P1600

The Tigecycline Evaluation Surveillance Trial (TEST) in Canada from 2004-2008

M. Renteria[1], J. Johnson[1], R. Badal[1], S. Hawser[2], M. Hackel[1], S. Bouchillon[1], B. Johnson[1], D. Hoban[1], M. Dowzicky[3]

[1]International Health Management Associates, Inc., Schaumburg, IL, USA
[2]International Health Management Associates Europe Sàrl, Epalinges, Switzerland
[3]Wyeth Pharmaceuticals, Collegeville, PA, USA

IHMA, Inc.
2122 Palmer Dr.
Schaumburg, IL
60173
Tel: 847.303.5003
Fax: 847.303.5601

Revised Abstract

Background: Tigecycline (TIG) is a new glycylcycline with enhanced activity against many multidrug resistant (MDR) pathogens including ESBL and AmpC producing *Enterobacteriaceae*, methicillin-resistant *S. aureus* (MRSA), carbapenem resistant *Acinetobacter* and fluoroquinolone resistant gram-negative rods. The TEST study evaluated the activity of TIG and comparators to pathogens in Canada 2004-2008. **Methods**: A total of 2578 pathogens were collected from 25 participating sites in Canada from 2004-2008. Isolates were identified to the species level and CLSI specified MICs were performed at each site. CLSI or FDA breakpoints were used, where applicable, to determine % susceptibility. **Results**: Tigecycline MICs are recorded in the following table:

Organisms (n=2578)	Tigecycline MICs (mg/L)		
	MIC_{50}	MIC_{90}	Range
Acinetobacter spp (n=204)	0.12	1	0.015 - 2
Enterobacter spp. (n=356)	0.5	1	0.12 - 8
E. coli (n=384)	0.12	0.5	≤0.008 – 2
Klebsiella spp. (n=371)	0.5	2	0.12 – 16
ESBLs (n=14)	1	2	0.25 – 2
Enterococcus spp. (n=230)	0.06	0.12	0.03 – 0.25
VREs (n=9)	0.03	0.06	0.03 – 0.06
Serratia spp (n=150)	1	2	0.25 – 8
H. influenzae (n=200)	0.25	0.5	0.03 – 2
S. aureus (n=341)	0.12	0.25	0.06 – 0.5
MRSA (n=46)	0.12	0.25	0.06 – 0.5
S. agalactiae (n=139)	0.03	0.06	≤0.008 – 0.12
S. pneumoniae (n=203)	0.03	0.03	≤0.008 – 0.12

Conclusions: Tigecycline showed excellent *in vitro* activity against a diverse collection of pathogens isolated in Canada between 2004-2008. MIC_{90} values of ≤0.5mg/L against most *Enterobacteriaceae* including ESBL and MIC_{90} of ≤0.12mg/L against most gram-positive pathogens document the *in vitro* potency of tigecycline, a new glycylcycline.

Introduction

Tigecycline belongs to the glycylcycline class of antimicrobials and demonstrates expanded broad-spectrum activity. Tigecycline inhibits protein synthesis by binding to the 30S ribosomal subunit and has shown some bactericidal activity against key targeted pathogens [1, 2]. Tigecycline was developed to provide activity against tetracycline and multi-drug-resistant pathogens and has demonstrated broad-spectrum activity against aerobic and anaerobic gram-positive and gram-negative microorganisms [2-4].

With the exception of *P. aeruginosa*, tetracycline-resistant bacteria with either tetracycline efflux pumps or ribosomal protective features are sensitive to tigecycline [2-4]. Tigecycline has shown to be active against multi-resistant *Acinetobacter* spp. Similar activity has been observed against *Enterobacteriaceae*, including extended-spectrum b-lactamase (ESBL) and AmpC producing strains [5]. Tigecycline has demonstrated MIC_{90} values of ≤0.5 mg/L against methicillin-resistant *Staphylococcus aureus* (MRSA) and other gram-positive organisms [2,4].

This study was designed to better define the *in vitro* activity of tigecycline in selected clinical isolates collected from 25 study centers in Canada and is part of the global Tigecycline Evaluation Surveillance Study (TEST).

Materials & Methods

- All isolates were derived from blood, respiratory tract, urine (no more than 25% of all isolates), skin, wound, fluids and few other defined sources. Only one isolate per patient was accepted.
- Clinical isolates (n=2578) were collected tested between January 2004 – December 2008 from 25 study centers across Canada.
- Custom broth microdilution panels were supplied by MicroScan (Dade MicroScan, Sacramento, CA, USA) with the following antimicrobial agents and concentrations (expressed in mg/L); gram-positive panel: amoxicillin/clavulanic acid (0.03/0.015-8/4, tested using a 2:1 ratio of amoxicillin:clavulanic acid; reported concentrations refer to amoxicillin); ampicillin (0.06-16); ceftriaxone (0.03-64); imipenem (0.06-16); linezolid (0.5-8); levofloxacin (0.06-32); minocycline (0.25-8); tigecycline (0.008-16); penicillin (0.06-8); piperacillin/tazobactam (0.25/4-16/4) and vancomycin (0.12-32). Gram-negative panel: amikacin (0.5-64); amoxicillin/clavulanic acid (0.12/0.06-32/16, tested using a 2:1 ratio of amoxicillin:clavulanic acid; reported concentrations refer to amoxicillin); ampicillin (0.5-32); cefepime (0.5-32); ceftriaxone (0.06-64); ceftazidime (8-32); imipenem (0.06-16); levofloxacin (0.008-8); minocycline (0.5-16); tigecycline (0.008-16) and piperacillin/tazobactam (0.06/4-128/4).
- MIC interpretive criteria followed published guidelines established by the CLSI where applicable [6].
- MIC interpretive criteria for Tigecycline followed published guidelines established by the FDA where applicable [7].
- Isolates were identified to genus and species by the local laboratory. Each site tested the isolates using broth microdilution.
- Quality control of broth microdilution panels followed manufacture's and CLSI guidelines using the following ATCC strains: *Enterococcus faecalis* ATTC 29212; *Escherichia coli* ATCC 25922; *Haemophilus influenzae* ATCC 49247; *Haemophilus influenzae* ATCC 49766; *Staphylococcus aureus* ATCC 29213; *Streptococcus pneumoniae* ATCC 49619; and *Pseudomonas aeruginosa* ATCC 27853.
- ESBL determinations: *E. coli, K. pneumoniae* and *K. oxytoca* were screened and confirmed for ESBL activity according to CLSI guidelines. Preliminary ESBL activity was determined by screening ceftriaxone with MICs >1 mg/L using broth microdilution panels. ESBL presence was confirmed by testing the following antibiotic disks: cefotaxime (30-mcg), cefotaxime/clavulanic acid (30/10-mcg) and ceftazidime (30-mcg), ceftazidime/clavulanic acid (30/10-mcg). Antimicrobial disks were manufactured by Oxoid, Inc. (Ogdensburg, NY, USA). Mueller-Hinton agar used in testing was manufactured by Remel, Inc. (Lenexa, KS, USA). An organism was interpreted as containing an ESBL if there was an increase of >5mm in the inhibition zone of the combination disk when compared to that of the cephalosporin alone. Quality control of antimicrobial disks followed manufacturer's guidelines (Oxoid) using the following ATCC strains: *K. pneumoniae* ATCC 700603 (positive ESBL control) and *E. coli* ATCC 25922.
- The collection and transportation of organisms and the confirmation of identification, as well as, construction and management of a centralized database were conducted and coordinated by Laboratories International for Microbiology Studies (LIMS), a subsidiary of International Health Management Associates, Inc. (IHMA, Schaumburg, IL, USA).

References

1 Sum, P.E. and P. Petersen, *Synthesis and structure-activity relationship of novel glycylcycline derivatives leading to the discovery of GAR-936.* Bioorg Med Chem Lett, 1999. **9**(10): p. 1459-62.
2 Abbanat, D., M. Macielag, and K. Bush, *Novel antibacterial agents for the treatment of serious gram-positive infections.* Expert Opin Investig Drugs, 2003. **12**(3): p. 379-99.
3 Betriu, C., et al., *In vitro activities of tigecycline (GAR-936) against recently isolated clinical bacteria in Spain.* Antimicrob Agents Chemother, 2002. **46**(3): p. 892-5.
4 Gales, A.C. and R.N. Jones, *Antimicrobial activity and spectrum of the new glycylcycline, GAR-936 tested against 1,203 recent clinical bacterial isolates.* Diagn Microbiol Infect Dis, 2000. **36**(1): p. 19-36.
5 Petersen, P.J., et al., *In vitro and in vivo antibacterial activities of a novel glycylcycline, the 9-t-butylglycylamido derivative of minocycline (GAR-936).* Antimicrob Agents Chemother, 1999. **43**(4): p. 738-44.
6 National Committee for Clinical Laboratory Standards (CLSI). Performance Standards for Antimicrobial Susceptibility Testing; Fourteenth Informational Supplement. CLSI document M100-S15. Wayne, PA, 2005.
7 Tygacil®, 2005. Tigecycline FDA package insert.

Acknowledgements

We gratefully acknowledge the contributions of the investigators, laboratory personnel, and all members of the Tigecycline Evaluation Study Trials program group. This study was sponsored by a grant from Wyeth Pharmaceuticals.

Results

Table 1. *In vitro* activity of tigecycline and comparative agents against various species of E*nterobacteriaceae*.

Organism	Drug	%Sus	%Int	%Res	MIC (mg/L) MIC_{50}	MIC_{90}
All *Enterobacteriaceae*	Tigecycline	97.4	1.8	0.8	0.5	1
(n=1,261)	Amikacin	99.8	0.1	0.2	2	4
	Amox/Clav	50.4	7.1	42.6	8	>32
	Ampicillin	15	8.5	76.5	>32	>32
	Cefepime	97.5	0.5	2	≤0.5	1
	Ceftazidime	93.3	2	4.7	≤8	≤8
	Ceftriaxone	92	2.5	5.5	≤0.06	4
	Imipenem	100	0	0	0.12	0.5
	Levofloxacin	88.2	2.1	9.8	0.06	4
	Minocycline	86.9	6.8	6.3	2	8
	Pip-Tazo	94.2	2.6	3.2	1	8
Enterobacter aerogenes	Tigecycline	96.9	0	3.1	0.5	2
(n=65)	Amikacin	100	0	0	2	4
	Amox/Clav	3.1	3.1	93.8	>32	>32
	Ampicillin	0	9.2	90.8	>32	>32
	Cefepime	100	0	0	≤0.5	≤0.5
	Ceftazidime	89.2	4.6	6.2	≤8	16
	Ceftriaxone	96.9	3.1	0	0.12	2
	Imipenem	100	0	0	0.25	1
	Levofloxacin	95.4	1.5	3.1	0.06	1
	Minocycline	87.7	10.8	1.5	2	8
	Pip/Tazo	93.8	6.2	0	2	16
Enterobacter cloacae	Tigecycline	96.5	2.8	0.7	0.5	1
(n=284)	Amikacin	100	0	0	2	2
	Amox/Clav	1.4	0.7	97.9	>32	>32
	Ampicillin	0.7	5.6	93.7	>32	>32
	Cefepime	98.2	1.1	0.7	≤0.5	2
	Ceftazidime	84.2	3.5	12.3	≤8	>32
	Ceftriaxone	83.1	4.2	12.7	0.25	64
	Imipenem	100	0	0	0.12	0.5
	Levofloxacin	96.1	2.1	1.8	0.03	0.25
	Minocycline	85.6	8.1	6.3	2	8
	Pip/Tazo	87.3	6	6.7	2	64
Escherichia coli	Tigecycline	100	0	0	0.12	0.5
(n=384)	Amikacin	99.5	0.3	0.3	2	4
	Amox/Clav	75.5	15.6	8.9	8	16
	Ampicillin	48.1	0.8	51.1	32	>32
	Cefepime	95.1	0.3	4.7	≤0.5	1
	Ceftazidime	95.8	1.6	2.6	≤8	≤8
	Ceftriaxone	93	1.3	5.7	≤0.06	0.5
	Imipenem	100	0	0	≤0.06	0.25
	Levofloxacin	74	1.3	24.7	0.03	>8
	Minocycline	87	7.3	5.7	1	8
	Pip/Tazo	97.1	1.6	1.3	1	4
Klebsiella pneumoniae	Tigecycline	94.6	4.4	1	0.5	2
(n=297)	Amikacin	100	0	0	1	2
	Amox/Clav	91.2	5.7	3	2	8
	Ampicillin	0.7	21.2	78.1	32	>32
	Cefepime	98.3	0.3	1.3	≤0.5	≤0.5
	Ceftazidime	95.6	1.7	2.7	≤8	≤8
	Ceftriaxone	95.3	2	2.7	≤0.06	0.25
	Imipenem	100	0	0	≤0.06	0.5
	Levofloxacin	93.3	1.7	5.1	0.06	1
	Minocycline	82.8	6.7	10.4	2	16
	Pip/Tazo	97.3	1	1.7	2	8
Klebsiella oxytoca	Tigecycline	98.6	0	1.4	0.25	0.5
(n=71)	Amikacin	100	0	0	2	2
	Amox/Clav	85.9	4.2	9.9	2	16
	Ampicillin	0	7	93	>32	>32
	Cefepime	100	0	0	≤0.5	0.5
	Ceftazidime	100	0	0	≤8	≤8
	Ceftriaxone	97.2	1.4	1.4	≤0.06	2
	Imipenem	100	0	0	≤0.06	0.5
	Levofloxacin	97.2	2.8	0	0.03	0.12
	Minocycline	97.2	2.8	0	1	4
	Pip/Tazo	88.7	0	11.3	1	4
S. marcescens	Tigecycline	97.9	1.4	0.7	1	2
(n=145)	Amikacin	99.3	0	0.7	2	4
	Amox/Clav	2.1	1.4	96.6	>32	>32
	Ampicillin	0.7	7.6	97.7	>32	>32
	Cefepime	99.3	0	0.7	≤0.5	≤0.5
	Ceftazidime	98.6	0.7	0.7	≤8	≤8
	Ceftriaxone	95.2	4.1	0.7	0.25	1
	Imipenem	100	0	0	0.12	1
	Levofloxacin	92.4	4.1	3.4	0.12	1
	Minocycline	92.4	4.1	3.4	2	4
	Pip/Tazo	96.6	2.1	1.4	1	4
All ESBL producers	Tigecycline	100	0	0	0.25	1
E. coli,	Amikacin	100	0	0	2	8
K. pneumoniae,	Amox/Clav	44.4	47.2	8.3	16	16
K. oxytoca	Ampicillin	0	0	100	>32	>32
(n=36)	Cefepime	44.4	5.6	50	16	>32
	Ceftazidime	38.9	19.4	41.7	16	>32
	Ceftriaxone	16.7	16.7	66.7	>64	>64
	Imipenem	100	0	0	0.25	0.25
	Levofloxacin	25	5.6	69.4	>8	>8
	Minocycline	61.1	19.4	19.4	>8	>8
	Pip/Tazo	91.7	5.6	2.8	4	16
Levofloxacin Resistant	Tigecycline	95.9	2.4	1.6	0.25	2
Enterobacteriaceae (n=123)	Amikacin	97.6	0.8	1.6	2	8
	Amox/Clav	50.4	28.5	21.1	8	>32
	Ampicillin	14	1.7	84.3	>32	>32
	Cefepime	80.5	1.6	17.9	≤0.5	>32
	Ceftazidime	81.3	4.9	13.8	≤8	>32
	Ceftriaxone	70.7	5.7	23.6	0.12	>64
	Imipenem	100	0	0	0.25	0.5
	Levofloxacin	0	0	100	>8	>8
	Minocycline	69.9	12.2	17.9	2	16
	Pip/Tazo	89.4	4.9	5.7	2	32
AmpC positive *Enterobacter spp.& S. marcescens*	Tigecycline	92	8	0	0.5	2
(n=50)	Amikacin	100	0	0	2	4
	Amox/Clav	0	0	100	>32	>32
	Ampicillin	0	0	100	>32	>32
	Cefepime	100	0	0	4	8
	Ceftazidime	0	0	100	>32	>32
	Ceftriaxone	0	0	100	>64	>64
	Imipenem	100	0	0	0.25	0.5
	Levofloxacin	88	4	8	0.03	4
	Minocycline	80	12	8	4	8
	Pip/Tazo	4	40	56	128	>128

Table 2. *In vitro* activity of tigecycline and comparative agents against *Acinetobacter baumannii* and *Pseudomonas aeruginosa.*

Organism	Drug	%SUS	%INT	%RES	MIC (mg/L) MIC_{50}	MIC_{90}
Acinetobacter	Tigecycline	na	na	na	0.25	1
baumannii (n=161)	Amikacin	91.9	1.9	6.2	2	16
	Cefepime	79.5	9.3	11.2	4	32
	Ceftazidime	82.0	10.6	7.5	≤8	16
	Ceftriaxone	45.3	41.0	13.7	16	64
	Imipenem	98.7	0	1.3	0.5	1
	Levofloxacin	83.9	6.8	9.3	0.12	4
	Minocycline	98.8	1.2	0	≤0.5	≤0.5
	Pip/Tazo	85.7	8.1	6.2	0.25	64
Acinetobacter spp.	Tigecycline	na	na	na	1	2
Carbapenem R (n=6)	Amikacin	66.7	0	33.3	16	>64
	Cefepime	0	0	100	>32	>32
	Ceftazidime	0	0	100	>32	>32
	Ceftriaxone	0	0	100	>64	>64
	Imipenem	0	0	100	>16	>16
	Levofloxacin	0	16.7	83.3	8	>8
	Minocycline	100	0	0	≤0.5	2
	Pip/Tazo	0	16.7	83.3	>128	>128
Pseudomonas	Tigecycline	na	na	na	8	16
aeruginosa (n=312)	Amikacin	96.2	1.0	2.9	4	8
	Cefepime	83.0	10.9	6.1	4	16
	Ceftazidime	87.8	5.8	6.4	≤8	16
	Ceftriaxone	19.2	37.2	43.6	32	>64
	Imipenem	87.8	2.9	9.4	1	8
	Levofloxacin	72.1	9.6	18.3	0.5	>8
	Minocycline	na	na	na	16	>16
	Pip/Tazo	93.6	0	6.4	4	32

Table 3. *In vitro* activity of tigecycline and comparative agents against non-fastidious gram-positive pathogens.

Organism	Drug	%SUS*	%INT	%RES	MIC (mg/L) MIC_{50}	MIC_{90}
S. aureus	Tigecycline	100	0	0	0.12	0.25
(n=341)	Amox-Clav	88.3	0	11.7	1	4
	Ampicillin	61.9	0	38.1	4	>16
	Ceftriaxone	87.1	3.2	9.7	4	16
	Imipenem	88.5	0	11.5	≤0.12	0.25
	Levofloxacin	86.2	2.3	11.4	0.12	16
	Linezolid	100	0	0	2	2
	Minocycline	99.1	0.6	0.3	≤0.25	≤0.25
	Penicillin	61.3	0	38.7	4	>8
	Pip-Tazo	88.3	0	11.7	1	8
	Vancomycin	100	0	0	0.5	1
S. aureus MRSA	Tigecycline	100	0	0	0.12	0.25
(n=46)	Amox-Clav	13	0	87.0	8	>8
	Ampicillin	0	0	100	16	>16
	Ceftriaxone	4.3	23.9	71.7	32	>64
	Imipenem	0	0	100	0.5	16
	Levofloxacin	23.9	4.3	71.7	16	>32
	Linezolid	100	0	0	2	2
	Minocycline	97.8	0	2.2	≤0.25	0.5
	Penicillin	0	0	100	>8	>8
	Pip-Tazo	13	0	87	16	>16
	Vancomycin	100	0	0	0.5	1
E. faecalis	Tigecycline	100	0	0	0.12	0.12
(n=151)	Ampicillin	100	0	0	0.5	1
	Levofloxacin	58.3	0	41.7	1	>32
	Linezolid	99.2	0.8	0	2	2
	Minocycline	43.0	43.7	13.2	8	>8
	Penicillin	100	0	0	2	4
	Vancomycin	100	0	0	1	2
E. faecium	Tigecycline	100	0	0	0.06	0.12
(n=48)	Ampicillin	18.8	0	81.3	>16	>16
	Levofloxacin	10.4	0	89.6	>32	>32
	Linezolid	100	0	0	2	2
	Minocycline	81.3	14.6	4.2	≤0.25	8
	Penicillin	18.8	0	81.3	>8	>8
	Vancomycin	79.2	0	20.8	1	>32
Vancomycin resistant	Tigecycline	100	0	0	0.03	0.06
E. faecium	Ampicillin	0	0	100	>16	>16
(n=9)	Levofloxacin	0	0	100	>32	>32
	Linezolid	100	0	0	1	2
	Minocycline	66.7	33.3	0	2	8
	Penicillin	0	0	100	>8	>8
	Vancomycin	0	0	100	>32	>32
S. agalactiae	Tigecycline	100	0	0	0.03	0.06
(n=139)	Ampicillin	100	0	0	0.12	0.12
	Ceftriaxone	100	0	0	0.06	0.12
	Levofloxacin	100	0	0	0.5	1
	Linezolid	100	0	0	1	1
	Penicillin	100	0	0	≤0.06	0.12
	Vancomycin	100	0	0	0.5	0.5

* FDA susceptible breakpoint of 0.25 mg/L for vancomycin-susceptible *E. faecalis* was expanded to include all enterococci for comparative purposes only.

Table 4. *In vitro* activity of tigecycline and comparative agents against *S. pneumoniae* and *H. influenzae.*

Organism (n)	Drug	%Sus	%Int	%Res	MIC (mg/L) MIC_{50}	MIC_{90}
S. pneumoniae	Tigecycline	na	na	na	0.03	0.03
(n=203)	Amox-Clav	98.5	1.0	0.5	≤0.03	0.25
	Ceftriaxone	97.0	2.5	0.5	≤0.03	0.25
	Imipenem	89.9	10.1	0	≤0.12	0.25
	Levofloxacin	99.5	0	0.5	1	1
	Linezolid	100	0	0	1	1
	Penicillin	80.3	16.3	3.4	≤0.06	0.5
	Vancomycin	100	0	0	0.25	0.5
S. pneumoniae	Tigecycline	na	na	na	0.03	0.03
Susceptible to penicillin	Amox-Clav	100	0	0	≤0.03	≤0.03
(n=163)	Ceftriaxone	100	0	0	≤0.03	≤0.03
	Imipenem	100	0	0	≤0.12	≤0.12
	Levofloxacin	99.3	0	0.7	1	1
	Linezolid	100	0	0	≤0.5	1
	Penicillin	100	0	0	≤0.06	≤0.06
	Vancomycin	100	0	0	0.25	0.5
S. pneumoniae	Tigecycline	na	na	na	0.03	0.06
Intermediate to penicillin	Amox-Clav	100	0	0	0.25	2
(n=33)	Ceftriaxone	96.8	3.2	0	0.12	0.5
	Imipenem	57.1	42.9	0	0.25	0.25
	Levofloxacin	100	0	0	1	1
	Linezolid	100	0	0	1	1
	Penicillin	0	100	0	0.5	1
	Vancomycin	100	0	0	0.25	0.5
S. pneumoniae Resistant	Tigecycline	na	na	na	0.03	0.03
to penicillin	Amox-Clav	57.1	28.6	14.3	2	8
(n=7)	Ceftriaxone	28.6	57.1	14.3	1	2
	Imipenem	66.7	33.3	0	0.5	1
	Levofloxacin	100	0	0	1	1
	Linezolid	100	0	0	1	2
	Penicillin	0	0	100	4	8
	Vancomycin	100	0	0	0.25	0.5
Haemophilus	Tigecycline	na	na	na	0.25	0.5
influenzae (n=200)	Amox/Clav	100	0	0	0.5	1
	Ampicillin	81.5	1.5	17.0	≤0.5	16
	Cefepime	99.5	0	0.5	≤0.5	≤0.5
	Ceftazidime	na	na	na	≤8	≤8
	Ceftriaxone	100	0	0	≤0.06	≤0.06
	Imipenem	100	0	0	0.12	0.5
	Levofloxacin	100	0	0	0.015	0.015
	Pip/Tazo	99.4	0	0.6	≤0.06	≤0.06
Haemophilus	Tigecycline	na	na	na	0.25	0.5
influenzae β-	Amox/Clav	100	0	0	1	1
Lactamase Positive	Ampicillin	0	2.9	97.1	32	>32
(n=35)	Cefepime	100	0	0	≤0.5	≤0.5
	Ceftazidime	na	na	na	≤8	≤8
	Ceftriaxone	100	0	0	≤0.06	≤0.06
	Imipenem	100	0	0	0.12	1
	Levofloxacin	100	0	0	0.015	0.015
	Pip/Tazo	100	0	0	≤0.06	≤0.06

Conclusions

- Tigecycline's MIC_{90} of 1 mg/L was similar to imipenem's MIC_{90} of 0.5mg/L and 4 to 64 fold better than the ß-lactams, ß-lactam/ß-lactamase inhibitor combinations and levofloxacin against all *Enterobacteriaceae* tested.
- Tigecycline demonstrated excellent *in vitro* activity against both ESBL and non-ESBL producing *E. coli, K. oxytoca* and *K. pneumonia* with MIC_{90} of 1mg/L.
- Tigecycline showed potent activity against *A. baumannii* with low MIC_{50}/ MIC_{90} values of 0.25 and 1 mg/L, respectively.
- Tigecycline demonstrated limited activity against *P. aeruginosa* with MIC_{90} of 16mg/L.
- Tigecycline showed potent inhibitory activity against *S. aureus* and retained this activity regardless of phenotype.
- At 0.12 mg/L Tigecycline had the lowest MIC_{90} value of all comparative agents against both vancomycin-sensitive and –resistant strains of *E. faecium* and *E. faecalis*.
- Tigecycline was very active against *S. pneumonia* regardless of resistant phenotype (0.03-0.06mg/L).
- Tigecycline presented a MIC_{90} value of 0.5 mg/L against *H. influenzae* and as expected was unaffected by the presence of ß-lactamase.