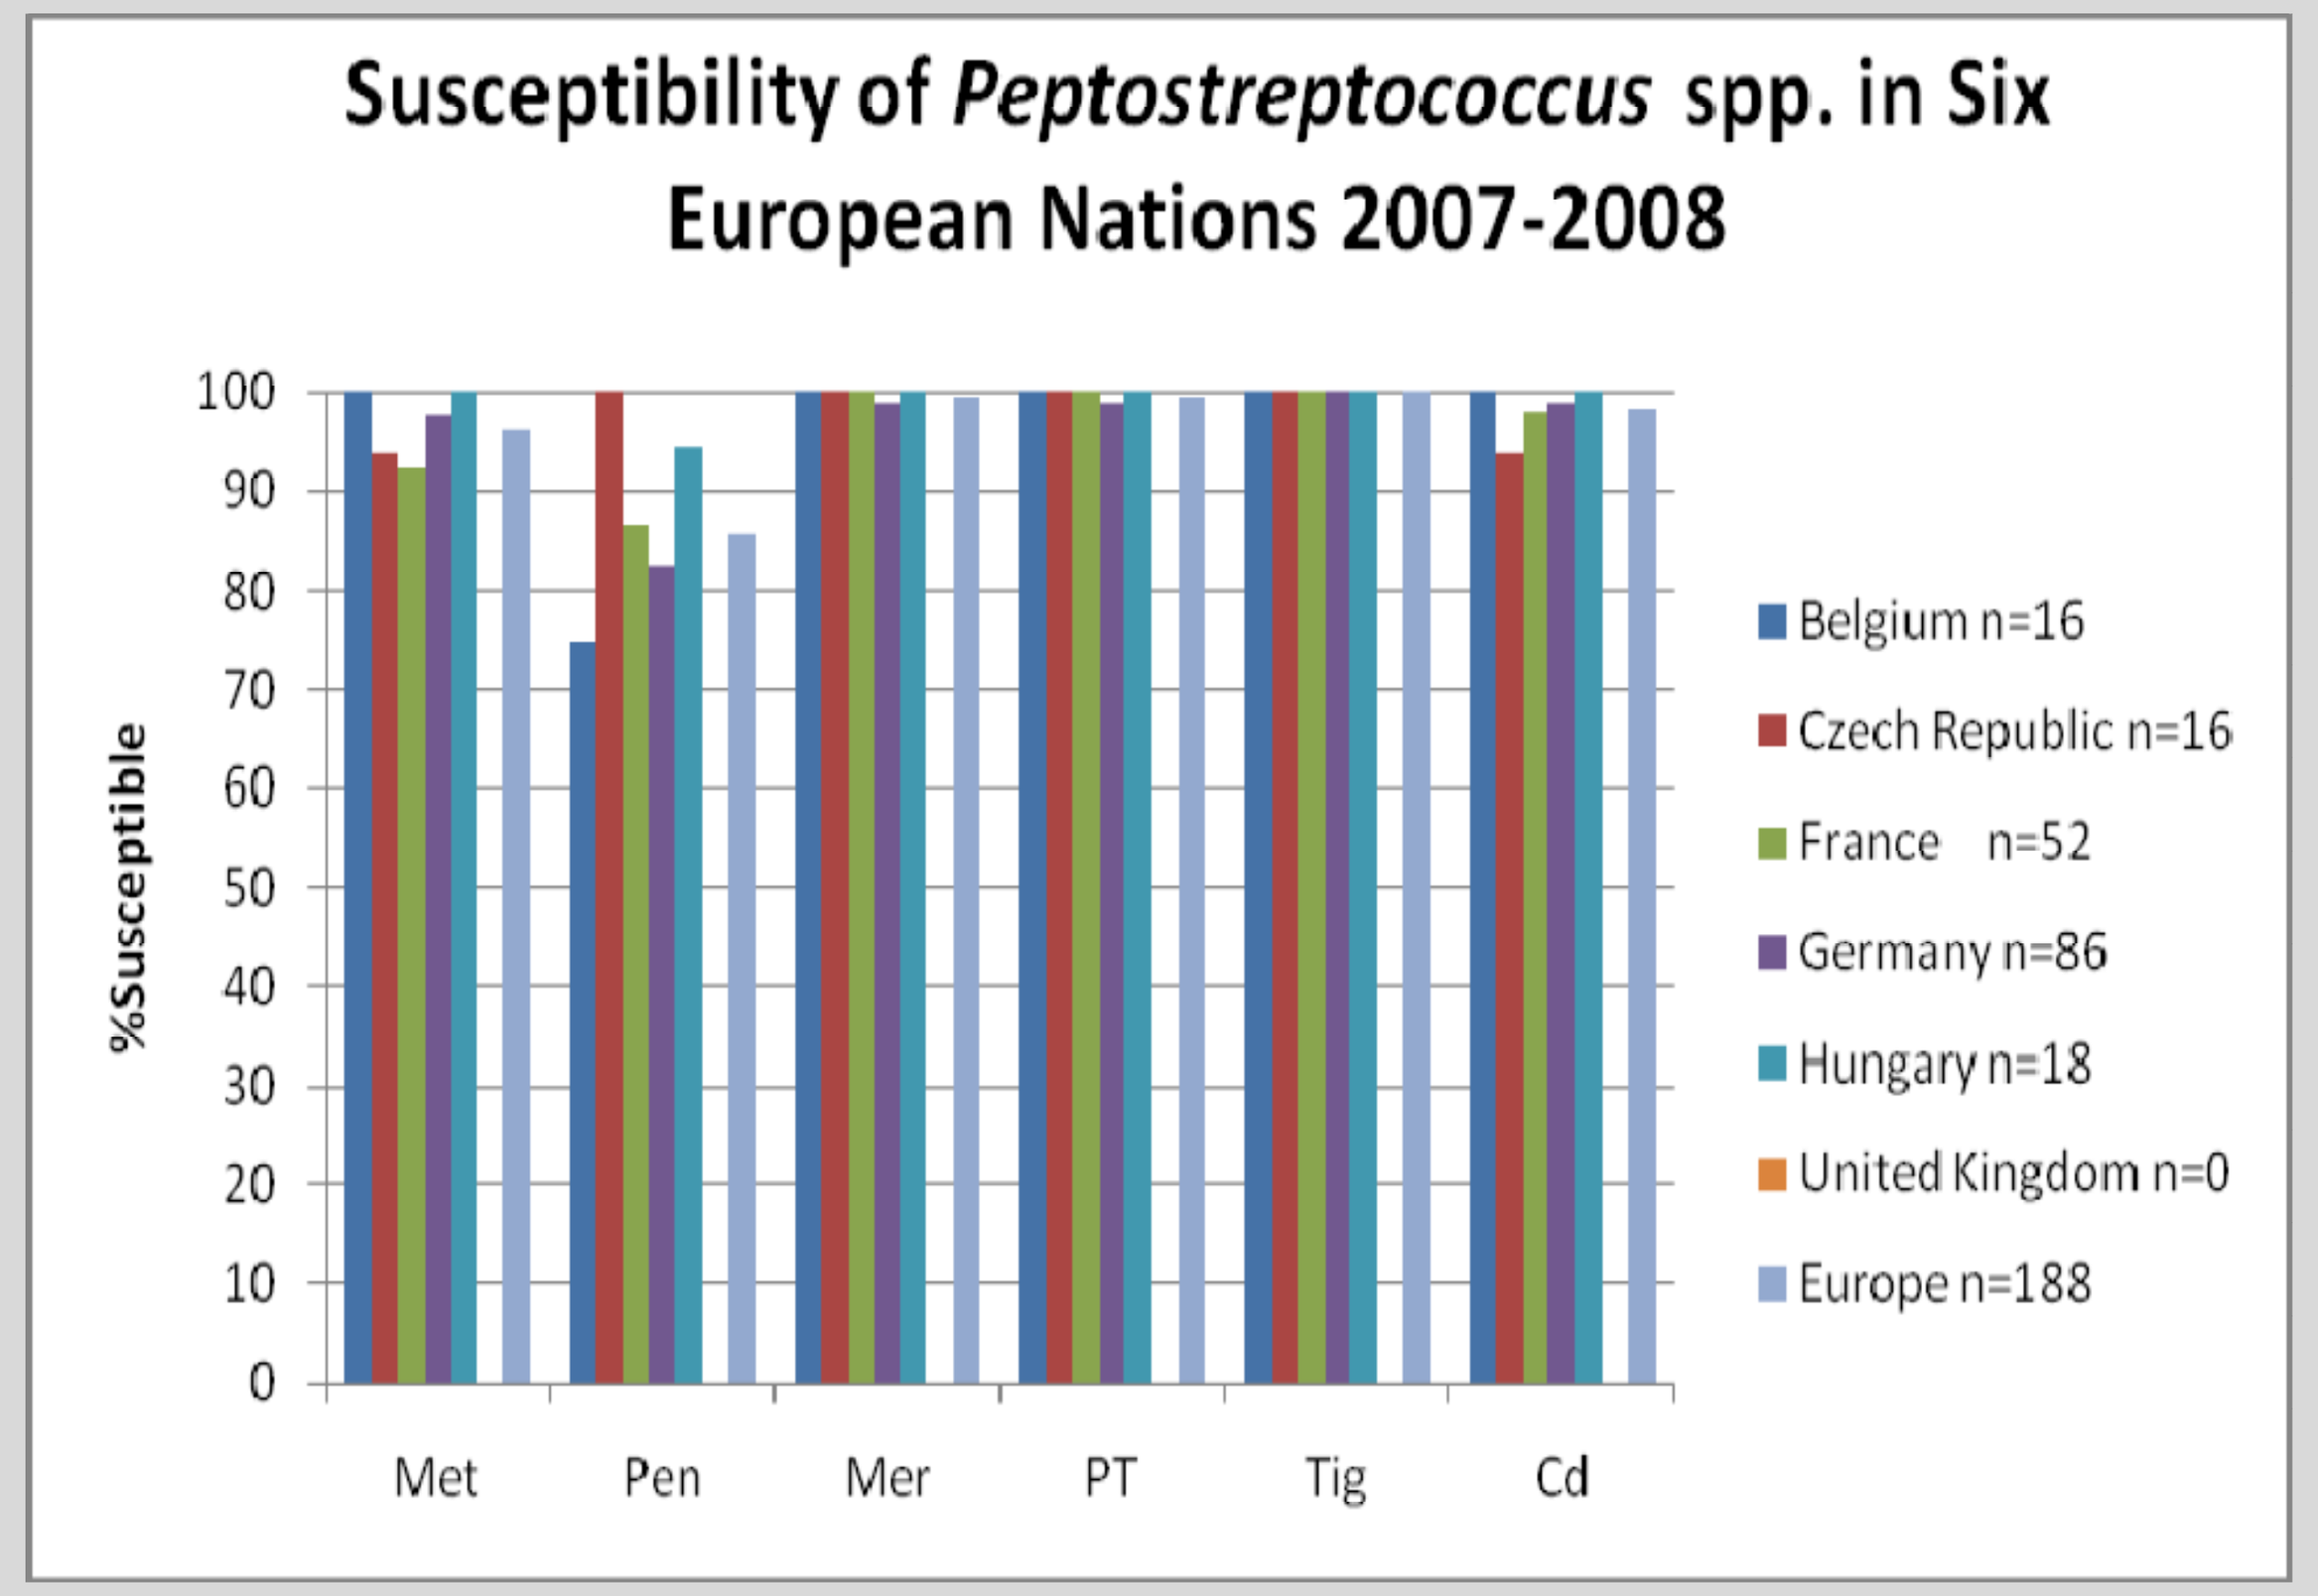
Susceptibility of *Peptostreptococcus* spp. in Six European Nations 2007-2008
%Susceptible
Met
Pen
Mer
PT
Tig
Cd
Belgium n=16
Czech Republic n=16
France n=52
Germany n=86
Hungary n=18
United Kingdom n=0
Europe n=188

Conclusions

- Tigecycline, piperacillin-tazobactam, meropenem, and metronidazole all inhibited >90% of isolates in all countries.
- Cefoxitin, clindamycin, and penicillin showed wide ranges in susceptibilities between countries and organism groups.
- No one country stood out as having lower or higher overall susceptibility levels than other European countries participating in this study.