P 1685

In Vitro Activity of Tigecycline and 10 Common Therapeutic Agents Against Methicillin-Resistant *Staphylococcus aureus* and Vancomycin-Resistant *Enterococcus* species - Global Data, 2004 - 2006

M. Hackel[1], R. Badal[1], J. Johnson[1], D. Hoban[1], B. Johnson[1], S. Bouchillon[1], M. Dowzicky[2]

[1]International Health Management Associates, Schaumburg, IL, USA
[2]Wyeth Pharmaceuticals, Collegeville, PA, USA

IHMA, Inc.
2122 Palmer Dr.
Schaumburg, IL
60173
Tel: (847) 303-5003
Fax: (847) 303-5601
www.ihmainc.com

REVISED ABSTRACT

Background: Tigecycline (TIG), a member of a new class of antimicrobials (glycylcyclines), has been shown to have potent activity against community and hospital acquired staphylococcal and enterococcal pathogens. The T.E.S.T. program determined the in vitro activity against methicillin-resistant *S. aureus* and vancomycin-resistant *Enterococcus* spp. of TIG and 10 antimicrobials commonly prescribed for serious gram-positive infections: amoxicillin-clavulanic acid (AUG), piperacillin-tazobactam (PT), levofloxacin (LVX), ceftriaxone (CAX), linezolid (LZD), minocycline (MIN), vancomycin (VAN), ampicillin (AMP), penicillin (P), and imipenem (IMP). Study strains were collected from 320 laboratories in 42 countries globally throughout 2004-2006. **Methods:** A total of 11,586 clinical isolates (4,482 enterococci, 7,104 *S. aureus*) were identified to the species level at each participating site and confirmed by a central laboratory. Minimum Inhibitory Concentrations (MICs) were determined by the local laboratory using broth microdilution panels. Antimicrobial resistance was interpreted according to CLSI breakpoints with TIG susceptible breakpoints defined as ≤0.5 mcg/ml for *S. aureus* and ≤0.25 mcg/ml for enterococci. **Results:** 15.6% (697/4482) of enterococci were resistant to vancomycin (VRE), and 44.8% (3181/7104) of *S. aureus* were resistant to oxacillin (MRSA). Among the *E. faecium* VRE, % resistance rates to other study drugs were LVX 99.2, P 98.5, AMP 97.5, VAN 100, MIN 7.3, and LZD 0.0. Resistance rates for MRSA were P 100, AMP 100, AUG 100, LVX 66, PT 100, CAX 100, IMP 100, MIN 0.2, LZD 0.0, and VAN 0.0. TIG inhibited 100% of the enterococci and *S. aureus* resistant to other drugs. Modal TIG MICs for VRE/nonVRE were 0.03/0.06, and 0.12/0.12 for MRSA/MSSA). **Conclusion:** TIG retained potent activity against drug-resistant *S. aureus* and enterococcal isolates, inhibiting 100% of all strains tested at their defined breakpoints of 0.5 and 0.25 mcg/ml, respectively. TIG should prove to be a useful drug for therapy of infections with these resistant gram-positive pathogens.

INTRODUCTION

Tigecycline is a broad-spectrum antimicrobial agent and first-in-class of the semisynthetic glycylcyclines to be approved for human use [1]. This synthetic analogue of the minocycline molecule exhibits significant antibacterial activity that is both bacteriostatic and, in certain instances, bactericidal with killing activity that is as much as fourfold better than vancomycin and daptomycin [2, 3]. The development of tigecycline is important in that tigecycline and other glycylcyclines are active against bacterial strains carrying either or both of the two major forms of tetracycline resistance: efflux and ribosomal protection. Certain substituents at the 9-position of the tetracycline molecule restored activity against bacteria harboring genes encoding either or both efflux and ribosomal protection. A single chemical modification of tigecycline overcomes the two molecularly distinct forms of resistance while maintaining activity against susceptible gram-positive, gram-negative, aerobic, and anaerobic bacteria [4]. Furthermore, resistance to tigecycline is difficult to produce even in the laboratory.

Previous studies have demonstrated excellent in vitro activity for tigecycline against clinical and laboratory strains of gram-positive and gram-negative bacteria with minimum inhibitory concentrations for the 90th percentile inhibited at or below 2 mcg/ml, including difficult to treat methicillin-resistant *S. aureus* (MRSA), vancomycin-resistant enterococci (VRE) and extended-spectrum beta-lactamase (ESBL) producing *Enterobacteriaceae* [5-9]. This study was undertaken to document the in vitro activity of tigecycline against significant numbers of *S. aureus* and *Enterococcus* spp. collected from 42 countries. This study is part of the larger ongoing global Tigecycline Evaluation and Surveillance Trials (T.E.S.T.) program.

MATERIALS & METHODS

- All isolates were derived from blood, respiratory tract, urine, skin, wound, body fluids and other defined sources. Only one isolate per patient was accepted into the study. Clinical isolates were collected and tested between January 2004 and December 2006 from 320 study centers in 42 countries throughout the world. Isolates were identified to the species level and tested at each site by the participating laboratory.
- Organism collection, transport, confirmation of organism identification, as well as, development and management of a centralized database was coordinated by Laboratories International for Microbiology Studies (LIMS), a division of International Health Management Associates, Inc. located in Schaumburg, IL, USA.
- All organisms were deemed clinically significant by local participant criteria. Isolate inclusion was independent of medical history, antimicrobial use, age or gender. All sites identified each study isolate utilizing local laboratory site criteria.
- Minimum inhibitory concentrations (MICs) were determined by the CLSI recommended broth microdilution testing method [10]. Tigecycline was supplied by Wyeth Pharmaceuticals (Collegeville, PA, USA). All other agents were supplied by the panel manufacturer, MicroScan (Dade Behring Inc., Sacramento, CA, USA). The following antimicrobial agents were included on the panels with their dilution ranges (expressed in mcg/ml): amoxicillin/clavulanic acid (0.12/0.06-32/16); ampicillin (0.06-16); ceftriaxone (0.06-64); imipenem (0.06-16); linezolid (0.5-8); levofloxacin (0.008-8); minocycline (0.5-16); tigecycline (0.008-16); penicillin (0.06-8); piperacillin/tazobactam (0.06/4-128/4) and vancomycin (0.12-32). MIC interpretive criteria followed published guidelines established by the Clinical and Laboratory Standards Institute [11] and recent US Food and Drug Administration packaging insert for tigecycline [12], where applicable.
- Quality controls (QC) were performed by each testing site on each day of testing using ATCC control strains *S. aureus* ATCC 29213 and *Enterococcus faecalis* ATCC 29212. Results were included in the analysis only when corresponding QC isolates tested within the acceptable range according to CLSI (2006) guidelines [11].

REFERENCES

1 FDA, Tygacil (R), NDA No. N021821 http://www.fda.gov/cder/rdmt/ndaaps05cy.htm June 15, 2005, United States Federal Drug Administration (FDA), 5600 Fishers Lane, Rockville, MD, USA.
2. Hoellman, D.B., et al., *Antipneumococcal activities of GAR-936 (a new glycylcycline) compared to those of nine other agents against penicillin-susceptible and -resistant pneumococci*. Antimicrob Agents Chemother, 2000. 44(4): p. 1085-8.
3. Labthavikul, P., P.J. Petersen, and P.A. Bradford, *In vitro activity of tigecycline against Staphylococcus epidermidis growing in an adherent-cell biofilm model*. Antimicrob Agents Chemother, 2003. 47(12): p. 3967-9.
4. Projan, S.J., *Preclinical pharmacology of GAR-936, a novel glycylcycline antibacterial agent*. Pharmacotherapy, 2000. 20(9 Pt 2): p. 219S-223S; discussion 224S-228S.
5. Gales, A.C. and R.N. Jones, *Antimicrobial activity and spectrum of the new glycylcycline, GAR-936 tested against 1,203 recent clinical bacterial isolates*. Diagn Microbiol Infect Dis, 2000. 36(1): p. 19-36.
6. Patel, R., et al., *In vitro activity of GAR-936 against vancomycin-resistant enterococci, methicillin-resistant Staphylococcus aureus and penicillin-resistant Streptococcus pneumoniae*. Diagn Microbiol Infect Dis, 2000. 38(3): p. 177-9.
7. Rupp, M.E. and P.D. Fey, *Extended spectrum beta-lactamase (ESBL)-producing Enterobacteriaceae: considerations for diagnosis, prevention and drug treatment*. Drugs, 2003. 63(4): p. 353-65.
8. Bouchillon, S.K., et al., *In Vitro Activity of Tigecycline Against 3,989 Gram-Negative and Gram-Positive Clinical Isolates from the United States Tigecycline Evaluation and Surveillance Trial (TEST Program; 2004)*. Diagn Microbiol Infect Dis, 2005. 52(3): p. 173-179.
9. Hoban, D.J., et al., *In Vitro Activity of Tigecycline Against 6,792 Gram-Negative and Gram-Positive Clinical Isolates from the Global Tigecycline Evaluation and Surveillance Trial (TEST Program; 2004)*. Diagn Microbiol Infect Dis, 2005. 52(3): p. 215-227.
10. CLSI, Methods for Dilution Antimicrobial Susceptibility Tests for Bacteria That Grow Aerobically; Approved Standard-Sixth Edition, in Document M7-A6. 2005: Clinical Laboratory Standards Institute (CLSI), 940 West Valley Road, Suite 1400, Wayne, Pennsylvania 19087-1898 USA.
11. CLSI, Performance Standards for Antimicrobial Susceptibility Testing, in Document M100-S16. 2006: Clinical Laboratory Standards Institute (CLSI), 940 West Valley Road, Suite 1400, Wayne, Pennsylvania 19087-1898 USA.
12. Tygacil, Product Insert. 2005: Wyeth Pharmaceuticals, Inc., Philadelphia, PA, USA.

ACKNOWLEDGEMENTS

We gratefully acknowledge the contributions of the investigators, laboratory personnel and all members of the Tigecycline Evaluation Study Trials program group. This study was sponsored by a grant from Wyeth Pharmaceuticals.

RESULTS

The results are listed in the following Tables.

Table 1. Study organisms with phenotypes.

Genus/Species/Phenotype[a]	Phenotype	Total N
Staphylococcus aureus		7104
Staphylococcus aureus, MRSA	3181 (44.8%)	
Staphylococcus aureus, MSSA	3923 (55.2%)	
Enterococcus faecalis		3259
Enterococcus faecalis, VRE	105 (3.2%)	
Enterococcus faecium		1223
Enterococcus faecium, VRE	592 (48.4%)	
Total		**11,586**

[a] Phenotypes: MRSA = methicillin-resistant *S. aureus*; MSSA = methicillin-susceptible *S. aureus*. Methicillin phenotype was determined by the susceptibility of *S. aureus* to cefoxitin 30 mcg disk; VRE = vancomycin-resistant enterococci

Table 2. The in vitro activity of tigecycline and comparative agents against 7,104 clinical isolates of *Staphylococcus aureus*.

Organism / Phenotype	Drug	%Sus[a]	%Res	MIC (mcg/ml) MIC_{50}	MIC_{90}	Range
Methicillin-Resistant *Staphylococcus aureus* (n=3,181)	Tigecycline	100	0	0.12	0.25	0.015 - 0.5
	AmoxClav	0	100	8	>8	0.25 - >8
	Ampicillin	0	100	>16	>16	0.5 - >16
	Ceftriaxone	0	100	32	>64	0.12 - >64
	Imipenem	0	100	0.5	>16	≤0.12 - >16
	Levofloxacin	20.8	66	8	>32	≤0.06 - >32
	Linezolid	100	0	2	2	≤0.5 - 4
	Minocycline	97.6	0.2	≤0.25	2	≤0.25 - >8
	Penicillin	0	100	>8	>8	0.25 - >8
	PipTazo	0	100	16	>16	≤0.25 - >16
	Vancomycin	100	0	1	1	0.25 - 2
Methicillin-Susceptible *Staphylococcus aureus* (n=3,923)	Tigecycline	100	0	0.12	0.12	≤0.008 - 0.5
	AmoxClav	100	0	1	2	≤0.03 - 4
	Ampicillin	19.5	80.5	4	>16	≤0.06 - >16
	Ceftriaxone	99.4	0	2	4	≤0.03 - >64
	Imipenem	100	0	≤0.12	0.25	≤0.12 - 8
	Levofloxacin	96.8	1	0.12	0.25	≤0.06 - >32
	Linezolid	100	0	2	2	≤0.5 - 4
	Minocycline	99.5	0.1	≤0.25	≤0.25	≤0.25 - >8
	Penicillin	16.7	83.3	8	>8	≤0.06 - >8
	PipTazo	100	0	0.5	1	≤0.25 - 8
	Vancomycin	100	0	0.5	1	≤0.12 - 4

[a] Susceptibilities are defined in CLSI document M100-S16 (2006) where applicable. Methicillin phenotype based upon cefoxitin 30 mcg disk results; beta-lactam susceptibilities based on methicillin phenotype. Tigecycline breakpoints are defined in FDA package insert (Tygacil®, 2005) as susceptible ≤0.5 mcg/ml.

Table 3. The frequency distribution (n) and cumulative percent inhibited (%) in vitro activity of tigecycline and comparative agents against 3,181 methicillin-resistant *S. aureus*.[a]

N / Cum%	MIC (mcg/ml) ≤0.008	0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	64	>64
Tigecycline		1	13	462	2084	484	137								
		0	0.4	15	80.5	95.7	100								
AmoxClav						6	32	80	204	554	1051	1254			
						0.2	1.2	3.7	10.1	27.5	60.6	100			
Ampicillin							16	36	65	117	305	641	2001		
							0.5	1.6	3.7	7.4	16.9	37.1	100		
Ceftriaxone					1		1	10	41	62	294	962	398	209	1203
					0		0.1	0.4	1.7	3.6	12.9	43.1	55.6	62.2	100
Imipenem					213	692	585	290	189	129	119	173	508		
					7.3	31.2	51.4	61.4	67.9	72.4	76.5	82.5	100		
Levofloxacin				35		163	54	19	105	632	424	444	394	519	
				1.1		18.5	20.2	20.8	24.1	44	57.3	71.3	83.7	100	
Linezolid							56	684	2170	271					
							1.8	23.3	91.5	100					
Minocycline						2623	171	37	106	167	72	5			
						82.5	87.8	89	92.3	97.6	99.8	100			
Penicillin						8	15	29	48	112	242	2727			
						0.3	0.7	1.6	3.1	6.7	14.3	100			
PipTazo						5	34	84	144	341	546	595	1432		
						0.2	1.2	3.9	8.4	19.1	36.3	55	100		
Vancomycin						41	1313	1707	120						
						1.3	42.6	96.2	100						

[a] Methicillin phenotype based upon cefoxitin 30 mcg disk results according to CLSI document M100-S16, 2006.

Table 4. The in vitro activity of tigecycline and comparative antimicrobial agents against 3,259 *Enterococcus faecalis* and 1,223 *Enterococcus faecium* isolates from a global population.

Organism / Phenotype	Drug	%Sus[a]	%Res	MIC (mcg/ml) MIC_{50}	MIC_{90}	Range
E. faecalis (n=3259)	Tigecycline	100	0	0.12	0.12	≤0.008 - 0.25
	Ampicillin	100	0	1	1	≤0.06 - 8
	Levofloxacin	62.2	36.9	1	>32	≤0.06 - >32
	Linezolid	98.3	0	2	2	≤0.5 - 4
	Minocycline	43.8	11.9	8	>8	≤0.25 - >8
	Penicillin	100	0	2	4	≤0.06 - 8
	Vancomycin	96.5	3.2	1	2	≤0.12 - >32
E. faecalis VRE (n=105)	Tigecycline	100	0	0.06	0.12	0.03 - 0.25
	Ampicillin	100	0	1	2	0.25 - 8
	Levofloxacin	3.8	96.2	32	>32	1 - >32
	Linezolid	100	0	1	2	≤0.5 - 2
	Minocycline	62.9	4.8	4	8	≤0.25 - >8
	Penicillin	100	0	2	8	0.5 - 8
	Vancomycin	0	100	>32	>32	32 - >32
E. faecium (n=1223)	Tigecycline	100	0	0.06	0.12	≤0.008 - 0.25
	Ampicillin	16.8	83.2	>16	>16	≤0.06 - >16
	Levofloxacin	13.4	84.1	>32	>32	0.12 - >32
	Linezolid	96.2	0	2	2	≤0.5 - 4
	Minocycline	73.9	8.3	≤0.25	8	≤0.25 - >8
	Penicillin	14.8	85.2	>8	>8	≤0.06 - >8
	Vancomycin	50.4	48.2	4	>32	≤0.12 - >32
E. faecium VRE (n=592)	Tigecycline	100	0	0.03	0.12	0.015 - 0.25
	Ampicillin	2.5	97.5	>16	>16	0.5 - >16
	Levofloxacin	0.7	99.2	>32	>32	2 - >32
	Linezolid	96.6	0	2	2	≤0.5 - 4
	Minocycline	74.2	7.3	≤0.25	8	≤0.25 - >8
	Penicillin	1.5	98.5	>8	>8	2 - >8
	Vancomycin	0	100	>32	>32	16 - >32

[a] Interpretive criteria as defined by CLSI, M100-S16 (2006), where applicable; Tigecycline FDA breakpoints for enterococci are approved for vancomycin-susceptible *E. faecalis*, only; susceptibilities for all other enterococci are entered for comparison purposes only. [10]

Table 5. The frequency distribution (n) and cumulative percent inhibited (%) in vitro activity of tigecycline and comparative agents against 105 isolates of vancomycin-resistant *Enterococcus faecalis* from a global population.

N / Cum%	MIC (mcg/ml) ≤0.008	0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	64	>64
Tigecycline			9	55	36	5									
			8.6	61	95.2	100									
Ampicillin						3	32	54	9	6	1				
						2.9	33.3	84.8	93.3	99	100				
Ceftriaxone				1			1				1	4	6	8	84
				1			1.9				2.9	6.7	12.4	20	100
Levofloxacin								3	1		1	15	40	45	
								2.9	3.8		4.8	19	57.1	100	
Linezolid							5	60	40						
							4.8	61.9	100						
Minocycline						32		2	7	25	34	5			
						30.5		32.4	39	62.9	95.2	100			
Penicillin							2	7	55	30	11				
							1.9	8.6	61	89.5	100				
Vancomycin													8	97	
													7.6	100	

Table 6. The frequency distribution (n) and cumulative percent inhibited (%) in vitro activity of tigecycline and comparative agents against 592 isolates of vancomycin-resistant *Enterococcus faecium* from a global population.

N / Cum%	MIC (mcg/ml) <0.008	0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	64	>64
Tigecycline		5	302	217	67	1									
		0.8	51.9	88.5	99.8	100									
Ampicillin							2	2		3	8	7	570		
							0.3	0.7		1.2	2.5	3.7	100		
Ceftriaxone			1							1	3	2	3	2	580
			0.2							0.3	0.8	1.2	1.7	2	100
Levofloxacin									4	1		22	66	499	
									0.7	0.8		4.6	15.7	100	
Linezolid							14	212	346	20					
							2.4	38.2	96.6	100					
Minocycline						332	12	10	16	69	110	43			
						56.1	58.1	59.8	62.5	74.2	92.7	100			
Penicillin									2	4	3	583			
									0.3	1	1.5	100			
Vancomycin												2	17	573	
												0.3	3.2	100	

CONCLUSIONS

- Tigecycline inhibited 100% of all *Staphylococcus aureus* and *Enterococcus* spp. at the FDA susceptible breakpoints of 0.5 and 0.25 mcg/ml, respectively, without regard to either methicillin- or vancomycin-resistant phenotypes.
- Tigecycline demonstrated equivalent in vitro potency to vancomycin and linezolid against all *Staphylococcus aureus* with MIC_{90} value of 0.25 mcg/ml.
- Tigecycline's MIC_{90} of 0.12 mcg/ml against both vancomycin-resistant *E. faecalis* and vancomycin-resistant *E. faecium* was the lowest of all comparator agents in this study.
- The in vitro activity of tigecycline in this study suggests that tigecycline is highly active against methicillin-resistant *Staphylococcus aureus* and vancomycin-resistant *Enterococcus* species and may be an effective treatment option for these increasingly difficult to treat phenotypes.