#P-1262

Susceptibility of Gram-negative Pathogens Isolated from Intra-abdominal Infections in Europe in 2008-2009 – The SMART Study

R. Badal, S. Bouchillon, D. Hoban, A. Johnson, M. Hackel
International Health Management Associates, Inc., Schaumburg, USA

IHMA, Inc.
2122 Palmer Dr.
Schaumburg, IL
60173
Tel: 847.303.5003
Fax: 847.303.5601

Revised Abstract

Objectives: The Study for Monitoring Antimicrobial Resistance Trends (SMART) has been monitoring the antimicrobial susceptibility of aerobic gram-negative bacteria from intra-abdominal infections (IAI) since 2002. This report summarizes susceptibility levels of key IAI pathogens in Europe to ertapenem (Etp), amikacin (Ak), cefepime (Cpe), cefoxitin (Cfx), ceftazidime (Caz), ceftriaxone (Cax), ciprofloxacin (Cp), imipenem (Imp), levofloxacin (Lvx), ampicillin-sulbactam (AS), and piperacillin/tazobactam (PT) during 2008-2009. **Methods:** 46 labs in 14 European countries each collected up to 100 consecutive gram-negative bacteria/year from IAI in 2008-2009. MICs were determined by broth microdilution, and interpreted using EUCAST guidelines if available. Susceptibility rates were determined for species with ≥10 isolates. **Results:** 6,320 isolates were collected; however, only those with N≥10 (6,239, 98.7% of the total) were included in this analysis. The remaining 81 isolates represented 37 species. The table below shows % susceptible for each drug; values ≥90% are shaded.

Organism	N	Imp	Etp	Ak	Cpe	PT	Cax	Cft	Caz	Lvx	Cp	AS
E. coli	3169	99.62	99.03	92.6	87	88.2	85.5	85.5	85.1	75.8	74.2	44.2
K. pneumoniae	712	94.06	91.16	88.8	74.6	71.1	73.2	74	71.7	75.4	71.4	52.8
P. aeruginosa	509	65.82		74.9	71.9	73.3			75.6	64.6	68.6	
E. cloacae	423	98.82	84.63	96.9	75.2	67.6	53.9	55.3	56.5	89.4	85.6	17
P. mirabilis	276	75.8	99.29	87.2	91.5	94.7	87.2	88.3	89.7	80.4	70.1	70.8
K. oxytoca	267	99.63	99.63	98.5	91.1	89.3	88.5	91.5	94.8	94.8	92.6	70
C. freundii	180	98.33	98.33	89.4	81.7	66.1	54.4	55	54.4	80.6	79.4	41.1
A. baumannii	178	43.82		46.6						18	18	
M. morganella	117	76.07	99.15	97.4	98.3	98.3	82.9	61.5	64.1	87.1	80.3	0.85
E. aerogenes	112	94.64	85.71	97.3	85.7	58.9	53.6	55.4	50.9	92.9	92	26.8
S. marcescens	76	97.37	97.37	92.1	88.2	79	79	80.3	84.2	88.2	77.6	15.8
P. vulgaris	63	77.78	100	98.4	95.2	100	46	79.4	87.3	100	100	57.1
S. maltophilia	51			9.8	1.96				25.5	74.5	3.92	
H. alvei	47	100	93.62	97.9	85.1	57.5	29.8	48.9	21.3	100	100	6.38
C. koseri	38	100	100	100	94.7	97.4	89.5	94.7	94.7	94.7	94.7	86.8
C. amalonaticus	11	100	100	100	100	100	81.8	81.8	90.9	90.9	100	36.4
A. hydrophila	10	60	60	100	100	70	100	100	100	100	90	20

Conclusions: *E. coli* (50.1% of all IAI pathogens) was ≥90% susceptible vs. only 3 drugs: Imp, Etp, and Ak. *K. pneumoniae* (11.4% of all IAI pathogens) was ≥90% susceptible vs. only 2 drugs (Imp and Etp), and just 1 other (Ak) was >80%. No drug achieved even 80% susceptible vs. *P. aeruginosa*. Until definitive identification and susceptibility testing results are known, options for effective empirical therapy of IAI in Europe have diminished to include very few (e.g., carbapenems, amikacin) of the agents evaluated in this study.

Introduction

Bacterial resistance to antimicrobials continues to evolve, and levels of such resistance and cross-resistance can vary dramatically in different geographic areas. Consequently, it is increasingly important to monitor susceptibility trends in different worldwide regions over time to detect, define, track, and communicate those trends so that effective therapeutic measures can be determined and customized to meet local needs. With frequent reports in the literature regarding increases in resistance, particularly that due to extended-spectrum beta-lactamase (ESBL) production, primarily in *Escherichia coli* and *Klebsiella pneumoniae*, it is important to know regional rates and prevalence of mechanisms of resistance to commonly-used antimicrobics.

Europe, like many other regions, has experienced increases in frequency of ESBL-producing organisms, especially those harboring CTX-M enzymes [1, 2, 3].

The Study for Monitoring Antimicrobial Resistance Trends (SMART) has monitored the activity of several antimicrobics against gram-negative aerobic bacteria recovered from intra-abdominal infections (IAI) worldwide since 2002. against. This report summarizes susceptibility data of ertapenem, imipenem, amikacin, cefepime, cefotaxime, cefoxitin, ceftazidime, ceftriaxone, ciprofloxacin, levofloxacin, ampicillin-sulbactam, and piperacillin-tazobactam against IAI pathogens collected from 39 laboratories in 12 European nations to in 2008-2009.

Materials & Methods

- All isolates were non-repeat isolates derived from IAIs. Only one isolate per species per patient was accepted into the study. 46 laboratories in 14 European countries each collected up to 100 consecutive non-selected gram-negative pathogens each year of the study. Isolates were identified to the species level and sent to a central laboratory (International Health Management Associates, Inc., in Schaumburg, Illinois), where identifications were confirmed and susceptibility testing was done.
- Minimum inhibitory concentrations (MICs) were determined using MicroScan dehydrated broth microdilution panels manufactured by Siemens Medical Solutions Diagnostics (West Sacramento, California, USA). All antimicrobial agents were supplied by the panel manufacturer. The following antimicrobial agents were included on the panels with their dilution ranges (expressed in mg/L): ertapenem 0.03-4, imipenem 0.06-8, cefepime 0.5-32, ceftazidime 0.5-128, ceftazidime/clavulanic acid 0.12-16, cefoxitin 2-16, ciprofloxacin 0.25-2, amikacin 4-32, levofloxacin 0.5-4, cefotaxime 0.5-128, cefotaxime/clavulanic acid 0.12/2-16/2, piperacillin/tazobactam 2/4-64/4, ampicillin/sulbactam 2/2-16/2, and ceftriaxone 1-32.
- MIC interpretive criteria followed published guidelines established by the European Committee on Antimicrobial Susceptibility Testing (EUCAST), www.eucast.org [4].
- *E. coli*, *K. pneumoniae, K. oxytoca*, and *Proteus mirabilis* isolates were classified as ESBL producers if there was at least an eight-fold reduction of MIC for ceftazidime or cefotaxime tested in combination with clavulanic acid versus their MICs when tested alone [5].
- Quality control testing (QC) was done by each testing site on each day of testing using the CLSI-recommended QC strains: *E. coli* ATCC 25922, *K. pneumoniae* ATCC 700603 (positive ESBL control), and *Pseudomonas aeruginosa* ATCC 27853 [5].
- Development of a centralized database of study results was managed by International Health Management Associates, Inc. located in Schaumburg, IL, USA.

References

1. Livermore, David M., Canton, Rafael, Gniadkowski, Marek, *et al*. *CTX-M: changing the face of ESBLs in Europe.* J Antimicrob Chemother (2007) **59:** 165-174.
2. Canton, R., Novais, A., Valverde, A., *et al. Prevalence and spread of extended-spectrum β-lactamase-producing Enterobacteriaceae in Europe.* Clin Microbiol Infect (2008) **14:** (Suppl.1), 144-153.
3. Coque TM, Baquero F, Canton R. *Increasing prevalence of ESBL-producing Enterobacteriaceae in Europe.* Euro Surveill. 2008 Nov 20:13(47).
4. European Committee on Antimicrobial Susceptibility Testing (EUCAST) website, http://www.eucast.org, 2009.
5. CLSI, *Performance Standards for Antimicrobial Susceptibility Testing, Document M100-S19*. 2009: Clinical Laboratory Standards Institute (CLSI), 940 West Valley Road, Suite 1400, Wayne, Pennsylvania 19087-1898 USA.

Acknowledgements

The contributions of all the SMART investigators and laboratories in Europe are greatly appreciated.
This study was funded by a grant from Merck & Co., Inc.

Results

Table 1. Susceptibility percentages of IAI pathogens for which n≥10 with percentages ≥90% highlighted.

Organism	N	Imp	Etp	Ak	Cpe	PT	Cax	Cft	Caz	Lvx	Cp	AS
E. coli	3169	99.62	99.03	92.6	87	88.2	85.5	85.5	85.1	75.8	74.2	44.2
K. pneumoniae	712	94.06	91.16	88.8	74.6	71.1	73.2	74	71.7	75.4	71.4	52.8
P. aeruginosa	509	65.82		74.9	71.9	73.3			75.6	64.6	68.6	
E. cloacae	423	98.82	84.63	96.9	75.2	67.6	53.9	55.3	56.5	89.4	85.6	17
P. mirabilis	276	75.8	99.29	87.2	91.5	94.7	87.2	88.3	89.7	80.4	70.1	70.8
K. oxytoca	267	99.63	99.63	98.5	91.1	89.3	88.5	91.5	94.8	94.8	92.6	70
C. freundii	180	98.33	98.33	89.4	81.7	66.1	54.4	55	54.4	80.6	79.4	41.1
A. baumannii	178	43.82		46.6						18	18	
M. morganella	117	76.07	99.15	97.4	98.3	98.3	82.9	61.5	64.1	87.1	80.3	0.85
E. aerogenes	112	94.64	85.71	97.3	85.7	58.9	53.6	55.4	50.9	92.9	92	26.8
S. marcescens	76	97.37	97.37	92.1	88.2	79	79	80.3	84.2	88.2	77.6	15.8
P. vulgaris	63	77.78	100	98.4	95.2	100	46	79.4	87.3	100	100	57.1
S. maltophilia	51			9.8	1.96				25.5	74.5	3.92	
H. alvei	47	100	93.62	97.9	85.1	57.5	29.8	48.9	21.3	100	100	6.38
C. koseri	38	100	100	100	94.7	97.4	89.5	94.7	94.7	94.7	94.7	86.8
C. amalonaticus	11	100	100	100	100	100	81.8	81.8	90.9	90.9	100	36.4
A. hydrophila	10	60	60	100	100	70	100	100	100	100	90	20

Imp=imipenem; Etp=ertapenem; Ak=amikacin; Cpe=cefepime; PT=piperacillin-tazobactam; Cax=ceftriaxone; Cft=cefotaxime; Caz=ceftazidime; Lvx=levofloxacin; Cp=ciprofloxacin; AS=ampicillin-sulbactam.
Blank=no EUCAST interpretive criteria exist for this organism/drug combination.

Figure 1. Frequency of occurrence of aerobic gram-negative IAI pathogens in 2008-2009.

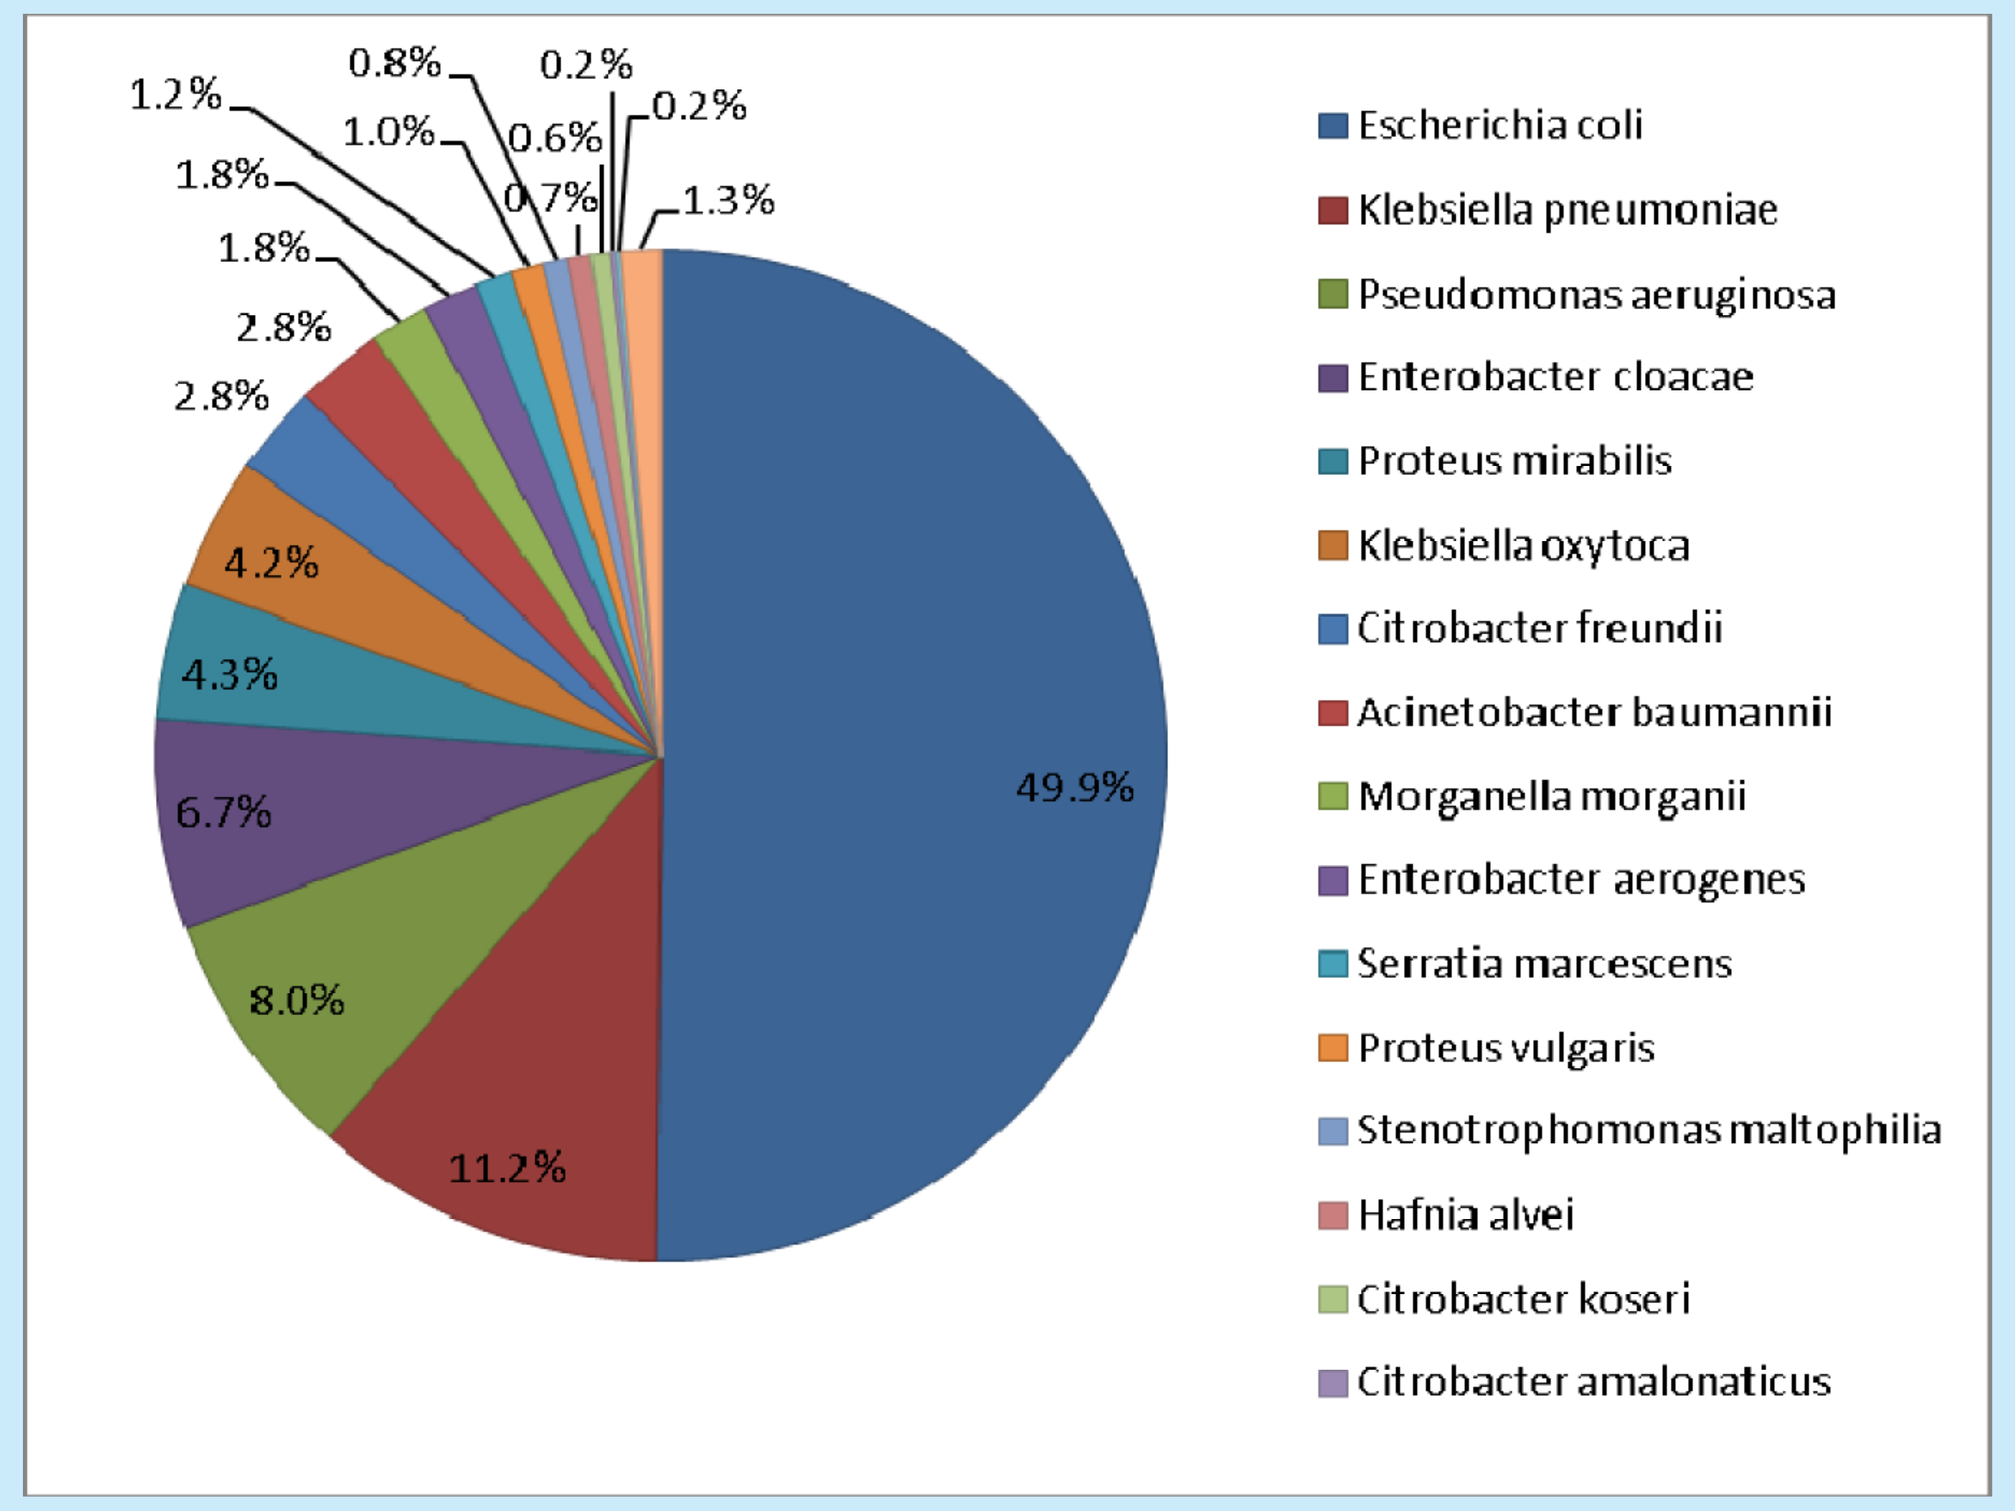
49.9%
11.2%
8.0%
6.7%
4.3%
4.2%
2.8%
2.8%
1.8%
1.8%
1.2%
1.0%
0.8%
0.7%
0.6%
0.2%
0.2%
1.3%
Escherichia coli
Klebsiella pneumoniae
Pseudomonas aeruginosa
Enterobacter cloacae
Proteus mirabilis
Klebsiella oxytoca
Citrobacter freundii
Acinetobacter baumannii
Morganella morganii
Enterobacter aerogenes
Serratia marcescens
Proteus vulgaris
Stenotrophomonas maltophilia
Hafnia alvei
Citrobacter koseri
Citrobacter amalonaticus

Table 2. Proportions of *E. coli*, *K. pneumoniae*, *K. oxytoca*, and *P. mirabilis* that were ESBL-positive in 2008-2009.

Species	#ESBL+/#ESBL-	% ESBL+
E. coli	(375/2794)	11.8%
K. pneumoniae	(155/557)	21.8%
K. oxytoca	(16/251)	6.0%
P. mirabilis	(14/262)	5.1%
All aerobic gram-negative IAI pathogens	(560/6320)	8.9%

Figure 2. Antimicrobic susceptibility of ESBL-negative (n=2794) and –positive *E. coli* (n=375).

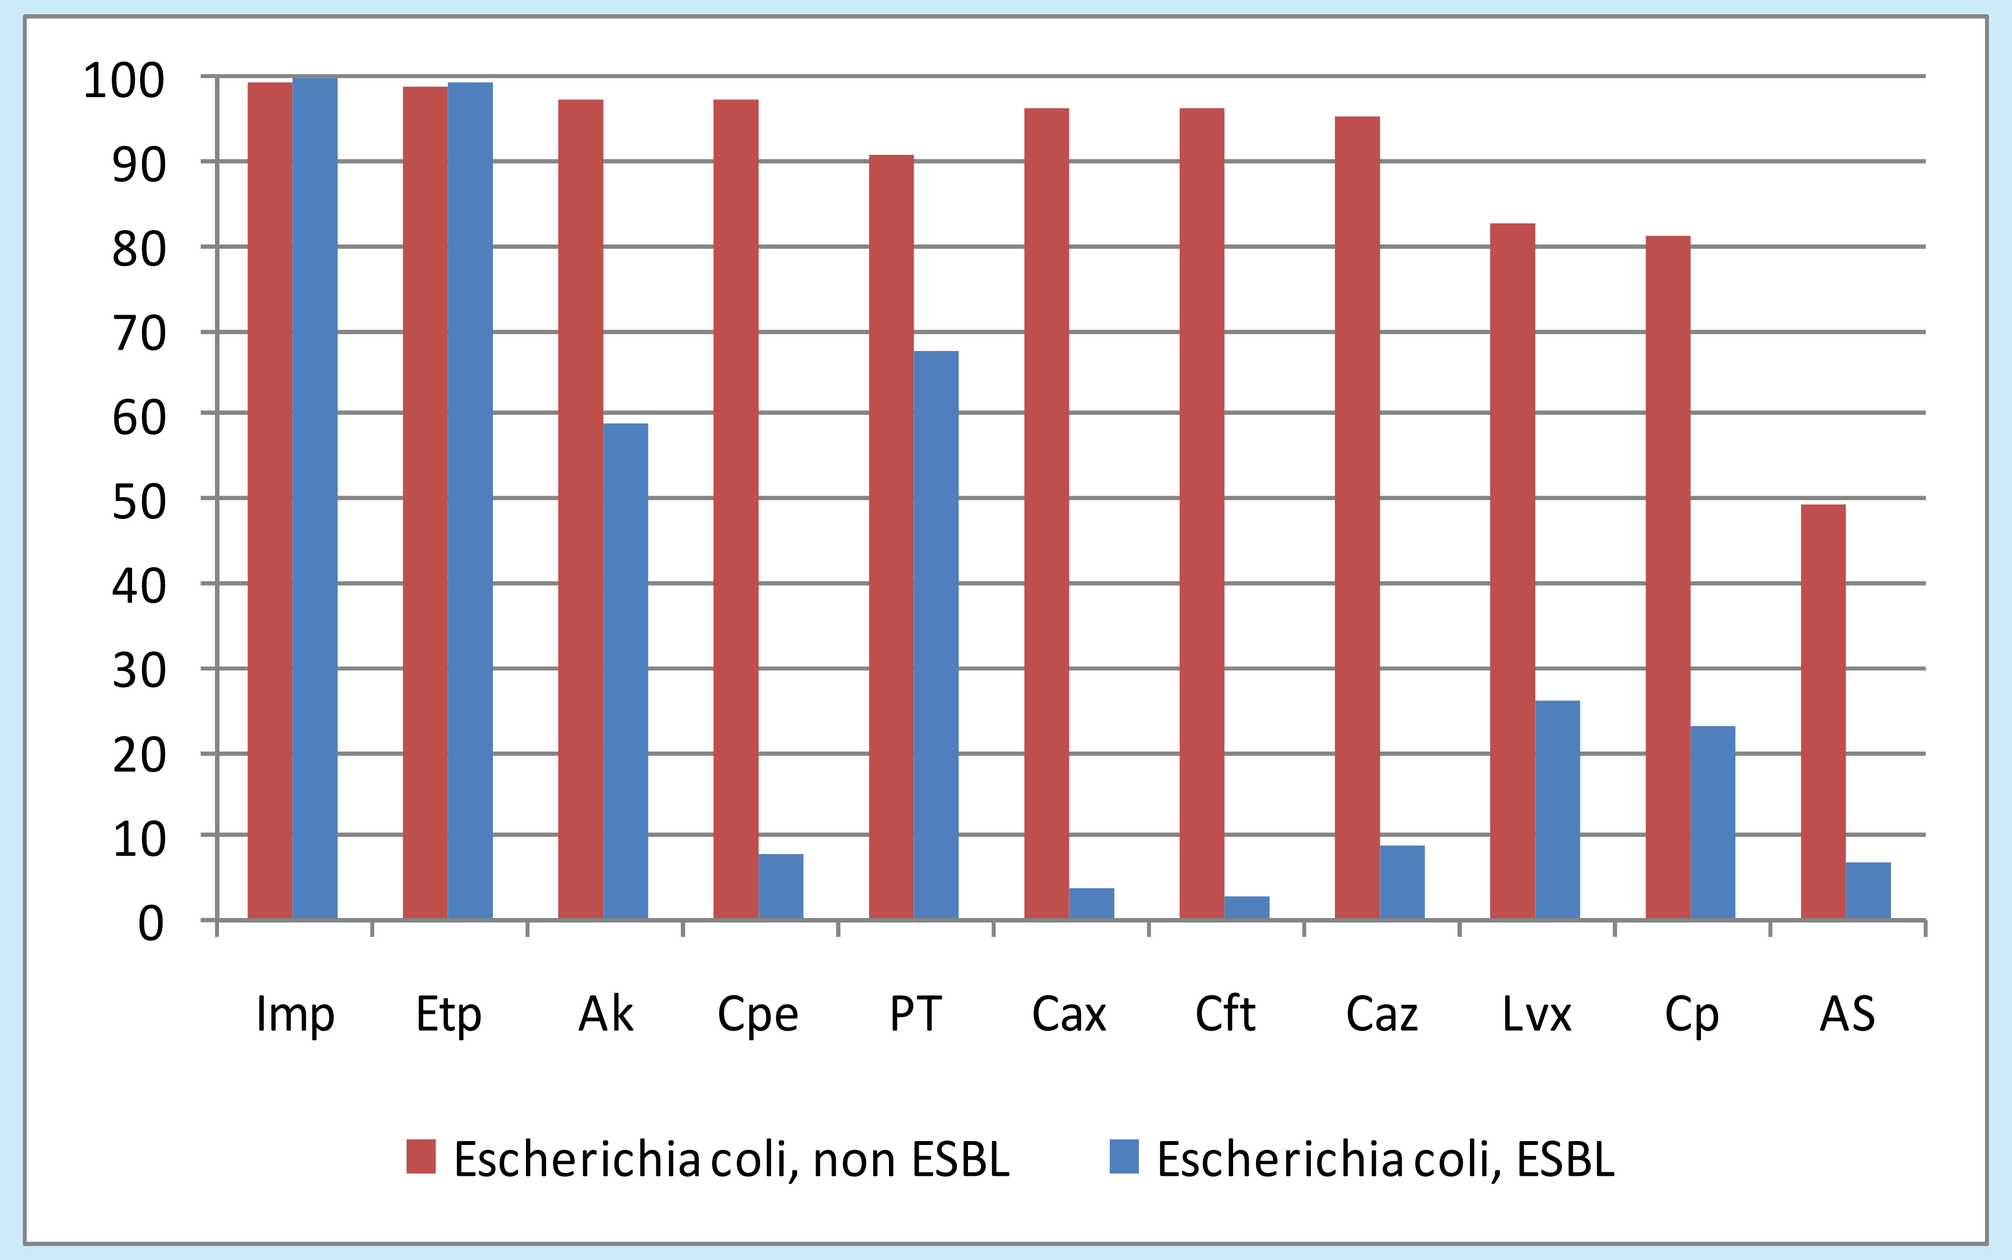
100
90
80
70
60
50
40
30
20
10
0
Imp
Etp
Ak
Cpe
PT
Cax
Cft
Caz
Lvx
Cp
AS
Escherichia coli, non ESBL
Escherichia coli, ESBL

Imp=imipenem; Etp=ertapenem; Ak=amikacin; Cpe=cefepime; PT=piperacillin-tazobactam; Cax=ceftriaxone; Cft=cefotaxime; Caz=ceftazidime; Lvx=levofloxacin; Cp=ciprofloxacin; AS=ampicillin-sulbactam.

Figure 3. Antimicrobic susceptibility of ESBL-negative (n=557) and –positive (n=155) *K. pneumoniae.*

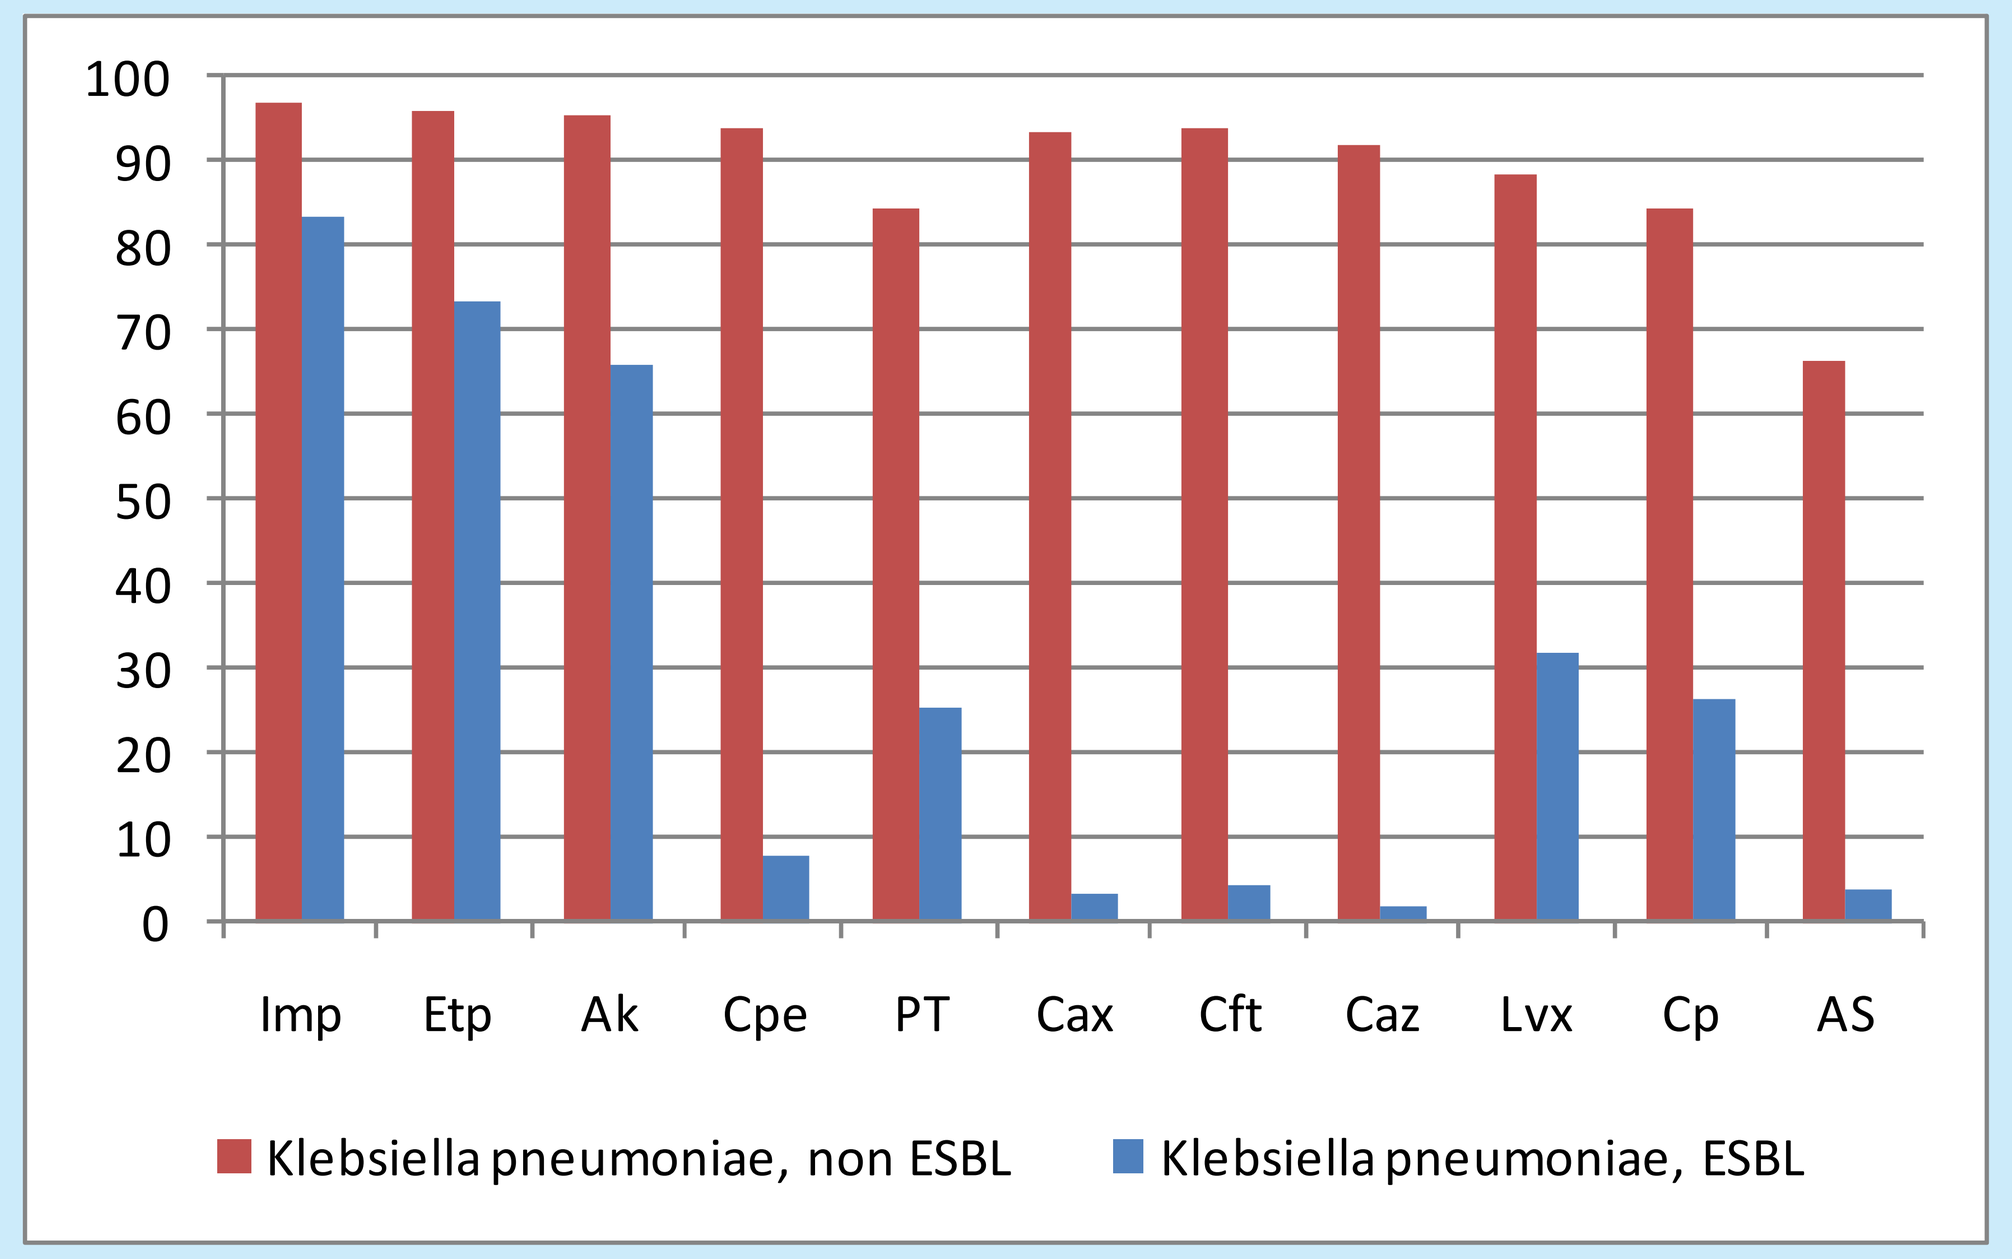
100
90
80
70
60
50
40
30
20
10
0
Imp
Etp
Ak
Cpe
PT
Cax
Cft
Caz
Lvx
Cp
AS
Klebsiella pneumoniae, non ESBL
Klebsiella pneumoniae, ESBL

Imp=imipenem; Etp=ertapenem; Ak=amikacin; Cpe=cefepime; PT=piperacillin-tazobactam; Cax=ceftriaxone; Cft=cefotaxime; Caz=ceftazidime; Lvx=levofloxacin; Cp=ciprofloxacin; AS=ampicillin-sulbactam.

Figure 4. Antimicrobic susceptibility of ESBL-negative (n=251) and –positive (n=216) *K. oxytoca.*

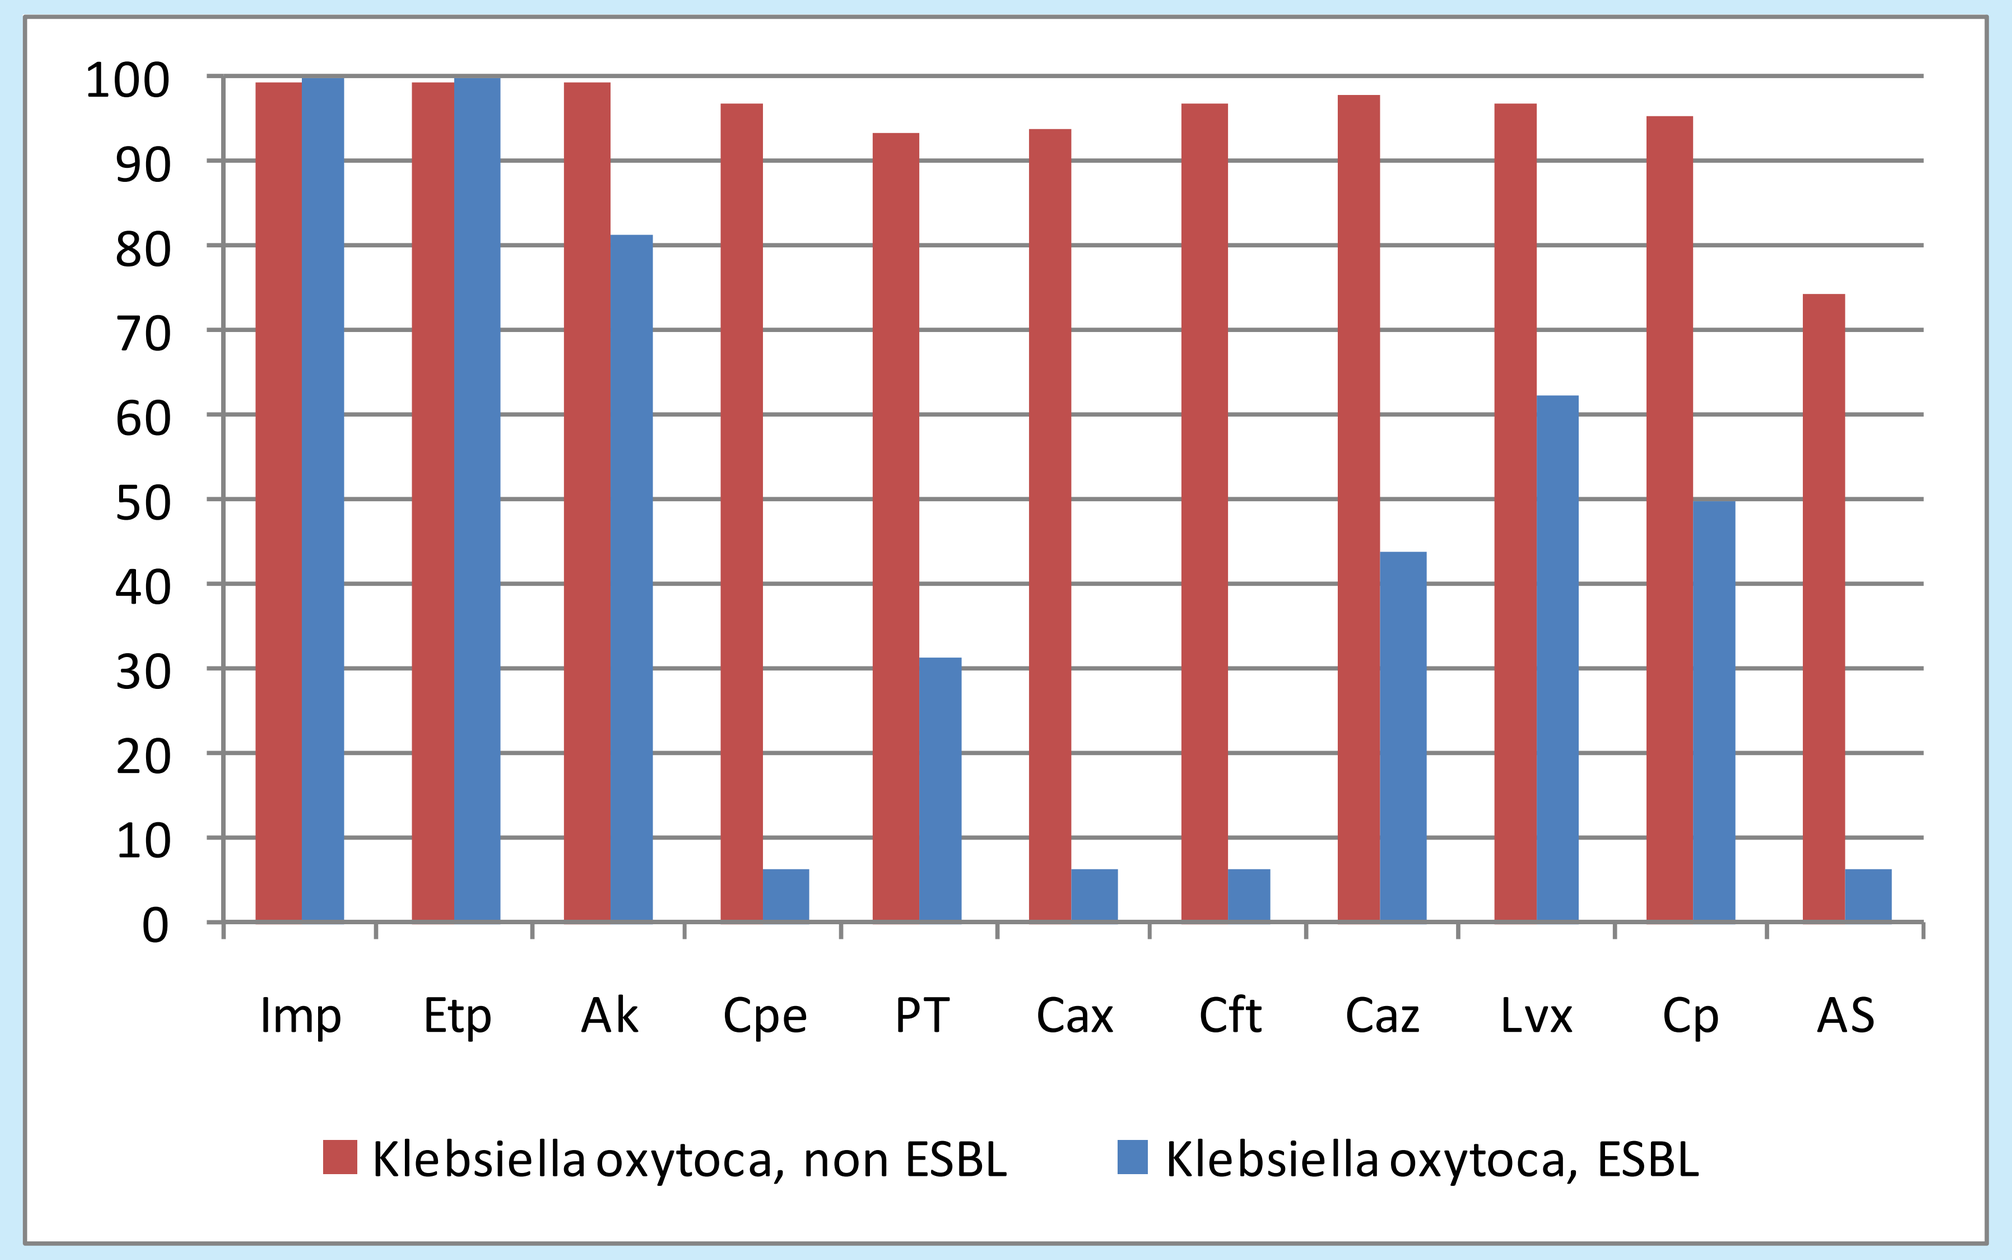
100
90
80
70
60
50
40
30
20
10
0
Imp
Etp
Ak
Cpe
PT
Cax
Cft
Caz
Lvx
Cp
AS
Klebsiella oxytoca, non ESBL
Klebsiella oxytoca, ESBL

Imp=imipenem; Etp=ertapenem; Ak=amikacin; Cpe=cefepime; PT=piperacillin-tazobactam; Cax=ceftriaxone; Cft=cefotaxime; Caz=ceftazidime; Lvx=levofloxacin; Cp=ciprofloxacin; AS=ampicillin-sulbactam.

Figure 5. Antimicrobic susceptibility of ESBL-negative (n=262) and –positive (n=14) *P. mirabilis.*

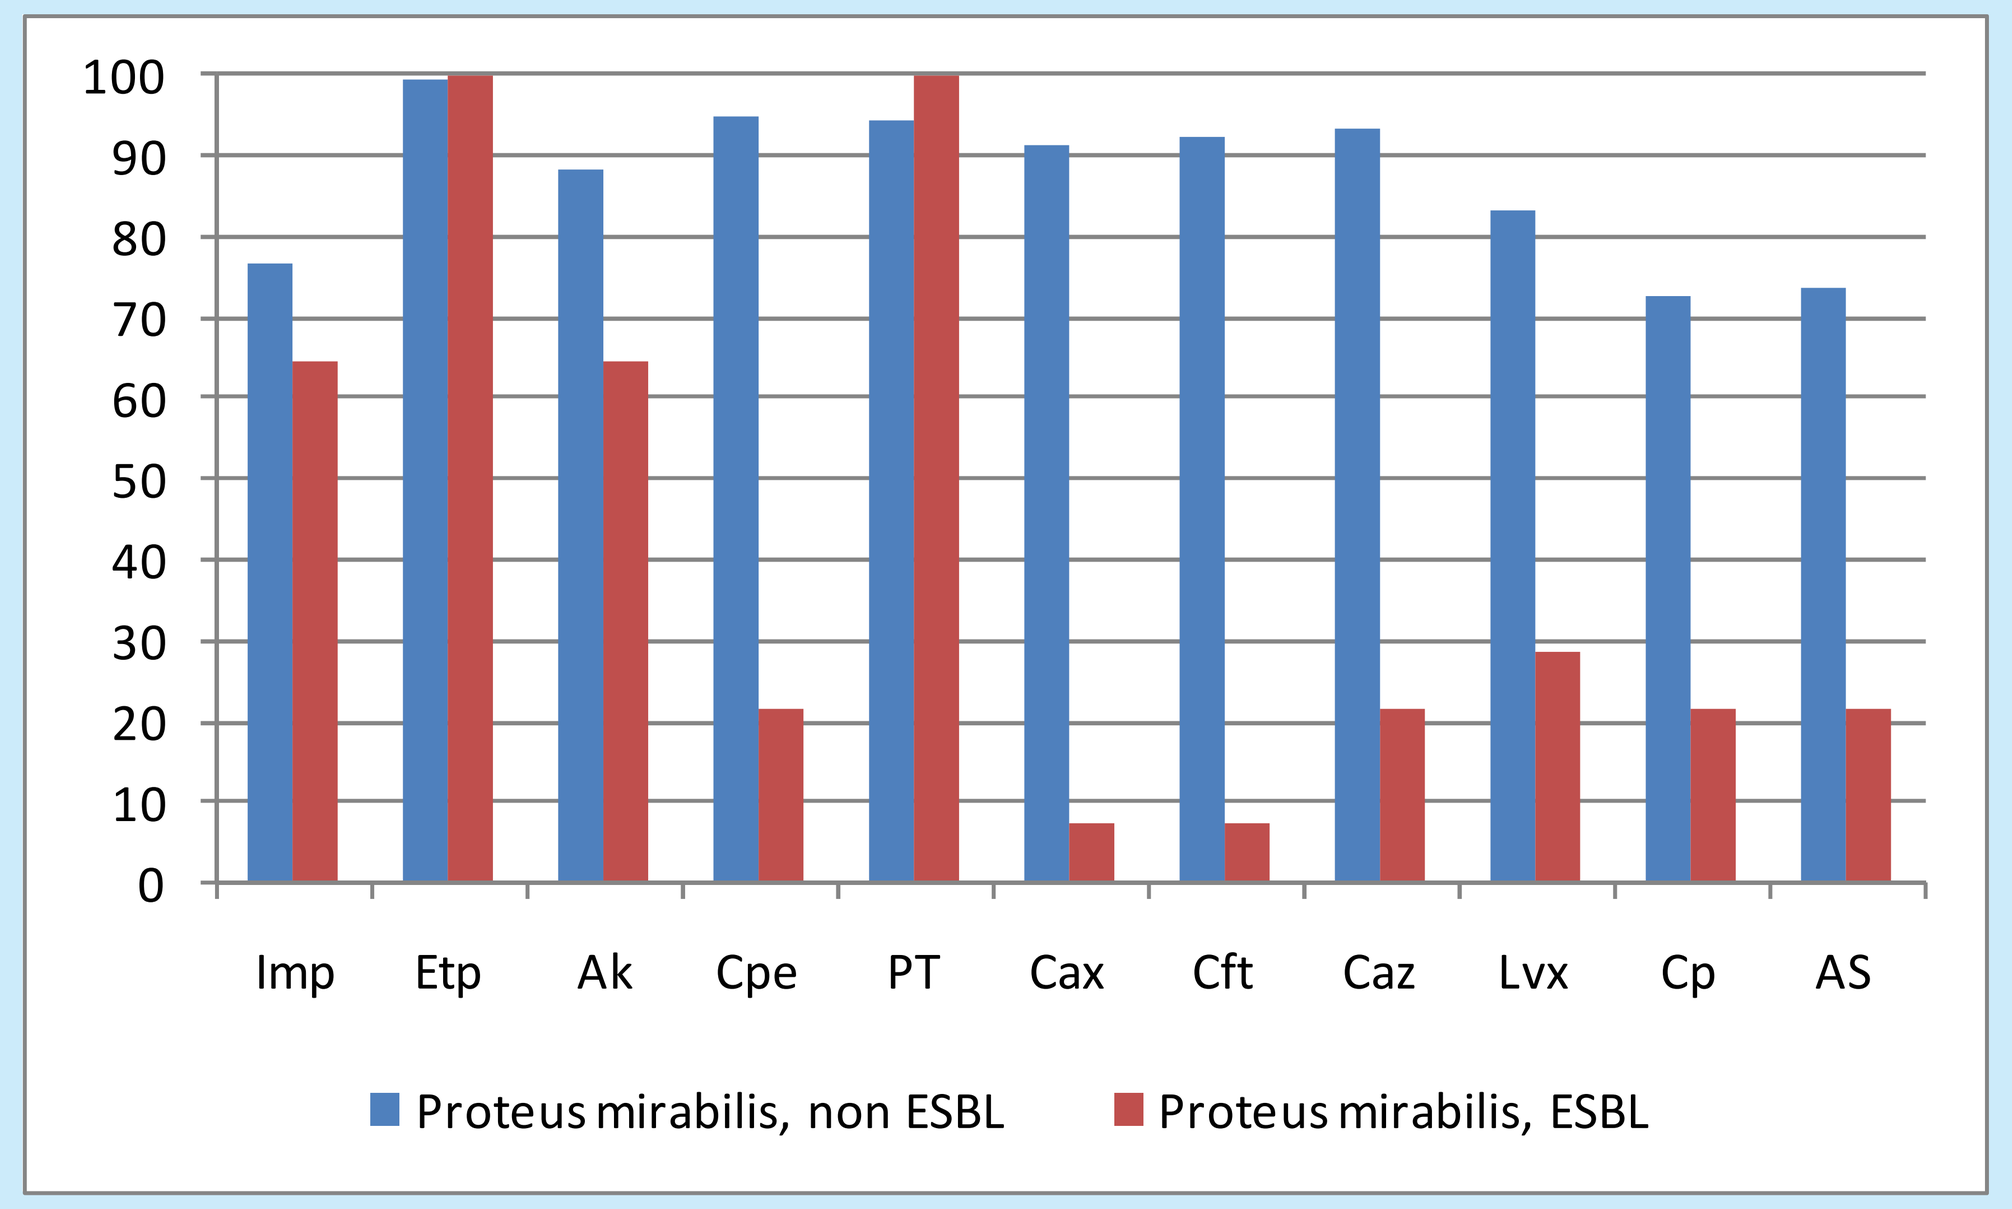
100
90
80
70
60
50
40
30
20
10
0
Imp
Etp
Ak
Cpe
PT
Cax
Cft
Caz
Lvx
Cp
AS
Proteus mirabilis, non ESBL
Proteus mirabilis, ESBL

Imp=imipenem; Etp=ertapenem; Ak=amikacin; Cpe=cefepime; PT=piperacillin-tazobactam; Cax=ceftriaxone; Cft=cefotaxime; Caz=ceftazidime; Lvx=levofloxacin; Cp=ciprofloxacin; AS=ampicillin-sulbactam.

Conclusions

- Only 3 drugs (imipenem, ertapenem and amikacin) had percent susceptibility levels ≥90% against *E. coli*, which represented nearly half (49.9%) of the aerobic pathogens found in IAI in Europe.
- Only 2 drugs (imipenem and ertapenem) had percent susceptibility levels ≥90% against *K. pneumoniae*, which represented 11.2% of the aerobic pathogens likely to be found in IAI in Europe.
- All drugs in this study were <80% effective *in vitro* at their respective susceptible breakpoints against *P. aeruginosa.*
- ESBL-positive rates in IAI in Europe were nearly 12% and 22% for *E. coli* and *K. pneumoniae*, respectively, and approached 9% of the aerobic gram-negative IAI pathogens overall. With ESBL-positive isolates usually being resistant to multiple drugs, physicians are faced with a dwindling choice of options for empiric therapy covering the most likely causative pathogens of IAI. Unless ESBL-positive organisms have been ruled out by laboratory testing, the most effective agents appear to be the carbapenems or amikacin for empiric therapy of IAI in Europe.