#P1-014

In Vitro Activity of Tigecycline and Comparators Against Pathogens from Switzerland, Sweden and The Netherlands

S. Bouchillon[1], T. Stevens[1], J. Johnson[1], D. Hoban[1], B. Johnson[1], R. Badal[1], M. Dowzicky[2]

[1]International Health Management Associates, Schaumburg, IL, USA

[2]Wyeth Pharmaceuticals, Collegeville, PA, USA

IHMA, Inc.
2122 Palmer Dr.
Schaumburg, IL 60173
Tel: (847) 303-5003
Fax: (847) 303-5601
www.ihmainc.com

REVISED ABSTRACT

Background: Tigecycline (TIG), a new glycylcycline, has been shown to have potent broad spectrum activity against most commonly encountered species responsible for community- and hospital-acquired infections. The T.E.S.T. program determined the in vitro activity of TIG and 10 comparators against respective gram positive/negative species. Isolates were collected during 2004 to 2006. **Methods:** A total of 570 clinically significant isolates from Switzerland, Sweden and The Netherlands were analyzed in this survey. Isolates were identified to the species level at the participating sites and confirmed by the central laboratory. MICs were determined by each site using supplied broth microdilution panels and interpreted according to CLSI guidelines. **Results:** Selected pathogens tested against tigecycline are shown in the table below:

	E. coli and *Klebsiella spp* (n=148)		*Enterobacter spp*. (n=60)		*Acinetobacter spp.** (n=37)		*P. aeruginosa** (n=62)	
	%S	MIC_{90}	%S	MIC_{90}	%S	MIC_{90}	%S	MIC_{90}
Tigecycline	98.6	0.5	100	0.5	n/a	0.25	n/a	16
Amikacin	100	2	100	2	100	4	98.4	8
Cefepime	97.3	0.5	100	0.5	91.9	8	90.3	8
Ceftazidime	99.3	8	90	16	91.9	8	90.3	8
Imipenem	100	0.5	100	1	97.3	1	93.5	2
Levofloxacin	93.9	0.5	98.3	0.06	97.3	0.12	93.5	2
Minocycline	92.6	4	93.3	4	100	0.5	25.8	>16
Pip-Tazo	96.6	2	95	8	97.3	4	96.8	8
	S. aureus (n=75)		*Enterococcus spp*. (n=48)		*S. pneumoniae** (n=46)		*S. agalactiae* (n=30)	
	%S	MIC_{90}	%S	MIC_{90}	%S	MIC_{90}	%S	MIC_{90}
Tigecycline	100	0.12	100	0.12	n/a	0.5	100	0.06
Levofloxacin	81.3	4	56.3	>32	100	1	96.7	1
Linezolid	100	2	100	2	100	1	100	1
Minocycline	100	0.25	50	8	n/a	4	n/a	8
Vancomycin	100	1	100	2	100	0.5	100	0.5

* na=breakpoints not available

Conclusion: Overall, the pathogens analyzed from these three countries are still very susceptible to most broad spectrum antimicrobials. Tigecycline's MIC_{90} of 0.5mcg/ml against gram-positive pathogens (including resistant phenotypes) and MIC_{90} of 0.5mcg/ml against overall *Enterobacteriaceae* and *Acinetobacter* spp. validate the potent inhibitory activity of TIG against community/hospital pathogens isolated from Switzerland, Sweden, and The Netherlands.

INTRODUCTION

Tigecycline (formerly GAR-936) is a member of a new class of antimicrobial agents, the glycylcyclines. This synthetic analogue of the tetracyclines exhibits significant antibacterial activity that is both bacteriostatic and, in certain instances, bactericidal with killing activity that is as much as four-fold better than vancomycin and daptomycin [1, 2]. The development of tigecycline is important in that it is active against bacterial strains carrying either or both of the two major forms of tetracycline resistance: efflux and ribosomal protection. Certain substituents at the 9-position of the tetracycline molecule restore activity against bacteria harboring genes encoding either or both efflux and ribosomal protection. A single chemical modification of tigecycline overcomes the two molecularly distinct forms of resistance while maintaining activity against susceptible gram-positive, gram-negative, aerobic, and anaerobic bacteria [3].

Previous studies have demonstrated excellent in vitro activity for tigecycline against clinical and laboratory strains of gram-positive and -negative bacteria with minimum inhibitory concentrations for the 90th percentile at or below 2 mcg/ml, including difficult to treat methicillin-resistant *Staphylococcus aureus* (MRSA), vancomycin-resistant enterococci (VRE) and extended-spectrum beta-lactamase (ESBL) producing *Enterobacteriaceae* [4-6]. This study was undertaken to document the in vitro activity of tigecycline against significant numbers of clinical pathogens collected in population centers in Switzerland, Sweden, and The Netherlands. This study is part of the larger ongoing global Tigecycline Evaluation and Surveillance Trials (T.E.S.T.) program.

MATERIALS & METHODS

- All isolates were derived from blood, respiratory tract, urine (no more than 25% of all isolates), skin, wound, body fluids, and other defined sources. Only one isolate per patient was accepted into the study. Clinical isolates were collected and tested between 2004 to 2006 from one study center in each of Switzerland, Sweden, and The Netherlands. Isolates were identified to the species level and tested at each site by the participating laboratory.
- Organism collection, transport, confirmation of organism identification, and development and management of a centralized database, were coordinated by Laboratories International for Microbiology Studies (LIMS), a division of International Health Management Associates, Inc. located in Schaumburg, IL, USA.
- All organisms were deemed clinically significant by local participant criteria. Isolate inclusion was independent of medical history, antimicrobial use, age, or gender. All sites identified each study isolate utilizing local laboratory criteria.
- Minimum inhibitory concentrations (MICs) were determined by the CLSI recommended broth microdilution testing method [7]. Tigecycline was supplied by Wyeth Pharmaceuticals (Collegeville, PA, USA). All other agents were supplied by the panel manufacturer, MicroScan (Dade Behring Inc., West Sacramento, CA, USA). The following antimicrobial agents were included on the panels with their dilution ranges (expressed in mcg/ml): amikacin (0.5-64); amoxicillin/clavulanic acid (0.12/0.06-32/16); ampicillin (0.5-32, gram-negative panel, and 0.06-16, gram-positive panel); cefepime (0.5-32); ceftriaxone (0.06-64); ceftazidime (8-32); imipenem (0.06-16); linezolid (0.5-8); levofloxacin (0.008-8); minocycline (0.5-16); tigecycline (0.008-16); penicillin (0.06-8); piperacillin/tazobactam (0.06/4-128/4), and vancomycin (0.12-32). MIC interpretive criteria followed published guidelines established by the Clinical and Laboratory Standards Institute [8] and the recent US Food and Drug Administration package insert for tigecycline [9], where applicable.
- *Escherichia coli*, *Klebsiella pneumoniae* and *Klebsiella oxytoca* were screened for ESBL activity when MIC results for ceftriaxone were >1 mcg/ml using broth microdilution panels. ESBL activity was confirmed using the CLSI (2006) phenotypic confirmatory disk test (Oxoid, Ogdensburg, NY, USA) on Mueller-Hinton agar (Remel Inc., Lenexa, KS, USA) according to CLSI (2006) guidelines. ESBL presence was confirmed by testing the following antibiotic disks: cefotaxime (30-mcg), cefotaxime/clavulanic acid (30/10-mcg), ceftazidime (30-mcg), and ceftazidime/clavulanic acid (30/10-mcg). Antimicrobial disks were manufactured by Oxoid, Inc. (Ogdensburg, NY, USA). Mueller-Hinton agar used in testing was manufactured by Remel, Inc. (Lenexa, KS, USA). An organism was interpreted as containing an ESBL if there was an increase of >5 mm in the inhibition zone of the combination disk when compared to that of the cephalosporin alone. *K. pneumoniae* ATCC 700603 was used to QC the ESBL confirmation tests.
- Quality controls (QC) were performed by each testing site on each day of testing using the corresponding ATCC control strains: *E. coli* ATCC 25922; *E. coli* ATCC 35218; *H. influenzae* ATCC 49766; *H. influenzae* ATCC 49247; *S. aureus* ATCC 29213; *Pseudomonas aeruginosa* ATCC 27853; *Enterococcus faecalis* ATCC 29212 and *S. pneumoniae* ATCC 49619. Results were included in the analysis only when corresponding QC isolates tested within the acceptable range according to CLSI (2006) guidelines [8].

REFERENCES

1. Hoellman, D.B., et al., *Antipneumococcal activities of GAR-936 (a new glycylcycline) compared to those of nine other agents against penicillin-susceptible and -resistant pneumococci*. Antimicrob Agents Chemother, 2000. 44(4): p. 1085-8.
2. Labthavikul, P., P.J. Petersen, and P.A. Bradford, *In vitro activity of tigecycline against Staphylococcus epidermidis growing in an adherent-cell biofilm model.* Antimicrob Agents Chemother, 2003. 47(12): p. 3967-9.
3. Projan, S.J., *Preclinical pharmacology of GAR-936, a novel glycylcycline antibacterial agent.* Pharmacotherapy, 2000. 20(9 Pt 2): p. 219S-223S; discussion 224S-228S.
4. Gales, A.C. and R.N. Jones, *Antimicrobial activity and spectrum of the new glycylcycline, GAR-936 tested against 1,203 recent clinical bacterial isolates.* Diagn Microbiol Infect Dis, 2000. 36(1): p. 19-36.
5. Patel, R., et al., *In vitro activity of GAR-936 against vancomycin-resistant enterococci, methicillin-resistant Staphylococcus aureus and penicillin-resistant Streptococcus pneumoniae.* Diagn Microbiol Infect Dis, 2000. 38(3): p. 177-9.
6. Rupp, M.E. and P.D. Fey, *Extended spectrum beta-lactamase (ESBL)-producing Enterobacteriaceae: considerations for diagnosis, prevention and drug treatment.* Drugs, 2003. 63(4): p. 353-65.
7. CLSI, *Methods for Dilution Antimicrobial Susceptibility Tests for Bacteria That Grow Aerobically; Approved Standard-Sixth Edition, in Document M7-A6.* 2005: Clinical Laboratory 65: Clinical Laboratory Standards Institute (CLSI), 940 West Valley Road, Suite 1400, Wayne, Pennsylvania 19087-1898 USA.
8. CLSI, *Performance Standards for Antimicrobial Susceptibility Testing, in Document M100-S15. 2005*: Clinical Laboratory Standards Institute (CLSI), 940 West Valley Road, Suite 1400, Wayne, Pennsylvania 19087-1898 USA.
9. Tygacil, *Product Insert. 2005: Wyeth Pharmaceuticals, Inc.*, Philadelphia, PA, USA.

ACKNOWLEDGEMENTS

We gratefully acknowledge the contributions of the investigators, laboratory personnel, and all members of the Tigecycline Evaluation Study Trials program group. This study was sponsored by a grant from Wyeth Pharmaceuticals.

RESULTS

The results are listed in the following tables.

Table 1. In vitro activity of tigecycline and comparative agents against *Enterobacteriaceae*.

Organism Name	Drug	%SUS [a]	%INT	%RES	MIC (mcg/ml) MIC_{50}	MIC_{90}
E. coli (n=73)	Tigecycline	100	0	0	0.12	0.12
	Amikacin	100	0	0	2	4
	AmoxClav	89	8.2	2.7	4	16
	Ampicillin	58.9	0	41.1	4	>32
	Cefepime	95.9	1.4	2.7	≤0.5	≤0.5
	Ceftazidime	98.6	1.4	0	≤8	≤8
	Ceftriaxone	94.5	0	5.5	≤0.06	0.12
	Imipenem	100	0	0	0.25	0.25
	Levofloxacin	87.7	6.8	5.5	0.03	4
	Minocycline	91.8	6.8	1.4	1	4
	PipTazo	97.3	2.7	0	0.5	2
K. pneumoniae (n=42)	Tigecycline	95.2	2.4	2.4	0.25	0.5
	Amikacin	100	0	0	1	2
	AmoxClav	97.6	2.4	0	2	8
	Ampicillin	0	33.3	66.7	32	>32
	Cefepime	100	0	0	≤0.5	≤0.5
	Ceftazidime	100	0	0	≤8	≤8
	Ceftriaxone	100	0	0	≤0.06	≤0.06
	Imipenem	100	0	0	0.25	0.5
	Levofloxacin	100	0	0	0.03	0.25
	Minocycline	88.1	0	11.9	1	16
	PipTazo	97.6	0	2.4	1	2
K. oxytoca (n=33)	Tigecycline	100	0	0	0.25	0.5
	Amikacin	100	0	0	1	2
	AmoxClav	100	0	0	2	4
	Ampicillin	0	21.2	78.8	>32	>32
	Cefepime	97	3	0	≤0.5	≤0.5
	Ceftazidime	100	0	0	≤8	≤8
	Ceftriaxone	97	0	3	≤0.06	0.25
	Imipenem	100	0	0	0.25	0.5
	Levofloxacin	100	0	0	0.03	0.06
	Minocycline	100	0	0	1	2
	PipTazo	93.9	6.1	0	1	4
E. aerogenes (n=17)	Tigecycline	100	0	0	0.25	0.5
	Amikacin	100	0	0	2	2
	AmoxClav	0	0	100	>32	>32
	Ampicillin	0	0	100	>32	>32
	Cefepime	100	0	0	≤0.5	≤0.5
	Ceftazidime	100	0	0	≤8	≤8
	Ceftriaxone	100	0	0	≤0.06	0.5
	Imipenem	100	0	0	1	1
	Levofloxacin	100	0	0	0.03	0.06
	Minocycline	100	0	0	2	2
	PipTazo	100	0	0	2	8
E. cloacae (n=40)	Tigecycline	100	0	0	0.5	0.5
	Amikacin	100	0	0	2	2
	AmoxClav	2.5	2.5	95	>32	>32
	Ampicillin	0	5	95	>32	>32
	Cefepime	100	0	0	≤0.5	2
	Ceftazidime	85	2.5	12.5	≤8	32
	Ceftriaxone	87.5	7.5	5	0.25	16
	Imipenem	100	0	0	0.5	0.5
	Levofloxacin	100	0	0	0.03	0.06
	Minocycline	92.5	0	7.5	2	4
	PipTazo	92.5	2.5	5	1	8
S. marcescens (n=26)	Tigecycline	100	0	0	0.5	1
	Amikacin	100	0	0	2	2
	AmoxClav	0	0	100	>32	>32
	Ampicillin	0	0	100	>32	>32
	Cefepime	100	0	0	≤0.5	≤0.5
	Ceftazidime	100	0	0	≤8	≤8
	Ceftriaxone	96.2	3.8	0	0.12	1
	Imipenem	100	0	0	0.5	1
	Levofloxacin	96.2	0	3.8	0.06	0.12
	Minocycline	100	0	0	2	4
	PipTazo	100	0	0	1	4

[a]Interpretive criteria as defined by CLSI, M100-S16 (2006), where available; Tigecycline susceptibility breakpoints are according to FDA package insert (Tygacil®, 2005), where applicable [9].
[b]Species with n <10 are omitted

Table 2. In vitro activity of tigecycline and comparative agents against *Acinetobacter* spp. and *P. aeruginosa*.

Organism Name	Drug	%SUS [a]	%INT	%RES	MIC (mcg/ml) MIC_{50}	MIC_{90}
Acinetobacter spp. (n=37)	Tigecycline	na	na	na	0.06	0.25
	Amikacin	100	0	0	1	4
	Cefepime	91.9	8.1	0	2	8
	Ceftazidime	91.9	2.7	5.4	≤8	≤8
	Ceftriaxone	78.4	18.9	2.7	4	16
	Imipenem	97.3	0	2.7	0.25	1
	Levofloxacin	97.3	2.7	0	0.06	0.12
	Minocycline	100	0	0	≤0.5	≤0.5
	PipTazo	97.3	0	2.7	≤0.06	4
P. aeruginosa (n=62)	Tigecycline	na	na	na	8	16
	Amikacin	98.4	1.6	0	2	8
	Cefepime	90.3	6.5	3.2	2	8
	Ceftazidime	90.3	1.6	8.1	≤8	≤8
	Ceftriaxone	29	6.5	64.5	>64	>64
	Imipenem	93.5	6.5	0	1	2
	Levofloxacin	93.5	4.8	1.6	0.5	2
	Minocycline	25.8	27.4	46.8	8	>16
	PipTazo	96.8	0	3.2	2	8

[a]Interpretive criteria as defined by CLSI, M100-S16 (2006), where available; Tigecycline susceptible breakpoint is according to FDA package insert (2005), where applicable [9]; na = not available - breakpoints are not yet established against this species.

Table 3. In vitro activity of tigecycline and comparative agents against gram-positive pathogens.

Organism Name	Drug	%SUS [a]	%INT	%RES	MIC (mcg/ml) MIC_{50}	MIC_{90}
S. aureus (n=75)	Tigecycline	100	0	0	0.12	0.12
	AmoxClav	88	0	12	0.5	8
	Ampicillin	24	0	76	2	>16
	Ceftriaxone	86.7	1.3	12	2	>64
	Imipenem	89.3	0	10.7	0.25	16
	Levofloxacin	81.3	0	18.7	0.12	4
	Linezolid	100	0	0	2	2
	Minocycline	100	0	0	≤0.25	≤0.25
	Penicillin	17.3	0	82.7	4	>8
	PipTazo	89.3	0	10.7	0.5	>16
	Vancomycin	100	0	0	0.5	1
S. aureus (MR) (n=12)	Tigecycline	100	0	0	0.12	0.25
	AmoxClav	25	0	75	>8	>8
	Ampicillin	0	0	100	>16	>16
	Ceftriaxone	16.7	8.3	75	>64	>64
	Imipenem	33.3	0	66.7	>16	>16
	Levofloxacin	16.7	0	83.3	4	16
	Linezolid	100	0	0	2	2
	Minocycline	100	0	0	≤0.25	≤0.25
	Penicillin	0	0	100	>8	>8
	PipTazo	33.3	0	66.7	>16	>16
	Vancomycin	100	0	0	1	1
Enterococcus spp [b]. (n=48)	Tigecycline	100	0	0	0.06	0.12
	Ampicillin	81.3	0	18.8	1	>16
	Levofloxacin	56.3	2.1	41.7	1	>32
	Linezolid	100	0	0	1	2
	Minocycline	50	41.7	8.3	4	8
	Penicillin	81.3	0	18.8	4	>8
	Vancomycin	100	0	0	1	2
E. faecalis [b] (n=37)	Tigecycline	100	0	0	0.12	0.12
	Ampicillin	100	0	0	1	2
	Levofloxacin	67.6	0	32.4	1	32
	Linezolid	100	0	0	1	2
	Minocycline	43.2	48.6	8.1	8	8
	Penicillin	100	0	0	2	8
	Vancomycin	100	0	0	1	2
E. faecium [b] (n=11)	Tigecycline	100	0	0	0.03	0.12
	Ampicillin	18.2	0	81.8	>16	>16
	Levofloxacin	18.2	9.1	72.7	32	>32
	Linezolid	100	0	0	1	2
	Minocycline	72.7	18.2	9.1	≤0.25	8
	Penicillin	18.2	0	81.8	>8	>8
	Vancomycin	100	0	0	0.5	1
S. agalactiae (n=30)	Tigecycline	100	0	0	0.03	0.06
	Ampicillin	100	0	0	≤0.06	0.12
	Ceftriaxone	100	0	0	0.06	0.12
	Levofloxacin	96.7	0	3.3	0.5	1
	Linezolid	100	0	0	1	1
	Penicillin	100	0	0	≤0.06	0.12
	Vancomycin	100	0	0	0.5	0.5

[a]Interpretive criteria as defined by CLSI, M100-S16 (2006), where available; tigecycline susceptible breakpoint is according to FDA package insert (2005), where applicable [9].
[b]FDA susceptible breakpoint of 0.25 mcg/ml for vancomycin-susceptible *E. faecalis* was used all enterococci for comparative purposes only.

Table 4. In vitro activity of tigecycline and comparative agents against respiratory pathogens.

Organism Name	Drug	%SUS [a]	%INT	%RES	MIC (mcg/ml) MIC_{50}	MIC_{90}
S. pneumoniae (n=46)	Tigecycline	na	na	na	0.03	0.5
	AmoxClav	97.8	0	2.2	≤0.03	1
	Ceftriaxone	97.8	2.2	0	≤0.03	1
	Imipenem	80	20	0	≤0.12	0.25
	Levofloxacin	100	0	0	0.5	1
	Linezolid	100	0	0	≤0.5	1
	Penicillin	76.1	13	10.9	≤0.06	2
	Vancomycin	100	0	0	0.25	0.5
H. influenzae (n=65) [b]	Tigecycline	na	na	na	0.12	0.12
	AmoxClav	100	0	0	0.25	0.5
	Ampicillin	86.1	5.6	8.3	≤0.5	2
	Cefepime	94.4	0	5.6	≤0.5	2
	Ceftriaxone	100	0	0	≤0.06	0.12
	Imipenem	100	0	0	0.5	1
	Levofloxacin	100	0	0	0.015	0.03
	PipTazo	100	0	0	≤0.06	0.12

[a]Interpretive criteria as defined by CLSI, M100-S16 (2006), where available; tigecycline susceptible breakpoint is according to FDA package insert (2005), where applicable [9]; na = not available.
[b]The 3 beta-lactamase positive strains were omitted from analysis.

Table 5. In vitro activity of tigecycline and comparators against *E. coli* and *Klebsiella* spp. (n=148) showing frequency distribution (n) and cumulative percent inhibited (%) at each MIC (mcg/ml).

MIC n %	≤0.008	0.015	0.03	≤0.06	0.06	0.12	0.25	≤0.5	0.5	1	2	4	≤8	8	>8	16	>16	32	>32	64	>64	>128
Tigecycline			8 5.4		22 20.3	47 52	45 82.4		22 97.3	1 98	1 98.6	1 99.3		1 100								
Amikacin								17 11.5		48 43.9	74 93.9	8 99.3		1 100								
AmoxClav									8 5.4	20 18.9	65 62.8	30 83.1		16 93.9		7 98.6		1 99.3	1 100			
Ampicillin								3 2		11 9.5	21 23.6	7 28.4		1 29.1		21 43.2		20 56.8	64 100			
Cefepime								140 94.6		2 95.9	1 96.6			1 97.3		2 98.6		1 99.3	1 100			
Ceftazidime													147 99.3			1 100						
Ceftriaxone				124 83.8		11 91.2	5 94.6		1 95.3		1 95.9			1 96.6						2 98	3 100	
Imipenem						27 19.4	92 85.6		20 100													
Levofloxacin	9 6.1	43 35.1	46 66.2		24 82.4	6 86.5	4 89.2		4 91.9	3 93.9		5 97.3		2 98.6	2 100							
Minocycline								49 33.1		61 74.3	17 85.8	10 92.6		5 95.9		3 98	3 100					
PipTazo						7 4.7	16 15.5		35 39.2	60 79.7	17 91.2	6 95.3		2 96.6				2 98		2 99.3		1 100

Table 6. In vitro activity of tigecycline and comparators against *Acinetobacter* spp. (n=37) showing frequency distribution (n) and cumulative percent inhibited (%) at each MIC (mcg/ml).

MIC n %	0.015	0.03	≤0.06	0.06	0.12	0.25	≤0.5	0.5	1	2	4	≤8	8	16	32	>32	>64	>128
Tigecycline	2 5.4	11 35.1		8 56.8	6 73	7 91.9		3 100										
Amikacin							9 24.3		10 51.4	14 89.2	3 97.3		1 100					
Cefepime							9 24.3		8 45.9	8 67.6	7 86.5		2 91.9	3 100				
Ceftazidime												34 91.9		1 94.6	2 100			
Ceftriaxone								2 5.4	3 13.5	4 24.3	14 62.2		6 78.4	6 94.6	1 97.3		1 100	
Imipenem					7 18.9	15 59.5		11 89.2	3 97.3					1 100				
Levofloxacin	3 8.1	10 35.1		15 75.7	6 91.9	1 94.6		1 97.3			1 100							
Minocycline							36 97.3		1 100									
PipTazo			23 62.2		4 73	1 75.7		3 83.8	1 86.5	1 89.2	1 91.9		1 94.6	1 97.3				1 100

Table 7. In vitro activity of tigecycline and comparators against *S. aureus* (n=75) showing frequency distribution (n) and cumulative percent inhibited (%) at each MIC (mcg/ml).

MIC n %	0.03	<0.06	0.06	≤0.12	0.12	≤0.25	0.25	≤0.5	0.5	1	2	4	8	>8	16	>16	32	>64
Tigecycline	3 4		31 45.3		38 96		3 100											
AmoxClav			6 8		9 20		13 37.3		23 68	12 84	3 88		2 90.7	7 100				
Ampicillin		8 10.7			5 17.3		5 24		6 32	5 38.7	13 56	12 72	2 74.7		8 85.3	11 100		
Ceftriaxone									1 1.3	15 21.3	29 60	19 85.3	1 86.7		1 88			9 100
Imipenem				37 49.3			28 86.7		2 89.3						1 90.7	7 100		
Levofloxacin		20 26.7			30 66.7		10 80			1 81.3		8 92	2 94.7		3 98.7		1 100	
Linezolid								2 2.7		32 45.3	40 98.7	1 100						
Minocycline						72 96			1 97.3	2 100								
Penicillin		12 16			1 17.3		6 25.3		4 30.7	7 40	3 44	15 64	7 73.3	20 100				
PipTazo						27 36			22 65.3	13 82.7	4 88		1 89.3			8 100		
Vancomycin							12 16		37 65.3	26 100								

CONCLUSIONS

- The strains studied from these three countries are generally susceptible to the broad spectrum antimicrobials evaluated in this study. Resistance rates of common phenotypes of all species were relatively low with only 3/148 (2.1%) *E. coli* and *Klebsiella spp.* strains harboring ESBL enzymes; 3/65(4.6%) *H. influenzae* beta-lactamase positive and 5/46 (10.9%) S. pneumoniae penicillin-resistant. No vancymycin-resistant enterococci were found.
- Tigecycline inhibited 100% of *E. coli* and *Klebsiella spp*. strains at ≤8 mcg/ml, with an MIC_{90} of 0.5 mcg/ml.
- Tigecycline's MIC_{90} of 0.25 mcg/ml against *Acinetobacter spp*. was at least four-fold lower than all other comparators except levofloxacin (0.12 mcg/ml) and minocycline (0.5 mcg/ml).
- Tigecycline inhibited the growth of all *S. aureus* at ≤ 0.25 mcg/ml, with an MIC_{90} (0.12 mcg/ml) at least 8-fold lower than linezolid, vancomycin, levofloxacin, imipenem, and the other beta-lactam antimicrobials.
- Tigecycline was as active as ampicillin, penicillin, vancomycin, and linezolid against *E. faecalis*, with 100% of strains susceptible to all 5 drugs, and as active as linezolid and vancomycin (100% of strains susceptible) against *E. faecium*.
- Tigecycline demonstrated excellent inhibitory against *H. influenzae*, without regard to beta-lactamase production.
- The in vitro activity of tigecycline in this study indicates its potential for the treatment of serious infections. Although the frequency of resistant strains in these three countries appears to be lower than that seen in many other parts of the world, most of the strains exhibiting resistance to one or more drugs remain fully susceptible to tigecycline.