GN061

Susceptibility of Meropenem Non-Susceptible *Enterobacteriaceae* from Asia to Tigecycline and Comparators: TEST 2009-2010

S. Hawser[1], D. Hoban[2], B. Johnson[2], S. Bouchillon[2], M. Dowzicky[3]

[1]IHMA Europe Sàrl, Epalinges, Switzerland
[2]International Health Management Associates, Inc., Schaumburg, IL, USA
[3]Pfizer Inc., Collegeville, PA, USA

IHMA, Inc.
2122 Palmer Dr.
Schaumburg, IL
60173
Tel: 847.303.5003
Fax: 847.303.5601

Revised Abstract

Background: The Tigecycline Evaluation and Surveillance Trial (TEST) monitors the activity of tigecycline and comparators against multiple pathogens collected worldwide. Such monitoring assists in investigating resistance rates either globally, regionally or by country. The current report describes susceptibility of *Enterobacteriaceae* from Asia, including meropenem non-susceptible isolates, to tigecycline and comparators. **Methods:** A total of 1,305 clinical isolates were collected from multiple infection sources in Asia during 2009-2010 of which 27 (2%) were non-susceptible to meropenem. Susceptibility testing was performed as per CLSI guidelines and interpreted using CLSI/FDA clinical breakpoints.

Results: Susceptibility of all and meropenem non-susceptible isolates were as follows.

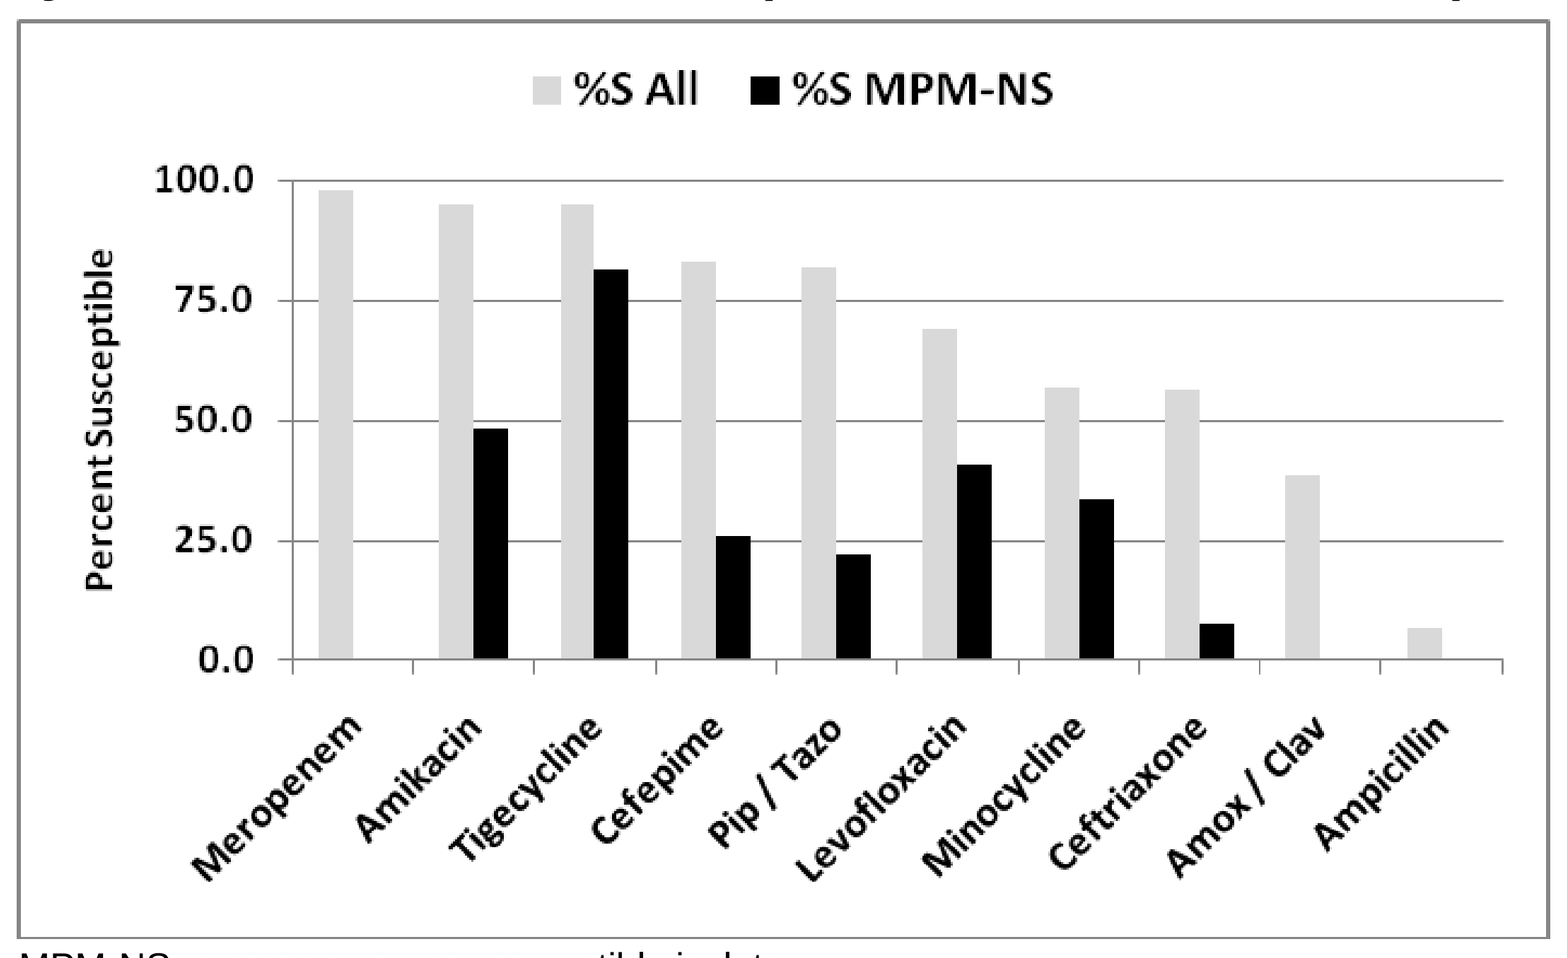

%S All
%S MPM-NS
Percent Susceptible
100.0
75.0
50.0
25.0
0.0
Meropenem
Amikacin
Tigecycline
Cefepime
Pip / Tazo
Levofloxacin
Minocycline
Ceftriaxone
Amox / Clav
Ampicillin

MPM-NS, meropenem non-susceptible isolates

Conclusions: Of the agents tested, only meropenem, amikacin and tigecycline had susceptibility >90% for the total pool of 1,305 isolates. However, against the sub-population of 27 meropenem non-susceptible isolates tigecycline was the only agent that had susceptibility >75%. While meropenem resistance is generally considered to be gradually increasing in *Enterobacteriaceae*, the promising activity of tigecycline against meropenem non-susceptible isolates is encouraging.

Introduction

There is tremendous variability of antimicrobial resistance not only in pathogens causing various clinical infections, in different geographic regions, but also over time. These situations make continuous surveillance of the extent and trends of antimicrobial resistance crucial to provide guidance in choosing optimal therapy. While most *Enterobacteriaceae* remain susceptible to the carbapenem class of antibiotics, non-susceptibility to these agents appears to be emerging [1, 2]. The present study describes the frequency of meropenem non-susceptible *Enterobacteriaceae* isolates, collected throughout Asia during 2009 – 2010 as part of the Tigecycline Evaluation and Surveillance Trial (TEST).

Materials & Methods

- ***Clinical isolates:*** Isolates collected from multiple infection sources were identified to the species level and tested at each participating laboratory. All organisms were deemed clinically significant by local participant criteria. Isolate inclusion was independent of medical history, antimicrobial use, age or gender. All sites identified each study isolate utilizing local laboratory criteria. All isolates were from the period 2009 - 2010 and originated from various countries in Asia.
- ***Susceptibility testing:*** Minimum inhibitory concentrations (MICs) were determined using plates manufactured by Trek Diagnostics, following manufacturer and Clinical and Laboratory Standards Institute (CLSI) instructions for broth microdilution testing [3]. Susceptibility was determined using clinical breakpoints published by the CLSI [4]. FDA breakpoints were used for tigecycline [5]. Tigecycline was supplied by Pfizer, Inc. (Collegeville, PA, USA). All other agents were supplied by the panel manufacturer TREK (TREK Diagnostic Systems, Cleveland, OH). The following antimicrobial agents were included on the panels with their dilution ranges (expressed in mcg/ml): amikacin (0.5-64); amoxicillin/clavulanic acid (0.12/0.06-32/16); ampicillin (0.06-16); cefepime (0.5-32); ceftazidime (8-32); ceftriaxone (0.06-64); meropenem (0.12-16); levofloxacin (0.008-8); minocycline (0.5-16); tigecycline (0.008-16); and piperacillin/tazobactam (0.06/4-128/4). *E. coli* ATCC 25922 and ATCC 35218 and *P. aeruginosa* ATCC 27853 were tested as quality control organisms.

References

1. Hsueh PR, Snyder TA, Dinubile MJ, Satischandran V, McCarroll K, Chow JW, et al. 2006. *In vitro susceptibilities of aerobic and facultative gram-negative bacilli isolated from patients with intra-abdominal infections in the Asia–Pacific region: 2004 results from SMART (Study for Monitoring Antimicrobial Resistance Trends)*. Int J Antimicrob Agents. 28: 238–243.
2. Hawser SP, Bouchillon SK, Hoban DJ, Badal RE, Hsueh PR, Paterson D. 2009. *Emergence of high levels of extended-spectrum beta-lactamase-producing gram-negative bacilli in Asia/Pacific: data from SMART 2007.* Antimicrob Agents Chemother. 53:3280-3284.
3. Clinical Laboratory Standards Institute. 2009. Methods for Dilution Antimicrobial Susceptibility Tests for Bacteria That Grow Aerobically; Approved Standards - Eighth Edition. CLSI document M07-A8. Wayne, PA.
4. Clinical and Laboratory Standards Institute. 2011. Performance Standards for Antimicrobial Susceptibility Testing; Twenty-First Informational Supplement. CLSI Document M100-S21. Wayne, PA.
5. Tygacil®, 2010 Federal Drug Administration (FDA) product information. Pfizer, Inc. Collegeville, PA, USA.

Acknowledgements

We gratefully acknowledge the contributions of the investigators, laboratory personnel, and all members of the Tigecycline Evaluation Surveillance Trial program group. This study was sponsored by Pfizer Inc.

Results

Figure 1. Number of all isolates (n = 1,305) and meropenem non-susceptible isolates (n = 27) by country.

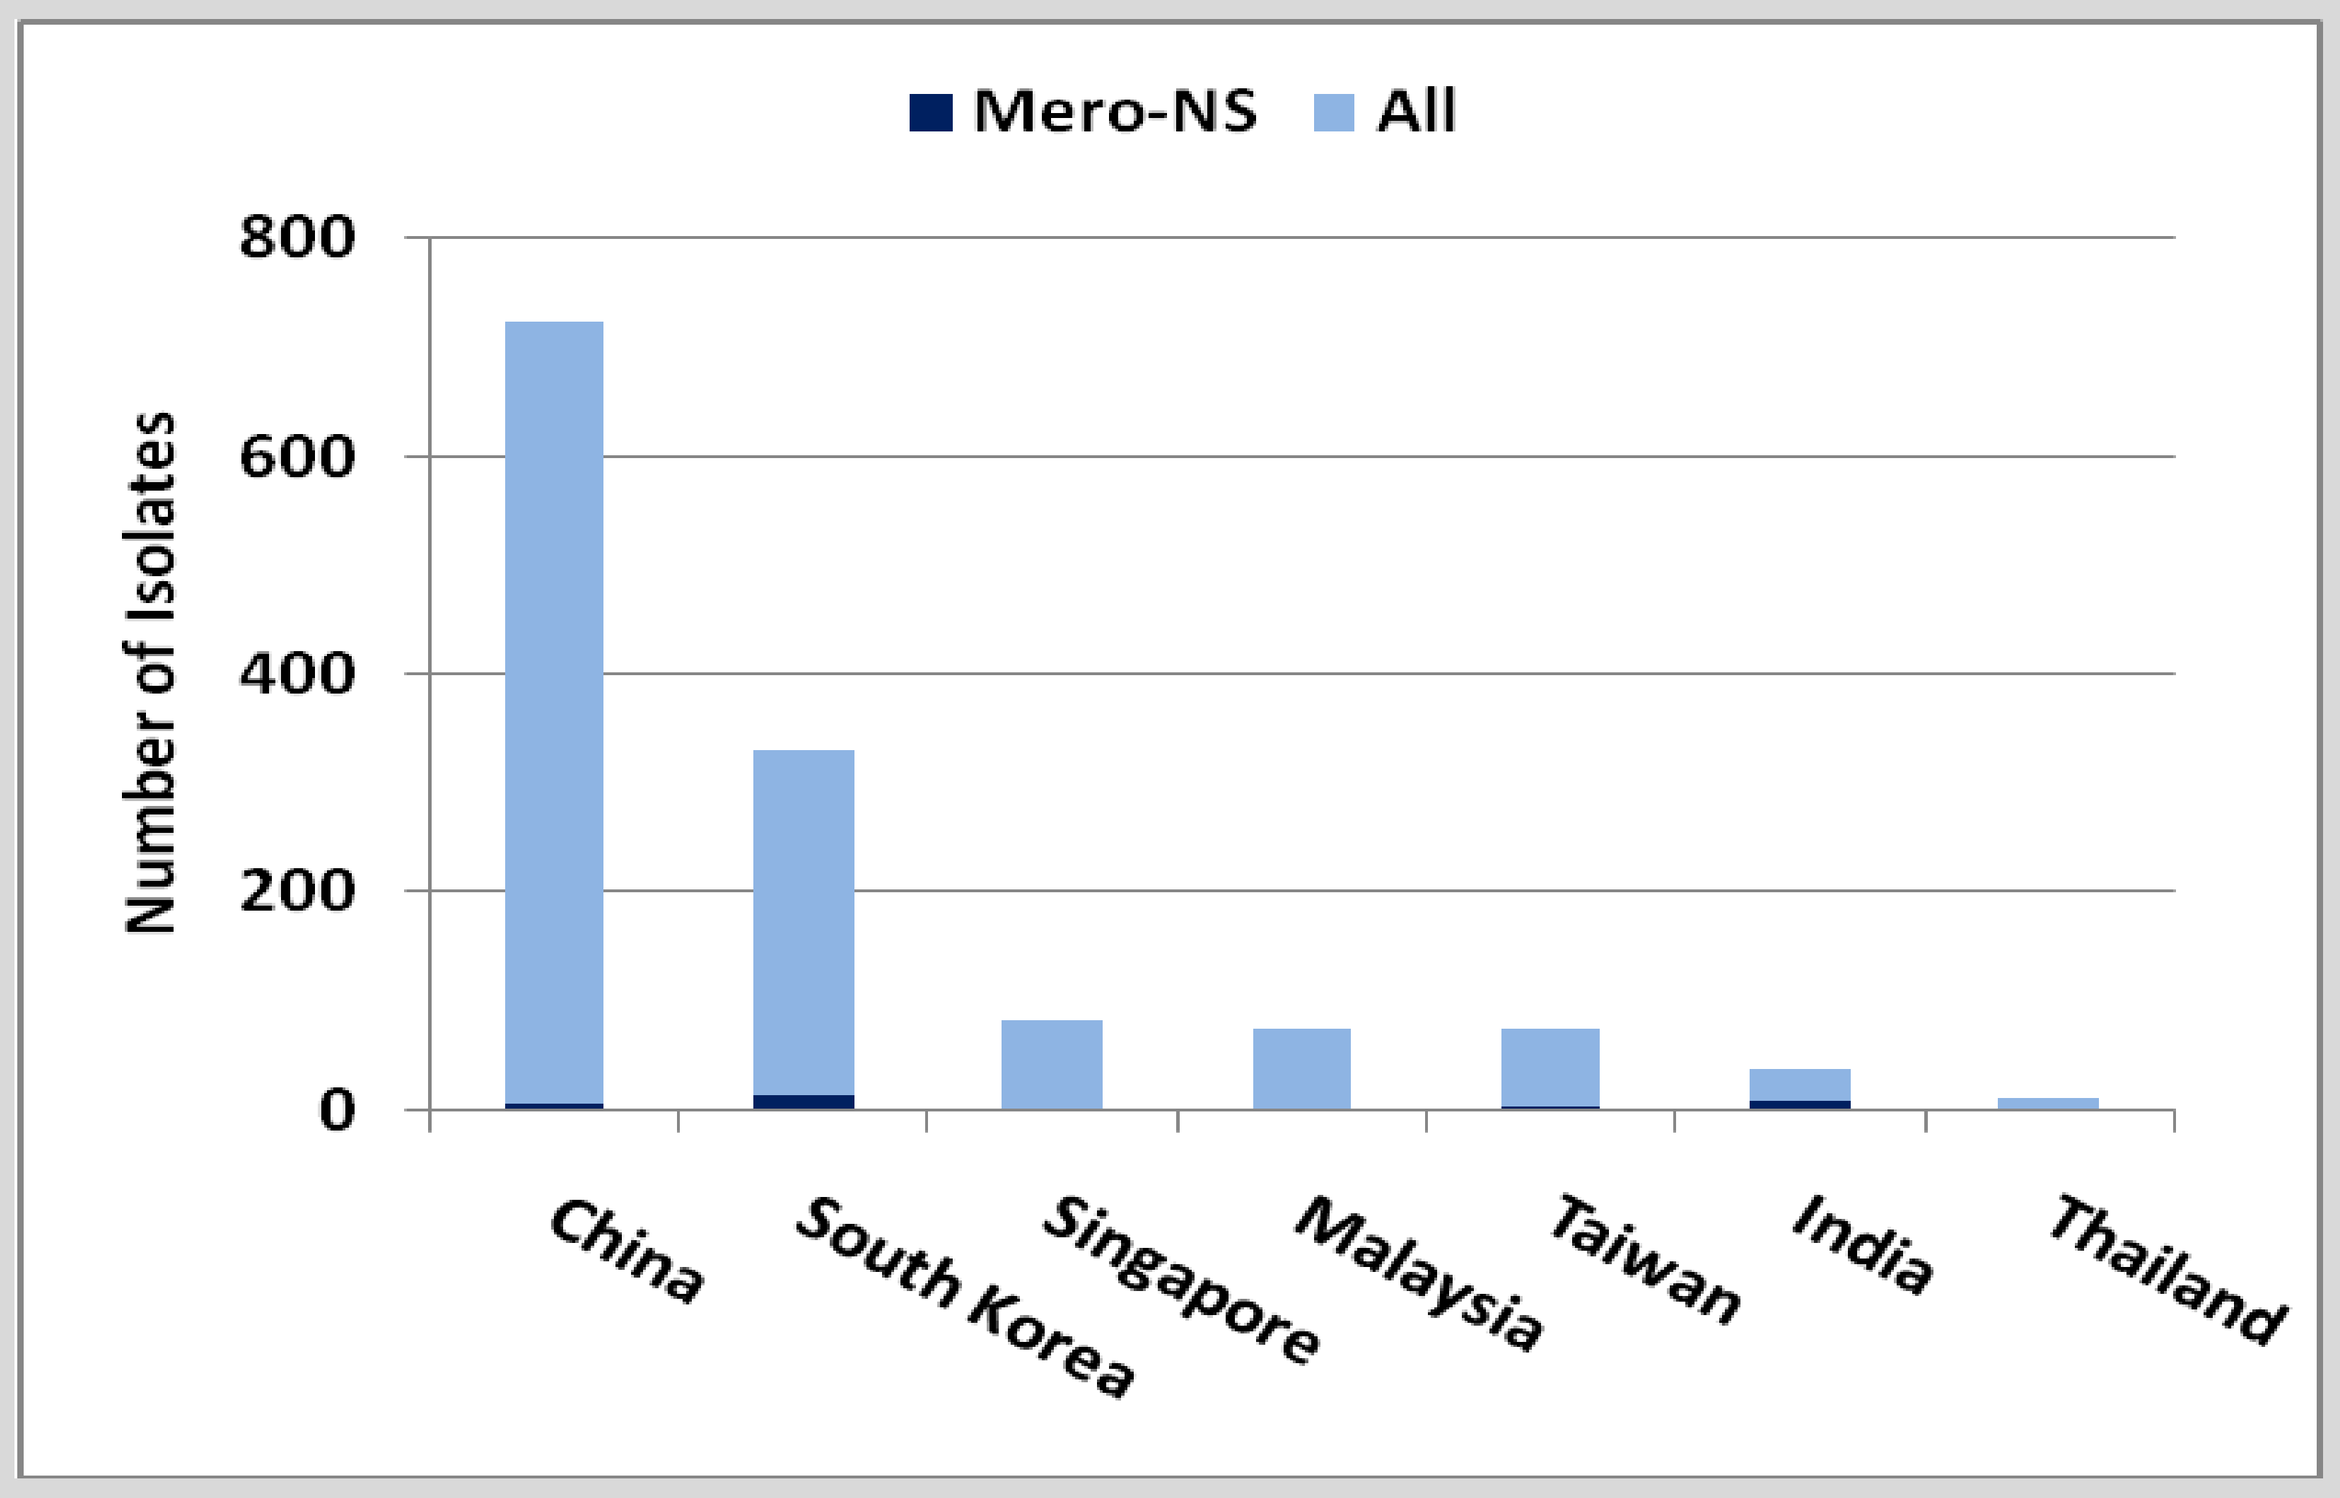

Mero-NS
All
Number of Isolates
800
600
400
200
0
China
South Korea
Singapore
Malaysia
Taiwan
India
Thailand

Figure 2. Percent of meropenem non-susceptible isolates (n = 27) by country.

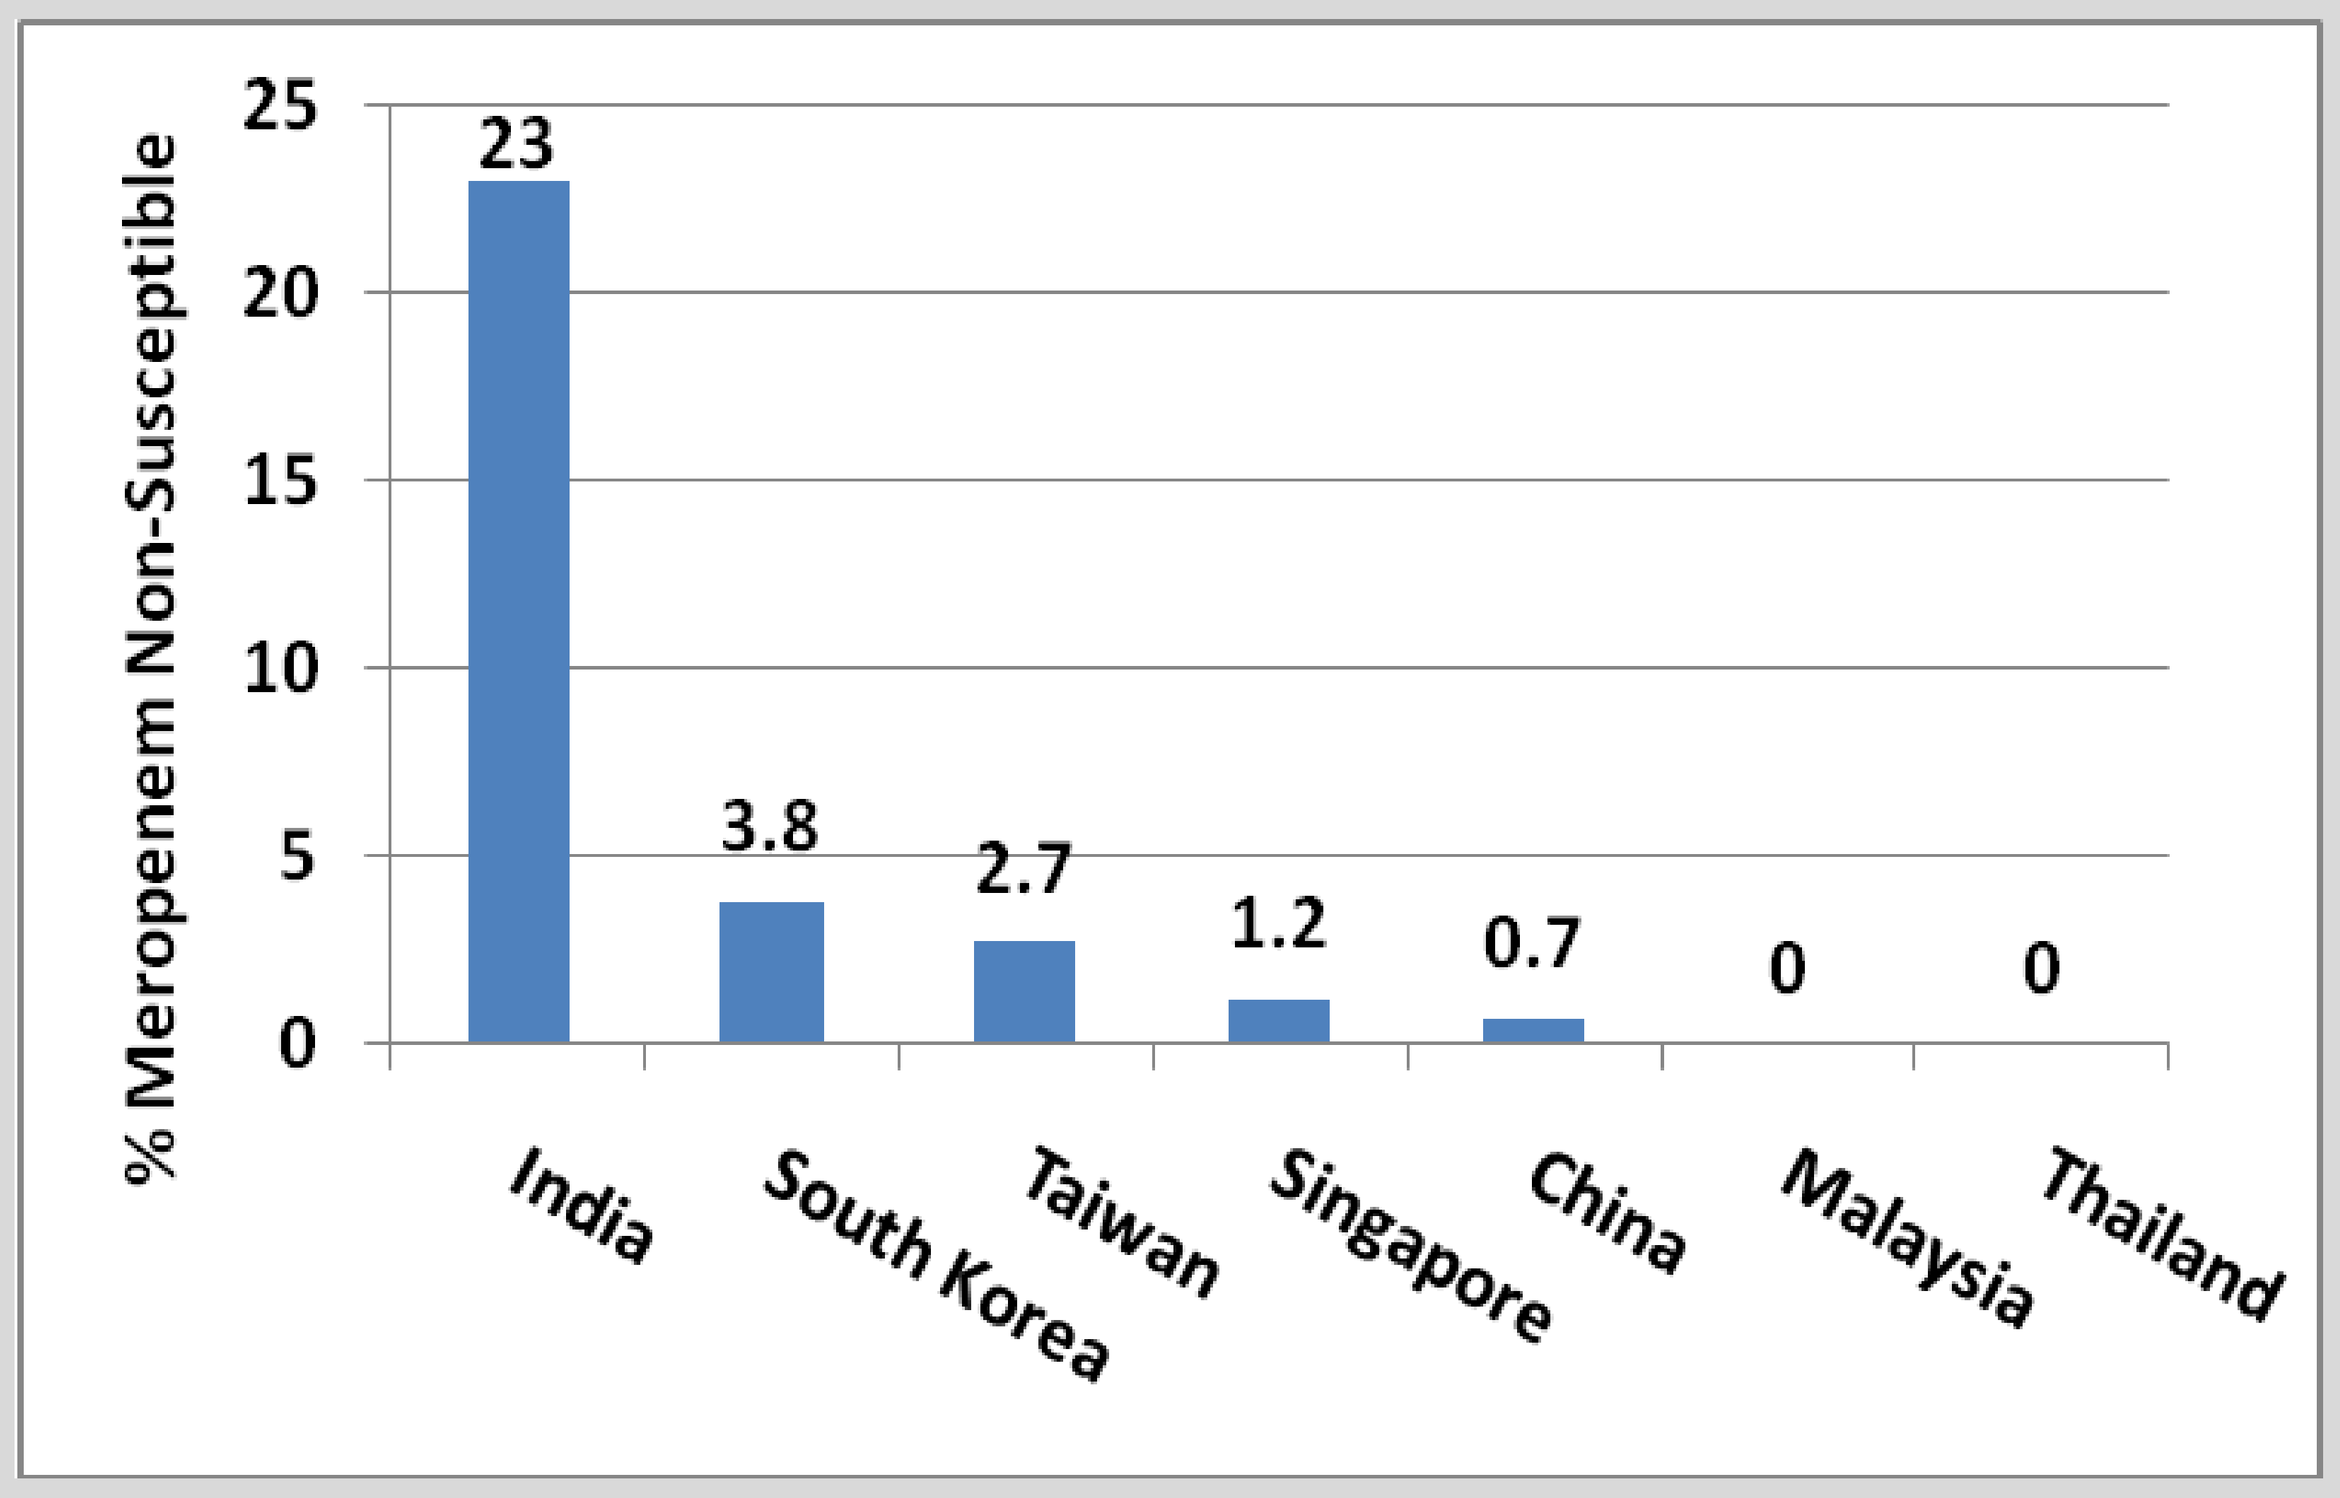

% Meropenem Non-Susceptible
25
20
15
10
5
0
23
3.8
2.7
1.2
0.7
0
0
India
South Korea
Taiwan
Singapore
China
Malaysia
Thailand

Figure 3. Number of all isolates (n = 1,305) and meropenem non-susceptible isolates (n = 27) by location.

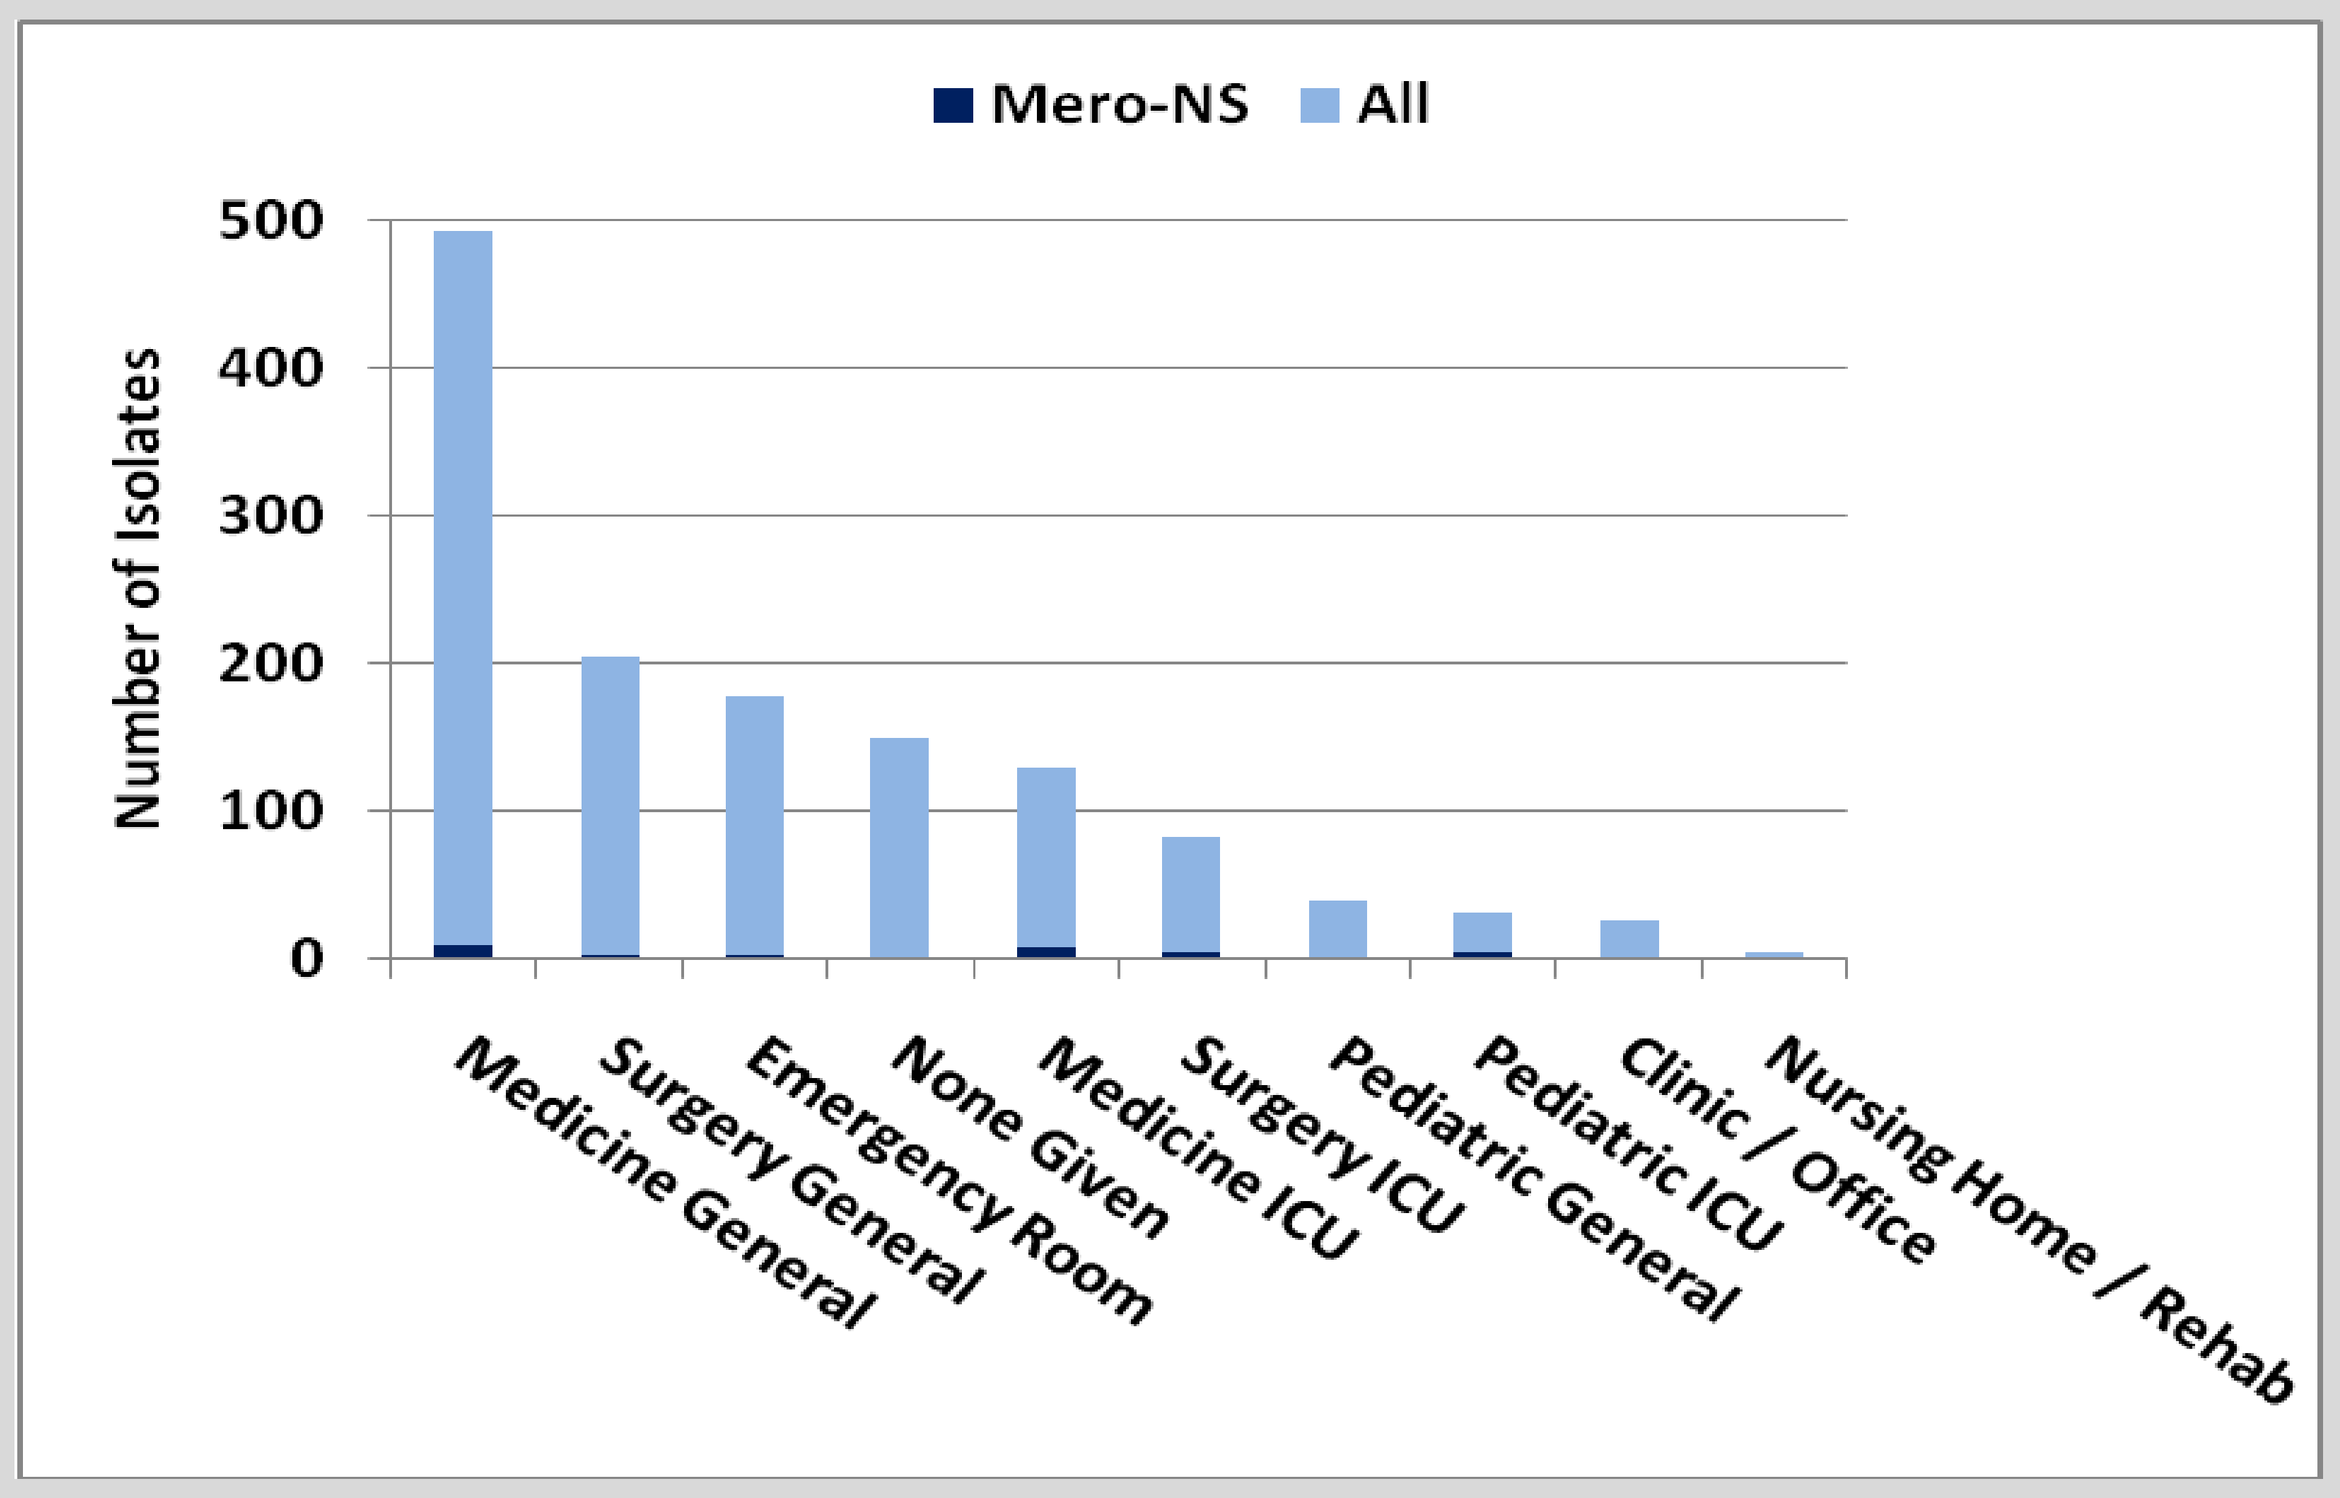

Mero-NS
All
Number of Isolates
500
400
300
200
100
0
Medicine General
Surgery General
Emergency Room
None Given
Medicine ICU
Surgery ICU
Pediatric General
Pediatric ICU
Clinic / Office
Nursing Home / Rehab

Figure 4. Percent of meropenem non-susceptible isolates (n = 27) by location.

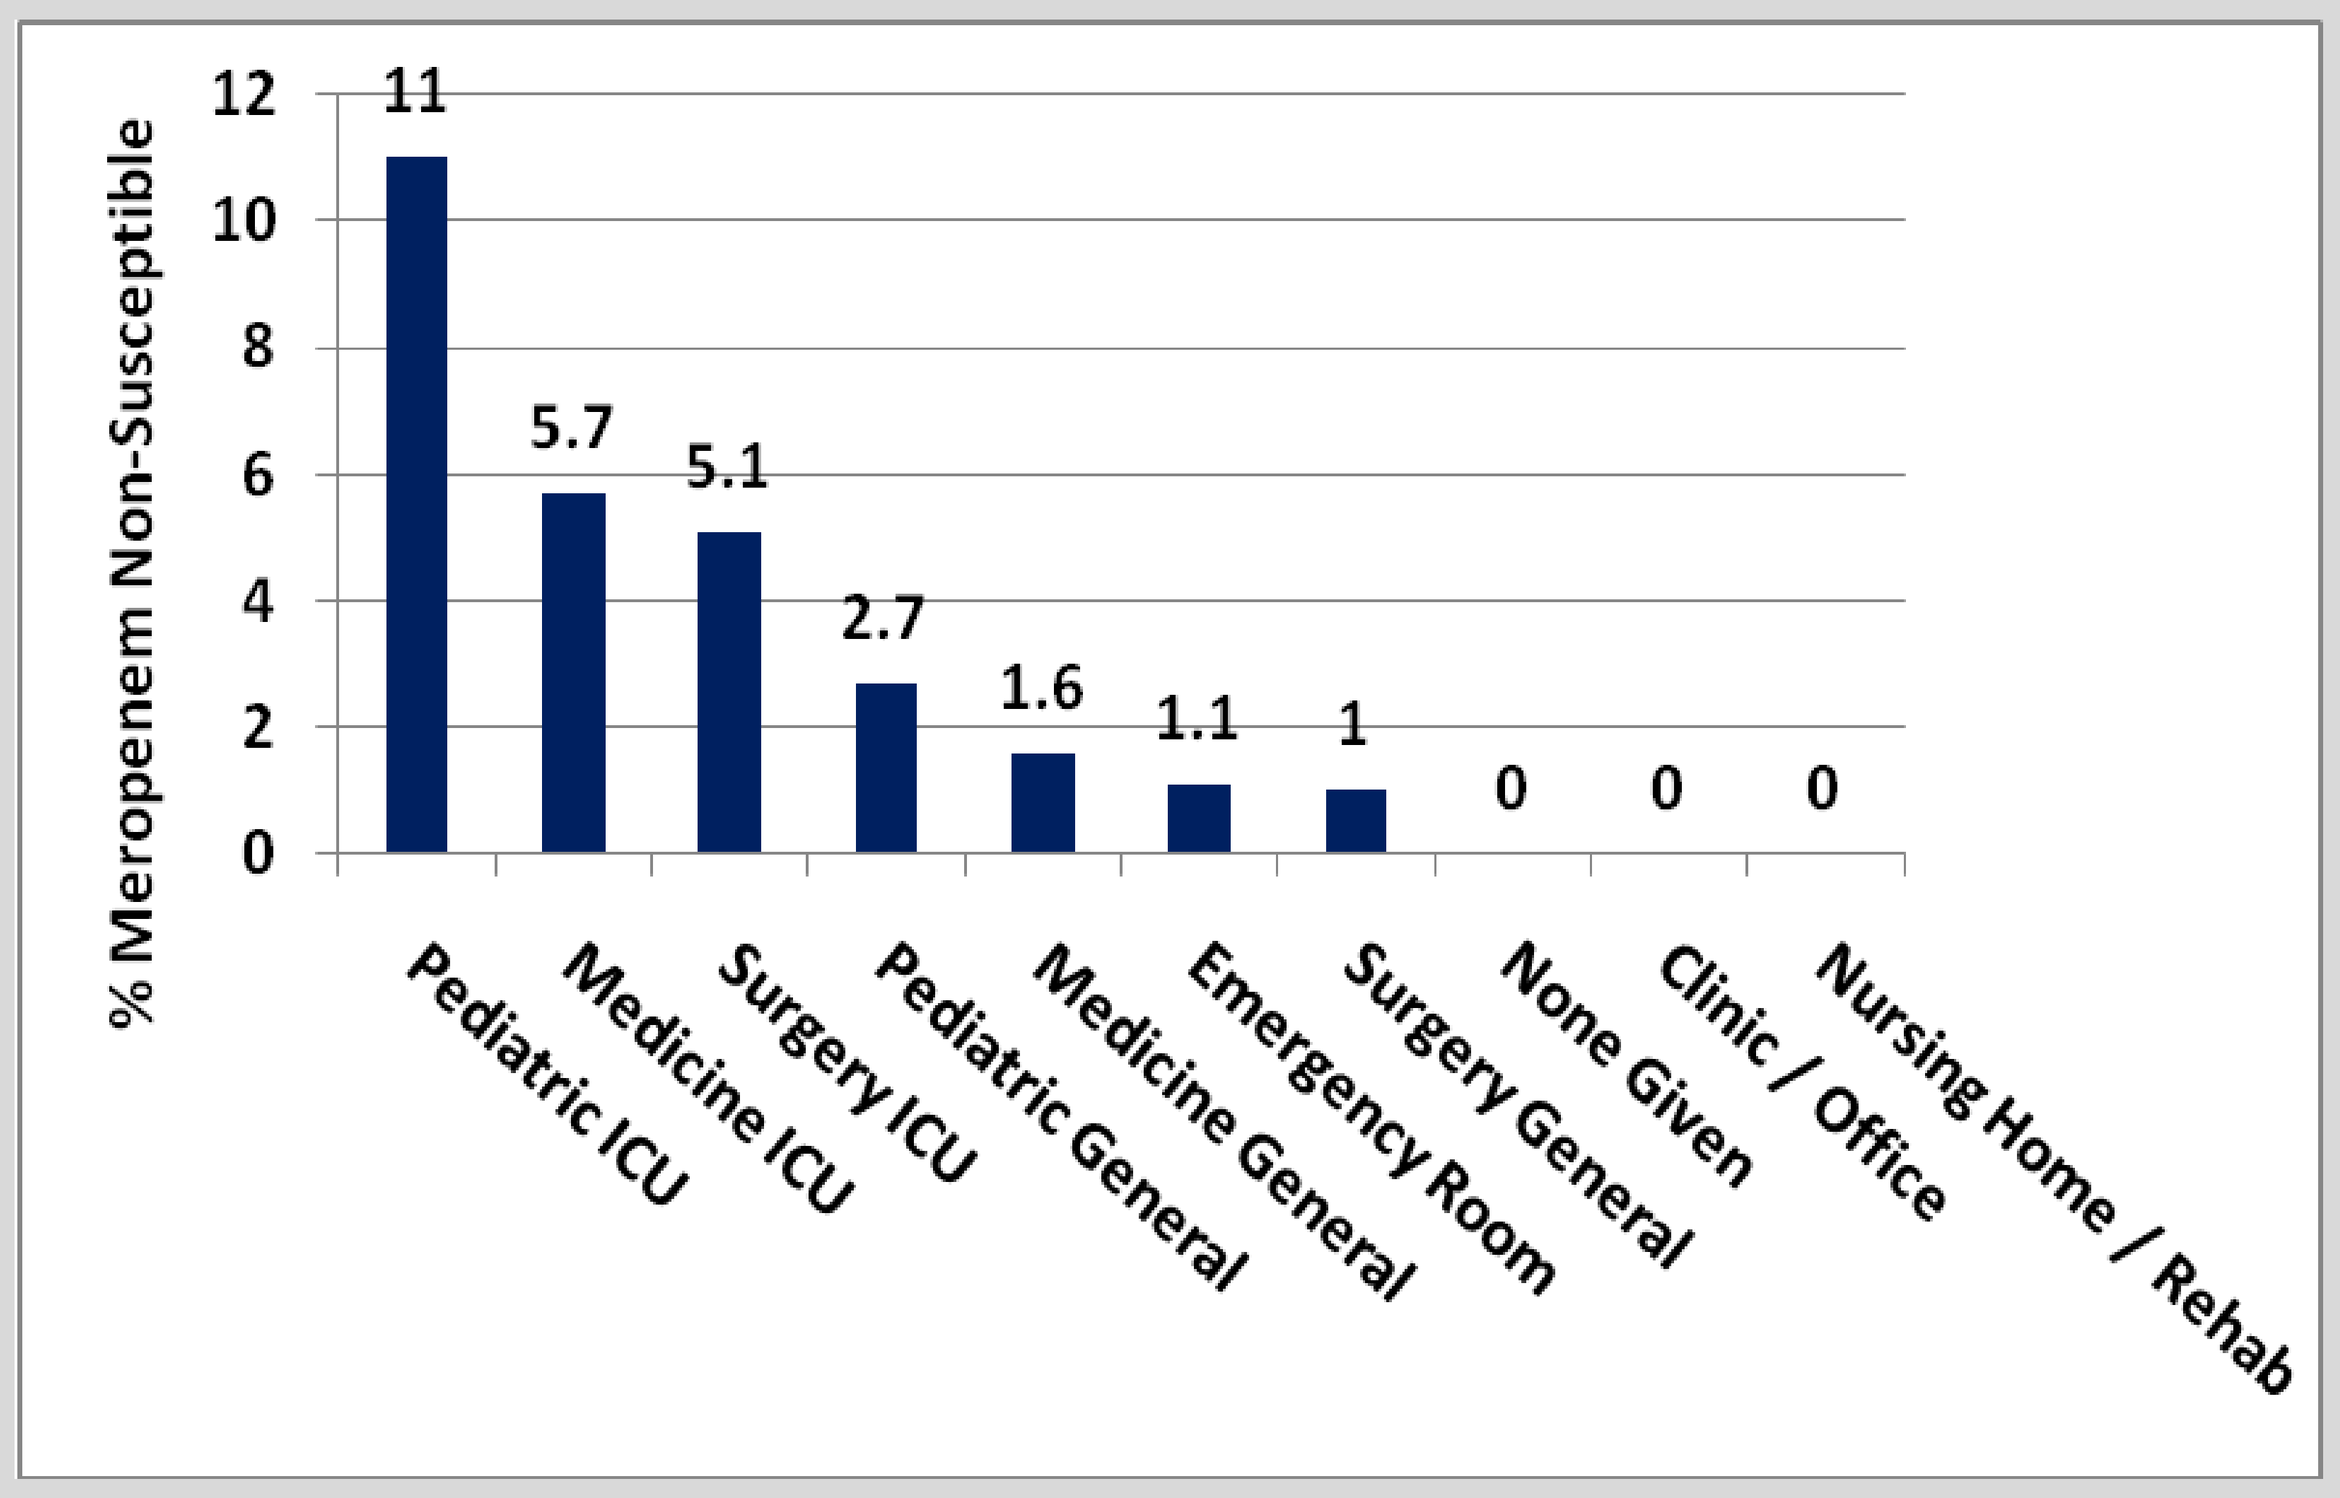

% Meropenem Non-Susceptible
12
10
8
6
4
2
0
11
5.7
5.1
2.7
1.6
1.1
1
0
0
0
Pediatric ICU
Medicine ICU
Surgery ICU
Pediatric General
Medicine General
Emergency Room
Surgery General
None Given
Clinic / Office
Nursing Home / Rehab

Figure 5. Number of all isolates (n = 1,305) and meropenem non-susceptible isolates (n = 27) by source.

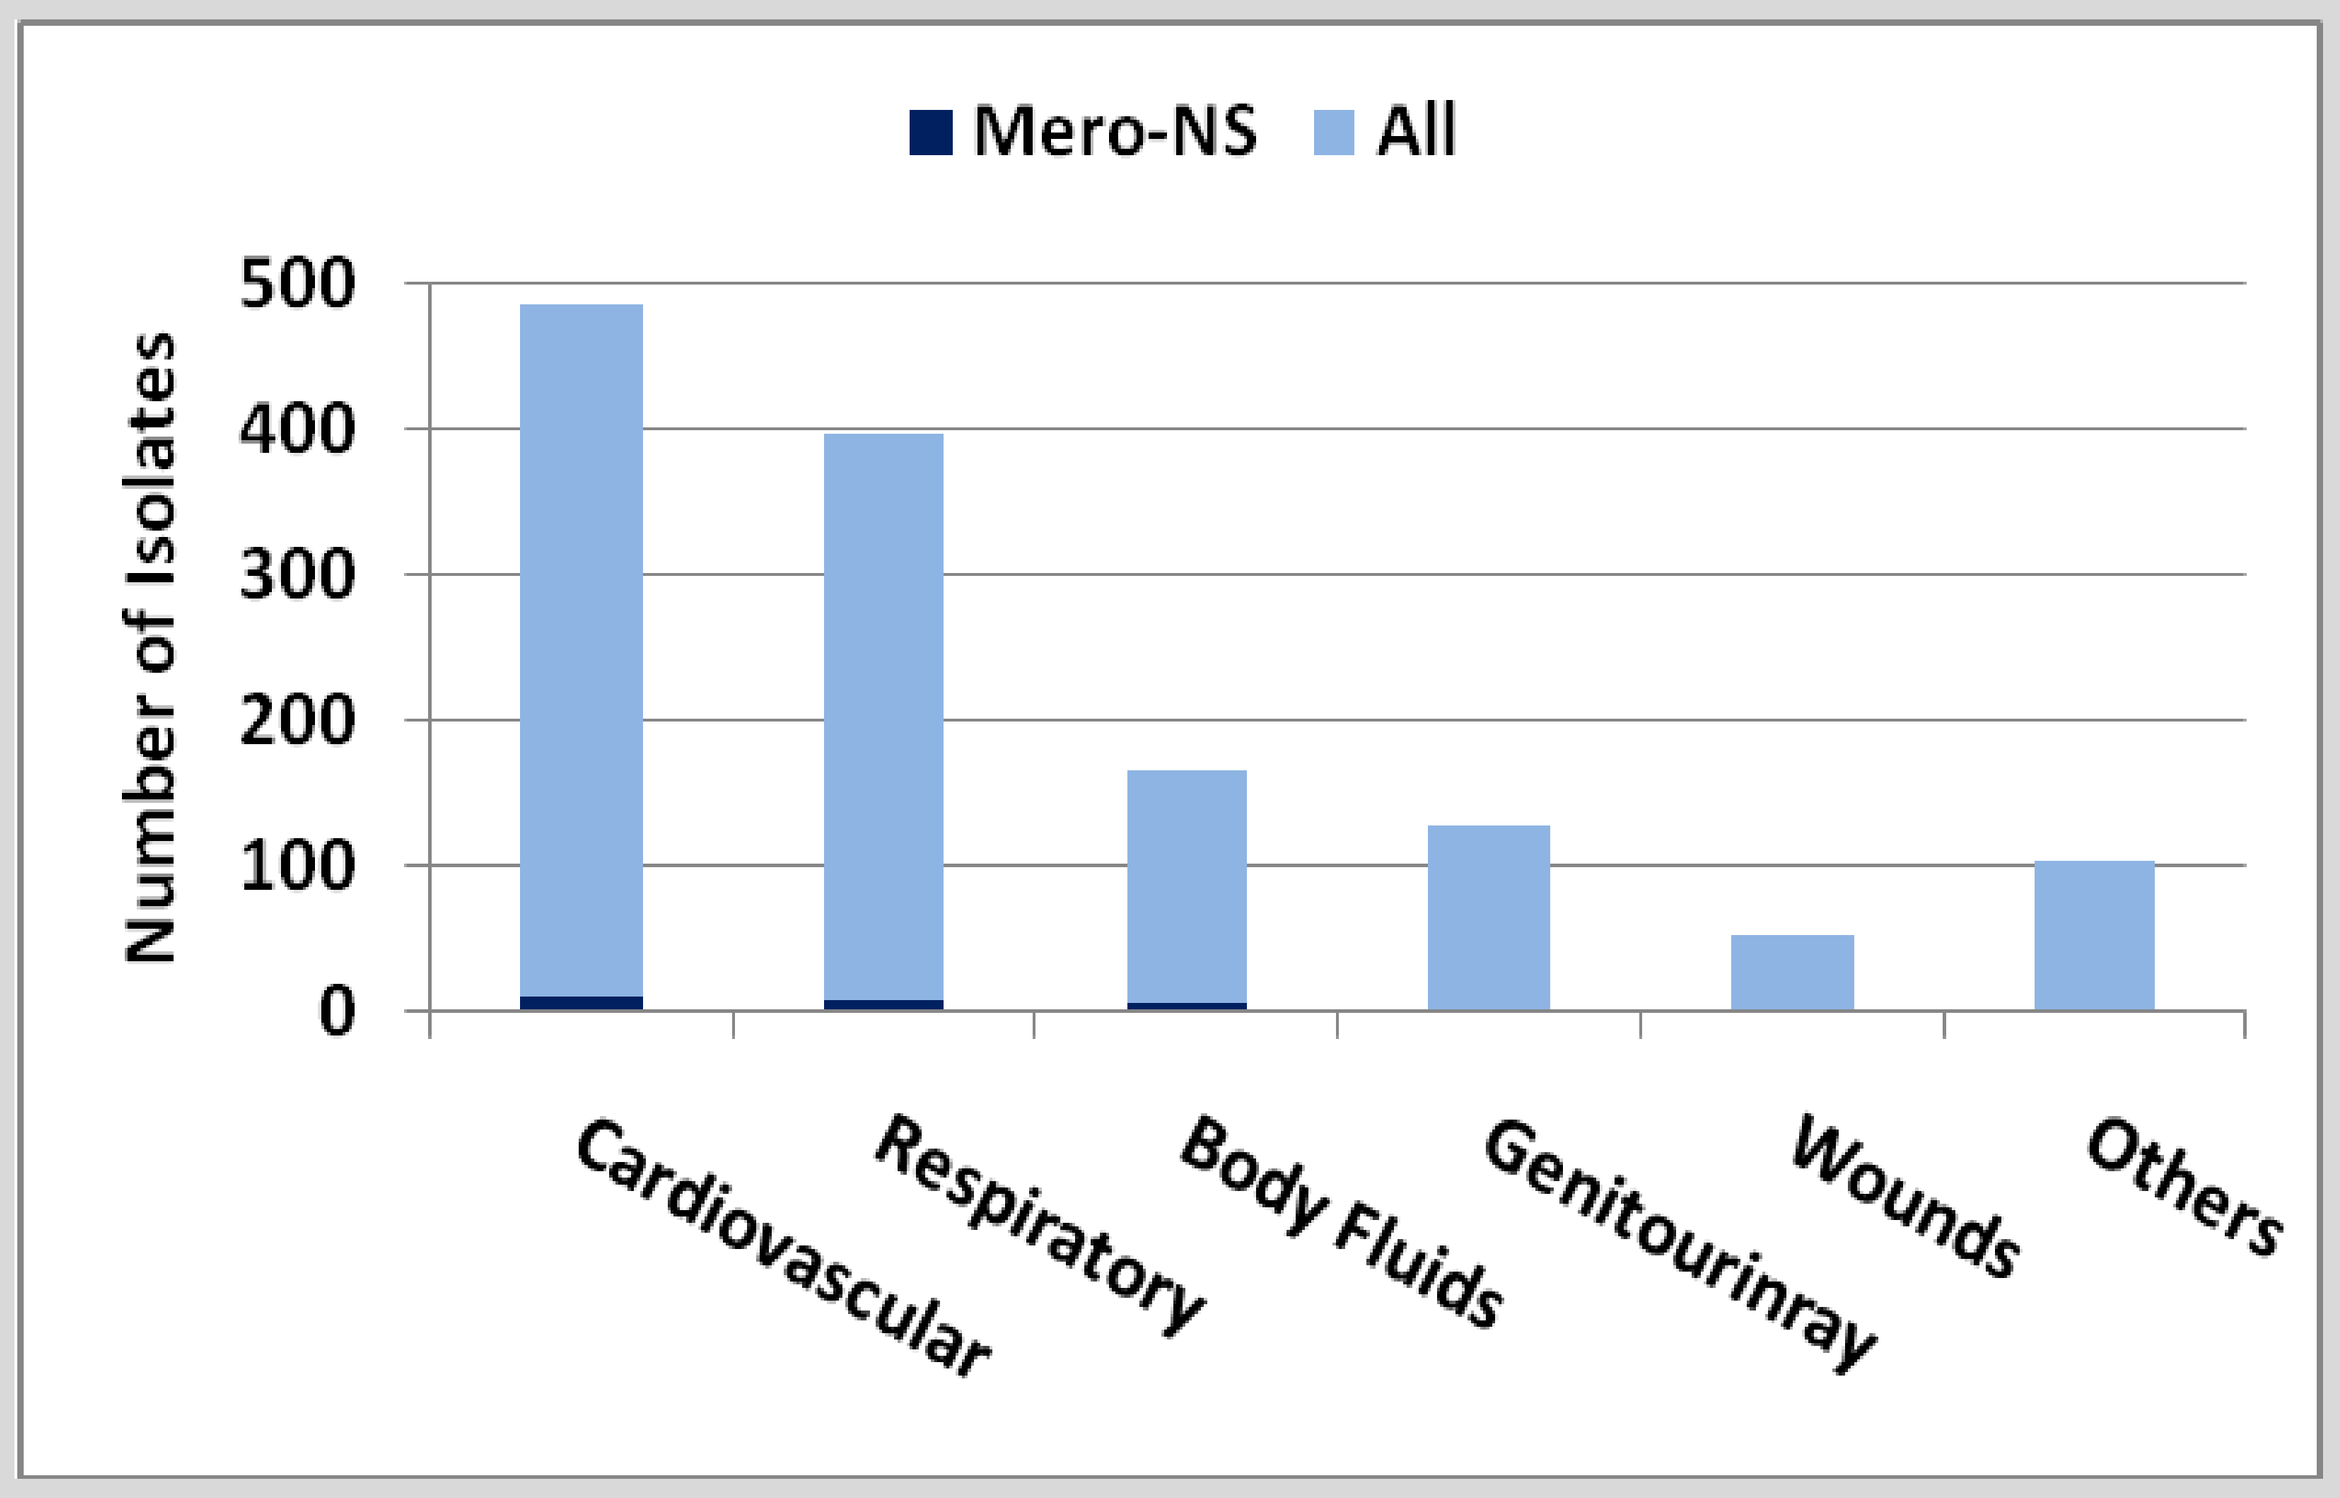

Mero-NS
All
Number of Isolates
500
400
300
200
100
0
Cardiovascular
Respiratory
Body Fluids
Genitourinray
Wounds
Others

Figure 6. Percent of meropenem non-susceptible isolates (n = 27) by source.

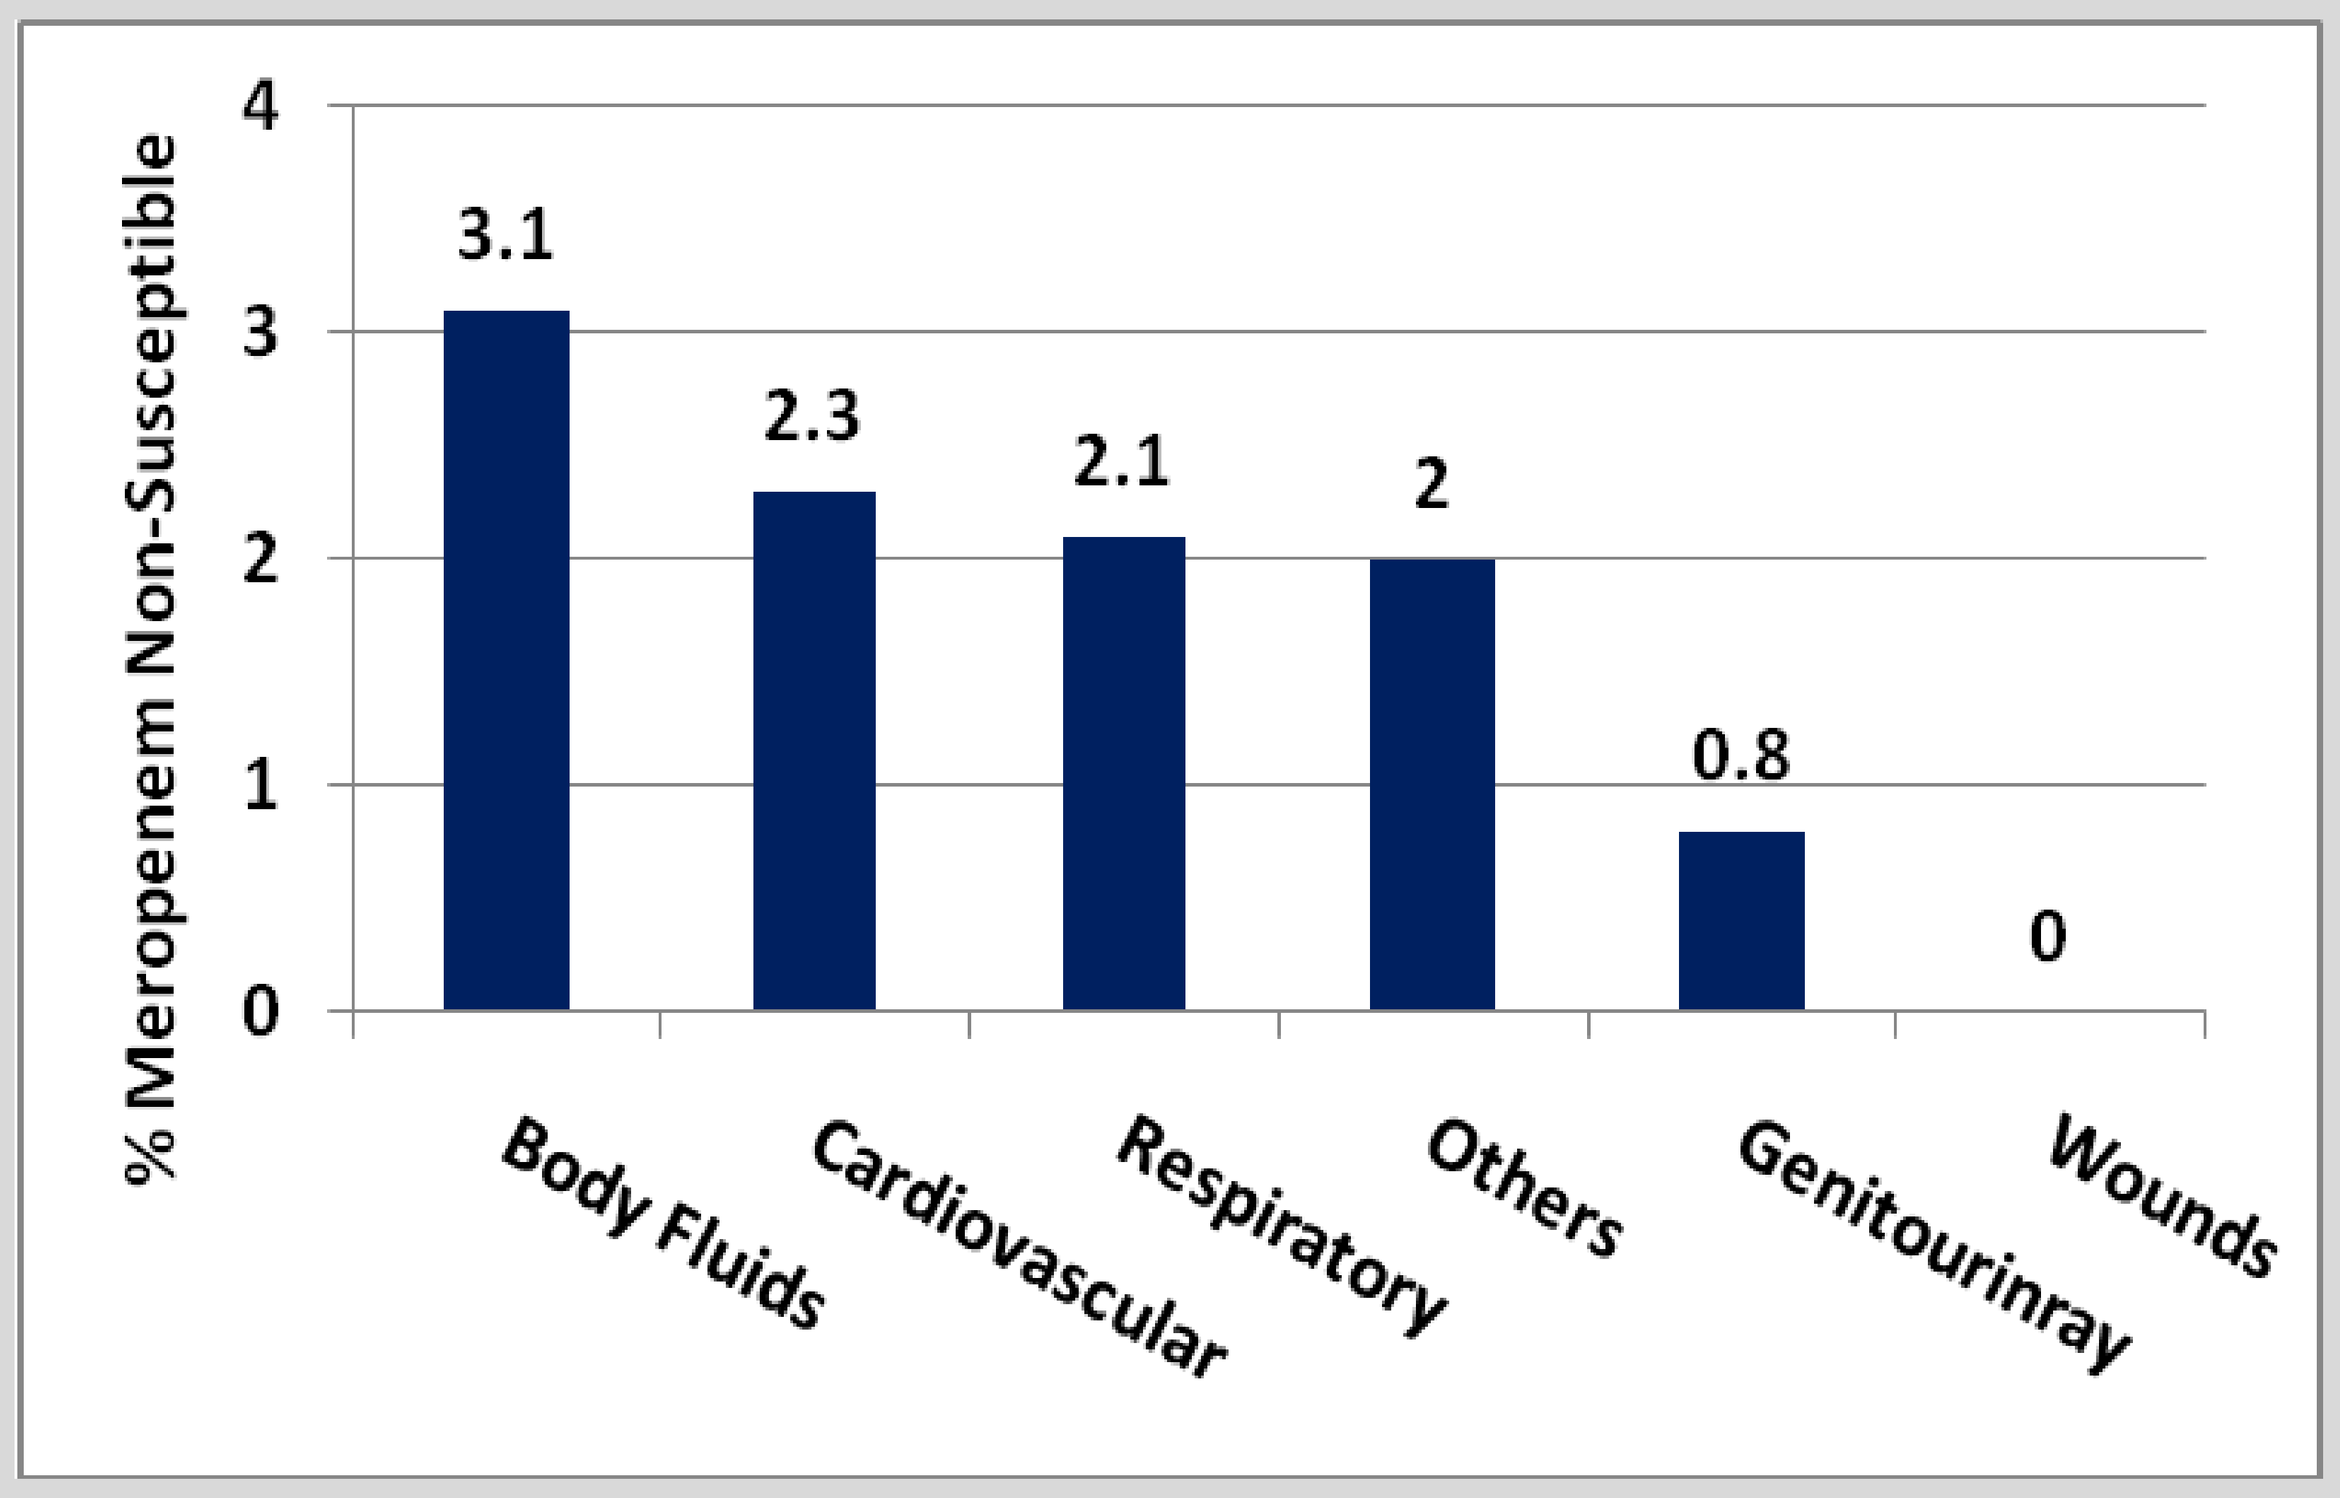

% Meropenem Non-Susceptible
4
3
2
1
0
3.1
2.3
2.1
2
0.8
0
Body Fluids
Cardiovascular
Respiratory
Others
Genitourinray
Wounds

Figure 7. Susceptibility of all isolates (n = 1,305) and meropenem non-susceptible isolates (n = 27).

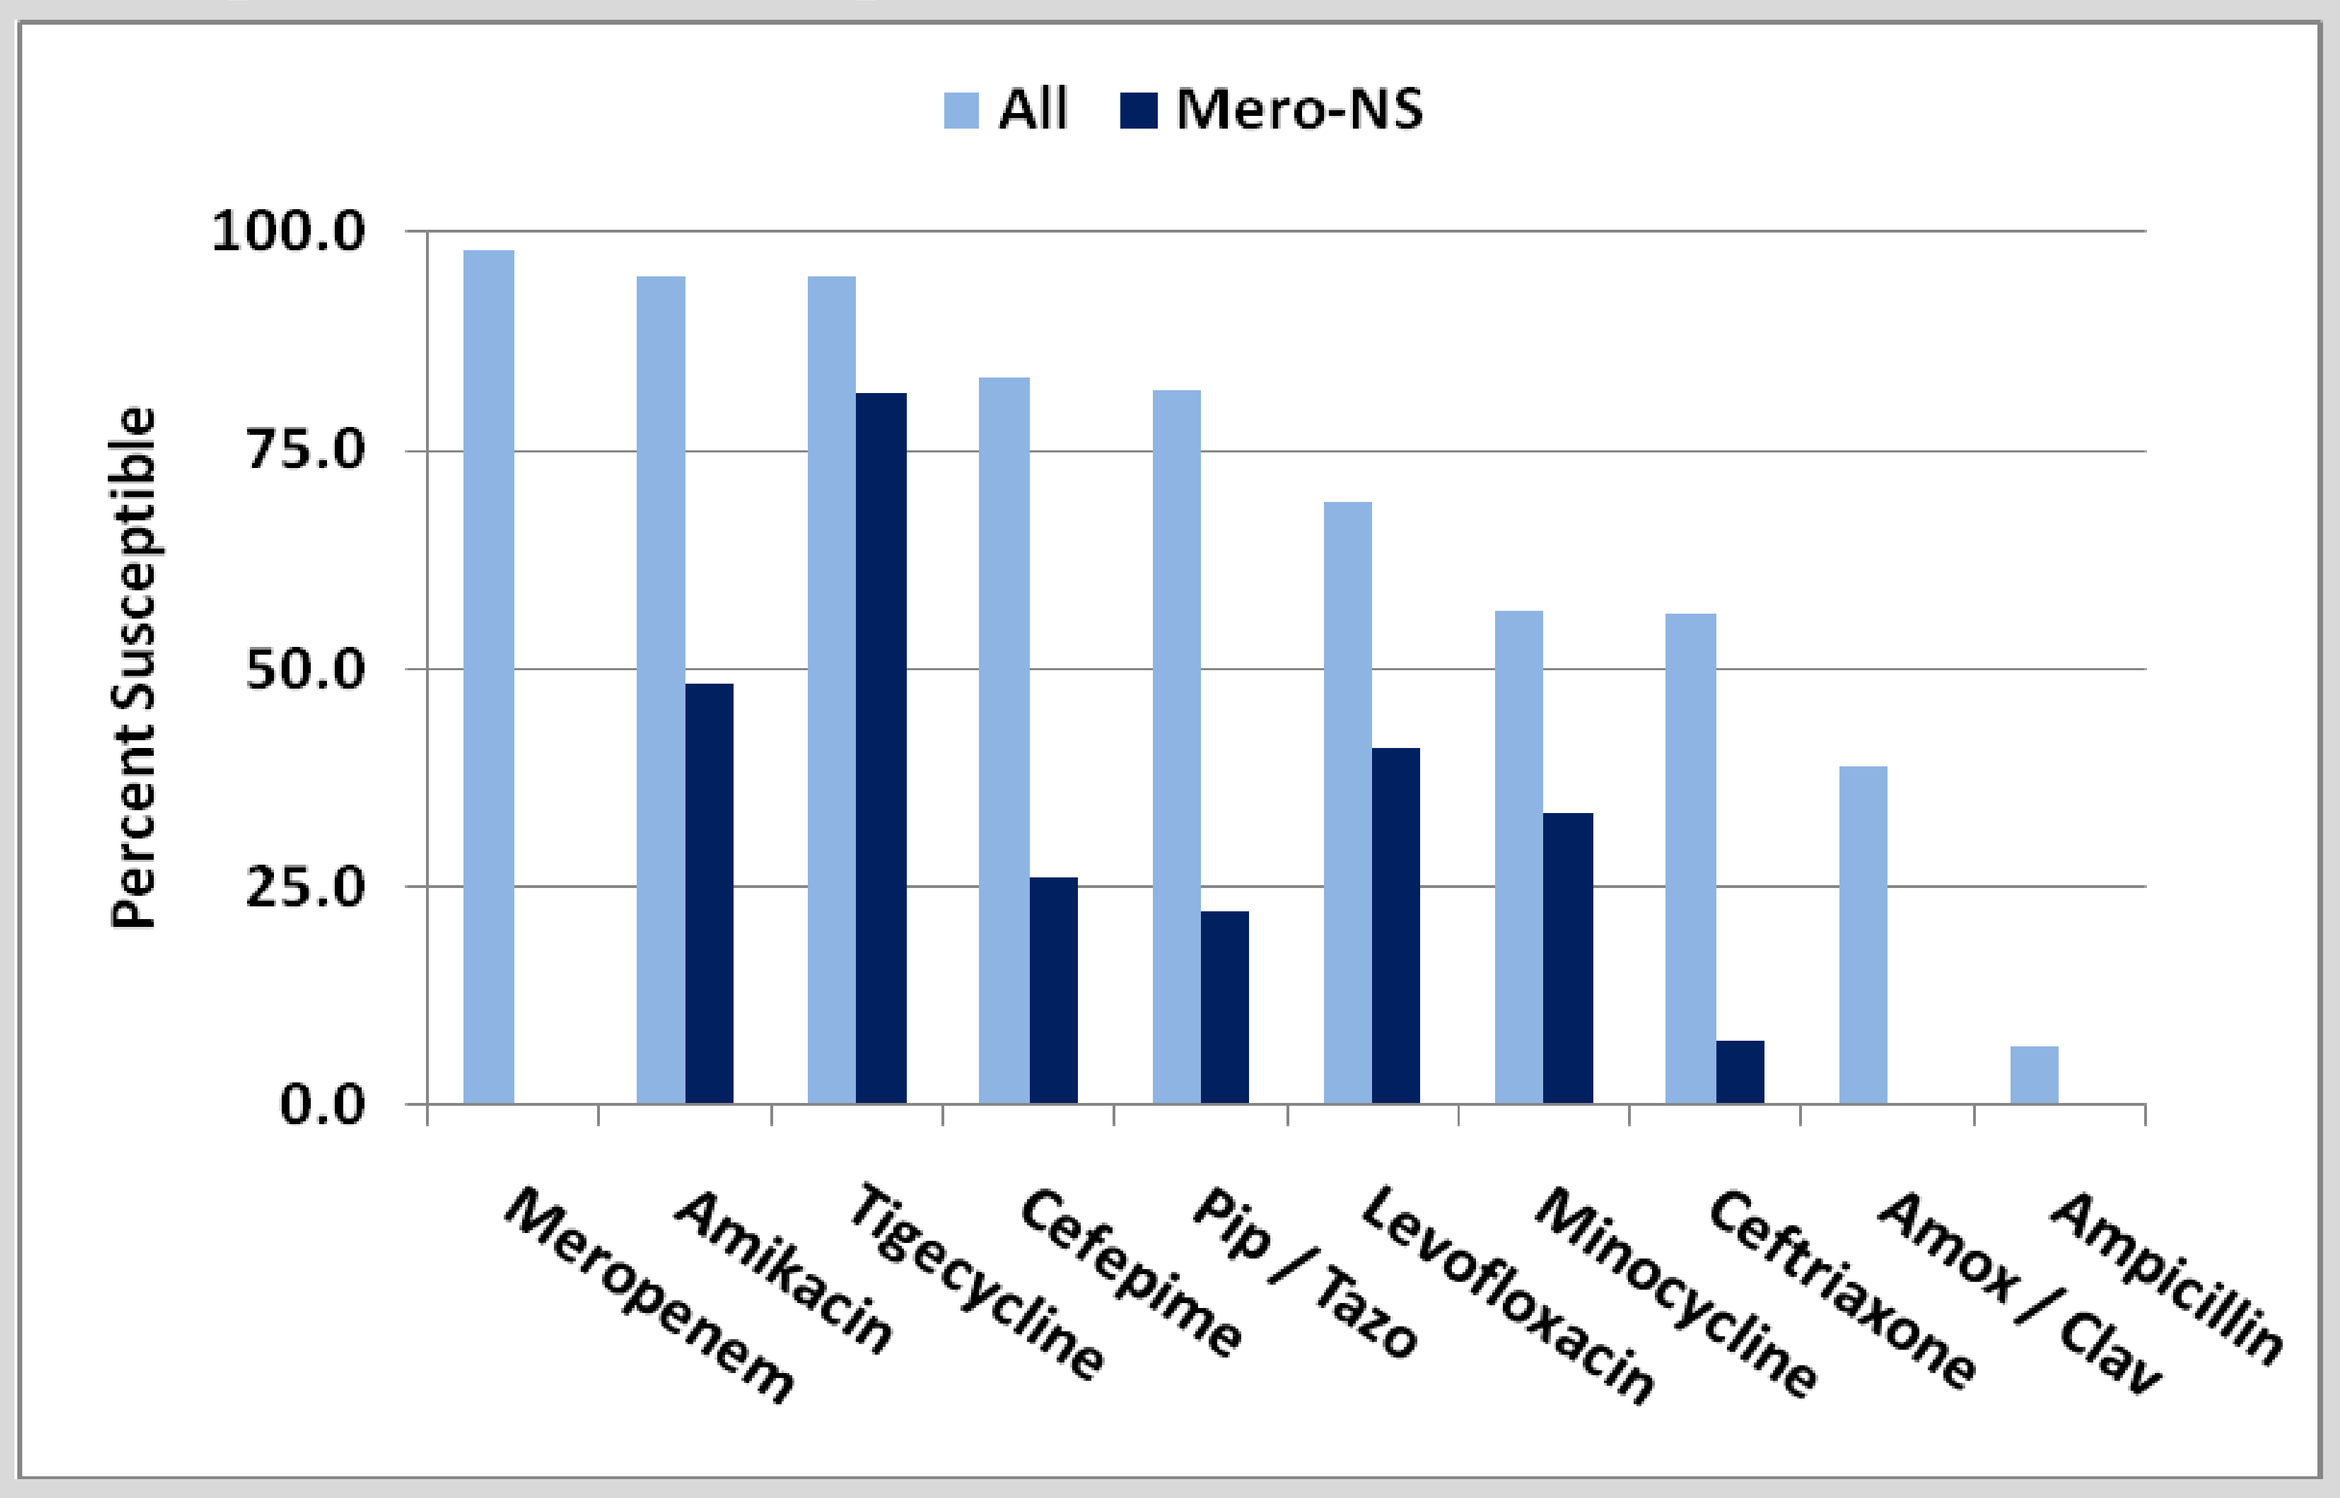

All
Mero-NS
Percent Susceptible
100.0
75.0
50.0
25.0
0.0
Meropenem
Amikacin
Tigecycline
Cefepime
Pip / Tazo
Levofloxacin
Minocycline
Ceftriaxone
Amox / Clav
Ampicillin

Conclusions

- Of the total of 1,305 isolates, 27 (2%) exhibited non-susceptibility to meropenem. Meropenem non-susceptible isolates were most common in India (23%) and South Korea (3.8%)
- Of the agents tested, only meropenem, amikacin and tigecycline had susceptibility >90% for the total pool of 1,305 isolates. However, against the sub-population of 27 meropenem non-susceptible isolates tigecycline was the only agent that had susceptibility >75%.
- While meropenem resistance is generally considered to be gradually increasing in *Enterobacteriaceae*, the promising activity of tigecycline against meropenem non-susceptible isolates is encouraging.