G3-774

Distribution of *Streptococcus pneumoniae* serotypes among populations in Europe

M. HACKEL[1], C. LASCOLS[1], S. BOUCHILLON[1], J. JOHNSON[1], B. HILTON[2], D. MORGENSTERN[2], J. PURDY[2]

[1]International Health Management Associates, Schaumburg, IL, USA
[2]Pfizer Inc., Collegeville, PA, USA

IHMA, Inc.
2122 Palmer Dr.
Schaumburg, IL 60173
Tel: 847.303.5003
Fax: 847.303.5601

Revised Abstract

Pneumococcal disease (PD) remains a leading cause of mortality and morbidity worldwide in children and adults. Data on serotype distribution of PD is needed to monitor and assess the pneumococcal burden of disease in the population, especially in the setting of pneumococcal vaccine immunization programs. In this study, serotyping of *Streptococcus pneumoniae* isolates collected in 22 countries in Europe was undertaken to document serotype distribution in children and adults of different age groups**. Methods:** The serotypes of 480 *S. pneumoniae* sterile body site clinical isolates collected through the Tigecycline Evaluation and Surveillance Trial (TEST) between 2004 and 2009 were determined. A PCR-based assay was used to identify 28 specific pneumococcal serotypes. All PCR results were confirmed by Quellung reactions. **Results:** The most common serotypes in Europe by age, N (%) are shown in the following table.

		Age in years		
Serotype	**Total N (%)**	**0-15**	**16-65**	**>65**
3	41(8.6)	4(4.3)	19(9.6)	18(9.5)
14	37(7.5)	15(15.9)	6(3.0)	16(8.4)
1	35(7.3)	7(7.6)	20(10.2)	8(4.2)
7F	35(7.3)	5(5.4)	14(7.1)	16(8.4)
19A	31(6.5)	9(9.8)	8(4.1)	14(7.4)
6A	28(5.8)	7(7.6)	12(6.1)	9(4.7)
6B	27(5.6)	6(6.5)	13(6.6)	8(4.2)
23F	24(5.0)	8(8.7)	9(4.6)	7(3.7)
9V	23(4.8)	2(2.2)	12(6.1)	9(4.7)
22F	21(4.4)	6(6.5)	8(4.1)	7(3.7)
4	20(4.2)	2(2.2)	6(3.0)	12(6.3)
19F	19(4.0)	7(7.6)	2(1.0)	10(5.1)
11A	18(3.8)	0	13(6.6)	5(2.6)
8	13(2.7)	0	5(2.5)	8(4.2)
18C	12(2.5)	3(3.3)	5(2.5)	4(2.1)
9N	12(2.5)	3(3.3)	5(2.5)	4(2.1)
12F	11(2.3)	0	5(2.5)	6(3.2)
35F	7(1.5)	1(1.1)	3(1.5)	4(2.1)
5	6(1.3)	3(3.3)	1(0.5)	2(1.1)
All Others	60(12.5)	6(6.5)	31(15.7)	23(12.1)
Total N	**480**	94	197	190

Conclusions: This analysis of organisms collected through the TEST program demonstrates that serotype 3 (8.6%) was the most common serotype isolated during 2004-09 in the 14 countries in Western Europe and 8 countries in Eastern Europe combined. The emerging serotype 19A (6.5%) and serotype 6A (5.8) were the fifth and sixth most common isotypes collected. These three serotypes (3, 19A and 6A) accounted for more than 20% of all *S. pneumoniae* clinical isolates, with 70.4% of all isolates collected among serotypes 1, 3, 4, 5, 6A, 6B, 7F, 9V, 14, 18C, 19A, 19F, 23F. The data, although limited in numbers, suggests that a significant burden of pneumococcal disease in these regions can be attributed to these pneumococcal serotypes.

Introduction

Streptococcus pneumoniae represents a leading cause of mortality and morbidity worldwide in children and adults, causing pneumonia, bacteremia and meningitis. The burden of disease is especially notable in the very young and those over 65 years of age [1]. *S. pneumoniae* has at least 92 known serotypes, although not all are associated with disease. Various serotypes vary greatly in invasiveness and virulence, and population distributions differ among age groups [2]. Additionally, most antibiotic-resistant pneumococcal infections are attributed to five specific serotypes (6B, 9V, 14, 19F, and 23F) [3]. A number of multivalent vaccines have been developed, using a variety of serotypes. Data on serotype distribution of pneumococcal disease is needed to monitor and assess the pneumococcal burden of disease in the population, especially in the setting of pneumococcal vaccination immunization programs. The Tigecycline European Surveillance Trial (TEST) is an on-going global surveillance program begun in 2004. In this analysis, serotyping of *Streptococcus pneumoniae* isolates collected in 22 countries in Europe through the TEST program was undertaken to document serotype distribution in children and adults of different age groups.

Materials & Methods

480 *S. pneumoniae* isolates from sterile body sites from the TEST program were included in this analysis. All isolates were collected from 2004-2009 in 14 countries in Western Europe (Austria n=2 , Belgium n=43, Denmark n=27, Finland n= 10, France n=56, Germany n=56, Ireland n=3, Italy n=95, Portugal n=4, Spain n=79, Sweden n=14, Switzerland n=13, the Netherlands n=8, and the United Kingdom n=10) and 8 countries in Eastern Europe (Croatia n=6, the Czech Republic n=9, Greece n=15, Hungary n=8, Latvia n=8, Lithuania n=4, Poland n=6, and Slovenia n=4). Sterile body sites included body fluids; cardiovascular system, including blood; central nervous system; lymph; ovary; lung; respiratory; sinus; and bone. Based on the seven- reaction sequential multiplex PCR described previously [4], we devised a PCR using only four reactions to identify the pneumococcal serotype, followed by confirmation via the conventional Quellung reaction. This PCR-based approach is based on serotype-specific genes within the capsular polysaccharide synthesis genes (the cps locus) of SPN. While this scheme could not detect all serotypes without doing a traditional serotyping checkerboard, it served to streamline the project and reduce cost by reducing the number of Quellung reactions needed. Using primers targeted at different loci within the cps gene, multiplex PCR reactions were set up in a sequential fashion, including an internal cps control. When the PCR assigned isolates to a serotype subset, conventional serotyping was performed based on the subset type. All PCR serotype results were confirmed by Quellung reaction. Isolates that were negative by PCR were serotyped by Quellung reactions alone. Pneumococcal antisera were purchased from Statens Serum Institute (Copenhagen, Denmark).

Minimum inhibitory concentrations (MICs) were determined by the Clinical and Laboratory Standards Institute (CLSI) recommended broth microdilution testing method [5] using panels prepared at IHMA, Inc. (Schaumburg, IL). MIC interpretive criteria followed published guidelines of the European Committee on Antimicrobial Susceptibility Testing (EUCAST) [6].

References

1. Obaro S, and Adegbola R. 2002. *The pneumococcus: carriage, disease and conjugate vaccines.* J. Med. Microbiol. 51:98-104.
2. Whitney CG, Farley MM, Hadler J, et al. 2003. *Decline in invasive pneumococcal diease after the introduction of protein-polysaccharide conjugate vaccine*. N. Engl. J. Med. 348:1737-1746.
3. Kyaw MH, Lynfield R, Schaffner W, et al. 2006. *Effect of introduction of the pneumococcal conjugate vaccine on drug-resistant Streptococcus pneumoniae.* N. Engl. J. Med. 354:1455-1463.
4. Pai R, Gertz RE, Beall B. 2006. *Sequential multiplex PCR approach for dertermining capsular serotypes of Streptococcus pneumoniae isolates.* J. Clin. Microbiol. 44:124-131.
5. Clinical Laboratory Standards Institute. 2009. Methods for Dilution Antimicrobial Susceptibility Tests for Bacteria That Grow Aerobically; Approved Standards – 8th Edition. CLSI document M07-A8 Wayne, PA.
6. European Committee on Antimicrobial Susceptibility Testing (EUCAST). 2011. Breakpoint tables for interpretation of MICs and zone diameters, version 1.3, http://www.eucast.org.

Acknowledgements

We gratefully acknowledge the contributions of the investigators, laboratory personnel, and all members of the Tigecycline Evaluation and Surveillance Trial group. This study was sponsored by a grant from Pfizer, Inc.

Results

Figure 1. Serotypes of 420 *S. pneumoniae* from sterile body sites from Western Europe, 2004 – 2009 (all ages).

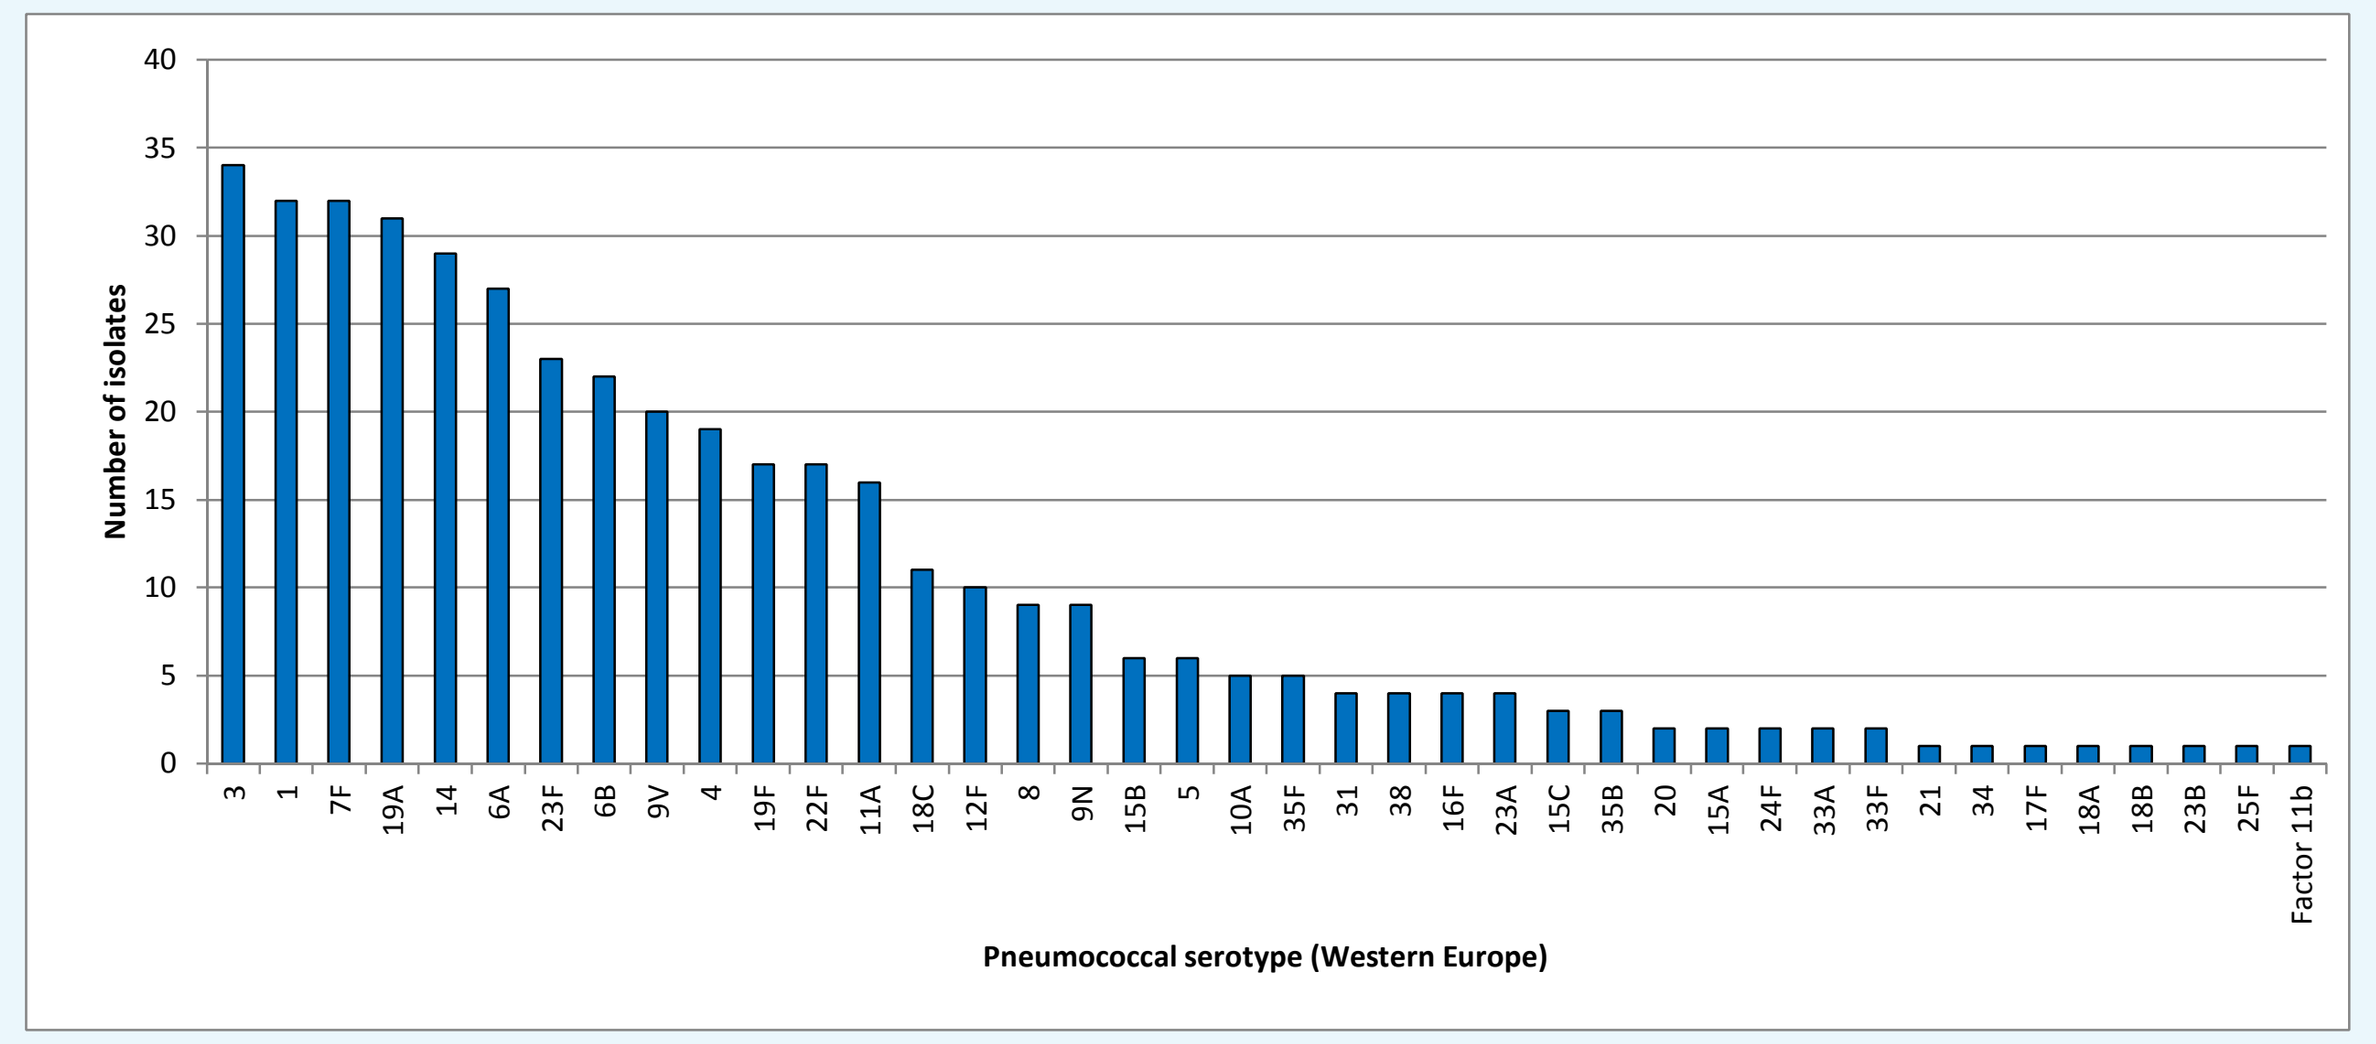
Number of isolates
Pneumococcal serotype (Western Europe)

Figure 2. Serotypes of 60 *S. pneumoniae* from sterile body sites in Eastern Europe, 2004 – 2009 (all ages).

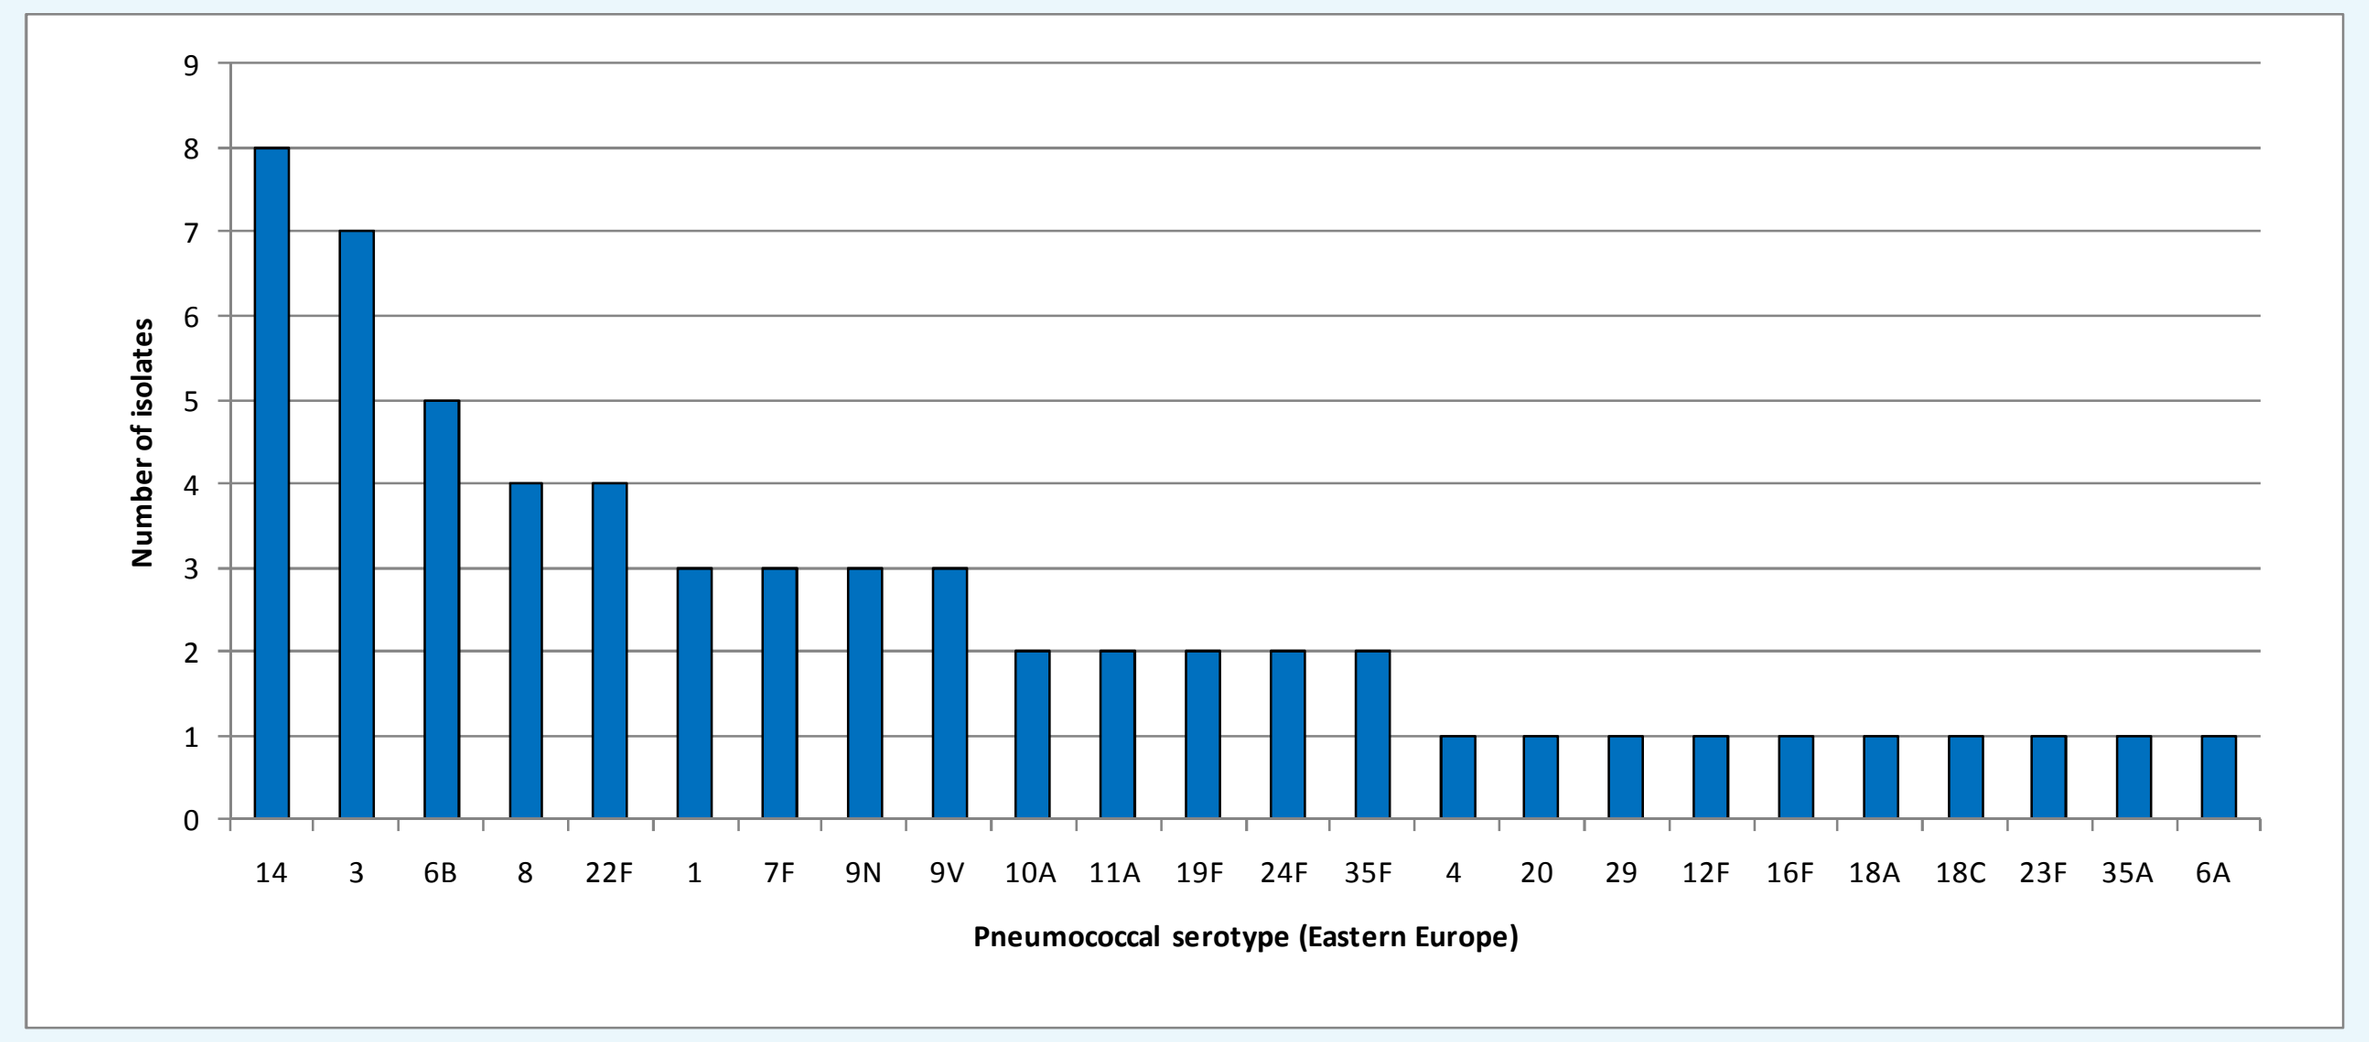
Number of isolates
Pneumococcal serotype (Eastern Europe)

Table 1. Age and serotype distribution of 420 *S. pneumoniae* isolates from Western Europe.

		Age in years				
Serotype	Total N (%)	<2	2-4	5-14	15-65	>65
3	34 (8.1)	1 (4.3)			16 (9.1)	17 (10.0)
1	32 (7.6)		2 (8.0)	4 (14.8)	18 (10.3)	8 (4.7)
7F	32 (7.6)			3 (11.1)	14 (8.0)	15 (8.8)
19A	31 (7.4)	3 (13.0)	3 (12.0)	3 (11.1)	8 (4.6)	14 (8.2)
14	29 (6.9)	7 (30.4)	3 (12.0)		5 (2.9)	14 (8.2)
6A	27 (6.4)	3 (13.0)	3 (12.0)	1 (3.7)	11 (6.3)	9 (5.3)
23F	23 (5.5)		3 (12.0)	5 (18.5)	8 (4.6)	7 (4.1)
6B	22 (5.2)	1 (4.3)		4 (14.8)	11 (6.3)	6 (3.5)
9V	20 (4.8)		1 (4.0)		12 (6.9)	7 (4.1)
4	19 (4.5)			1 (3.7)	6 (3.0)	12 (7.1)
19F	17 (4.0)	5 (21.7)	2 (8.0)		2 (1.1)	8 (4.7)
22F	17 (4.0)	1 (4.3)	1 (4.0)	3 (11.1)	6 (3.0)	6 (3.5)
11A	16 (3.8)				11 (6.3)	5 (2.9)
18C	11 (2.6)		1	1 (3.7)	5 (2.9)	4 (2.4)
12F	10 (2.4)				5 (2.9)	5 (2.9)
8	9 (2.1)				3 (1.7)	6 (3.5)
9N	9 (2.1)		1 (4.0)		5 (2.9)	3 (1.8)
5	6 (1.4)		3 (12.0)		1 (0.5)	2 (1.2)
15B	6 (1.4)	1 (4.3)			2 (1.1)	3 (1.8)
10A	5 (1.2)				2 (1.1)	3 (1.8)
35F	5 (1.2)				3 (1.7)	2 (1.2)
31	4 (1.0)				3 (1.7)	1 (0.6)
38	4 (1.0)		1 (4.0)		1 (0.5)	2 (1.2)
16F	4 (1.0)				2 (1.1)	2 (1.2)
23A	4 (1.0)				2 (1.1)	2 (1.2)
15C	3 (0.7)		1 (4.0)		2 (1.1)	
35B	3 (0.7)				2 (1.1)	1 (0.6)
20	2 (0.5)				1 (0.5)	1 (0.6)
15A	2 (0.5)				1 (0.5)	1 (0.6)
24F	2 (0.5)				1 (0.5)	1 (0.6)
33A	2 (0.5)				1 (0.5)	1 (0.6)
33F	2 (0.5)	1 (4.3)			1 (0.5)	
21	1 (0.2)			1(3.7)		
34	1 (0.2)					1 (0.6)
17F	1 (0.2)					1 (0.6)
18A	1 (0.2)			1(3.7)		
18B	1 (0.2)				1 (0.5)	
23B	1 (0.2)				1 (0.5)	
25F	1 (0.2)				1 (0.5)	
Factor 11b	1 (0.2)				1 (0.5)	
Total N	**420**	**23 (5.5)**	**25 (6.0)**	**27 (6.4)**	**175 (41.7)**	**170 (40.5)**

Table 2. Age and serotype distribution of 60 *S. pneumoniae* isolates from Eastern Europe.

		Age in years				
Serotype	Total N (%)	<2	2-4	5-14	15-65	>65
14	8 (13.3)	2 (33.3)	3 (50.0)		1 (4.5)	2 (10.0)
3	7 (11.7)	2 (33.3)	1 (16.7)		3 (13.6)	1 (5.0)
6B	5 (8.3)	1 (16.7)			2 (9.1)	2 (10.0)
8	4 (6.7)				2 (9.1)	2 (10.0)
22F	4 (6.7)			1 (16.7)	2 (9.1)	1 (5.0)
1	3 (5.0)			1 (16.7)	2 (9.1)	
7F	3 (5.0)			2 (33.3)		1 (5.0)
9N	3 (5.0)		2 (33.3)			1 (5.0)
9V	3 (5.0)			1 (16.7)		2 (10.0)
10A	2 (3.3)				2 (9.1)	
11A	2 (3.3)				2 (9.1)	
19F	2 (3.3)					2 (10.0)
24F	2 (3.3)				1 (4.5)	1 (5.0)
35F	2 (3.3)					2 (10.0)
4	1 (1.7)	1 (16.7)				
20	1 (1.7)					1 (5.0)
29	1 (1.7)					1 (5.0)
12F	1 (1.7)					1 (5.0)
16F	1 (1.7)				1 (4.5)	
18A	1 (1.7)				1 (4.5)	
18C	1 (1.7)			1 (16.7)		
23F	1 (1.7)				1 (4.5)	
35A	1 (1.7)				1 (4.5)	
6A	1 (1.7)				1 (4.5)	
Total N	**60**	**6 (10)**	**6 (10)**	**6 (10)**	**22 (36.7)**	**20 (33.3)**

Figure 3. Percent of isolates from Europe by serotype in 2004 compared to 2009 (all ages).

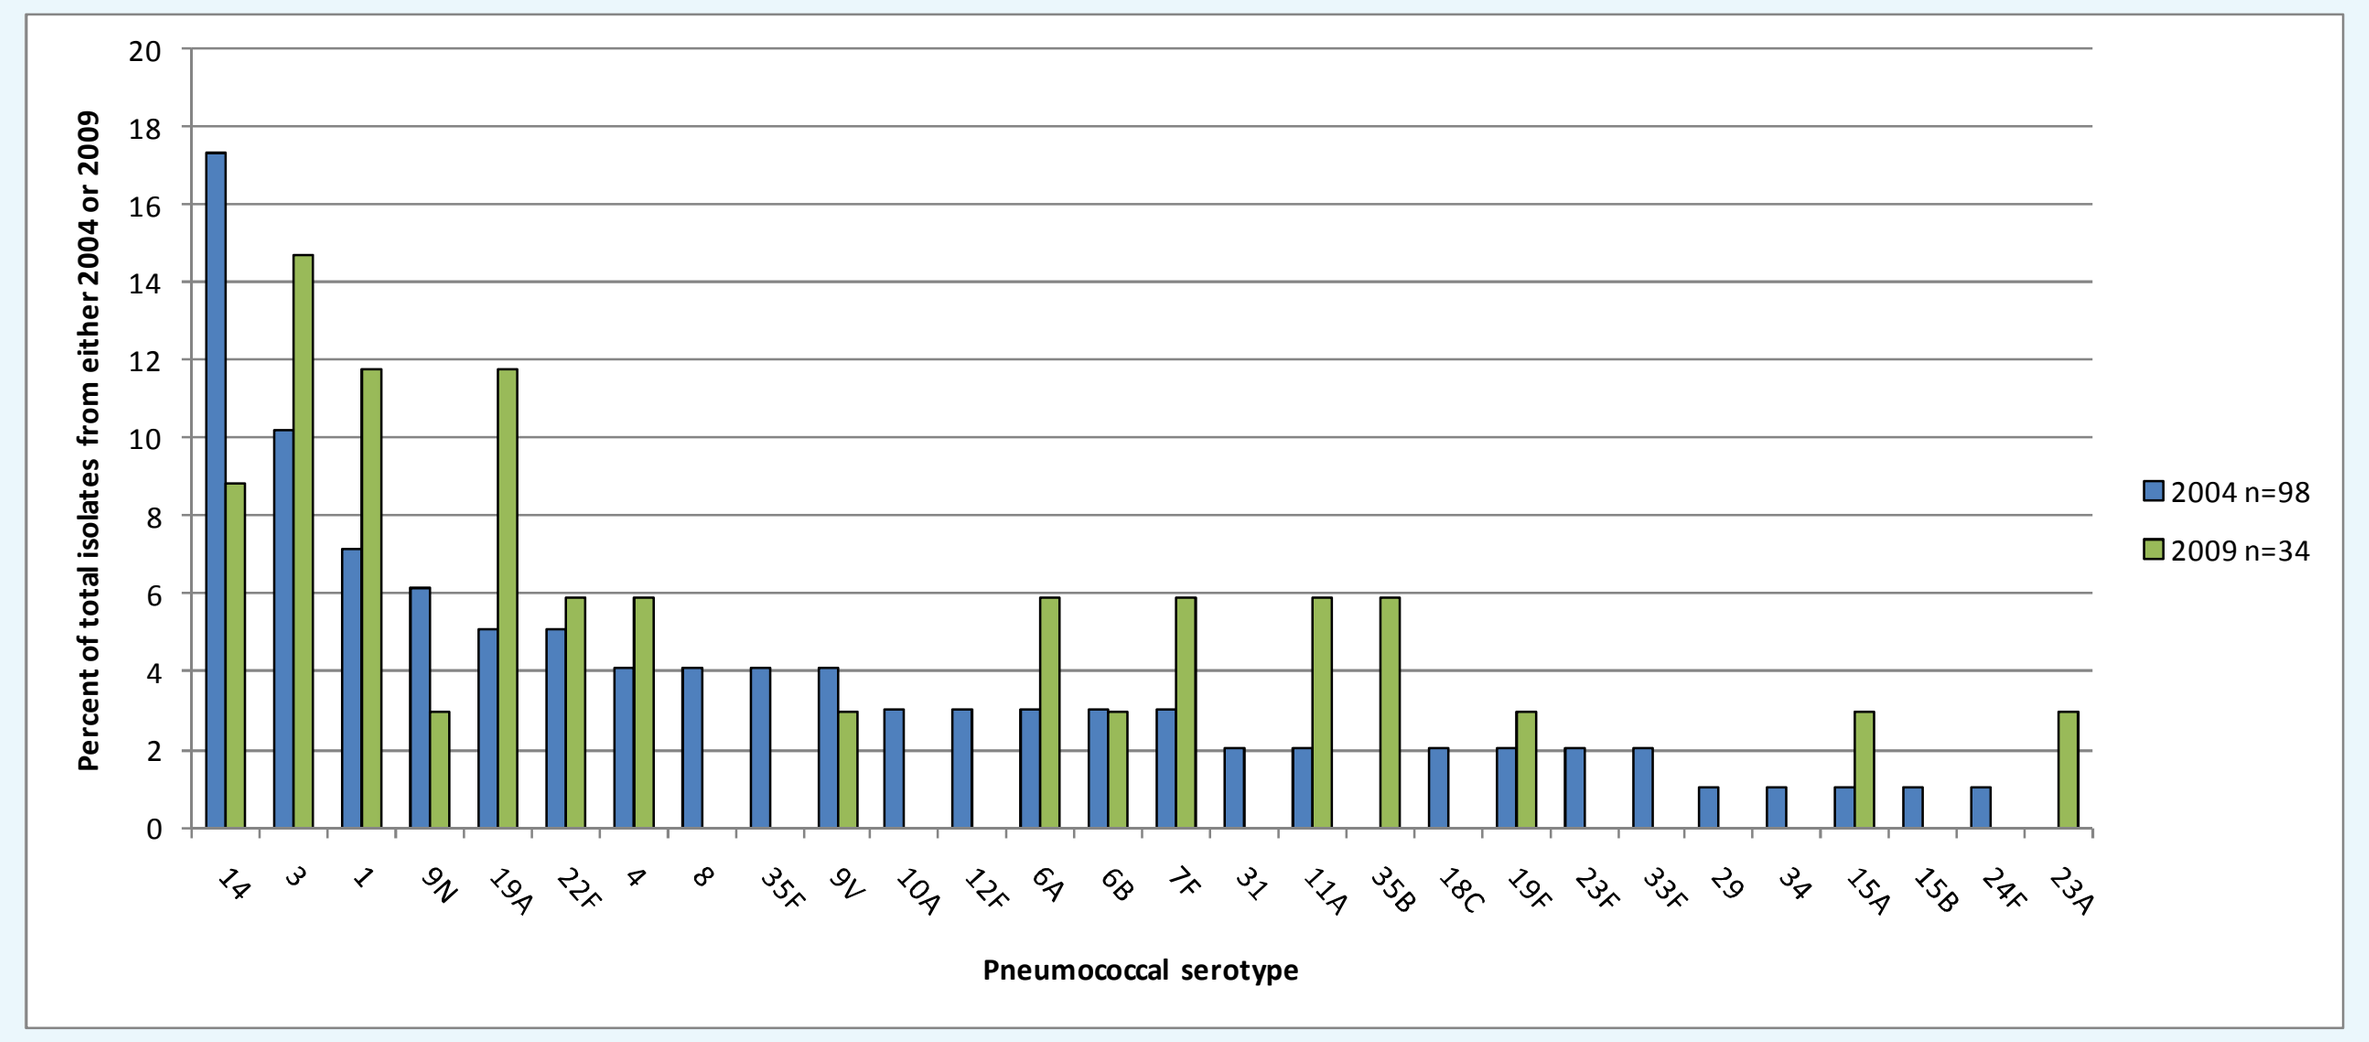
Percent of total isolates from either 2004 or 2009
Pneumococcal serotype
2004 n=98
2009 n=34

Figure 4. Resistance to erythromycin of select serotypes from Western Europe.

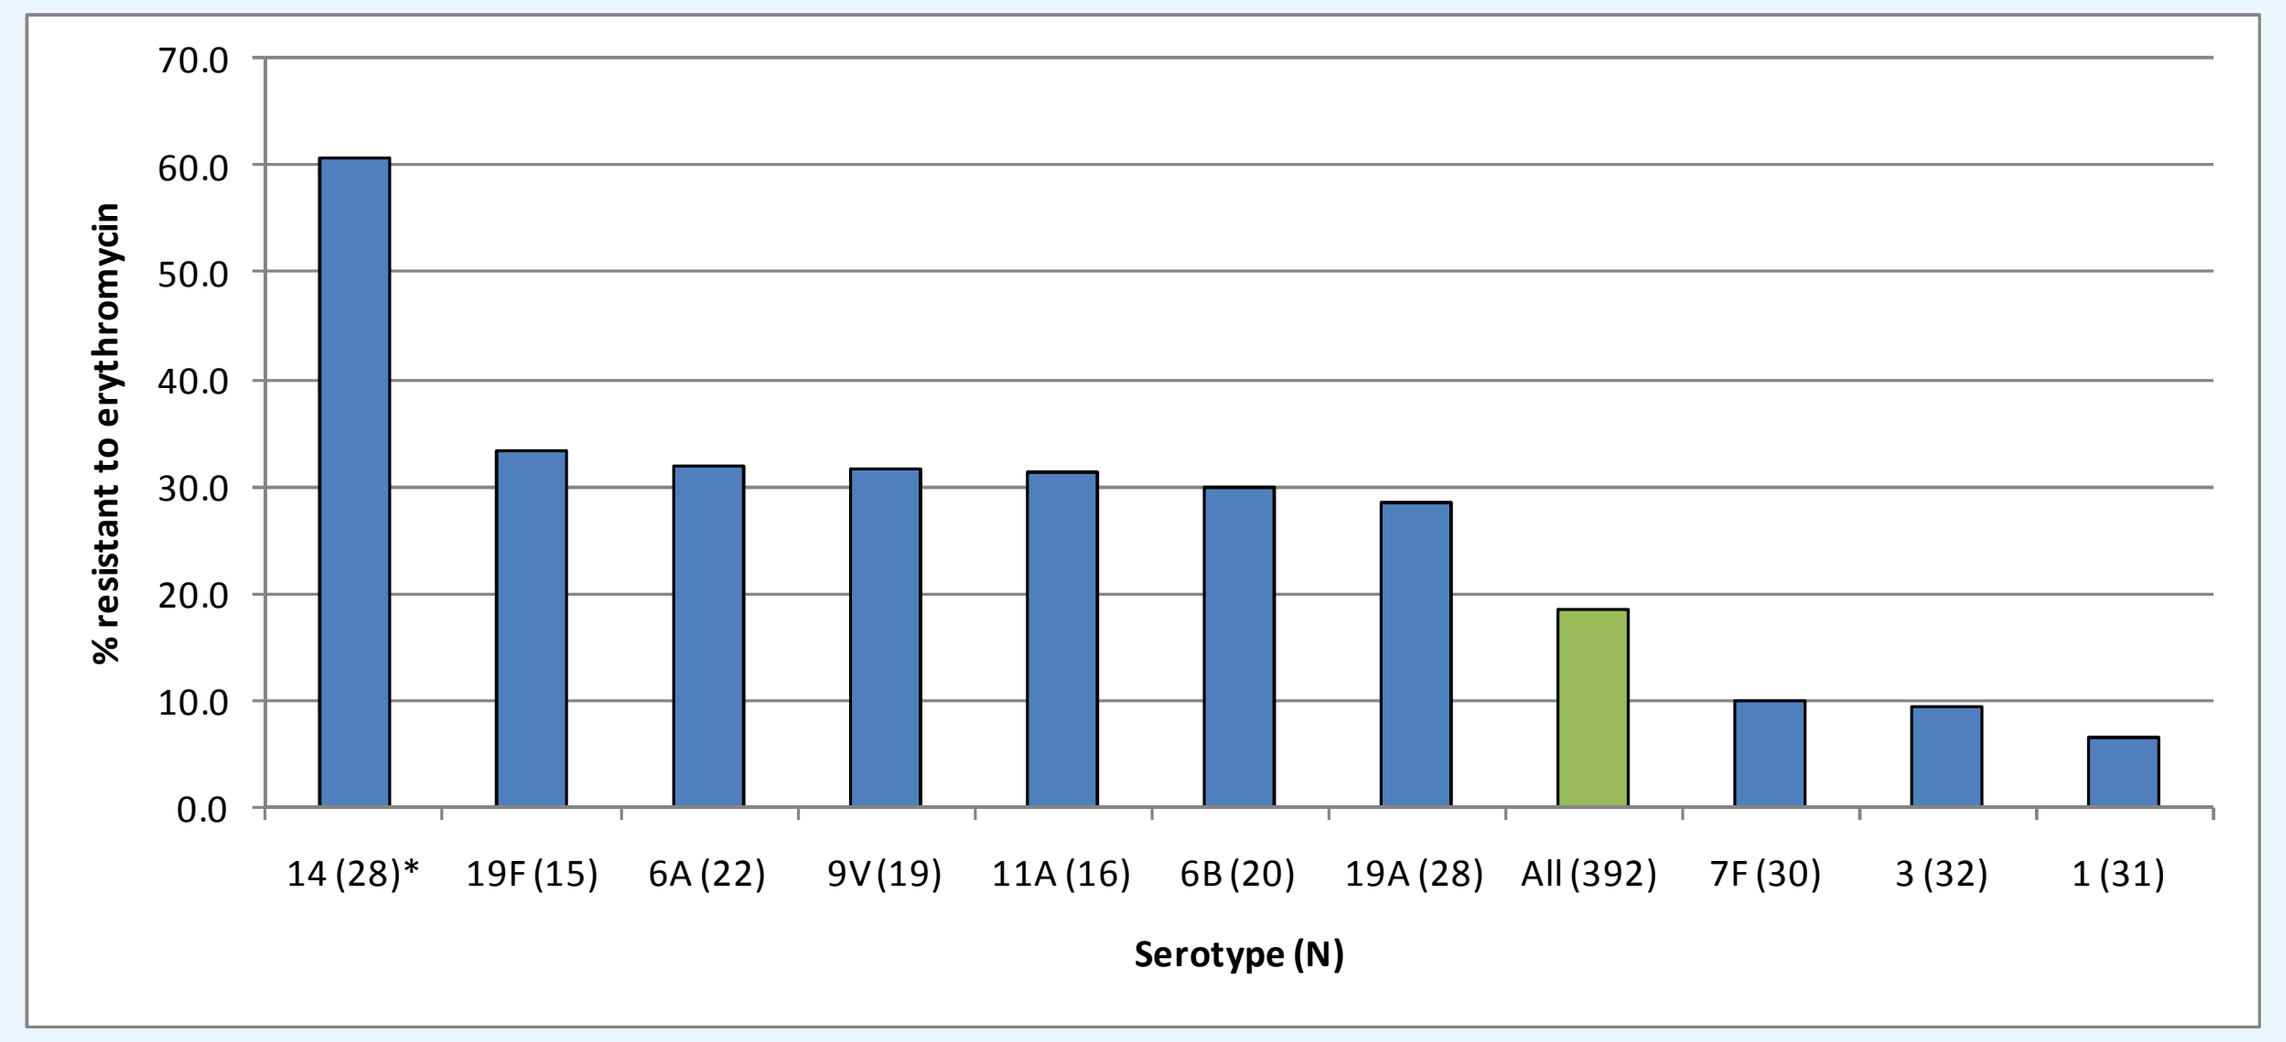
% resistant to erythromycin
14 (28)*
19F (15)
6A (22)
9V (19)
11A (16)
6B (20)
19A (28)
All (392)
7F (30)
3 (32)
1 (31)
Serotype (N)

Conclusions

- This analysis of organisms collected through the TEST program demonstrates that across all ages, serotypes 3 (8.1%), 1 (7.6%) and 7F (7.6%) were the most common serotypes isolated during 2004-09 in Western Europe. Serotypes 14 (13.3%) and 3 (11.7%) were the most common in Eastern Europe. Serotype distribution varied by age group, with 31% of European isolates from children under two years exhibiting serotype 14, and 17% exhibiting 19F. For adults over 15 years, serotype 3 was most common (9.6%), followed by serotypes 7F (7.8%) and 1 (7.2%).
- Antibiotic susceptibility testing showed serotype 14 to be associated with increased erythromycin resistance ($p<0.0001$, Fisher's exact test). Two isolates from Europe were levofloxacin resistant and none were penicillin resistant (data not shown).
- In this study, three emerging serotypes (3, 19A and 6A) account for 21.9% of all *S. pneumoniae* clinical isolates from Western Europe, and 13.3% (serotypes 3 and 6A only) from Eastern Europe for all ages. Overall, 70.4% of all serotypes collected were among serotypes 1, 3, 4, 5, 6A, 6B, 7F, 9V, 14, 18C, 19A, 19F, 23F. Conversely, the prevalence of serotypes 4, 6B, 9V, 14, 18C, 19F, 23F has been reduced to less than 34% overall.
- The serotype distribution and association of serotypes with antibiotic resistance highlights the need for ongoing monitoring of the seroepidemiology of this important pathogen.