P 644

Rapid Molecular Detection of Carbapenemase-producers in *Enterobacteriaceae* Including the Newly Described NDM-1 Carbapenemase (TEST 2009-2010)

C. Lascols[1], M. Hackel[1], S. Hawser[2], R. Badal[1], S Bouchillon[1], D. Hoban[1], M. Dowzicky[3]

[1]International Health Management Associates, Schaumburg, IL, USA
[2]IHMA Europe Sàrl, Epalinges, Switzerland
[3]Pfizer Inc., Collegeville, PA, USA

IHMA, Inc.
2122 Palmer Dr.
Schaumburg, IL
60173
Tel: 847.303.5003
Fax: 847.303.5601

Revised Abstract

Background: β-lactamases conferring resistance to carbapenems, such as metallo-β-lactamases (MBLs), KPC and class D β-lactamases (e.g. OXA-48) have emerged in *Enterobacteriaceae*. Moreover, the latest threat in gram-negative resistance is the recent emergence of NDM-1 multidrug-resistant isolates. A fast multiplex-PCR for the detection of all these versatile enzymes was designed in this study.

Methods: A total of 343 recent carbapenem-resistant *Enterobacteriaceae* isolates from 2009 – 2010 were investigated for the presence of KPC, OXA-48, VIM and NDM-1. A rapid molecular test was developed using multiplex-PCR and previously described primers for the KPC, OXA-48 and VIM genes. NDM-1 specific primers were designed for identification of the NDM-1 gene. Minimum inhibitory concentrations (MICs) were determined using broth microdilution method following CLSI guidelines.

Results: The 343 isolates were collected from multiple infection sources and originated from various countries in Europe (43.1%), Latin America (30.3%), and North America (19%), the remaining were from Middle East (5%), Asia (2.3%) and Africa (0.3%). The majority of isolates included *K. pneumoniae* (74.3%), *E. coli* (16.3%), *K. oxytoca* (4.1%), *E. cloacae* (4.1%), and *E. aerogenes* (1.2%). Of the 343 isolates, 135 isolates (39%) carrying at least one of these carbapenemases were identified. Among the carbapenemase-producers, 114 (84.5%) were KPC-positive isolates, followed by a significantly lower proportion for other genes: 13 (9.6%) VIM, 7 (5.2%) OXA-48 and 1 (0.7%) NDM-1. All the VIM and OXA-48-producers were isolated in Europe and in the Middle East. KPC-producers were isolated in Europe (45.6%), North America (28.1%), Latin America (15.8%) and Middle East (10.5%). The new NDM-1 gene was identified in a *K. pneumoniae* isolate from South Africa, from a urinary tract infection (UTI) in a 39-year old male patient. This multi-drug resistant isolate, carrying an ESBL (CTX-M15) and an AmpC β-lactamase (CMY-2), was only susceptible to colistin.

Conclusions: Carbapenem resistance has become a potential health care issue, intensified by the rapid emergence of KPC carbapenemases and the newly described NDM-1 carbapenemase. Molecular epidemiological studies are crucial for monitoring the dissemination of all these carbapenemase-producers worldwide.

Introduction

The most common carbapenemases reported in *Enterobacteriaceae*, are KPC, OXA-48 and more rarely the VIM metallo-β-lactamase [1].

The latest threat in gram-negative resistance is the recent emergence of NDM-1 producing multidrug-resistant isolates which were first identified in India [2], and rapidly disseminated worldwide. Monitoring the prevalence of these enzymes is essential. In this study, we evaluated isolates from the Tigecycline Evaluation and Surveillance Trial (TEST) for the following carbapenemase genes: bla_{KPC}, $bla_{OXA\text{-}48}$, bla_{VIM} and $bla_{NDM\text{-}1}$ by multiplex-PCR.

Materials & Methods

Bacterial strains

- 343 meropenem-resistant (meropenem MICs ≥ 2 mg/L), [3]) *Enterobacteriaceae* isolates from 2009-2010 collected through the Tigecycline Evaluation and Surveillance Trial (TEST) were screened for carbapenemase genes.
- Strains were isolated in the following regions: Europe (43.1%), Latin America (30.3%), North America (19%), Middle East (5%), Asia (23%), and Africa (0.3%),
- Isolates came from blood (25.7%), urine (19.2%), respiratory (16.9%), body fluids (14.9%) and other (23.3%) sources.

Susceptibility testing

- Minimum inhibitory concentration (MIC) testing followed criteria established by the Clinical Laboratory Standards Institute (CLSI) [4] using custom broth microdilution panels supplied by TREK (TREK Diagnostic Systems, Cleveland, OH).

Molecular detection

- DNA extraction: suspension from overnight colonies was heated for 5 min at 95˚C and immediately frozen at -20˚C for at least 5 min in 96 wells-plates

Table 1: Oligonucleotide primers used in this study [5, 6]

Target	Primer name	Sequence 5' to 3'	PCR product size (bp)
VIM	VIMF	GATGGTGTTTGGTCGCATA	390
	VIMR	CGAATGCGCAGCACCAG	
KPC	KPCF	TGTCACTGTATCGCCGTC	1,011
	KPCR	CTCAGTGCTCTACAGAAAACC	
OXA-48	OXA-48F	GCTTGATCGCCCTCGATT	281
	OXA-48R	GATTTGCTCCGTGGCCGAAA	
NDM-1	NDM-1F	CCGTATGAGTGATTGCGGCG	779
	NDM-1R	GCCCAATATTATGCACCCGG	

Table 2: Qiagen Multiplex PCR-Reagents

Components	Description of components	Final Concentration
2X QIAGEN Multiplex PCR Master Mix (6 mM $MgCl_2$)	-Hot Start Taq® DNA Polymerase -Multiplex PCR buffer -dNTP Mix	1X (3 mM $MgCl_2$)
10X Primer Mix (2 µM each primer)	-Primers Mix (4 sets of primers)	0.2 µM
RNase-free water	-	-
Template DNA	-bacterial suspension	-

Figure 1: PCR results illustration

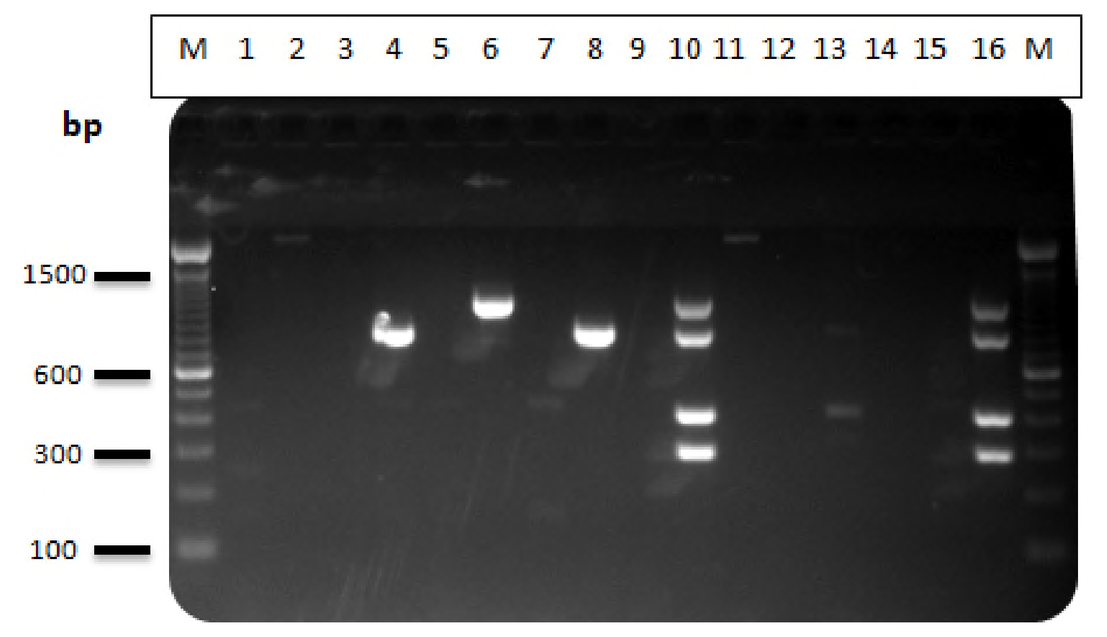
M 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 M
bp
1500
600
300
100

Legend:

1, 2, 3, 5, 7, 9, 11, 12, 13, 14 and 15: carbapenemase-negative isolates

4 and 8: NDM-1-producers

6: KPC-producer

10 and 16: positive control mix: $bla_{OXA\text{-}48}$ (281 bp), bla_{VIM} (390 bp), $bla_{NDM\text{-}1}$ (779 bp) and bla_{KPC} (1,011 bp)

Further characterization of isolates via the Check-Points system (Check MDR CT 101, Check-Points, The Netherlands) after extraction with the QIAamp DNA Mini Kit using the QIAcube instrument (Qiagen, Valencia, CA) was performed for the NDM-1-producer.

References

1. Nordmann, P. and A. Carrer. 2010. *Carbapenemases in enterobacteriaceae*. Arch Pediatr. 17 Suppl 4: S154-62.
2. Kumarasamy, K.K., et al. 2010. *Emergence of a new antibiotic resistance mechanism in India, Pakistan, and the UK: a molecular, biological, and epidemiological study.* Lancet Infect. Dis. 10: 597-602.
3. Clinical and Laboratory Standards Institute. (2010) Performance Standards for Antimicrobial Susceptibility Testing, CLSI Document M100-S20-U. Wayne, PA.
4. Clinical Laboratory Standards Institute. 2008. Methods for Dilution Antimicrobial Susceptibility Tests for Bacteria That Grow Aerobically; Approved Standard—Eighth Edition, CLSI Document M7-A8. 2008: Wayne, PA.
5. Yigit, H., et al. 2001. *Novel carbapenem-hydrolyzing beta-lactamase, KPC-1, from a carbapenem-resistant strain of Klebsiella pneumoniae*. Antimicrob. Agents Chemother. 45: 1151-61.
6. Dallenne, C., et al. 2010. *Development of a set of multiplex PCR assays for the detection of genes encoding important beta-lactamases in Enterobacteriaceae*. J. Antimicrob. Chemother. 65: 490-5.

Acknowledgements

We gratefully acknowledge the contributions of the investigators, laboratory personnel, and all members of the Tigecycline European Surveillance Trial program group. This study was sponsored by Pfizer Inc.

Results

Figure 2: Geographical distribution of 343 meropenem non-susceptible *Enterobacteriaceae* isolates.

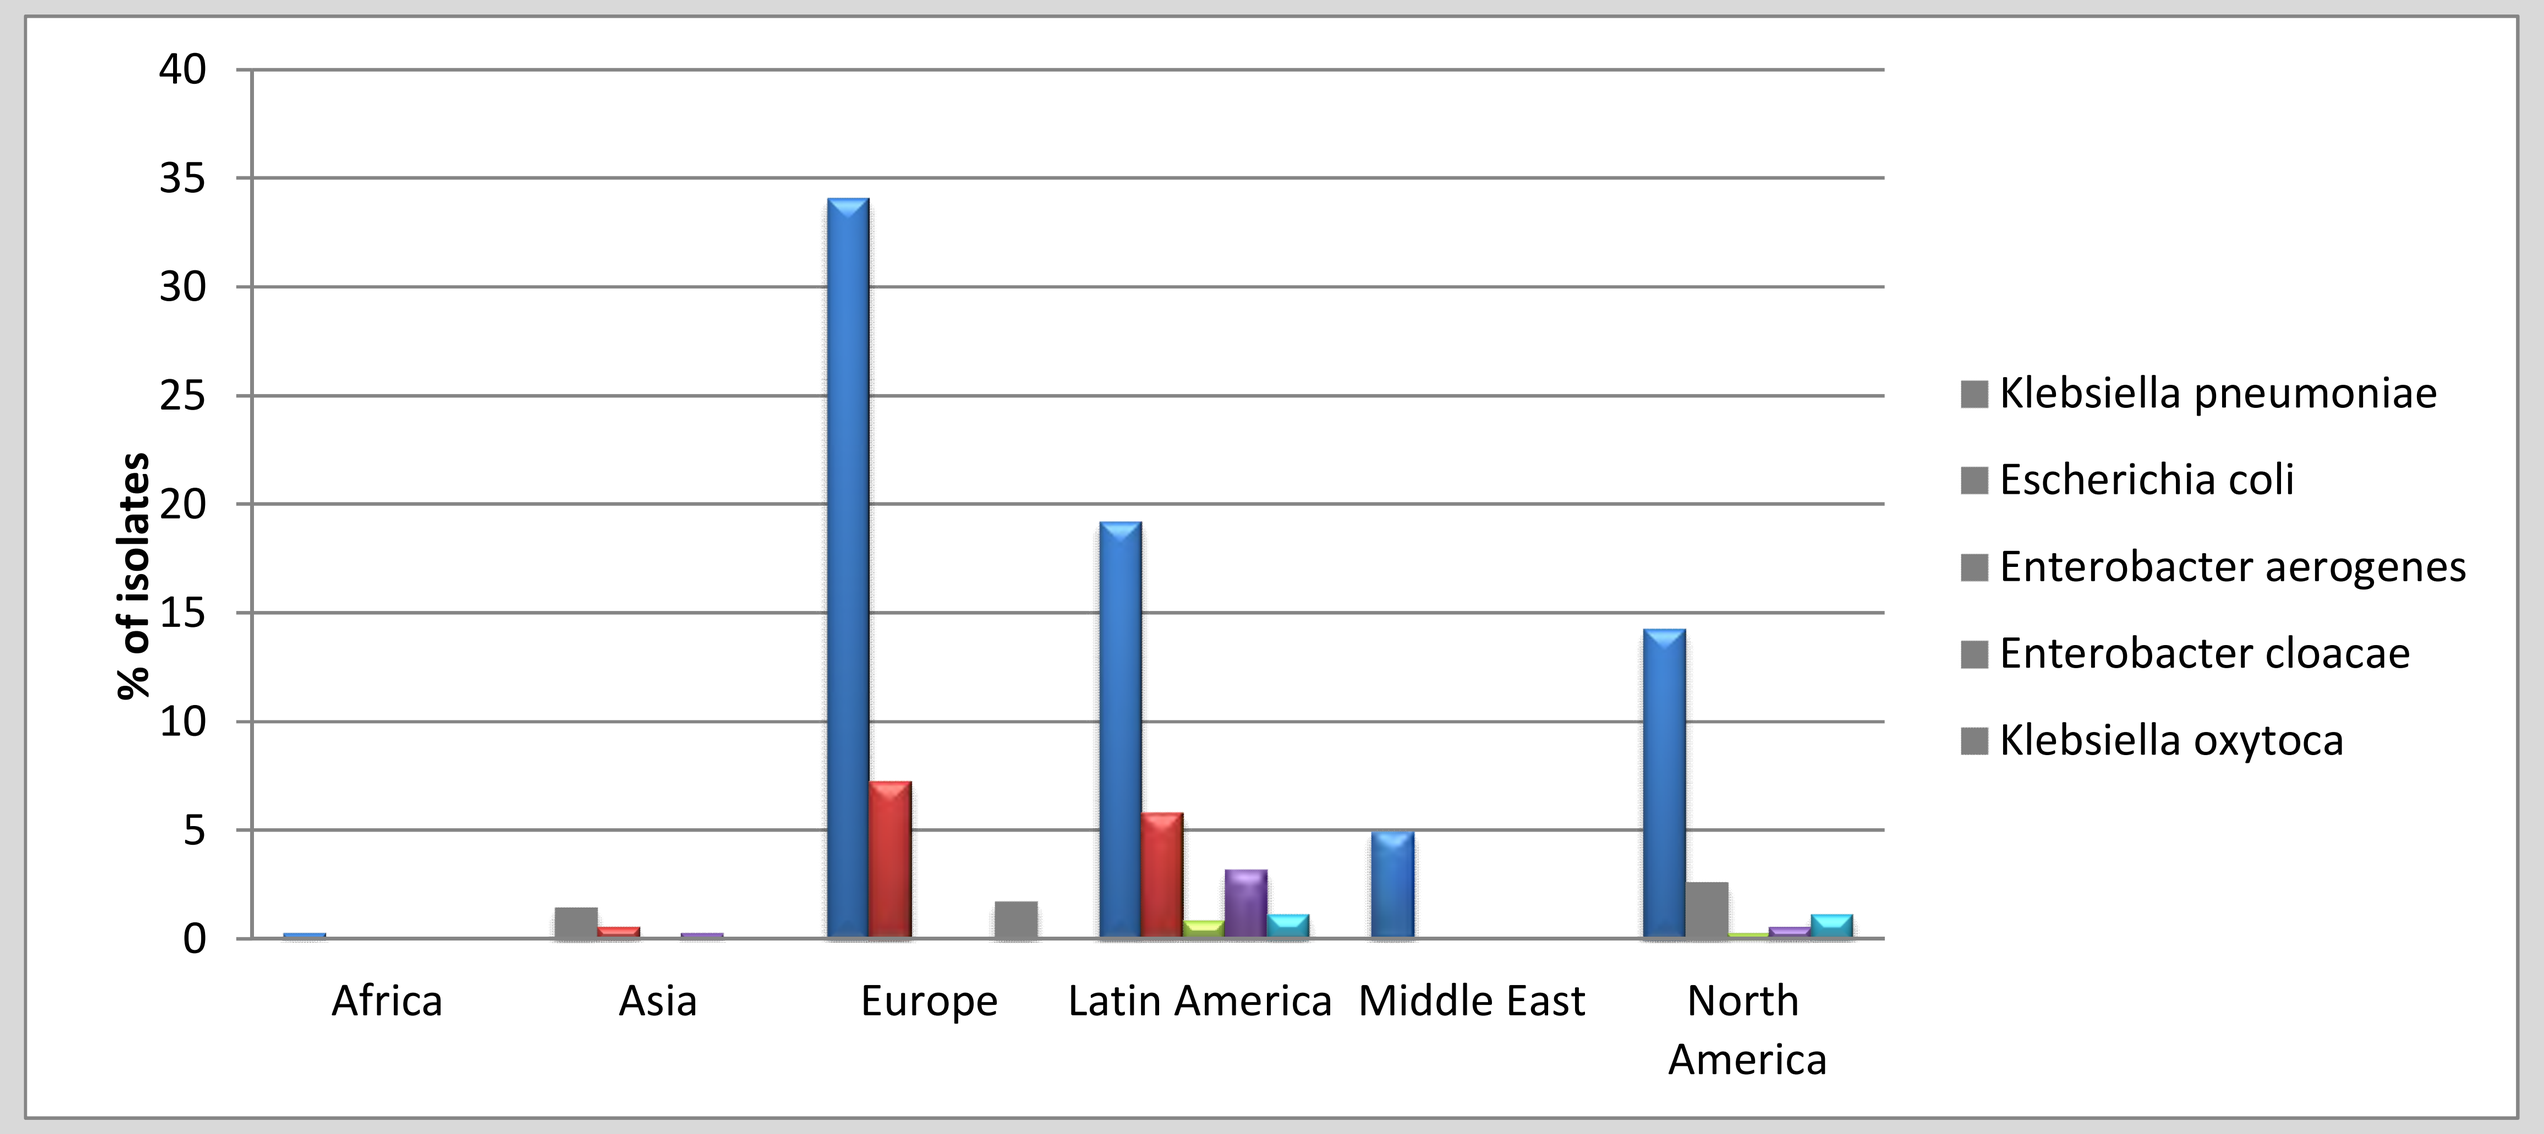
40
35
30
25
20
15
10
5
0
% of isolates
Africa
Asia
Europe
Latin America
Middle East
North America
Klebsiella pneumoniae
Escherichia coli
Enterobacter aerogenes
Enterobacter cloacae
Klebsiella oxytoca

-*K. pneumoniae* (n=255, 74.3%), -*E. coli* (n=56, 16.3%), -*K. oxytoca* (n=14, 4.1%), -*E. cloacae* (n=14, 4.1%), -*E. aerogenes* (n=4, 1.2%)

Figure 3: Distribution of enzymes among the 135 carbapenemase-producers

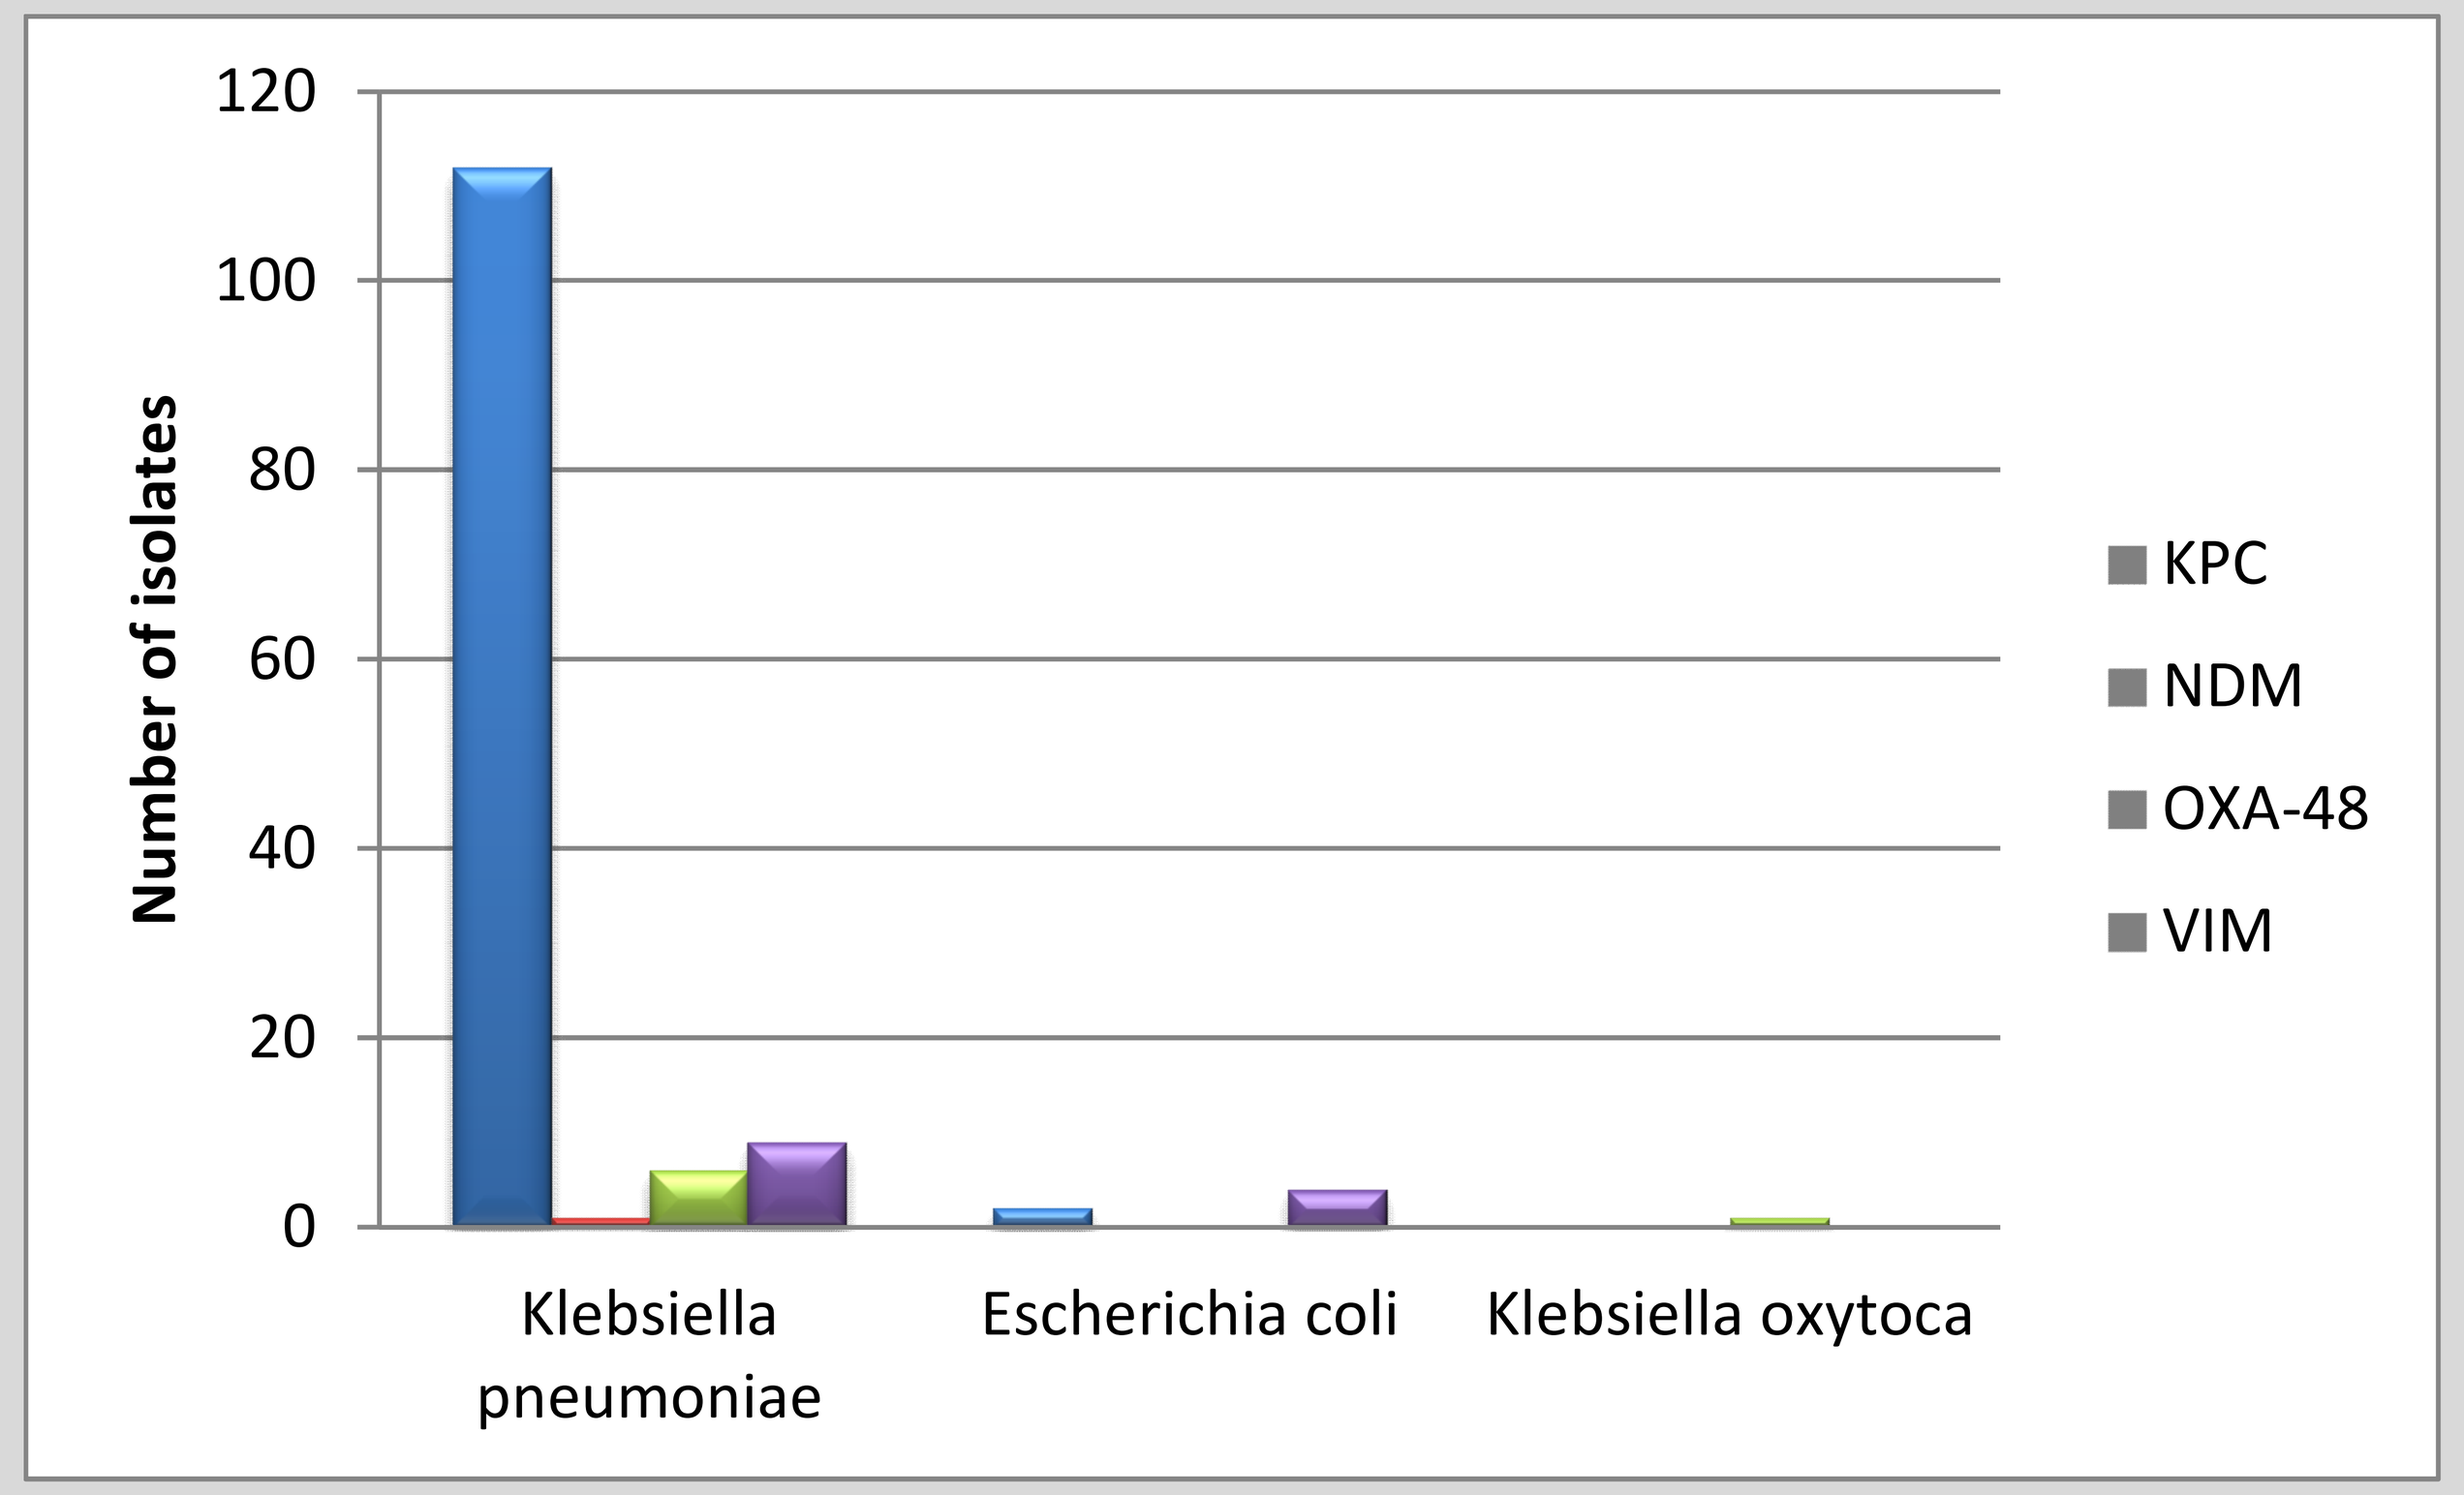
120
100
80
60
40
20
0
Number of isolates
Klebsiella pneumoniae
Escherichia coli
Klebsiella oxytoca
KPC
NDM
OXA-48
VIM

KPC-producers (84.5%): *K. pneumoniae (n=112, 98%) and E. coli (n=2, 2%)*
NDM-1-producer (0.7%): *K. pneumoniae (n=1, 100%)*
OXA-48-producers (5.2%): *K. pneumoniae (n=6, 86%) and K. oxytoca (n=1, 14%)*
VIM-producers (9.6%): *K. pneumoniae (n=9, 69%) and E. coli (n=4, 31%)*

Figure 4: Geographical distribution of enzymes among the 135 carbapenemase-producers.

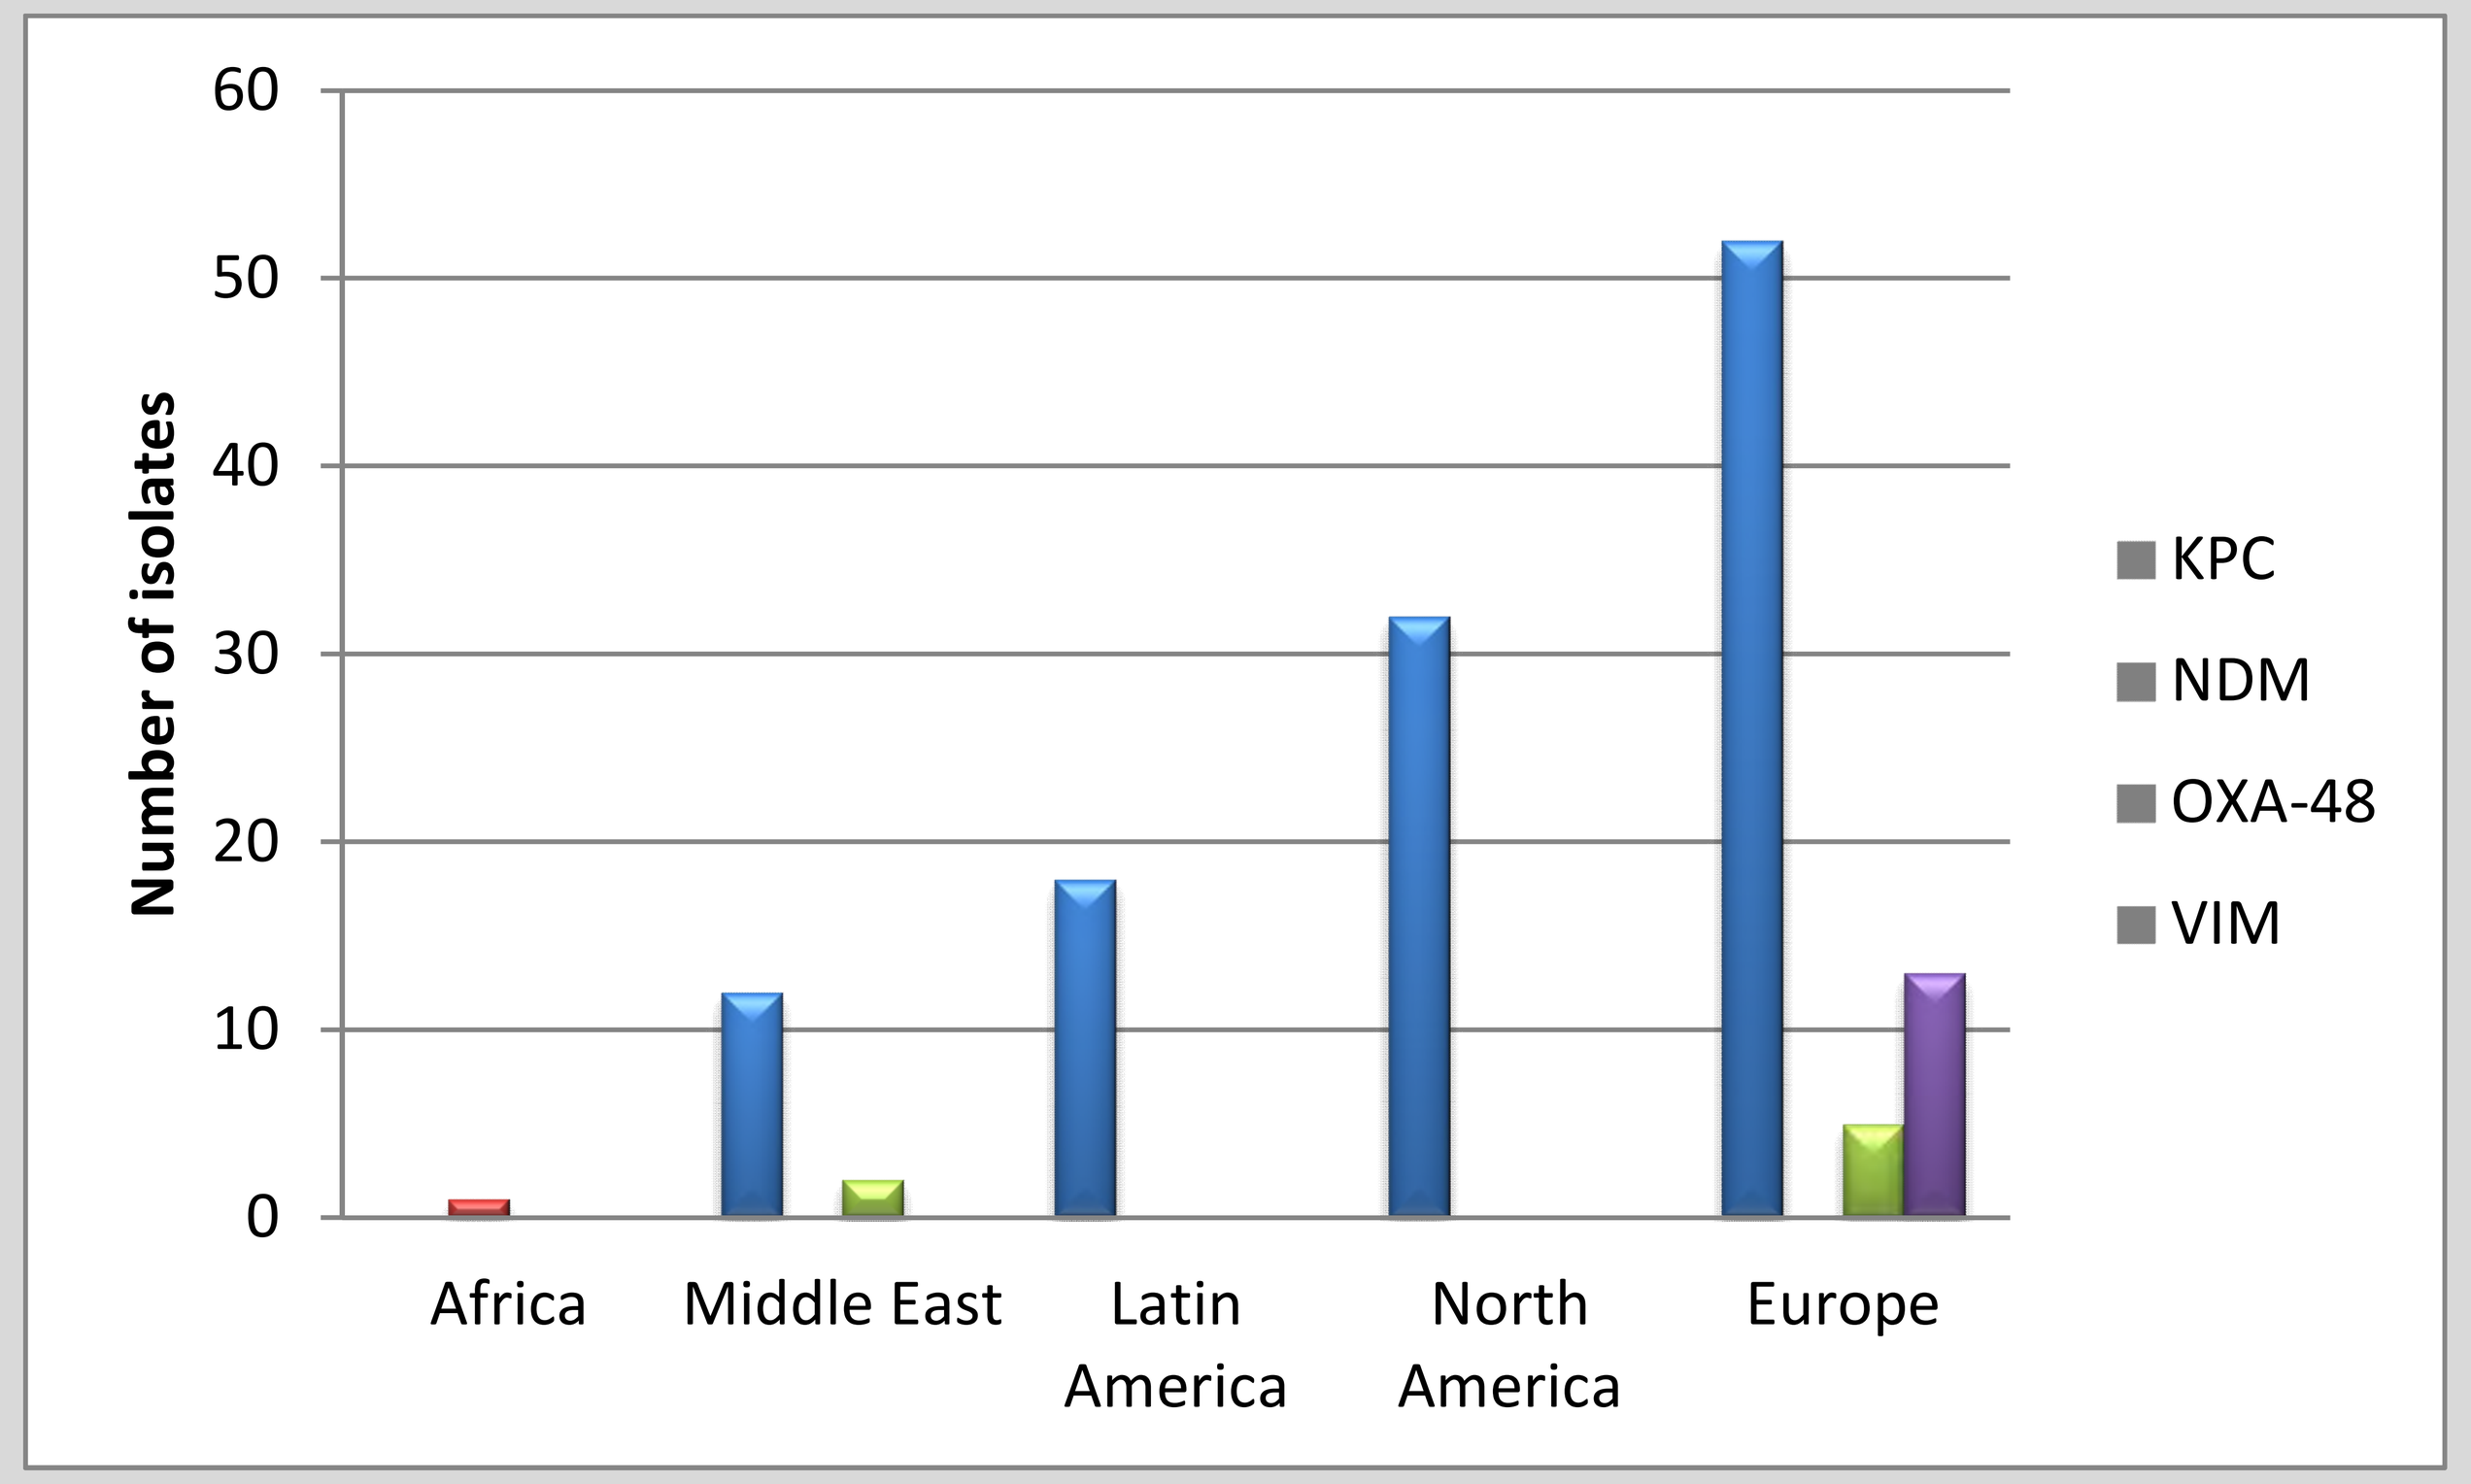
60
50
40
30
20
10
0
Number of isolates
Africa
Middle East
Latin America
North America
Europe
KPC
NDM
OXA-48
VIM

KPC-producers: -Europe (n=52, 46%), *Greece (n=25, 48%), Italy (n=15, 29%), Poland (n=9, 17%), Spain (n=2, 4%), Germany (n=1, 2%);*
-United States (n=32, 28%); -Latin America (n=18, 18%); -Middle East (n=12, 11%).
VIM-producers: -Europe (n=13)
OXA-48-producers: -Europe, *Turkey (n=5)*; -Middle East (n=2)
NDM-1-producer: -South Africa (n=1)

Figure 5: Meropenem MIC distribution among the 135 carbapenemase-producers.

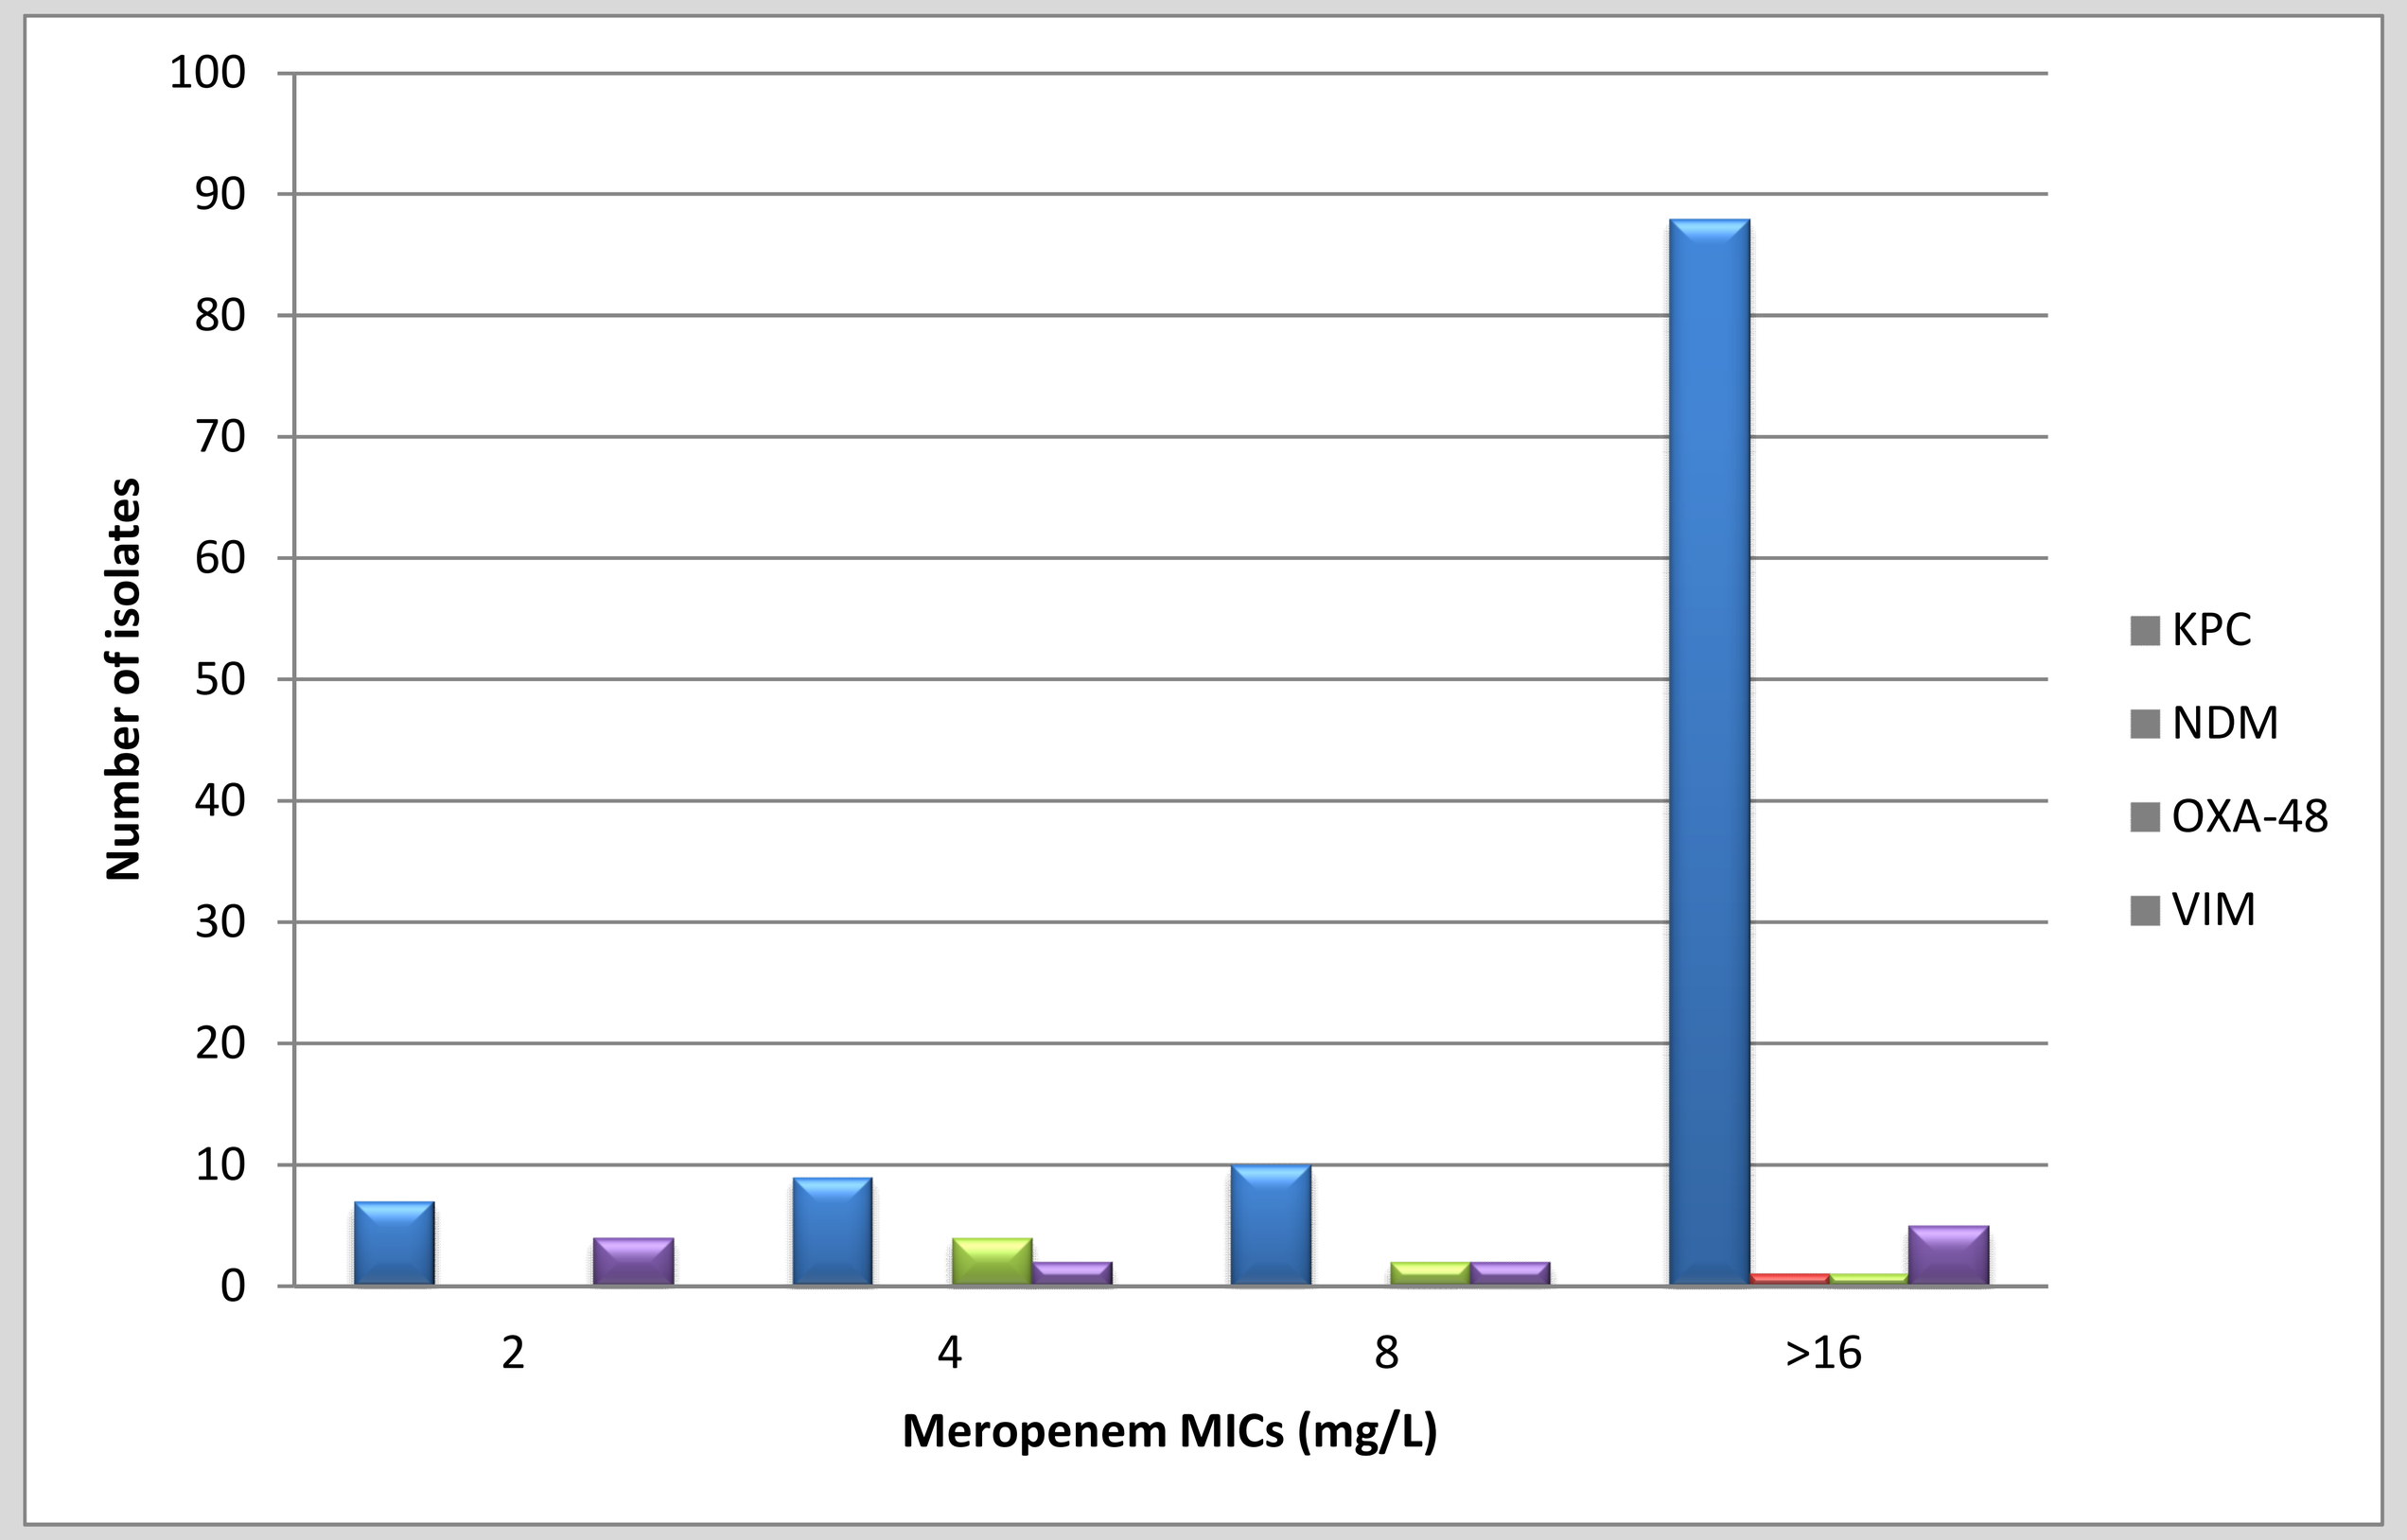
100
90
80
70
60
50
40
30
20
10
0
Number of isolates
2
4
8
>16
Meropenem MICs (mg/L)
KPC
NDM
OXA-48
VIM

Table 3: Characteristics of the *K. pneumoniae* NDM-1-producer (Kp4.14)

PATIENT	
Age	39
Sexe	Male
Source	Urine
Department	Surgery General

MICs (mg/L)	PTZ	CTX	CAZ	CPM	MERO	AK	LEVO	TIG	COL
Kp4.14	>128	>64	>32	>32	>16	>64	>8	2	1

β-lactamases	ESBL	AmpC
Kp4.14	CTX-M-15	CMY-2

Conclusions

- Of the species to produce carbapenemases, *K. pneumoniae* (74.3%) and *E. coli* (16.3%) were the most common.
- KPC was the most common carbapenemase identified in this study, with bla_{KPC} found in 84.5% of isolates producing a carbapenemase.
- Molecular epidemiological studies remain crucial for monitoring the dissemination of all these carbapenemases worldwide.
- Rapid and simple molecular tests that can easily be implemented in clinical laboratories for detection of these growing enzymes are essential.