P 491

Tigecycline In Vitro Activity Against Multi-Drug Resistant Acinetobacter spp. In The T.E.S.T. Program Global Data

D. Hoban[1], B. Johnson[1], R. Badal[1], S. Bouchillon[1], T. Stevens[1], J. Johnson[1], M. Dowzicky[2]

[1]International Health Management Associates, Schaumburg, IL, USA
[2]Wyeth Pharmaceuticals, Collegeville, PA, USA

IHMA, Inc.
2122 Palmer Dr.
Schaumburg, IL
60173
Tel: (847) 303-5003
Fax: (847) 303-5601
www.ihmainc.com

REVISED ABSTRACT

Background: Tigecycline is a glycylcycline, a new generation of tetracyclines significantly different to be classified as a separate antimicrobial class. Glycylcyclines are being developed to overcome the problem of bacterial resistance to tetracyclines and other antimicrobials. Tigecycline is better tolerated and is more active than tetracyclines against a wide variety of Gram-positive and Gram-negative bacteria including *Acinetobacter* spp. The T.E.S.T. program determined the in vitro activity of TIG against *Acinetobacter* resistant to one or more of piperacillin-tazobactam (PT), levofloxacin (LVX), ceftriaxone (CAX), cefepime (CPE), amikacin (AK), minocycline (MIN), ceftazidime (CAZ) and imipenem (IMP). Study strains were collected from hospitals globally throughout 2004-2006.

Methods: A total of 1,798 clinical isolates of *Acinetobacter* spp. were identified to species level from 150 participating sites and confirmed by the central laboratory. Minimum Inhibitory Concentrations (MICs) were determined by the local laboratory using supplied broth microdilution panels and interpreted according to CLSI guidelines, with tigecycline susceptible breakpoint defined as <u><</u>2 mcg/ml.

Results: TIG inhibited 97% of *Acinetobacter* resistant to 2 or more drug classes at 2 mcg/mL and inhibited 184/192 (95.8%) and 39/42 (92.9%) of imipenem- and minocycline-resistant strains, respectively. Resistance rates for comparator drugs against all *Acinetobacter* were CAX 45%, CAZ 45%, LVX 45%, CPE 37%, PT 24%, AK 8%, IMP 6.3%, and MIN 2.2%. Only one strain had a MIC of 8 mcg/mL against TIG. The modal TIG MIC for strains resistant to 2 or more drug classes was 1 mcg/ml compared to 0.12 mcg/ml strains with no resistant parameters, indicating an 8-fold diminishment of activity.

Conclusion: It has been seen in some species that existing multi-drug efflux pumps may also pump TIG. In spite of this, TIG remained effective and inhibited most *Acinetobacter* strains resistant to three or more other drug classes in this study, although the higher TIG MICs seen for these strains suggests some linkage to resistance mechanisms for other drugs. TIG remained effective in inhibiting multi-drug resistant *Acinetobacter* spp., further broadening its wide spectrum of activity vs. drug-resistant bacteria.

INTRODUCTION

Tigecycline (formerly GAR-936) is a member of a new class of antimicrobial agents, the glycylcyclines. This synthetic analogue of the tetracyclines exhibits significant antibacterial activity that is both bacteriostatic and, in certain instances, bactericidal with killing activity that is as much as fourfold better than vancomycin and daptomycin [1, 2]. The development of tigecycline is important in that it and other glycylcyclines are active against bacterial strains carrying either or both of the two major forms of tetracycline resistance: efflux and ribosomal protection. Certain substituents at the 9-position of the tetracycline molecule restored activity against bacteria harboring genes encoding either or both efflux and ribosomal protection. A single chemical modification of tigecycline overcomes the two molecularly distinct forms of resistance while maintaining activity against susceptible gram-positive, gram-negative, aerobic, and anaerobic bacteria [3]. Furthermore, resistance to tigecycline is difficult to produce even in the laboratory.

Infections secondary *Acinetobacter* present a challenge to clinicians with increasing multi-drug resistance worldwide. Resistance of *Acinetobacter* to cephalosporins, aminoglycosides and ciprofloxacin is now widespread but carbapenems, colistin, sulbactam and minocycline remain effective in over 80% of most strains [4, 5]. The emergence of imipenem resistance in this species is of considerable concern leaving relatively limited treatment options for infections due these resistant *Acinetobacter* and has led to a search for new compounds with activity against these problematic pathogens.

This study was undertaken to document the in vitro activity of tigecycline against multi-drug resistant *Acinetobacter* in a large diverse population from multiple global investigative sites. This study is part of the larger ongoing global Tigecycline Evaluation and Surveillance Trials (T.E.S.T.) program.

MATERIALS & METHODS

- All isolates were derived from blood, respiratory tract, urine, skin, wound, body fluids and other defined sources. Only one isolate per patient was accepted into the study. Clinical isolates were collected and tested between January 2004 - January 2006 from 150 sites in 27 countries. Isolates were identified to the species level and tested at each site by the participating laboratory.
- Organism collection, transport, confirmation of organism identification, and development and management of a centralized database were coordinated by Laboratories International for Microbiology Studies (LIMS), a division of International Health Management Associates, Inc. located in Schaumburg, IL, USA.
- All organisms were deemed clinically significant by local participant criteria. Isolate inclusion was independent of medical history, antimicrobial use, age or gender. All sites identified each study isolate utilizing local laboratory site criteria.

ANTIMICROBIAL SUSCEPTIBILITY TESTING

- Minimum inhibitory concentrations (MICs) were determined by the CLSI recommended broth microdilution testing method [6]. Tigecycline was supplied by Wyeth Pharmaceuticals (Collegeville, PA, USA). All other agents were supplied by the panel manufacturer, MicroScan (Dade Behring Inc., West Sacramento, CA, USA). The following antimicrobial agents and dilution ranges (expressed in mcg/mL) were included on the panels: tigecycline (0.008-16), imipenem (0.06-16), levofloxacin (0.008-8), minocycline (0.5-16), piperacillin/tazobactam (0.06/4-128/4), amikacin (0.5-32), ceftazidime (8-32), ceftriaxone (0.06-64), and cefepime (0.5-32). MIC interpretive criteria followed published guidelines established by the Clinical and Laboratory Standards Institute [7], where applicable. Tigecycline breakpoints were defined conservatively as susceptible <u><</u>2, intermediate = 4 and resistant <u>></u>8 mcg/mL.
- Quality controls (QC) were performed by each testing site on each day of testing using the corresponding ATCC control strains: *Escherichia coli* ATCC 25922, *Escherichia coli* ATCC 35218, and *Pseudomonas aeruginosa* ATCC 27853. Results were included in the analysis only when corresponding QC isolates tested within the acceptable range according to CLSI (2005) guidelines [7].

REFERENCES

1. Hoellman, D.B., et al., *Antipneumococcal activities of GAR-936 (a new glycylcycline) compared to those of nine other agents against penicillin-susceptible and -resistant pneumococci*. Antimicrob Agents Chemother, 2000. 44(4): p. 1085-8.
2. Labthavikul, P., P.J. Petersen, and P.A. Bradford, *In vitro activity of tigecycline against Staphylococcus epidermidis growing in an adherent-cell biofilm model*. Antimicrob Agents Chemother, 2003. 47(12): p. 3967-9.
3. Projan, S.J., *Preclinical pharmacology of GAR-936, a novel glycylcycline antibacterial agent.* Pharmacotherapy, 2000. 20(9 Pt 2): p. 219S-223S; discussion 224S-228S.
4. Henwood, C.J., et al., *Antibiotic resistance among clinical isolates of Acinetobacter in the UK, and in vitro evaluation of tigecycline (GAR-936)*. J Antimicrob Chemother, 2002. 49(3): p. 479-87.
5. Hanberger, H., et al., *Antibiotic susceptibility among aerobic gram-negative bacilli in intensive care units in 5 European countries. French and Portuguese ICU Study Groups*. Jama, 1999. 281(1): p. 67-71.
6. CLSI, *Methods for Dilution Antimicrobial Susceptibility Tests for Bacteria That Grow Aerobically; Approved Standard-Sixth Edition, in Document M7-A6.* 2005: Clinical Laboratory Standards Institute (CLSI), 940 West Valley Road, Suite 1400, Wayne, Pennsylvania 19087-1898 USA.
7. CLSI, *Performance Standards for Antimicrobial Susceptibility Testing, in Document M100-S15*. 2005: Clinical Laboratory Standards Institute (CLSI), 940 West Valley Road, Suite 1400, Wayne, Pennsylvania 19087-1898 USA.

ACKNOWLEDGEMENTS

This study was sponsored by a grant from Wyeth Pharmaceuticals. We gratefully acknowledge the contributions of the investigators, laboratory personnel and all 150 current members of the Tigecycline Evaluation Study Trials program group.

RESULTS

TIG inhibited 97% of *Acinetobacter* spp resistant to 2 or more drug classes at 2 mcg/mL (Table 2) and inhibited 184/192 (95.8%) and 39/42 (92.9%) of imipenem- and minocycline-resistant strains, respectively (no table). Other results are shown in the following tables.

Table 1. In Vitro Activity of Tigecycline and Comparators against 1,798 Isolates of *Acinetobacter* from a Global Population.

Drug	%Sus [a]	%Int	%Res	MIC (mcg/mL) MIC50	MIC90	Range
Tigecycline	98.4	1.5	0.1	0.25	1	<u><</u>0.008 - 8
Amikacin	85.1	6.9	8.0	4	32	<u><</u>0.5 - >64
Cefepime	47.8	15.7	36.5	16	>32	<u><</u>0.5 - >32
Ceftazidime	49.3	5.3	45.3	16	>32	<u><</u>8 - >32
Ceftriaxone	32.5	22.9	44.6	32	>64	<u><</u>0.06 - >64
Imipenem	87	6.7	6.3	0.5	8	<u><</u>0.06 - >16
Levofloxacin	50.4	5.1	44.5	2	>8	<u><</u>0.008 - >8
Minocycline	88.7	9.1	2.2	<u><</u>0.5	8	<u><</u>0.5 - >16
PipTazo	76.2	0	23.8	8	>128	<u><</u>0.06 - >128

[a]Interpretive criteria as defined by CLSI, M100-S15 (2005); tigecycline breakpoints are defined as susceptible <u><</u>2, intermediate = 4 and resistant <u>></u>8 mcg/mL, for comparison purposes only.

Table 2. Frequency Distribution and Cumulative Percent Inhibited (%) at each MIC (mcg/mL) for Tigecycline and Comparators for 503 Multi-Drug Resistant* *Acinetobacter* Isolates from the United States.

n/Cum%	MIC (mcg/mL) <u><</u>0.008	0.06	0.12	0.25	0.5	1	2	4	8	16	32	64	128	>128
Tigecycline	1		14	48	139	225	63	12	1					
	0.2		3.0	12.5	40.2	84.9	97.4	99.8	100.0					
Amikacin						8	39	89	47	50	64	76	130	
						1.6	9.3	27.0	36.4	46.3	59.0	74.2	100.0	
AmoxClav									1	8	23	471		
									0.2	1.8	6.4	100.0		
Ampicillin							1			3	3	496		
							0.2			0.8	1.4	100.0		
Cefepime								1	20	93	167	222		
								0.2	4.2	22.7	55.9	100.0		
Ceftazidime									11	21	71	400		
									2.2	6.4	20.5	100.0		
Ceftriaxone									1	5	9	32	456	
									0.2	1.2	3.0	9.3	100.0	
Imipenem				2	32	107	56	63	60	90	93			
				0.4	6.8	28.0	39.2	51.7	63.6	81.5	100.0			
Levofloxacin		1		1			9	39	144	309				
		0.2		0.4			2.2	9.9	38.6	100.0				
Minocycline					140	138	65	42	84	31	3			
					27.8	55.3	68.2	76.5	93.2	99.4	100.0			
PipTazo		2	1	2	1	1		4	1	11	17	22	159	282
		0.4	0.6	1.0	1.2	1.4		2.2	2.4	4.6	8.0	12.3	43.9	100.0

*Multi-drug resistance is defined as any bacterial strain that is resistant to 3 or more drug classes of antimicrobial agents.

Figure 1. Cumulative Percent Inhibited (%) for Tigecycline and Comparators at each MIC against 503 Multi-drug Resistant *Acinetobacter* spp.*

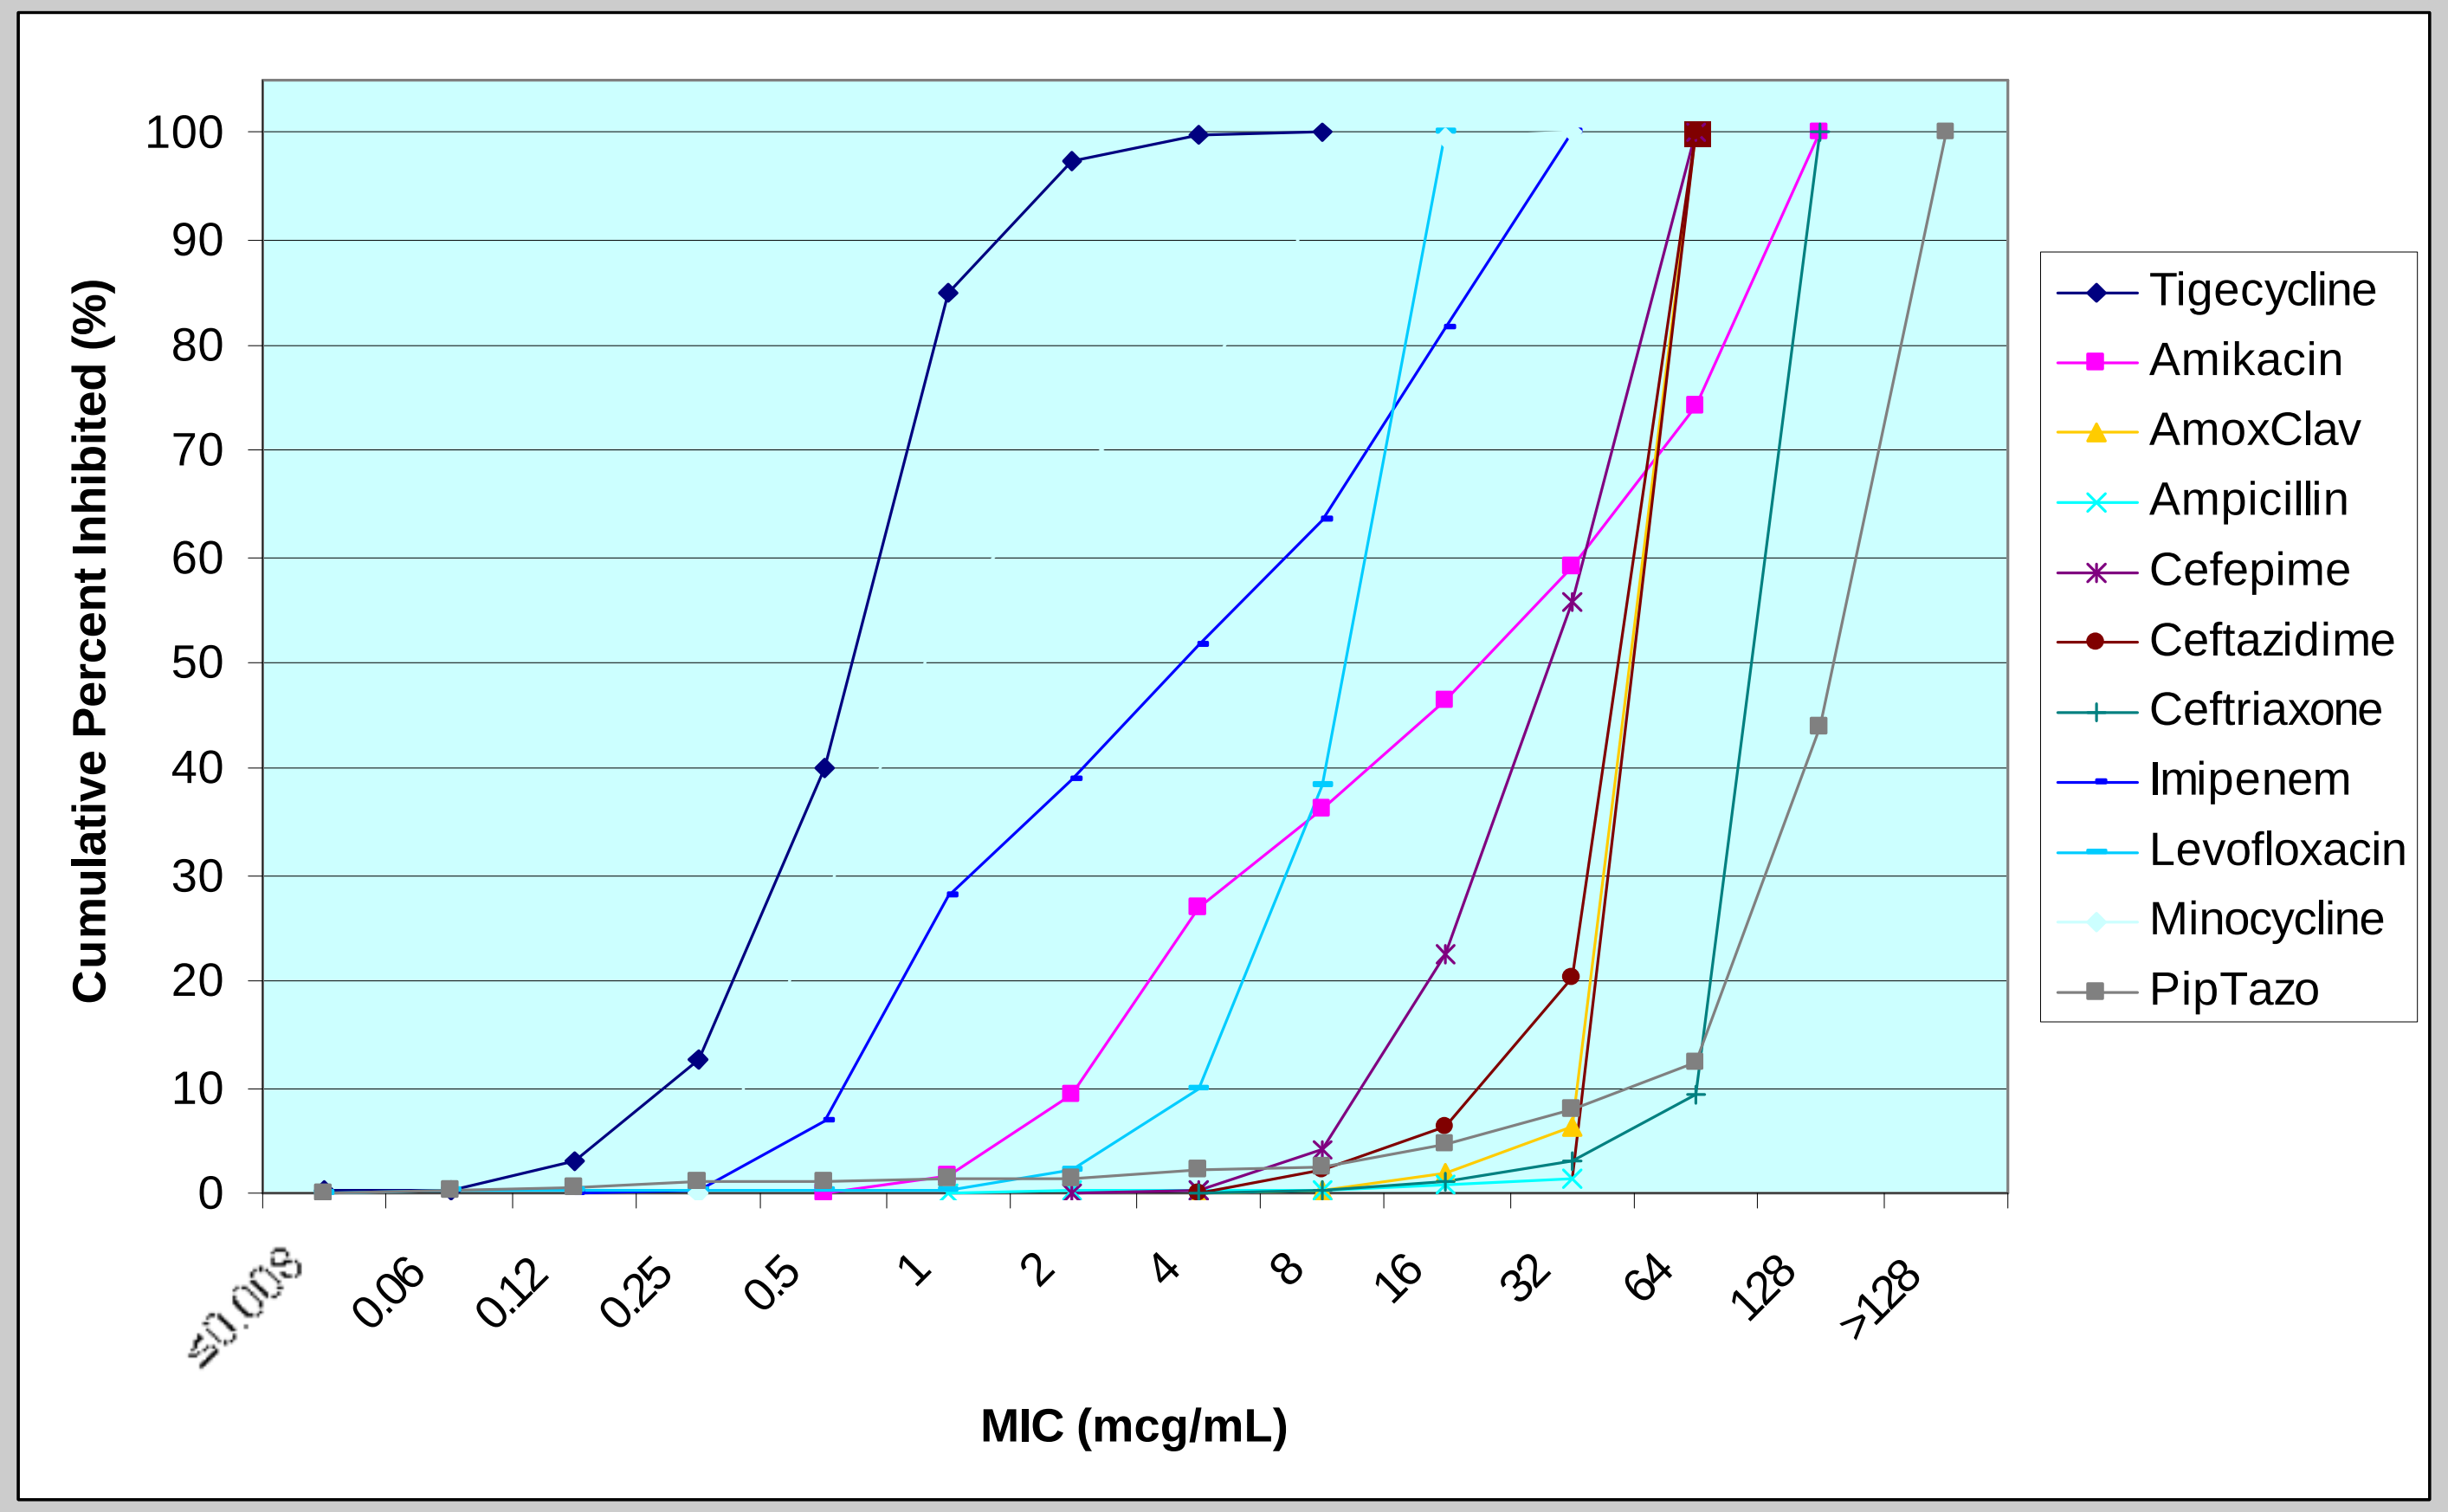

Cumulative Percent Inhibited (%)
0
10
20
30
40
50
60
70
80
90
100
≤0.008
0.06
0.12
0.25
0.5
1
2
4
8
16
32
64
128
>128
MIC (mcg/mL)
Tigecycline
Amikacin
AmoxClav
Ampicillin
Cefepime
Ceftazidime
Ceftriaxone
Imipenem
Levofloxacin
Minocycline
PipTazo

*Multi-drug resistance is defined as any bacterial strain that is resistant to 3 or more drug classes of antimicrobial agents.

CONCLUSIONS

- Tigecycline had the lowest in vitro MIC_{90} of all study drugs against all strains of *Acinetobacter* and at 1 mcg/mL. This value was 8- to 64-fold lower than imipenem, levofloxacin and cefepime.
- Tigecycline inhibited 97.4% of all multi-drug resistant (resistant to 2 or more drug classes) *Acinetobacter* strains at a MIC of 2 mcg/mL.
- The in vitro activity or tigecycline was superior to imipenem, cefepime, ceftazidime, ceftriaxone, levofloxacin, minocycline and piperacillin-tazobactam against all *Acinetobacter* resistant to 2 or more drug classes.
- The in vitro activity of tigecycline in this study suggests that tigecycline is a potent antimicrobial agent that may be beneficial in the treatment of infections due to drug resistant and multi-drug resistant *Acinetobacter*.