#P2024

Invasive *Streptococcus pneumoniae* Serotypes Associated with Adult and Pediatric Isolates from the United States

M. Hackel[1], C. Lascols[1], D. Hoban[1], S. Bouchillon[1], J. Johnson[1], M. Dowzicky[2]

[1]International Health Management Associates, Schaumburg, IL, USA
[2]Pfizer Inc, Collegeville, PA, USA

IHMA, Inc.
2122 Palmer Dr.
Schaumburg, IL
60173
Tel: 847.303.5003
Fax: 847.303.5601

Revised Abstract

Background: *Streptococcus pneumoniae* (SPN) is a major cause of invasive diseases and upper respiratory tract infections. Introduction of the pneumococcal conjugate 7-valent vaccine (PV7) into the US childhood vaccine schedule in 2000 has significantly reduced invasive pneumococcal disease in children and adults, with concurrent reduction in the seven vaccine serotypes. Consistent monitoring of possible replacement serotypes is essential to determine possible antibiotic resistance patterns as certain serotypes are more closely associated with antibiotic resistance, as well as to determine targets for future vaccines. In this study we evaluate the serotypes of invasive SPN isolates from pediatric and adult patients from 2004. **Methods:** The capsular serotypes of 368 invasive SPN isolates collected in the US through the Tigecycline Evaluation Surveillance Trial were determined using sequential multiplex PCR and confirmed using the Quellung reaction. Invasive isolates were defined as those from normally sterile sites, such as blood, CSF and other body fluids. **Results:** Results are presented in the following table.

	Total N	PV7 serotypes N(%)	Non-PV7 Serotypes N(%)							
			19A	**22F/A**	**3**	**35**	**15**	**7**	**others**	
Pediatric	57	13 (23)	16(28)	5(9)	2(4)	0(0)	2(4)	3(5)	16(28)	
Adult	311	85 (27)	50(16)	19(6)	18(6)	16(5)	11(3)	13(4)	99(32)	

Conclusions: While PV7 serotypes have declined since 2000, in 2004 approximately 25% of the invasive isolates from this study were from the seven vaccine types (4, 6B, 9V, 14, 18C, 19F, 23F). Pediatric and adult patients were equally likely to carry PV7 serotypes. Serotype 19A was the most common serotype in both groups, which is cause for concern as this serotype is often non-susceptible to penicillin and erythromycin. Continued monitoring of post-vaccine serotype trends is on-going and will be vital to the management of pneumococcal disease.

Introduction

Streptococcus pneumoniae is an important cause of invasive disease and upper respiratory infections in children and the elderly. Contributing to the pathogenicity of this organism is a capsular polysaccharide (cps), the typing of which can differentiate isolates into more than 90 distinct serotypes, with about 15 of these serotypes causing the majority of invasive disease worldwide [1, 2]. The introduction of multi-valent pneumococcal conjugate vaccines has led to a decline in the serotypes covered by the vaccines, however the long-term impact on the prevalence of these serotypes and the emergence of non-vaccine serotypes is unknown[3-6]. The 7-valent conjugate vaccine (PV7) was introduced in the United States in 2000. This vaccine includes serotypes 4, 6B, 9V, 14, 18C, 19F, and 23F [4]. Replacement of vaccine-serotypes by non-vaccine serotypes has already been reported [5, 6]. Post-vaccine surveillance studies of serotype prevalence patterns worldwide are necessary to monitor the changing seroepidemiology of this organism in order to evaluate the appropriate formulation for current and future vaccines. In addition monitoring of possible replacement serotypes is essential to determine possible antibiotic resistance patterns as certain serotypes are more closely associated with antibiotic resistance [9]. The established method for SPN serotyping is the Quellung reaction [7], which is costly and requires technical expertise. For this study, we utilized a combination of multiplex PCR, confirmed by the Quellung reation, to determine the capsular serotypes of invasive SPN from adult and pediatric patients in the United States.

Materials & Methods

368 SPN isolates from select sterile body sites from the Tigecycline Evaluation Surveillance Trial (T.E.S.T.) study were included in this analysis. All isolates were collected in 2004 in the United States (US). Sterile body sites include body fluids, cardiovascular system, including blood, central nervous system, lymph, ovary, lung, respiratory sinus and bone. Based on the seven- reaction sequential multiplex PCR described previously [8], we devised a PCR using only four reactions to identify the pneumococcal serotype, followed by confirmation via the conventional Quellung reaction [7]. This PCR-based approach is based on serotype-specific genes within the capsular polysaccharide synthesis genes (the *cps* locus) of SPN. While this scheme could not detect all serotypes without doing a traditional serotyping checkerboard, it served to streamline the project and reduce cost by reducing the number of Quellung reactions needed. Using primers targeted at different loci within the *cps* gene, multiplex PCR reactions were set up in a sequential fashion, including an internal *cps* control. When the PCR assigned isolates to a serotype subset, conventional serotyping was performed based on the subset type. All PCR serotype results were confirmed by Quellung reaction. Isolates that were negative by PCR were serotyped by Quellung reactions alone. All PCR reactions were run with positive and negative controls, including the internal *cps* locus. Pneumococcal antisera were purchased from Statens Serum Institute (Copenhagen, Denmark).

References

1. Hsu K, Pelton S, Karumuri S, Heisey-Grove D, Klein J. *Population based surveillance for childhood invasive pneumoncoccal disease in the era of conjugate vaccine.* Pediatr Infect Dis J. 2005; **24**:17-23.
2. Hausdorff WP, Bryant J, Paradiso PR, Siber GR. *Which pneumococcal serogroups cause the most invasive disease: implications for conjugate vaccine formulation and use, part I*. Clin Infect Dis. 2000; **30**:100-121.
3. Hager HL, Wooley TW, Berk SL. *Review of recent pneumococcal infections with attention to vaccine and nonvaccine serotypes*. Rev Infect Dis.1990; **12**:267-272
4. Whitney CG, Farley MM, Hadler J, et al. *Decline in invasive pneumococcal diease after the introduction of protein-polysaccharide conjugate vaccine*. N Engl J Med. 2003; **348**:1737-1746.
5. Mahjoub-Messai F, Doit C, Koeck J-L, et al. *Population Snapshot of Streptococcus pneumoniae Serotype 19A Isolates before and after Introduction of Seven-Valent Pneumococcal Vaccination for French Children*. J. Clin. Microbiol. 2009; **47**: 837 - 840.
6. Jacobs MR, Good CE, Beall B, et al. Changes in Serotypes and Antimicrobial Susceptibility of Invasive *Streptococcus pneumoniae* Strains in Cleveland: a Quarter Century of Experience**.** J. Clin. Microbiol. 2008; **46**: 982 - 990.
7. Lund E. *Laboratory diagnosis of pneumococcus infections*. Bull WHO. 1960; **23**:5-13
8. Pai R, Gertz RE, Beall B. *Sequntial multiplex PCR approach for dertermining capsular serotypes of Streptococcus pneumoniae isolates*. J Clin Microbiol. 2006; **44**:124-131
9. Kyaw MH, Lynfield R, Schaffner W, et al. *Effect of introduction of the pneumococcal conjugate vaccine on drug-resistant Streptococcus pneumoniae*. N Engl J Med. 2006; **354**:1455-1463

Acknowledgements

This study was sponsored by a grant from Pfizer Inc. We gratefully acknowledge the contributions of all investigators, laboratory personnel and from all participants in the Tigecycline Evaluation Study Trials program.

Results

Figure 1. Percentage of US SPN isolates in each capsular serotype.

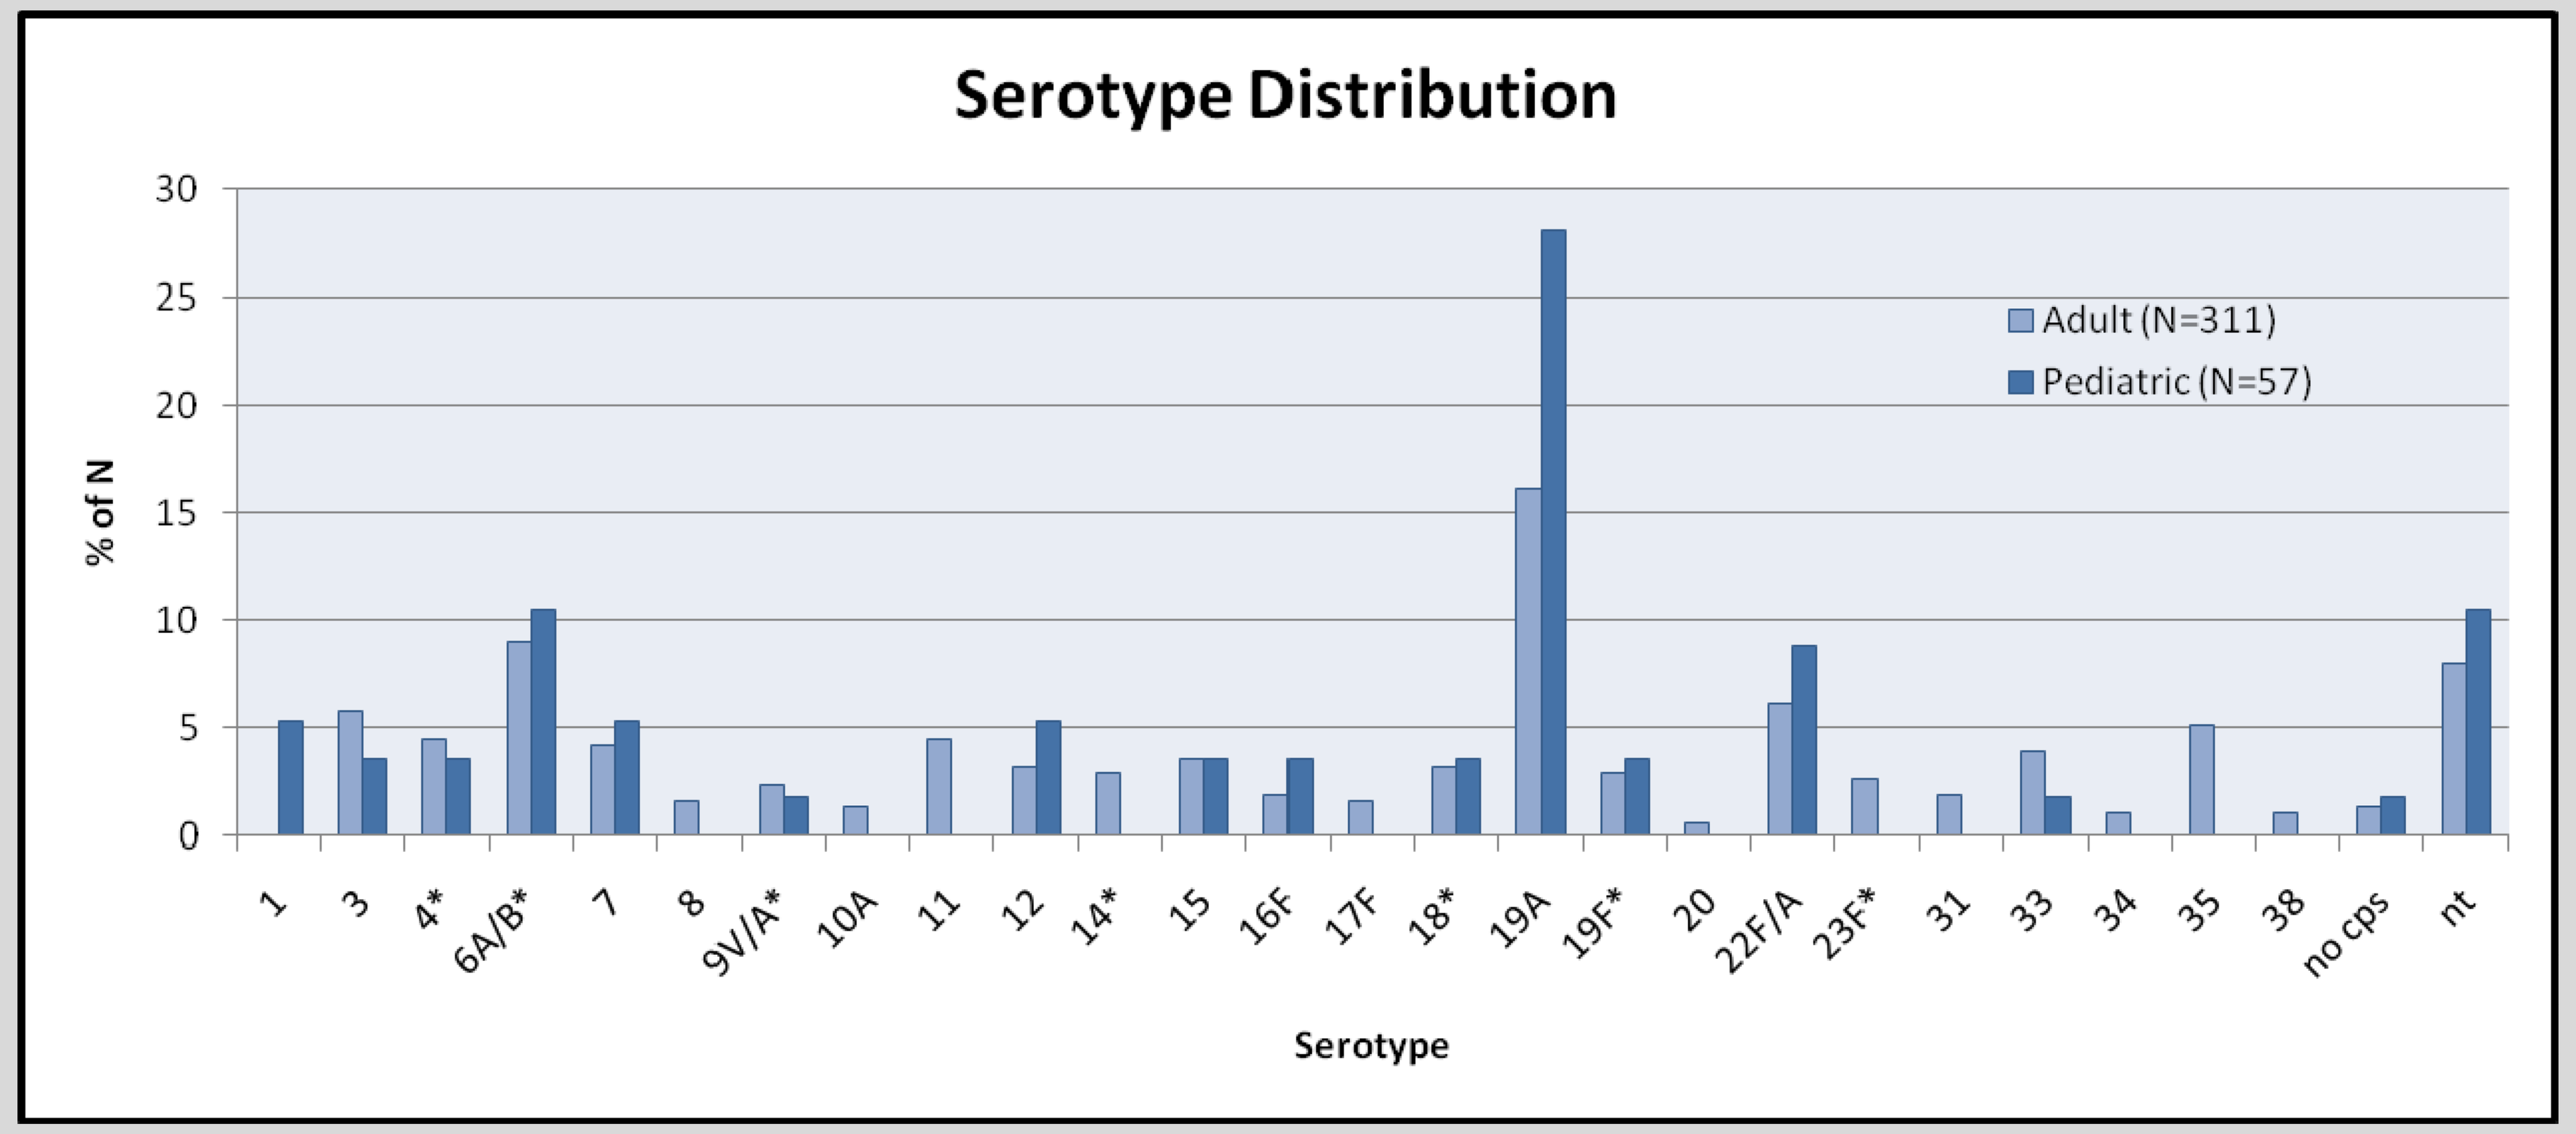
Serotype Distribution
% of N
30
25
20
15
10
5
0
Adult (N=311)
Pediatric (N=57)
1
3
4*
6A/B*
7
8
9V/A*
10A
11
12
14*
15
16F
17F
18*
19A
19F*
20
22F/A
23F*
31
33
34
35
38
no cps
nt
Serotype

*PV7 serotype

Table 1. Distribution of SPN PV7 capsular serotypes.

	Serotype							
	N	4	6A/B	9V	14	18	19F	23F
Pediatric	13	2	6	1	0	2	2	0
Adult	85	14	28	7	9	10	9	8

Table 2. Distribution of SPN non-vaccine capsular serotypes.

Serotype	N	1	3	7	8	10A	11	12	15	16F	17F
Pediatric	44	3	2	3	0	0	0	3	2	2	0
Adult	226	0	18	13	5	4	14	10	11	6	5

Serotype	19A	20	22F/A	31	33	34	35	38	no CPS	nt*
Pediatric	16	0	5	0	1	0	0	0	1	6
Adult	50	2	19	6	12	3	16	3	4	25

* nt = non-typeable

Conclusions

- In this study approximately 25% of the invasive SPN isolate serotypes were from the seven vaccine types (4, 6B, 9V, 14, 18C, 19F, 23F). Adult and pediatric patients were equally likely to carry PV7 serotypes.
- Serotype 19A, a common cause of respiratory infections, was the most common serotype in both adult and pediatric patients, which is cause for concern as this serotype is often non-susceptible to penicillin and erythromycin.
- Replacement disease caused by non-vaccine serotypes remains an important clinical issue. Continued monitoring of invasive pneumococcal disease is essential for future vaccine development.